Baseline Characteristics of Patients Switching to Prophylactic Efanesoctocog Alfa Therapy Evaluated for Joint Health in Hemophilia A in Taiwan (PROTECT-ALT Study)

PB3293

Yeu-Chin Chen,[1,2] Ming-Ching Shen,[3,4] Cheng-Wei Huang,[3] Hung Chang,[5] Jiaan-Der Wang,[6] Sheng-Chieh Chou,[7] Ching-Tien Peng,[8] Chih-Cheng Hsiao,[9] Chia-Yau Chang,[10,11] Te-Fu Weng,[12] Tong-Ling Chien,[13] Ching-Min Chang,[13,14] Shyh-Shin Chiou[15,16]

[1]Hemophilia Care and Research Center, Tri-Service General Hospital, Taipei, Taiwan; [2]Division of Hematology/Oncology, Department of Internal Medicine, National Defense Medical University, Taipei, Taiwan; [3]Department of Internal Medicine, Changhua Christian Hospital, Changhua, Taiwan; [4]Department of Laboratory Medicine and Department of Internal Medicine, National Taiwan University Hospital, Taipei, Taiwan; [5]Division of Hematology-Oncology, Chang Gung Memorial Hospital, Taipei, Taiwan; [6]The Center for Rare Disease and Hemophilia, Taichung Veterans General Hospital, Taichung, Taiwan; [7]Department of Internal Medicine, National Taiwan University Hospital, Taipei, Taiwan; [8]Department of Pediatric Hemato/Oncology, China Medical University Children's Hospital, Taichung, Taiwan; [9]Division of Pediatric Hematology/Oncology, Kaohsiung Chang Gung Memorial Hospital, Kaohsiung, Taiwan; [10]Hemophilia Center, Department of Pediatric Hematology/Oncology, Taipei Medical University Hospital, Taipei, Taiwan; [11]Department of Pediatrics, School of Medicine, College of Medicine, Taipei Medical University, Taipei, Taiwan; [12]Department of Pediatrics, Chung Shan Medical University Hospital, Taichung, Taiwan; [13]Sanofi, Taiwan; [14]Sanofi, Hong Kong; [15]Graduate Institute of Clinical Medicine, College of Medicine, Kaohsiung Medical University, Kaohsiung, Taiwan; [16]Division of Hematology and Oncology, Department of Pediatrics, Kaohsiung Medical University Hospital, Kaohsiung, Taiwan.

BACKGROUND

- Efanesoctocog alfa is a first-in-class, high-sustained factor VIII (FVIII) replacement therapy (also referred to as ultra-long half-life FVIII) with established efficacy and safety in clinical trials.[1,2]
- Prospective real-world data linking bleed control to structural and functional joint outcomes remains limited.
- Data assessing clinical and subclinical joint status using standardized joint scores and imaging can inform important patient outcomes.

AIM

- **To characterize baseline joint health and establish a real-world longitudinal framework to evaluate the impact of efanesoctocog alfa prophylaxis on joint outcomes in patients with hemophilia A.**

METHODS

- PROTECT-ALT (NCT06684314) is a prospective observational study enrolling ~100 participants who initiated efanesoctocog alfa within 3 months prior to enrollment in Taiwan (**Figure 1**).
- Inclusion criteria included a Pettersson score (PS) ≤6 at either ankle.
- Primary endpoints: changes from baseline in Hemophilia Joint Health Score (HJHS) and annualized joint bleeding rate, as well as number/percentage of target joint development/resolution.
- Secondary endpoints: annualized bleeding rate (ABR), treatment adherence, factor consumption, patient-reported outcomes (quality of life), healthcare resource utilization, and magnetic resonance imaging (MRI) changes in index joints.
- Target joint: major joint in which ≥3 spontaneous bleeding episodes occurred in a consecutive 6-month period.
- Index joints: Ankles with PS ≤6 (monitored by MRI regardless of target joint definition).
 - Additional index joints: the most vulnerable joint by principal investigator's judgment in addition to the ankles with PS ≤6 or one-sided ankle with a PS >6.
- This analysis summarizes baseline data from patients enrolled during the first 11 months of the study; the entire study period is 5 years.
- Ethics approval and informed consent were obtained.

Figure 1. Schematic representation of study design

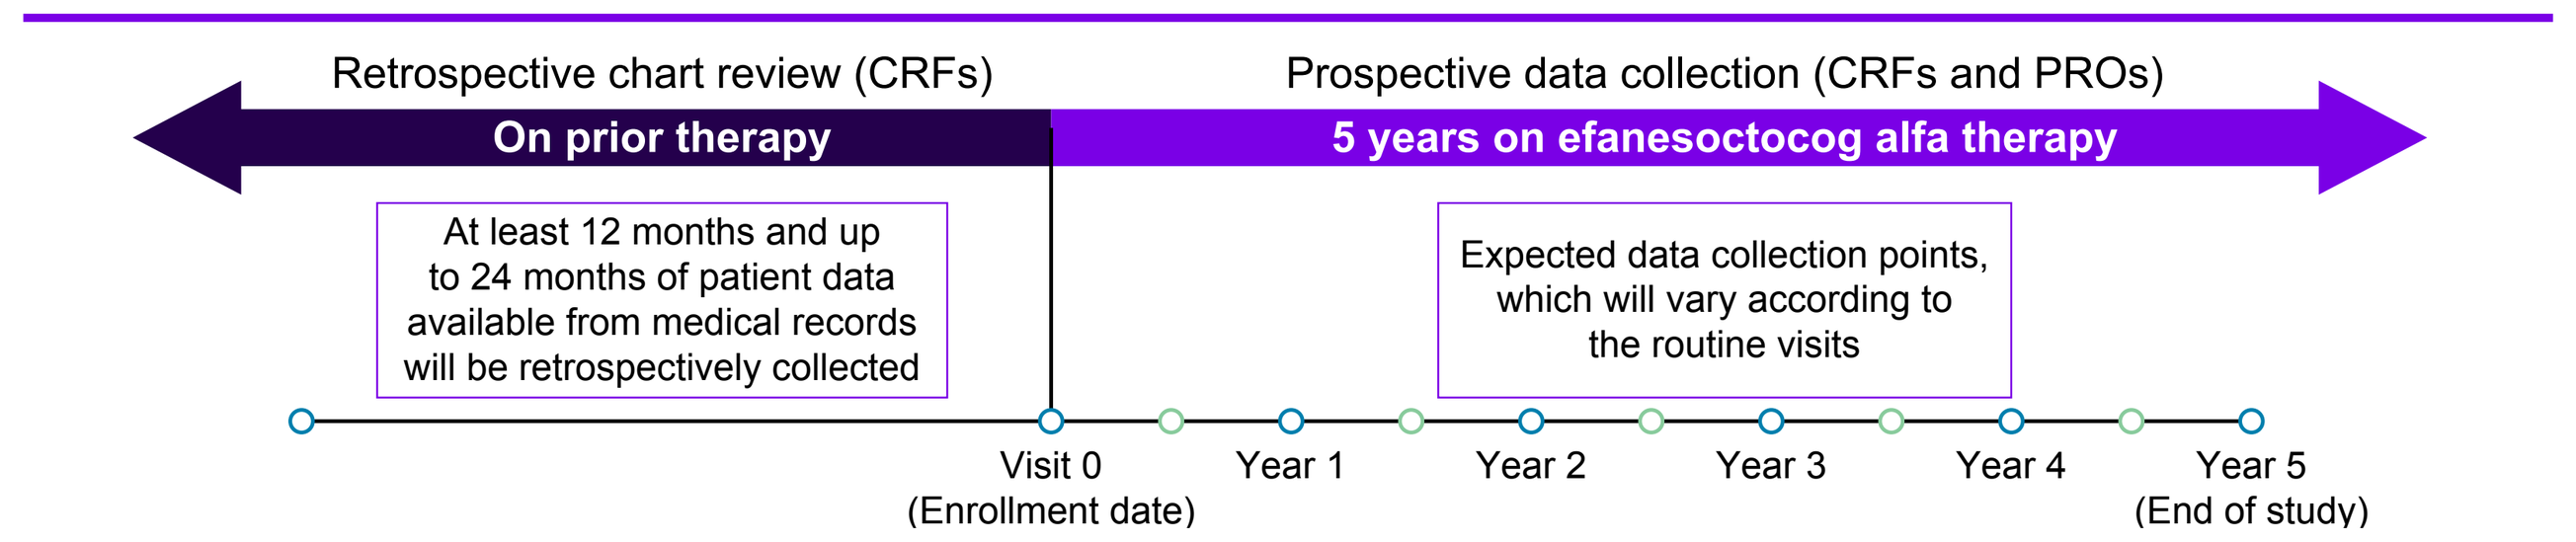
Retrospective chart review (CRFs)
Prospective data collection (CRFs and PROs)
On prior therapy
5 years on efanesoctocog alfa therapy
At least 12 months and up to 24 months of patient data available from medical records will be retrospectively collected
Expected data collection points, which will vary according to the routine visits
Visit 0
(Enrollment date)
Year 1
Year 2
Year 3
Year 4
Year 5
(End of study)

The enrollment date is the date on which the patient signs the informed consent form. Patients will be followed up until the end of the prospective data collection period, efanesoctocog alfa discontinuation, loss to follow-up, study withdrawal, enrollment in a clinical trial, or death, whichever occurs first.
CRF, case report form; PRO, patient-reported outcome.

RESULTS: BASELINE PATIENT CHARACTERISTICS

- Baseline demographics and disease characteristics are shown in **Table 1**.
- As of September 30, 2025, 64 male patients, including 11 adolescent (12 to <18 years) and 5 pediatric (6 to <12 years) patients, were enrolled.
- Most had severe hemophilia A (n=55) with mean (SD) age at diagnosis of 5 (10) years.
- Musculoskeletal and connective tissue conditions were the most frequently reported (n=31).
- Prior to efanesoctocog alfa, 60 patients were treated with factor replacement prophylaxis (standard half-life (SHL), n=11; extended half-life (EHL), n=53) and 3 patients with on-demand treatment (SHL, n=1; EHL, n=2). Data was missing in 1 patient by the data cutoff.
 - Patients may have received different prophylactic drugs during different periods between 0 and 24 months.
- The mean (SD) starting dose of efanesoctocog alfa was 46 (4) IU/kg weekly.

PLAIN LANGUAGE SUMMARY

- This research study, called PROTECT-ALT, is designed to investigate **joint health** in people with **hemophilia A** in Taiwan who switch to **efanesoctocog alfa** as their preventive therapy. The focus is on ankle joint health.
- Participants who entered the study had, on average, **mild joint disease** primarily in the ankles.

Table 1. Baseline patient demographics and disease characteristics

	All-Patient Population (N=64)
Mean (SD) age at enrollment, years	28 (13)
Median (range) age at enrollment, years	26 (17–34)
Age category at enrollment, n (%)	
6 to <12 years	5 (7.8)
12 to <18 years	11 (17.2)
18 to <50 years	42 (65.6)
≥50 years	6 (9.4)
Sex, n (%)	
Male	64 (100)
Mean (SD) BMI at enrollment, kg/m²	24.5 (4.5)
Participants with hemophilia medical history, n (%)	63 (98.4)
Mean (SD) age at hemophilia diagnosis, years	5 (10)
Severity of hemophilia at diagnosis, n (%)	
Mild[a]	1 (1.6)
Moderate	7 (10.9)
Severe	55 (85.9)
Missing	1 (1.6)
Relevant medical history at enrollment[b,c], n (%)	
Any medical history reported	43 (67.2)
Musculoskeletal and connective tissue disorders	31 (48.4)
Arthropathy	29 (45.3)
Infections and infestations	11 (17.2)
Blood and lymphatic system disorders	6 (9.4)
Hemorrhagic disorder	6 (9.4)
Respiratory, thoracic, and mediastinal disorders	5 (7.8)
Allergic rhinitis	5 (7.8)
Relevant comorbidities at enrollment[b,d], n (%)	
Any relevant comorbidities	17 (26.6)
Hepatitis C virus infection	7 (10.9)
History of FVIII inhibitor testing	
Yes	63 (98.4)
No	1 (1.6)

[a]This patient had FVIII activity of 6.4% at diagnosis and FVIII activity of 3.6% at the follow-up visit. According to the site, the patient presented more like a moderate hemophilia patient with severe phenotype. Differences in FVIII activity levels may be due to lab variations; [b]Conditions occurring in >5% of patients; [c]A patient can have >1 medical history reported; [d]A patient can have >1 relevant comorbidity reported.
BMI, body mass index; SD, standard deviation.

KEY CONCLUSIONS

1. Among patients starting efanesoctocog alfa in Taiwan, baseline assessments indicated **low HJHS scores and mild arthropathy** on MRI, predominantly in the **ankles.**
2. **Longitudinal follow-up** will provide additional real-world experience on the **effectiveness of efanesoctocog alfa regarding joint outcomes.**

RESULTS: BLEEDING HISTORY

- Retrospective bleeding history (≥12 months prior to initiation) was available for 42 patients, with 34 traumatic and 24 spontaneous bleeds recorded.
 - ABR was available for 55 patients (**Figure 2**). Baseline bleeding data indicate ongoing bleed burden despite prior therapy, supporting the need for improved prophylactic strategies.
- At enrollment, 8 patients had target joints (6 ankles, 1 knee, and 5 elbows with ≥3 spontaneous bleeding episodes in a consecutive 6-month period), and 64 patients had index joints (55 left ankles and 47 right ankles with PS ≤6).
 - In patients with additional index joints, 4 left ankles (PS >6), 13 right ankles (PS >6), 3 left elbows, 1 right elbow, 1 left knee, and 1 left hip were reported.
- All 63 patients with FVIII inhibitor testing history (**Table 1**) had negative inhibitor titers.
- During the prospective period, inhibitor testing was performed in 18 patients, of whom 17 patients had negative inhibitor titers and 1 patient had missing results.

Figure 2. Annualized bleeding rate (ABR) across bleed types in the previous 12 months (n=55)[a]

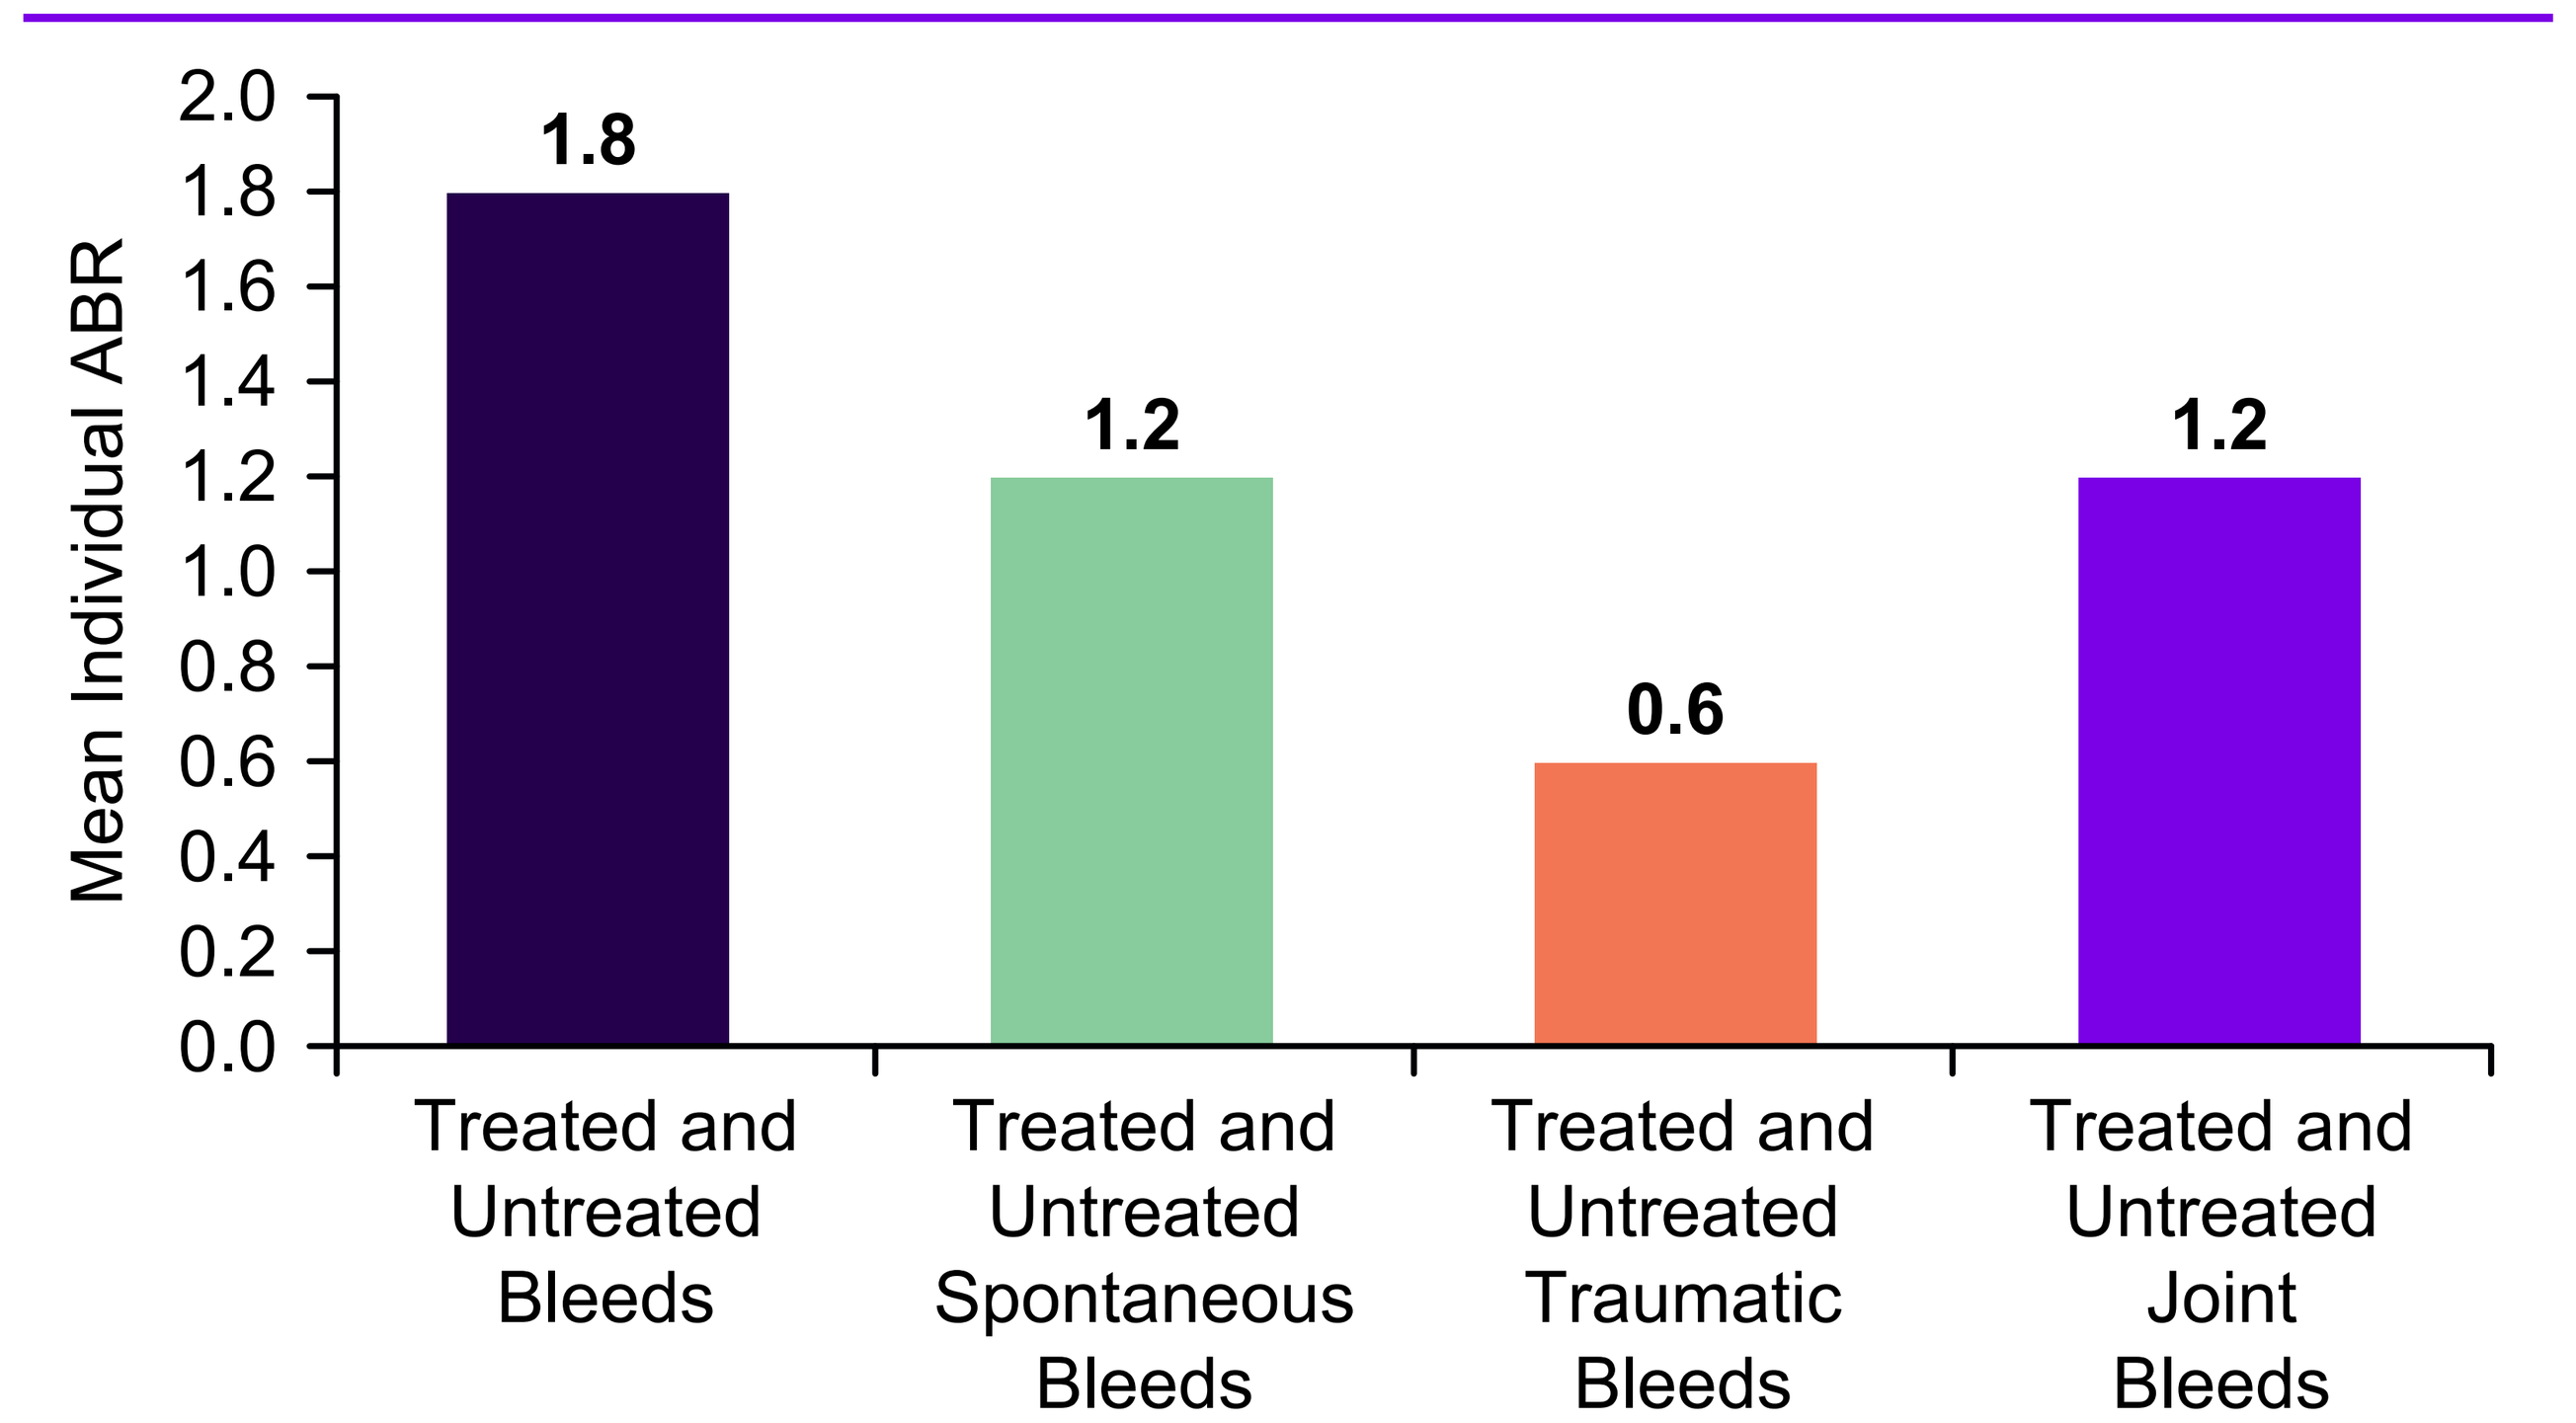
Mean Individual ABR
2.0
1.8
1.6
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0.0
1.8
1.2
0.6
1.2
Treated and Untreated Bleeds
Treated and Untreated Spontaneous Bleeds
Treated and Untreated Traumatic Bleeds
Treated and Untreated Joint Bleeds

[a]Data was missing in 9 patients by the data cutoff. The baseline period for bleeding event data collection is defined as up to 24 months prior to the first efanesoctocog alfa injection. Individual ABR = (Number of bleeding episodes occurring during the treatment period / Duration of the treatment period [days]) * 365.25.

RESULTS: BASELINE ASSESSMENTS

- Baseline joint health assessments indicated low HJHS (mean total, 9.2; **Table 2**).
- Baseline imaging assessments showed low soft-tissue and osteochondral changes sub-scores, mainly in the ankles (n=49; **Table 3**).

Table 2. Baseline joint health assessments

HJHS Total Scores (All-Patient Population, N=64)

	Left Elbow	Right Elbow	Left Knee	Right Knee	Left Ankle	Right Ankle	**Sum of Joint Total Score**	**Global Gait Score**	**HJHS Total Score**
n	58	58	58	58	58	58	58	58	58
Mean (SD)	1.3 (2.17)	1.3 (2.61)	1.4 (2.53)	1.1 (2.46)	1.4 (2.02)	1.9 (2.73)	7.9 (10.11)	0.7 (1.30)	9.2 (10.97)
Median	0	0	0	0	1	0	6	0	6
Q1, Q3	0, 1	0, 1	0, 2	0, 1	0, 2	0, 4	0, 11	0, 1	1, 13
Min, Max	0, 9	0, 12	0, 13	0, 12	0, 11	0, 10	0, 48	0, 4	0, 52
Missing[a]	6	6	6	6	6	6	6	6	6

[a]Missing data reflects data that had not yet been collected by the data cutoff.
HJHS, Hemophilia Joint Health Score; Max, maximum; Min, minimum; Q, quartile; SD, standard deviation.

Table 3. Baseline imaging assessments

MRI IPSG Score (All-Patient Population, N=64)

	Left Ankle	Right Ankle	Additional Index Joint[a]
Soft-tissue changes subscore			
n	49	49	16
Mean (SD)	3.1 (1.53)	3.3 (1.53)	3.9 (1.77)
Median	3	3	4
Min, Max (range)	0, 7 (7)	0, 7 (7)	1, 7 (6)
Missing	15	15	48
Osteochondral changes subscore			
n	49	49	16
Mean (SD)	2.3 (2.98)	3.0 (3.29)	5.1 (3.44)
Median	0	1	7
Min, Max (range)	0, 8 (8)	0, 8 (8)	0, 8 (8)
Missing[b]	15	15	48

[a]Additional index joint defined by principal investigator's judgment. [b]Missing data reflects data that had not yet been collected by the data cutoff.
IPSG, International Prophylaxis Study Group; Max, maximum; Min, minimum; MRI, magnetic resonance imaging; SD, standard deviation.

DISCLOSURES:
Yeu-Chin Chen has received speaker fees from AstraZeneca, Bayer, BeiGene, BMS, Chugai, CSL Behring, Kirin, Novartis, PharmaEssentia, Roche, Sanofi, and Takeda, and received grant support from Sanofi and Shire; **Ming-Ching Shen, Cheng-Wei Huang,** and **Hung Chang** have no conflicts to disclose; **Jiaan-Der Wang** has received honoraria from and conducted clinical trials on behalf of Bayer, BioMarin, Chugai, CSL Behring, Novo Nordisk, Pfizer, and Sanofi; **Sheng-Chieh Chou** has received honoraria and consulting fees from AstraZeneca, Bayer, BioMarin, Chugai, CSL Behring, Daiichi Sankyo, Novartis, Novo Nordisk, Pfizer, Roche, Sanofi, Sysmex, and Takeda; **Ching-Tien Peng** and **Chih-Cheng Hsiao** have no conflicts to disclose; **Chia-Yau Chang** has received research funding from Bayer and Sanofi, honoraria from Bayer, Chugai, Novo Nordisk, Pfizer, Sanofi, and Takeda, and participated in advisory boards for Bayer, Chugai, Novo Nordisk, and Sanofi; **Te-Fu Weng** has received honoraria, served on advisory boards, and consulted for Sanofi; **Tong-Ling Chien** and **Ching-Min Chang** are employees of Sanofi and may hold shares/stock options; **Shyh-Shin Chiou** has received research funding from Sanofi.

FUNDING:
This study and medical writing support were funded by Sanofi.

ACKNOWLEDGMENTS:
Medical writing support for the development of this poster was provided by Kimberly Fung, PhD, CMPP™, of IMPRINT Science, and was funded by Sanofi. The authors thank Monique Bidell, PharmD, CMPP™ (Sanofi), and Nick Fulcher, PhD, CMPP™ (Sobi), for publication coordination. Sanofi and Sobi personnel reviewed this poster. The authors had full editorial control of the poster and provided their final approval of all content.

REFERENCES:
1. von Drygalski A, et al. *N Engl J Med*. 2023;388(4):310–318.
2. Malec L, et al. *N Engl J Med*. 2024;391(3):235–246.

International Society on Thrombosis and Haemostasis (ISTH) Congress; July 11–15 2026; Paris, France