Pegcetacoplan Versus Iptacopan for the Treatment of Patients with C3 Glomerulopathy: Indirect Treatment Comparisons

Bradley P. Dixon,[1] Andrew S. Bomback,[2] Carly Rich,[3] Mingyi Huang,[4] Piotr Wojciechowski,[5] Rose Chang,[6] Fernando Caravaca-Fontán,[7] Fadi Fakhouri[8]

[1]University of Colorado School of Medicine, Aurora, CO, USA; [2]Columbia University Irving Medical Center, New York, NY, USA; [3]Sobi, Stockholm, Sweden; [4]Apellis Pharmaceuticals, Inc., Waltham, MA, USA; [5]Clever Access, Kraków, Poland; [6]Analysis Group Inc., Boston, MA, USA; [7]Instituto de Investigación Hospital 12 de Octubre (imas12), Madrid, Spain; [8]Lausanne University Hospital and University of Lausanne, Lausanne, Switzerland

SA-PO0826

INTRODUCTION

- C3G is a rare complement-mediated kidney disease, characterized by dysregulation of the complement system, leading to deposition of C3 fragments and its downstream effectors within the glomeruli[1]
- Current guidelines recommend supportive care treatments that generally have limited or unconfirmed clinical benefit;[1,2] prognosis remains poor, with progression to kidney failure within 10 years of C3G diagnosis reported in up to 50% of patients[3,4]
- A positive clinical benefit has been reported in Phase 3 placebo-controlled trials of two complement pathway inhibitors, pegcetacoplan (targeted C3/C3b inhibitor) and iptacopan (factor B inhibitor)[5–8]
- Currently, there is no direct clinical evidence comparing the relative efficacy of pegcetacoplan and iptacopan in patients with C3G

OBJECTIVE

- To assess the relative efficacy of pegcetacoplan and iptacopan in patients with C3G using two anchored indirect treatment comparisons (ITCs): Bucher (primary analysis – preserves randomization) and matching-adjusted indirect comparison (MAIC; supportive analysis – adjusts for trial differences)

METHODS

Data sources

- The VALIANT trial for pegcetacoplan and the APPEAR-C3G trial for iptacopan had similar designs; however, there were some key differences in eligibility criteria (**Table 1**)

Table 1: Overview of the study characteristics and key inclusion criteria of the VALIANT and APPEAR-C3G trials

Name	Design	Treatments[a]	Key differences in eligibility criteria
VALIANT (NCT05067127)[5,9]	Phase 3, multicenter, randomized (1:1), double-blind, placebo-controlled (6 months) with open-label extensions (6 months)	Pegcetacoplan (up to 1080 mg biw)[b] by subcutaneous infusion (n=63) or placebo (n=61)	• Adolescents (≥12 years and ≥30 kg) or adults (≥18 years) • Native or post-transplant recurrent C3G or primary IC-MPGN • No restrictions based on baseline serum C3 levels • Prednisone doses up to 20 mg/day permitted
APPEAR-C3G (NCT04817618)[10,11]		Iptacopan (200 mg bid) orally (n=38) or placebo (n=36)	• Adults (≥18 to ≤60 years)[c] • Native C3G • Reduced serum C3 level (<0.85 × LLN [<77 mg/dL]) • Prednisone doses up to 7.5 mg/day permitted

[a] During the double-blind period; all patients received open-label active treatment (pegcetacoplan or iptacopan) during the open-label extension
[b] Lower weight-based dose of 648 mg or 810 mg used for some adolescents. [c] Enrollment ongoing in a separate cohort of adolescents

- Both ITCs used baseline and 6-month outcome data. Individual patient data (IPD) from VALIANT were used to derive aggregate data inputs for the Bucher and the IPD were used directly for the MAIC. Published aggregate data were used for APPEAR-C3G[7,8] The Bucher analysis and the MAIC were anchored to the common placebo arm

Bucher analysis

- This methodology assumes the studies are comparable with respect to study design, effect modifiers, and outcomes measured[12–14]
- The Bucher analyzed data for all available patients (i.e. the ITT populations) to preserve randomization

MAIC

- In MAICs, IPD are reweighted so that baseline characteristics match the comparator trial's published aggregates data, reducing between-trial differences[15,16]
- In this MAIC, the trials were first aligned by excluding patients in VALIANT who would be ineligible under the APPEAR-C3G criteria (**Table 1**), yielding comparable analysis populations. Propensity score weights were then estimated via logistic regression, using age as a prespecified prognostic variable, to approximate each VALIANT patient's probability of enrollment in APPEAR-C3G and to balance baseline characteristics across trials. Outcomes were subsequently analyzed on the weighted populations

Outcomes

- Relative efficacy at Month 6 was assessed for five key outcomes: mean reduction in log-transformed UPCR from baseline; proportion of patients who achieved UPCR <1 g/g; proportion of patients who achieved ≥50% UPCR reduction from baseline; mean reduction in eGFR from baseline; and proportion of patients who achieved the composite renal endpoint (≥50% reduction in UPCR from baseline and ≤15% reduction in eGFR from baseline)
- A comparative assessment of glomerular C3 staining was not feasible due to differences in outcome measurement in the two trials

RESULTS

Efficacy of pegcetacoplan vs. iptacopan

- The Bucher analysis showed that pegcetacoplan was associated with a **statistically significant improvement in therapeutic benefit** compared with iptacopan at Month 6 across three clinically relevant proteinuria outcomes and the composite endpoint (**Figures 1** and **2**)

Figure 1: Summary of proteinuria outcomes and the composite endpoint for pegcetacoplan vs. iptacopan at Month 6 (Bucher)

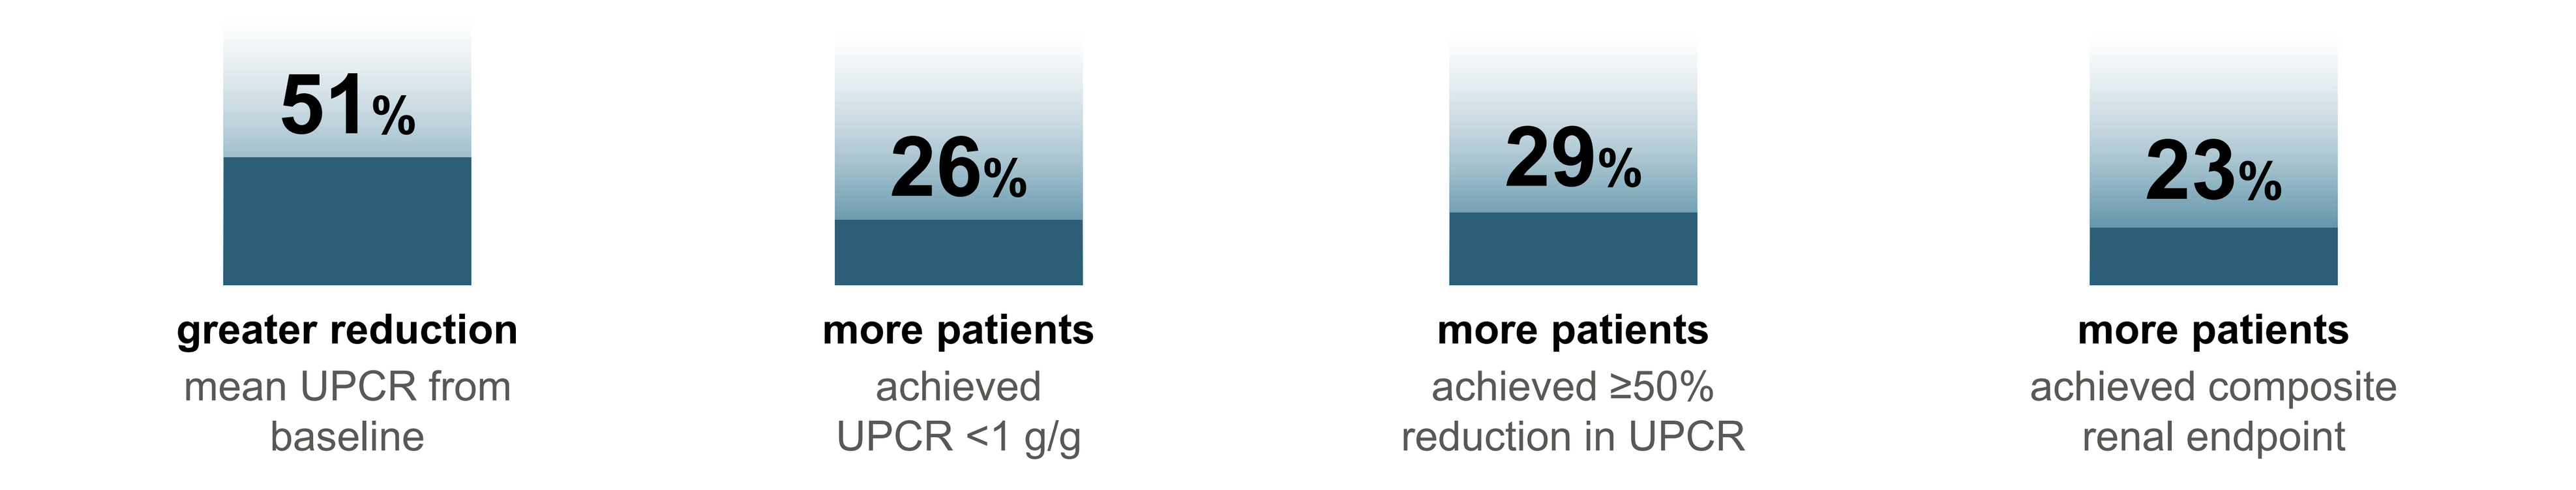
51%
greater reduction
mean UPCR from baseline
26%
more patients
achieved UPCR <1 g/g
29%
more patients
achieved ≥50% reduction in UPCR
23%
more patients
achieved composite renal endpoint

- In the Bucher analysis, change from baseline in eGFR **favored pegcetacoplan vs. iptacopan numerically**, although the difference did not reach statistical significance (MD 4.16 [95% CI −3.45, 11.76]; p=0.284)
- Results from the **MAIC were generally consistent** with the Bucher analysis across the five endpoints (**Figure 2**)

LIMITATIONS

- IPD were available only for the pegcetacoplan trial and not for the iptacopan trial; thus, patient-level adjustment between trials was not feasible
- Anchored ITCs are potentially limited by assumptions about differences in trial design, reliance on reporting of trial characteristics and unknown differences that were not accounted for

Figure 2: Efficacy outcomes for pegcetacoplan vs. iptacopan at Month 6 (Bucher and MAIC)

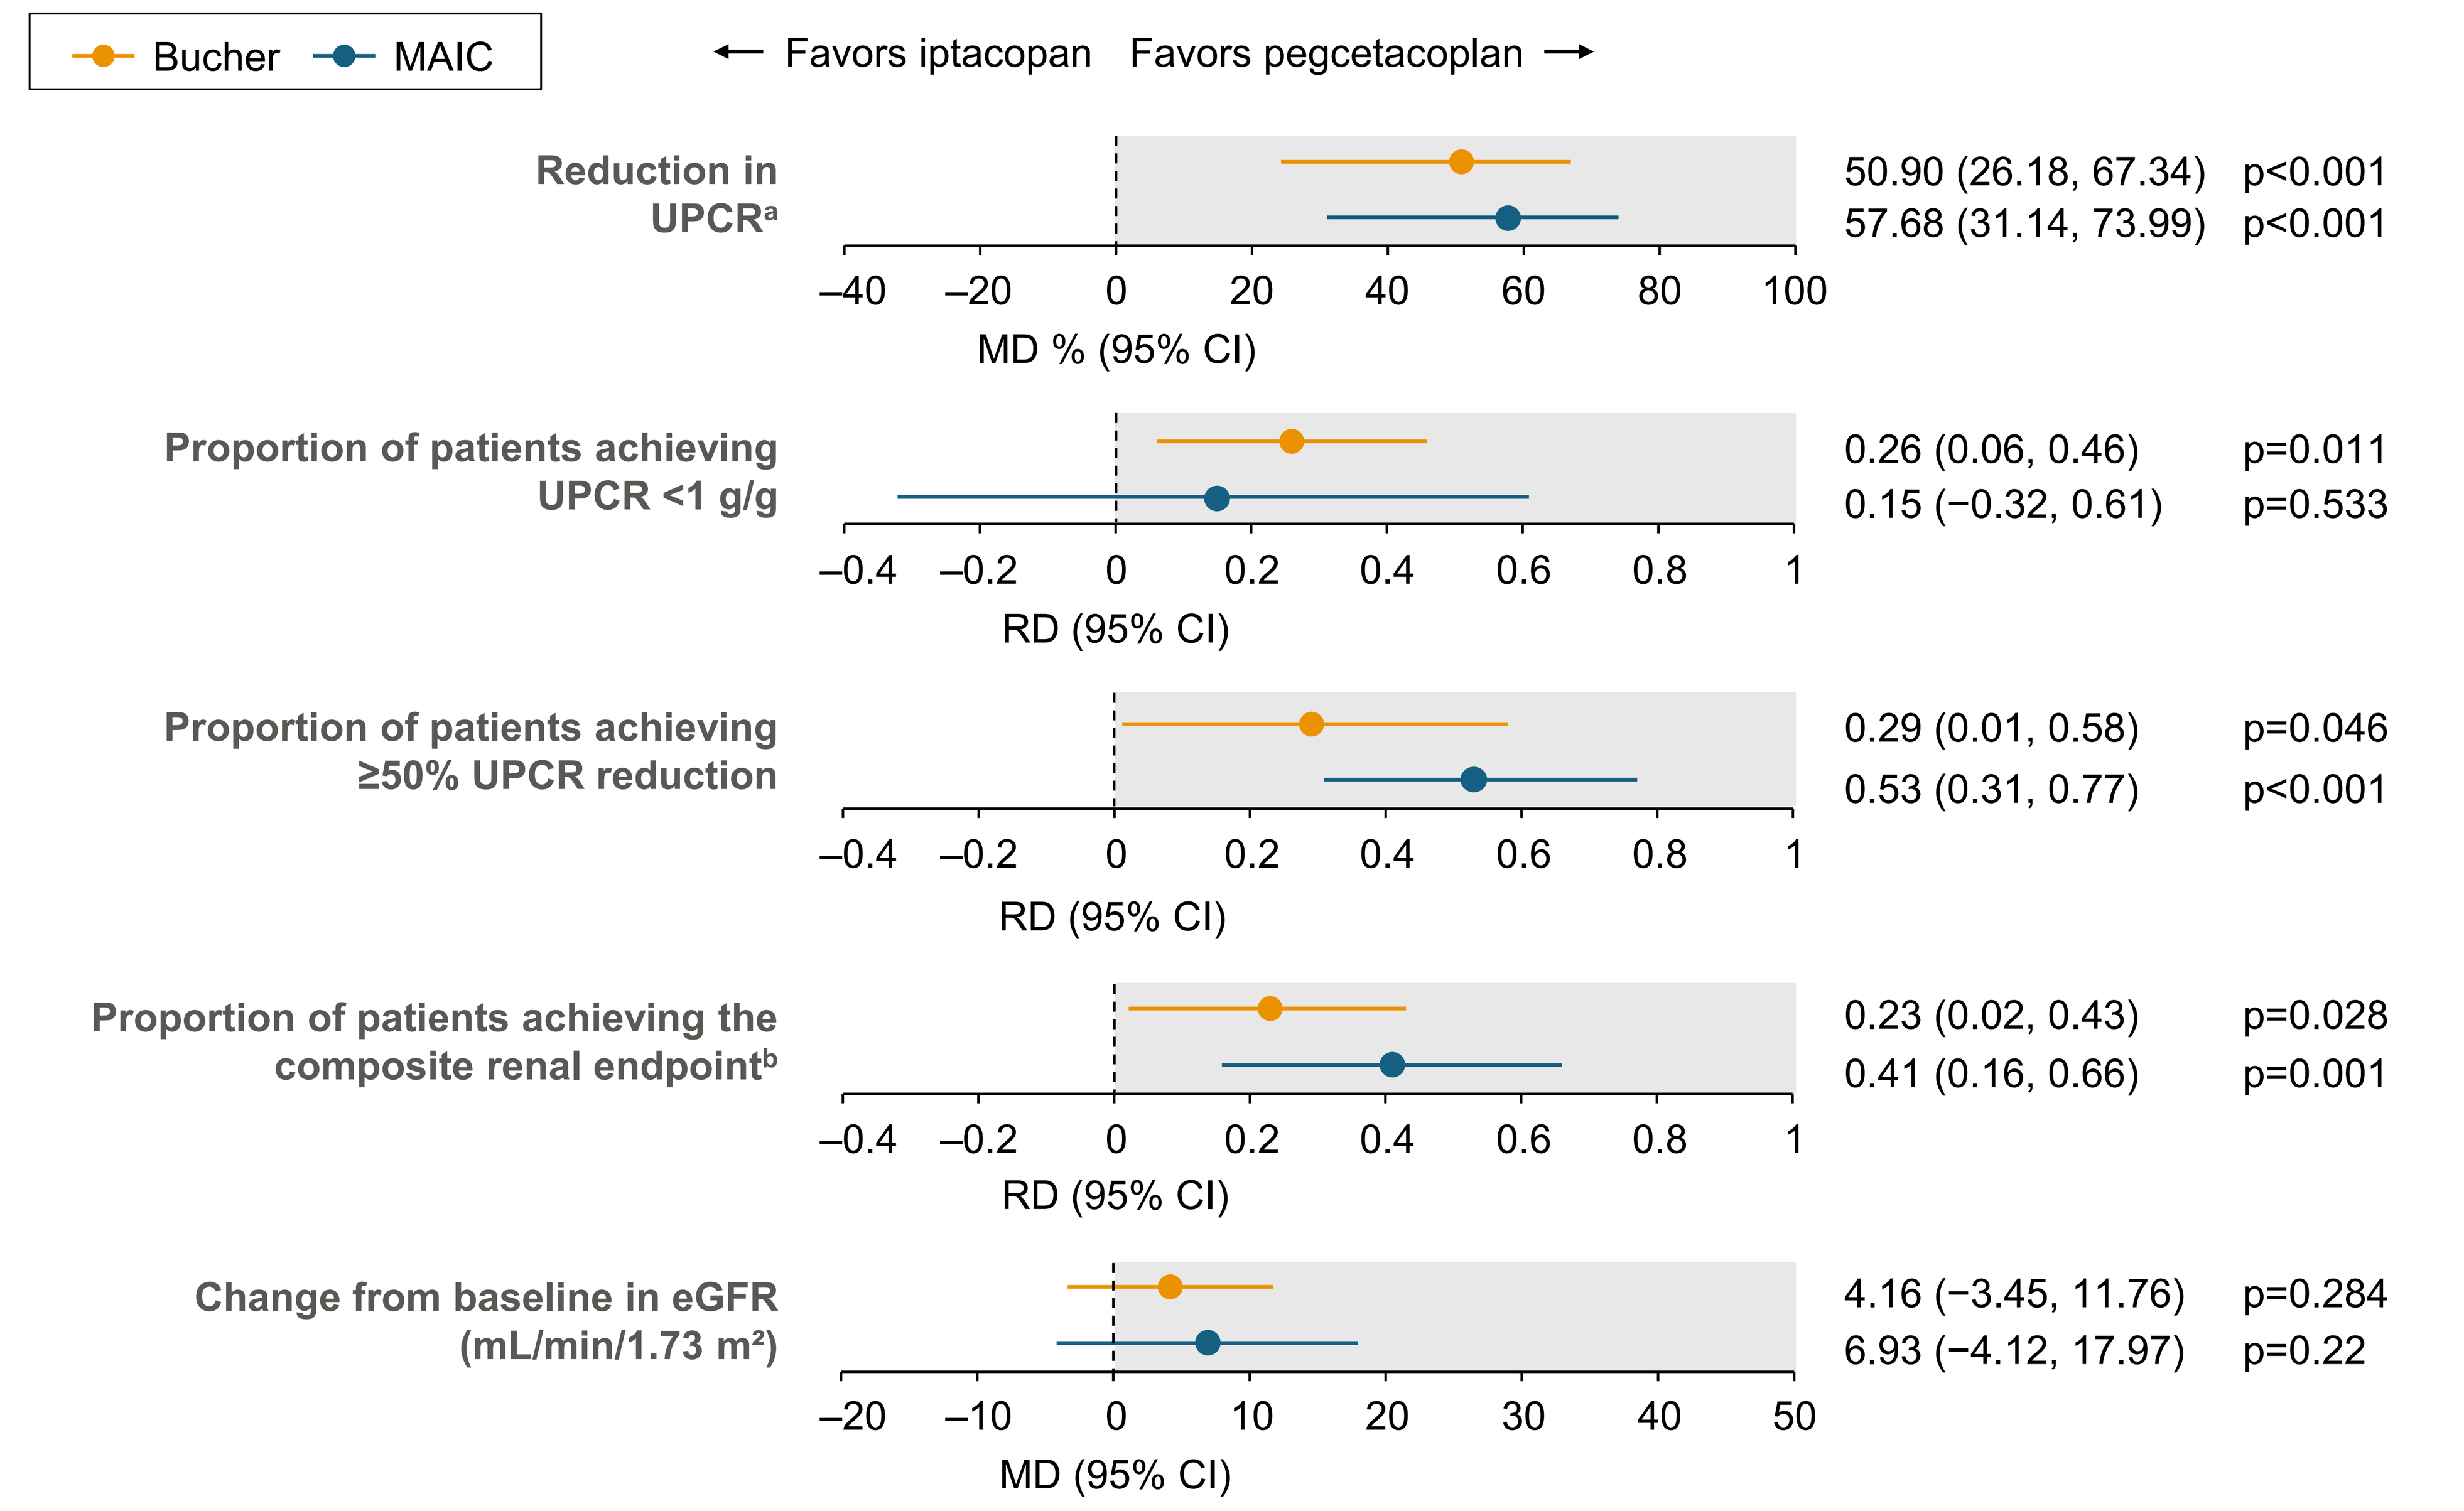
Bucher
MAIC
← Favors iptacopan
Favors pegcetacoplan →
Reduction in UPCR[a]
50.90 (26.18, 67.34) p<0.001
57.68 (31.14, 73.99) p<0.001
–40 –20 0 20 40 60 80 100
MD % (95% CI)
Proportion of patients achieving UPCR <1 g/g
0.26 (0.06, 0.46) p=0.011
0.15 (−0.32, 0.61) p=0.533
–0.4 –0.2 0 0.2 0.4 0.6 0.8 1
RD (95% CI)
Proportion of patients achieving ≥50% UPCR reduction
0.29 (0.01, 0.58) p=0.046
0.53 (0.31, 0.77) p<0.001
–0.4 –0.2 0 0.2 0.4 0.6 0.8 1
RD (95% CI)
Proportion of patients achieving the composite renal endpoint[b]
0.23 (0.02, 0.43) p=0.028
0.41 (0.16, 0.66) p=0.001
–0.4 –0.2 0 0.2 0.4 0.6 0.8 1
RD (95% CI)
Change from baseline in eGFR (mL/min/1.73 m²)
4.16 (−3.45, 11.76) p=0.284
6.93 (−4.12, 17.97) p=0.22
–20 –10 0 10 20 30 40 50
MD (95% CI)

[a] Changes in UPCR from baseline at Month 6 were assessed using first-morning urine samples in VALIANT but 24-hour urine collections in APPEAR-C3G
[b] Composite renal endpoint was defined as proportion of patients achieving **≥50% reduction from baseline in UPCR** and **stable/improved (≤15% reduction) eGFR**

CONCLUSIONS

- **In the absence of head-to-head studies, two separate ITCs using Bucher and MAIC methodologies indicate that pegcetacoplan provides a therapeutic benefit compared with iptacopan in patients with C3G**
- **Pegcetacoplan was associated with greater reductions in proteinuria levels, and more pegcetacoplan-treated patients achieved the composite renal endpoint (≥50% reduction in UPCR and stable/improved eGFR) vs. iptacopan**
- **This analysis highlights the potential of pegcetacoplan to significantly improve the lives of patients with C3G through sustained reduction in proteinuria, thereby reducing the risk of long-term outcomes such as kidney disease progression, kidney failure, and kidney transplant**
- **These comparative effectiveness findings may help guide clinicians and payers managing patients with this rare disease**

References

1. Tarragon Estebanez B, Bomback AS. *Kidney Int Rep.* 2023;9(3):569–579; **2.** Bomback AS, et al. *Kidney Int Rep.* 2024;10(1):17–28; **3.** Guzman GL, Perry KW. *Nephrology (Carlton).* 2025;30(2):e70001; **4.** Magliulo EK, Ravipati P. *Glomerular Dis.* 2024;4(1):200–210; **5.** Nester CM, et al. Presented at ASN Annual Meeting 2024; **6.** Vivarelli M, et al. Presented at 62nd ERA Congress. 2025; **7.** Kavanagh D, et al. Presented at 61st ERA Congress. 2024; **8.** Kavanagh D, et al. *Lancet.* 2025;406(10512):1587–1598; **9.** Clinicaltrials.gov. Available from: https://clinicaltrials.gov/study/NCT05067127. Accessed September 2025; **10.** Bomback AS, et al. *Kidney Int Rep.* 2022;7(10):21502159; **11.** Clinicaltrials.gov. Available from: https://clinicaltrials.gov/study/NCT04817618. Accessed September 2025; **12.** Bucher HC, et al. *J Clin Epidemiol.* 1997;50(6):683–691; **13.** Macabeo B, et al. *Pharmacoecon Open.* 2024;8(1):5–18; **14.** Merkel M, et al. *Expert Opin Pharmacother.* 2023;24(10):1205–1214; **15.** Signorovitch JE, et al. *Value Health.* 2012;15(6):940–947; **16.** Phillippo D, et al. Available from: https://research-information.bris.ac.uk/ws/portalfiles/portal/94868463/Population_adjustment_TSD_FINAL.pdf. Accessed September 2025

Abbreviations

ASN, American Society of Nephrology; bid, twice per day; biw, twice per week; C3, complement component 3; C3G, C3 glomerulopathy; CI, confidence interval; eGFR, estimated glomerular filtration rate; ERA, European Renal Association; IC-MPGN, immune-complex membranoproliferative glomerulonephritis; IPD, individual patient data; ITC, indirect treatment comparison; ITT, intent-to-treat; LLN, lower limit of normal; MAIC, matching-adjusted indirect comparison; MD, mean difference; RD, risk difference; UPCR, urine protein-creatinine ratio.

Acknowledgments

The authors gratefully acknowledge the patients, investigators and study teams involved in the VALIANT and APPEAR-C3G trials. The authors thank Maryaline Catillon, Anaïs Lemyre, Chunyi Xu, and Alice Qu of the Analysis Group, Inc. for study conceptualization, data processing, statistical analyses, and/or interpretation of results. The authors also thank Wojciech Margas and Katarzyna Jamróz-Dolinska from Clever Access for data processing and statistical analyses. The authors acknowledge Sophia Miliara from Sobi for publication coordination and Tyrone Daniel from Genesis Medical Writing Ltd (Manchester, UK) for medical writing. Sobi and Apellis reviewed and provided feedback on the poster. The authors had full editorial control of the poster and provided their final approval of all content. This study and medical writing support were funded by Sobi and Apellis Inc.

Disclosures

BPD reports consultancy from Apellis, Novartis, Alexion, AstraZeneca, Arrowhead, Calliditas; ASB reports consultancy and/or honoraria from Achillion, Alexion, Amgen, Apellis, Aurinia, Calliditas, Catalyst, Genentech, GSK, Kezar, Novartis, Otsuka, Q32, Silence Therapeutics, UpToDate; CR is an employee of Sobi; MH is an employee of Apellis; PW is an employee of Clever Access; RC is an employee of the Analysis Group, Inc.; FC-F reports consultancy and/or honoraria from Alexion, Apellis, AstraZeneca, Novartis, Otsuka, Roche, Sobi, Vifor; FF reports consultancy and/or honoraria from Alexion, Apellis, AstraZeneca, Novartis, Roche, Sanofi, Sobi.

Kidney Week 2025, November 6–9, Houston, Texas, USA