Final Surgery Data from B-MORE: A 24-Month, Prospective, Non-Interventional Study of the Real-World Effectiveness and Usage of rFIXFc in Haemophilia B

Heidi Glosli,[1] Anna Olsson,[2] Saket Badle,[3] Flora Peyvandi,[4] Håkan Malmström,[5] Eva Bednar,[5] Stefan Lethagen[5]

[1]Norwegian Centre for Rare Diseases, Division of Paediatric and Adolescent Medicine, Oslo University Hospital, Oslo, Norway; [2]Department of Medicine, Region Västra Götaland, Götaland, Sahlgrenska University Hospital, Gothenburg, Sweden; [3]Paediatric Haemostasis and Thrombosis Unit, Oxford University Hospitals NHS Foundation Trust, Oxford, United Kingdom; [4]Angelo Bianchi Bonomi Hemophilia and Thrombosis Center, Fondazione IRCCS Ca' Granda Ospedale Maggiore Policlinico, Milan, Italy; [5]Sobi, Stockholm, Sweden

PB2117

CONCLUSION

Final surgical data from B-MORE confirm extended half-life recombinant factor IX Fc fusion protein is efficacious and well tolerated in people with haemophilia B when used in a real-world surgical setting.

INTRODUCTION

- People with haemophilia B (PwHB) are at risk of excessive bleeding during surgery; factor replacement is the standard of care for the perioperative management of patients without inhibitors to establish effective bleed resolution and haemostasis.
- Extended half-life recombinant factor IX Fc fusion protein (rFIXFc) has established efficacy and safety profiles in PwHB across surgeries in Phase 3 trials;[1–4] however, comprehensive real-world data can guide perioperative management.
- B-MORE was a 24-month, prospective, non-interventional study evaluating the real-world effectiveness and usage of rFIXFc in PwHB across Europe and the Middle East.[5]
- Here, we report final surgical data for patients treated with rFIXFc over a prolonged period in B-MORE.

METHODS

- This analysis included PwHB who had ≥1 surgery whilst exclusively receiving rFIXFc during the retrospective and prospective periods of B-MORE.
- Major surgeries were defined as one or more of the following:
 - Any elective or emergent surgical procedure involving general anaesthesia.
 - Respiratory assistance in which a major body cavity is penetrated and exposed, or for which a substantial impairment of physical or physiological functions is produced.
- Minor surgeries were all other invasive procedures.
- The perioperative period was defined as the time prior to, during and post-surgery until return to regular treatment.
- Outcomes reported include haemostatic efficacy, hospitalisation duration, blood loss, rFIXFc usage and concomitant medication use.
- rFIXFc usage data reflect surgeries with bolus injections, while haemostatic efficacy outcomes (where available) also include surgeries with continuous infusion.
- All patients provided informed consent; B-MORE was approved by an independent ethics committee.

RESULTS

Study population

- B-MORE enrolled 151 PwHB; of these, 32 PwHB had ≥1 surgery and are included in this analysis, with 52 surgeries in total.
 - Baseline characteristics of these patients are presented in **Table 1**.
- Overall, 46 surgeries were managed with bolus injections (9 major, 37 minor), 2 surgeries were managed with continuous infusion (1 major, 1 minor) and 4 minor surgeries were managed with regular prophylaxis.
- Ten surgeries were major (n=9 PwHB), as shown in **Table 2**, and 42 surgeries were minor (n=25 PwHB), including 9 dental procedures and 8 central venous access device removals/insertions.

Major surgeries

- Outcomes including injection frequency and rFIXFc consumption for individual major surgeries at 24, 72 and 168 hours post-surgery are shown in **Table 2**.
- Haemostatic efficacy was rated excellent or good for all major surgeries with available data (n=1 surgery with bolus injection missing).
- Most patients were hospitalised for less than a week and had no intraoperative or postoperative blood loss.
 - One total knee replacement required 2 red blood cell unit transfusions.

Table 1: Baseline characteristics

	Patients with ≥1 surgery on rFIXFc (n=32)
Age (years), median (range)	33.3 (1–81)
Age categories (years), n (%)	
<6	4 (12.5)
6–<12	7 (21.9)
12–<18	3 (9.4)
18–<65	16 (50.0)
≥65	2 (6.3)
Weight (kg), median (range)	72.5 (11–104)
BMI (kg/m²), median (range)	24.4 (16–33)
Haemophilia B severity, n (%)	
Severe	24 (75.0)
Moderate	5 (15.6)
Mild	3 (9.4)
History of inhibitors, n (%)	0

All characteristics are presented at B-MORE enrolment. Percentages may not sum to 100 due to rounding.

Table 2: Patient characteristics and perioperative outcomes for individual major surgeries

Patient characteristics			Perioperative outcomes												
Major surgery type	Age (years)[a]	Haemophilia B severity	Haemostatic efficacy[b]	Hospitalisation duration (days)	Return to regular treatment (days)	Intraoperative blood loss (mL)	Postoperative blood loss (mL)	Number of rFIXFc injections (n)				rFIXFc consumption (IU/kg)			
								Total[c]	During 24h[d]	During 72h[d]	During 168h[d]	Total[c]	During 24h[d]	During 72h[d]	During 168h[d]
Orthopaedic															
Total knee replacement[e,f]	18–<65	Severe	Good	10	21	N/A	500	19	4	8	12	688.9	144.4	266.7	444.4
Ankle arthroscopy	18–<65	Severe	Excellent	2	3	0	0	5	3	5	–	155.6	111.1	155.6	–
Knee arthroplasty	18–<65	Severe	N/A	N/A	2	N/A	N/A	3	3	–	–	226.6	226.6	–	–
Ankle arthroscopy osteophyte removal	12–<18	Severe	Good	2	11	0	0	4	1	3	4	175.5	75.2	125.3	175.5
Ankle arthrodesis[g]	18–<65	Severe	Good	9	7	N/A	N/A	N/A	N/A	N/A	N/A	N/A	N/A	N/A	N/A
Non-orthopaedic															
Appendectomy	6–<12	Moderate	Excellent	4	5	0	0	5	2	4	5	335.6	134.2	268.5	335.6
Evacuation of haematoma	18–<65	Severe	Excellent	9	17	200	0	4	1	2	4	182.9	73.2	109.8	182.9
Low anterior resection with diverting stoma[f,h]	18–<65	Severe	Excellent	8	12	750	0	11	3	5	9	464.4	134.2	227.1	361.2
Reversal of loop ileostomy	18–<65	Severe	Excellent	4	6	0	0	7	3	5	7	318.1	186.5	241.3	318.1
L4–L5 laminectomy[f]	18–<65	Severe	Good	6	21	N/A	N/A	12	3	6	8	682.5	182.0	318.5	409.5

N/A represents data that are not available. [a]Age range at the time of surgery. [b]As rated by the surgeon/physician: Excellent (intraoperative and postoperative blood loss similar [within 10%] to patients without haemophilia; good (intraoperative and postoperative blood loss slightly increased [10–25% of expected], but the difference is judged by the involved surgeon/anaesthetist to be clinically insignificant); fair (intraoperative and/or postoperative blood loss increased over expectation [25–50%] for patients without haemophilia and additional treatment is needed); or poor (significant intraoperative and/or postoperative blood loss that is substantially increased over expectation [>50%] for patients without haemophilia, requires intervention and is not explained by a surgical/medical issue other than haemophilia). [c]During the perioperative period. [d]From preoperative loading dose to 24 hours, 72 hours and 168 hours after surgery, respectively. [e]This surgery required 2 red blood cell unit transfusions. [f]These patients received intensified dosing following surgery, prior to return to regular prophylaxis. [g]This surgery was performed under continuous infusion; a loading dose of 60.3 IU/kg was reported with no further information provided regarding infusion rate. [h]Thromboprophylaxis was given for this surgery.

Minor surgeries

- A summary of outcomes for minor surgeries, including haemostatic efficacy, hospitalisation duration, time to regular prophylaxis and rFIXFc usage, are shown in **Figure 1**.
 - Haemostatic efficacy was excellent or good for all minor surgeries with available data except 1 (rated fair; n=6 surgeries missing).
- No blood loss, transfusions or thromboprophylaxis use were reported.

Concomitant medication

- Concomitant anti-fibrinolytic therapy was used in 6 major surgeries and 26 minor surgeries.
- Thromboprophylaxis was given for 1 major surgery.

Safety

- No inhibitor development was reported in this study.
- No surgery-related serious adverse events or serious adverse events leading to treatment discontinuation were reported.

Figure 1: Perioperative outcomes for minor surgeries

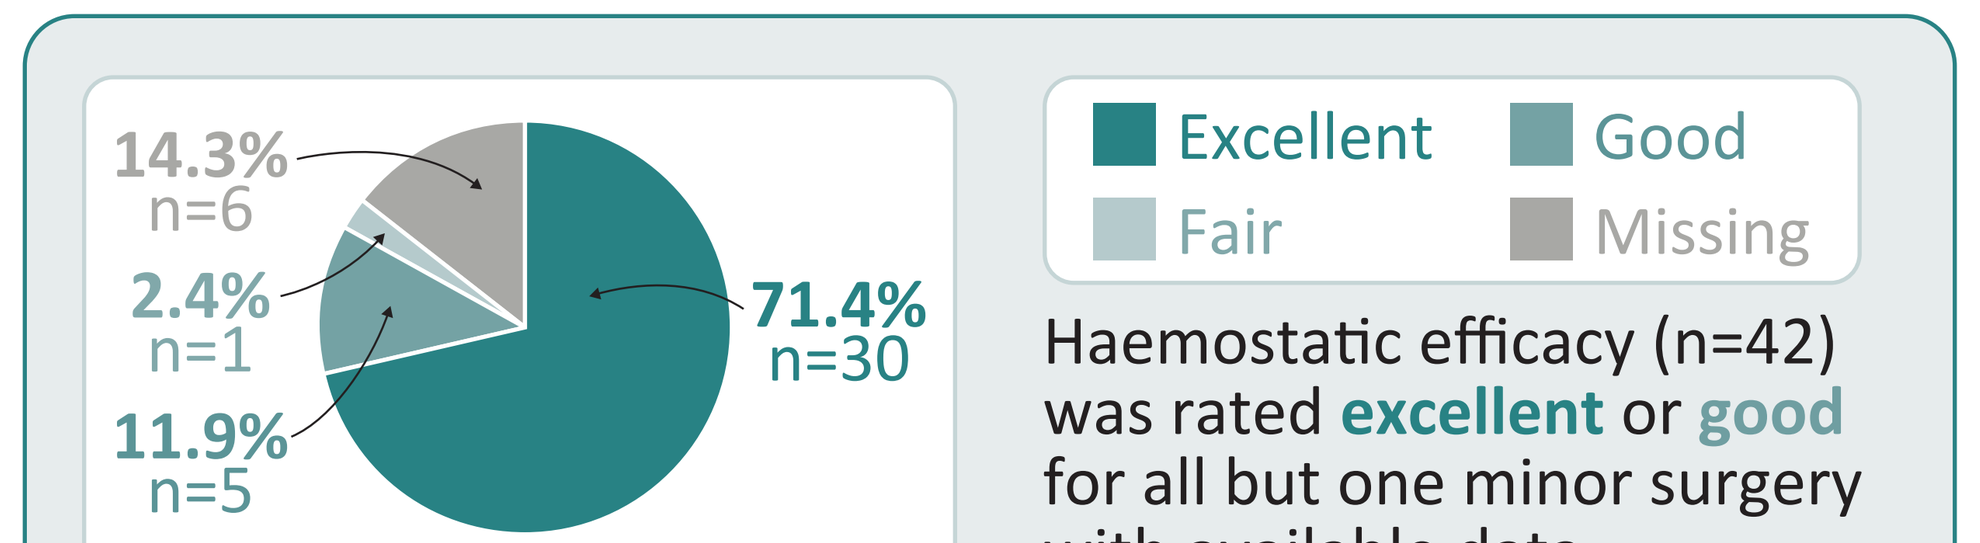
14.3%
n=6
2.4%
n=1
11.9%
n=5
71.4%
n=30
Excellent
Good
Fair
Missing

Haemostatic efficacy (n=42) was rated **excellent** or **good** for all but one minor surgery with available data

Haemostatic efficacy as rated by the surgeon/physician.

Hospitalisation duration:[a] **2.0 (1–7) days**

Time to regular prophylaxis:[b] **5.5 (0–15) days**

Values are reported as median (range). [a]n=32. [b]n=36.

Median (IQR) rFIXFc usage (n=37)	During 24h	During 72h	During 168h
Factor consumption, IU/kg	66.7 (39.7–96.4)	81.7 (42.3–151.3)	83.9 (43.8–151.3)
Injection frequency	1.0 (1–1)	1.0 (1–2)	1.0 (1–3)

Time periods are from preoperative loading dose to 24 hours, 72 hours and 168 hours after surgery, respectively.

References

1. Powell JS, et al. *N Engl J Med* 2013;369:2313–23; **2.** Fischer K, et al. *Lancet Haematol* 2017;4:e75–82; **3.** Pasi KJ, et al. *Haemophilia* 2020;26:e262–71; **4.** Nolan B, et al. *Blood Adv* 2021;5:2732–9; **5.** ClinicalTrials.gov (NCT03901755).

Disclosures

HG: Principal investigator for studies by Baxalta (Takeda), Bayer, Novo Nordisk, Octapharma, Roche and Sobi; writing support from Bayer and Sobi. **AO:** Research grant from CSL Behring. **SB:** Educational meeting support and/or advisory boards for CSL Behring, LFB, Novo Nordisk, Roche/Chugai, Roche and Sobi. **FP:** Participation at educational meetings and advisory boards of BioMarin, Grifols, Roche, Sanofi, Sobi and Takeda. **HM, EB, SL:** Employees of Sobi and may hold shares and/or stock options in the company.

Abbreviations

BMI: body mass index; **FIX:** factor IX; **IU:** International Unit; **IQR:** interquartile range; **PwHB:** people with haemophilia B; **rFIXFc:** recombinant factor IX Fc fusion protein.

Acknowledgements

We thank the patients and investigators who participated in the study. The authors acknowledge Nick Fulcher, PhD, CMPP from Sobi for publication coordination. The authors also acknowledge Isabel Merrien, PgDip, Costello Medical, London, UK for medical writing and editorial assistance, and Peter Lang, Costello Medical, for design assistance, funded by Sobi. Sobi and Sanofi reviewed and provided feedback on the poster. This research is funded by Sobi.

Presented at the 34th Congress of the International Society on Thrombosis and Haemostasis (ISTH) | 11–15 July 2026 | Paris, France