WFH
WORLD CONGRESS
KUALA LUMPUR • APRIL 19-22, 2026

Plain Language Summary: What is Extravascular Distribution of Factor IX and How Does it Affect Management of Those With Haemophilia B?

C. HERMANS[1], C. ESCURIOLA ETTINGSHAUSEN[2], M. LAFFAN[3], D. MATINO[4], N. O'CONNELL[5], R.F. SIDONIO JR[6], R. LJUNG[7]

1 Division of Haematology, Saint-Luc University Hospital, Université catholique de Louvain, Brussels, Belgium; 2 Haemophilia Centre Rhein Main – HZRM, Frankfurt, Germany; 3 Centre for Haematology, Imperial College London, London, UK; 4 McMaster University, Hamilton, Canada; 5 National Coagulation Centre, St James's Hospital, Dublin, Ireland; 6 Emory University and HOG Center for Bleeding and Clotting Disorders, Atlanta, USA; 7 Lund University, Department of Clinical Sciences – Paediatrics, Lund, Sweden

Abstract ID: 210

Poster Number: PP-68

Access poster via the Sobi Science Library. Copies of this poster obtained through QR Code are for personal use only.

INTRODUCTION

- People with haemophilia B have low levels of a protein called 'factor IX' (FIX), or the FIX they do have does not function properly.[1,2]
- FIX is found in blood vessels (the 'bloodstream' or 'vasculature') but can also move into other parts of the body; this is called 'extravascular distribution'.[3,4]
- FIX has a role in helping blood to clot.[5] Products containing FIX are used as treatment to prevent/stop bleeding in people with haemophilia B.[6] For continued protection from bleeding, ongoing regular therapy ('prophylaxis') is needed.[6]
- There are different types of FIX products. Some, termed 'extended half-life', are modified to stay longer in the body so can be given less often.[6,7] However, these modifications can influence how much product remains in the bloodstream or is extravascular, as shown by several studies.[8–11]

AIM

- To collect information from scientific literature to help understand the implications of the extravascular distribution of FIX when treating people with haemophilia B.

METHODS

- We searched the scientific literature to find articles about the extravascular distribution of FIX/FIX products and its relevance for preventing bleeding.
- A total of 476 articles were found, published between 1957 and October 2025.
- After the title and summary of each article were screened, seven reviewers from seven countries with expertise in haemophilia B collected and critically reviewed and evaluated information from selected articles.
- We summarised our findings from the selected articles and any additional material, as required, and considered what this means for treating people with haemophilia B.

RESULTS

- Our summary of the information collected from the literature is shown below.

Extravascular distribution of FIX and management of haemophilia B

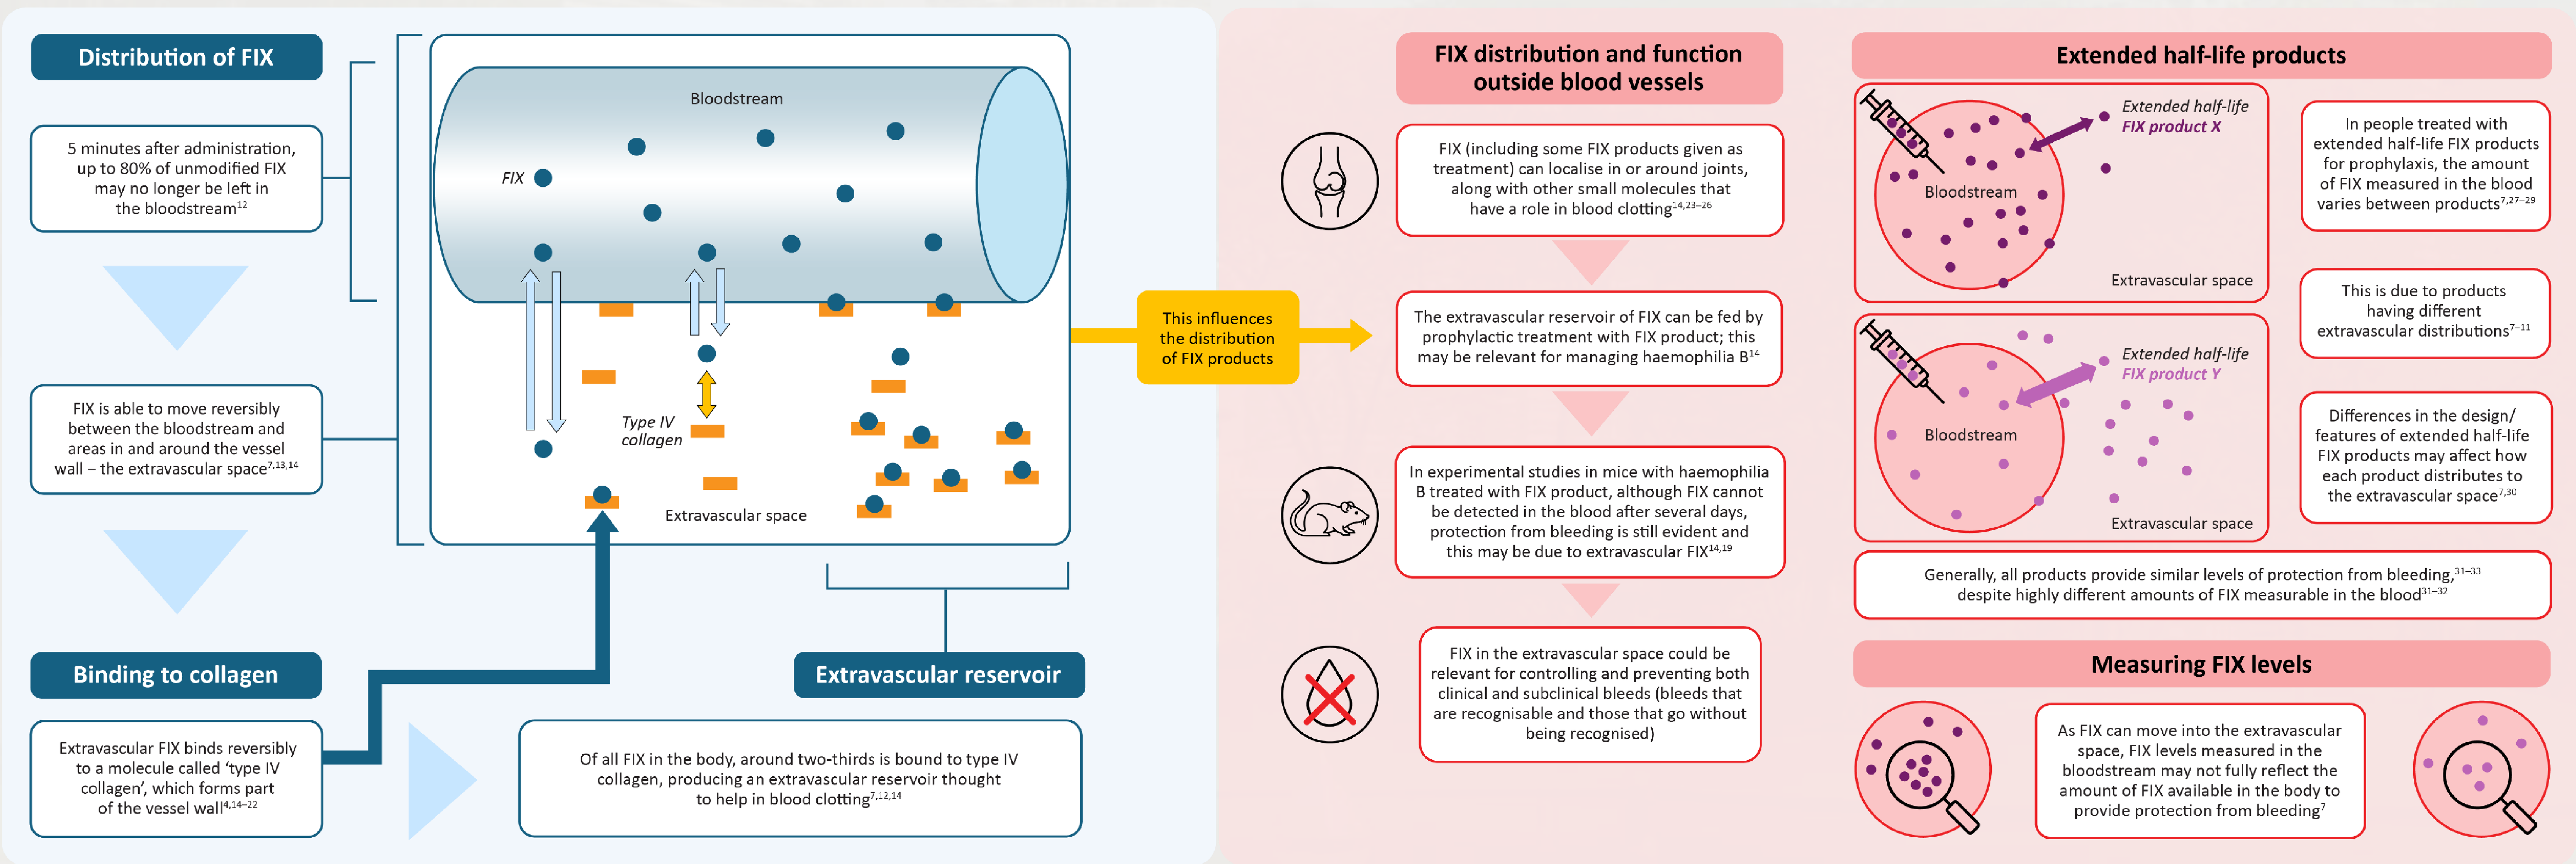

Distribution of FIX
5 minutes after administration, up to 80% of unmodified FIX may no longer be left in the bloodstream[12]
FIX is able to move reversibly between the bloodstream and areas in and around the vessel wall – the extravascular space[7,13,14]
Binding to collagen
Extravascular FIX binds reversibly to a molecule called 'type IV collagen', which forms part of the vessel wall[4,14–22]
Bloodstream
FIX
Type IV collagen
Extravascular space
Extravascular reservoir
Of all FIX in the body, around two-thirds is bound to type IV collagen, producing an extravascular reservoir thought to help in blood clotting[7,12,14]
This influences the distribution of FIX products
FIX distribution and function outside blood vessels
FIX (including some FIX products given as treatment) can localise in or around joints, along with other small molecules that have a role in blood clotting[14,23–26]
The extravascular reservoir of FIX can be fed by prophylactic treatment with FIX product; this may be relevant for managing haemophilia B[14]
In experimental studies in mice with haemophilia B treated with FIX product, although FIX cannot be detected in the blood after several days, protection from bleeding is still evident and this may be due to extravascular FIX[14,19]
FIX in the extravascular space could be relevant for controlling and preventing both clinical and subclinical bleeds (bleeds that are recognisable and those that go without being recognised)
Extended half-life products
Extended half-life FIX product X
Bloodstream
Extravascular space
Extended half-life FIX product Y
Bloodstream
Extravascular space
In people treated with extended half-life FIX products for prophylaxis, the amount of FIX measured in the blood varies between products[7,27–29]
This is due to products having different extravascular distributions[7–11]
Differences in the design/features of extended half-life FIX products may affect how each product distributes to the extravascular space[7,30]
Generally, all products provide similar levels of protection from bleeding,[31–33] despite highly different amounts of FIX measurable in the blood[31–32]
Measuring FIX levels
As FIX can move into the extravascular space, FIX levels measured in the bloodstream may not fully reflect the amount of FIX available in the body to provide protection from bleeding[7]

CONCLUSIONS

- Evidence suggests FIX has a role in blood clotting outside of the bloodstream. The extravascular presence of FIX may provide protection from bleeding, with benefits for treatment of haemophilia B.
- FIX measured in blood may not fully reflect the degree of protection from bleeding; it is not currently possible to measure FIX in the extravascular space. Measuring other outcomes, such as how often bleeds occur, joint health and function and quality of life, will provide valuable information about how well treatment is working.

REFERENCES

1. Pezeshkpoor B, et al. Hamostaseologie. 2022;42:390–9; 2. Kizilocak H and Young G. Clin Adv Hematol Oncol. 2019;17:344–51; 3. Stafford D. Thromb J. 2016;14(Suppl. 1):35; 4. Castaman G and Matino D. Haematologica. 2019;104:1702–9; 5. Peters R and Harris T. Nat Rev Drug Discov. 2018;17:493–508; 6. Srivastava A, et al. Haemophilia. 2020;26(Suppl. 6):1–158; 7. Mann DM, et al. Haemophilia. 2021;27:332–9; 8. Preijers T, et al. Eur J Clin Pharmacol. 2021;77:1193–200; 9. Diao L, et al. Clin Pharmacokinet. 2014;53:467–77; 10. Escuriola Ettingshausen C, et al. Res Pract Thromb Haemost. 2019;3:268–76; 11. Zhang Y, et al. J Thromb Haemost. 2016;14:2132–40; 12. Feng D, et al. J Thromb Haemost. 2013;11:2176–8; 13. Iorio A, et al. Thromb Haemost. 2017;117:1023–30; 14. Leuci A, et al. J Thromb Haemost. 2024;22:700–8; 15. Cheung WF, et al. Proc Natl Acad Sci USA. 1996;93:11068–73; 16. Wolberg AS, et al. J Biol Chem. 997;272:16717–20; 17. Cooley B, et al. Blood. 2016;128:286–92; 18. Machado SK, et al. Thromb Haemost. 2023;123:751–62; 19. Chu B, et al. Res Pract Thromb Haemost. 2023;7:98; 20. Morrow G, et al. Res Pract Thromb Haemost. 2024;8(Suppl. 2):11 (Abstract OC36.11); 21. Stern DM, et al. Br J Haematol. 1987;66:227–32; 22. Manon-Jensen T, et al. J Thromb Haemost. 2016;14:438–48; 23. Chang P, et al. Am J Hematol. 1995;50:79–83; 24. Enjolras N, et al. Haemophilia. 2023;29(Suppl. 1):36; 25. van der Flier A, et al. Blood Coagul Fibrinolysis. 2023;34:353–63; 26. Salas J, et al. Blood. 2017;130(Suppl. 1):1061 (Abstract 1321); 27. Collins PW, et al. Blood. 2014;124:3880–6; 28. Powell JS, et al. N Engl J Med. 2013;369:2313–23; 29. Santagostino E, et al. Blood 2016;127:1761–9; 30. Kleiboer B, et al. Haemophilia. 2020;26:e23–5; 31. Mancuso ME, et al. J Blood Med. 2023;14:427–34; 32. Astermark J, et al. J Blood Med. 2021;12:613–21; 33. Guillet B, et al. Adv Ther. 2024;41:649–58.

ACKNOWLEDGEMENTS/DISCLOSURES

This study was sponsored by Sobi. Medical writing support, funded by Sobi, was provided by Hayley Fowkes and Andy Lockley, Bioscript Group, Macclesfield, UK. Publication coordination support was provided by Nick Fulcher, PhD, CMPP, of Sobi. Sobi and Sanofi reviewed and provided feedback on the poster; however, the authors remained responsible for the final content.
CH has received research funding from CSL Behring and Sobi, as well as honoraria and speakers' bureau fees from LFB, CAF-DCF, CSL Behring, Hoffmann-La Roche, Novo Nordisk, Octapharma, Pfizer, Sobi, Sanofi, Regeneron and Biomarin. CEE has provided consultancy (including expert testimony) for Bayer Healthcare, Biomarin, CSL Behring, LFB, Novo Nordisk, Octapharma, Roche/Chugai, Sobi and Takeda, declares membership on an entity's Board of Directors or advisory committees for Biomarin, LFB, Novo Nordisk, Octapharma, Roche/Chugai, Sobi, Takeda, and speakers' bureau for Octapharma and Sobi, as well as receiving research funding from Biotest and Octapharma, honoraria from Bayer Healthcare, Biomarin, Biotest, CSL Behring, Kedrion, LFB, Novo Nordisk, Octapharma, Pfizer, Roche/Chugai, Sanofi, Sobi and Takeda, and travel funding from Bayer Healthcare, Biomarin, Biotest, CSL Behring, LFB, Novo Nordisk, Octapharma, Roche/Chugai, Sanofi, Sobi and Takeda. ML has received grant/research/speaker fees from Takeda, Pfizer, Roche-Chugai, Sobi, AstraZeneca and BioMarin, and has acted as a Consultant for AstraZeneca and Biomarin. DM has received research grants paid directly to the Institution (McMaster University) from Bayer, Pfizer, Novo Nordisk, Sanofi, Octapharma and Roche, as well as personal fees/honoraria from Sanofi, Sobi, Novo Nordisk, Bayer, Pfizer, Octapharma and Roche for participation in advisory boards, lectures and preparation of educational material. NO has received research support from or acted as Principal Investigator for CSL Behring, F. Hoffmann-La Roche, Novo Nordisk, Sanofi and Sobi; has acted as a consultant for AstraZeneca, F. Hoffmann-La Roche Ltd, uniQure, Sobi and CSL Behring; has received speaker fees from CSL Behring, Bayer, F. Hoffmann-La Roche, Novo Nordisk, Sobi and Takeda. All fees are donated to an institutional charitable body that supports education in haemostasis and thrombosis. RFS Jr has received grants from Takeda, Octapharma and Hema Biologics/LFB, as well as provided consultancy for Takeda, Octapharma, Hema Biologics/LFB, Pfizer, Sanofi/Sobi, Novo Nordisk, Guardian Therapeutics, Vega, HEMAb, Genentech/Roche and Bayer. RL has during the last 3 years received research grant funding from Sobi, and consultancy or speaker's fees from Idogen AB, Sobi, Novo Nordisk, Takeda, Sanofi and Bayer. None of these are related to the current poster.

CONTACT INFORMATION

Correspondence: Cédric Hermans

Email: cedric.hermans@uclouvain.be

Affiliation: Division of Haematology, Saint-Luc University Hospital, Université catholique de Louvain, Brussels, Belgium