Association between FIX levels & bleeding rates in hemophilia B patients receiving rFIXFc or N9-GP

Poster Number:
PB0788

Alfonso Iorio[1], Emma Iserman[1], Alix Arnaud[2], Kalyani Hawaldar[2], Daisy Ng-Mak[2], Quazi Ibrahim[1], Alejandro Fernandez[3], Graham Neill[4], Arun Keepanasseril[1]

[1]McMaster University, Hamilton, Ontario, Canada; [2]Sanofi, Cambridge, MA, USA; [3]Sanofi, Zurich, Switzerland; [4]Sanofi, Reading, UK

Background

- Congenital hemophilia B is a rare bleeding caused by dysfunctional or absent blood clotting factor IX (FIX); it affects approximately 1 in 50,000 people in Canada, predominantly men.[1,2]
- Prophylactic infusion of FIX is essential for managing moderate-to-severe hemophilia B. Extended half-life (EHL) FIX concentrates, such as recombinant factor IX Fc fusion protein (rFIXFc) or nonacog beta pegol (N9-GP), have exhibited safety and efficacy in clinical trials and real-world settings.[3,4,5]
- Differences in the EHL pharmacokinetic (PK)/pharmacodynamic properties suggest that plasma FIX levels may not completely explain hemostatic control levels. rFIXFc, with a greater volume of distribution than N9-GP, transiently distributes into the extravascular space, whereas N9-GP remains largely confined to the intravascular compartment.[6,7,8]

Objective

To explore the association between plasma FIX levels and bleeding rates in people with hemophilia B (PWHB) treated with rFIXFc or N9-GP.

Methods

Study design

- This study was a retrospective analysis of real-world patient data obtained from the Canadian Bleeding Disorders Registry (CBDR) and the Web-Accessible Pharmacokinetic Service-Hemophilia Service (WAPPS-Hemo) platform (**Figure 1**).

Figure 1. Study design

Study Population

Inclusion Criteria
- Patients of all ages who received prophylactic treatment with either rFIXFc (2016–2018) or N9-GP (2018 onward) for ≥3 months
- Available WAPPS-Hemo PK data

Cohort Overlap
- Treatment groups were not mutually exclusive, as some patients switched from rFIXFc to N9-GP
- Correlations due to repeated observations from a patient within and across products were considered using patient specific random intercepts in the regression
- Natural heterogeneity among patients was used to explain intra-patient correlations

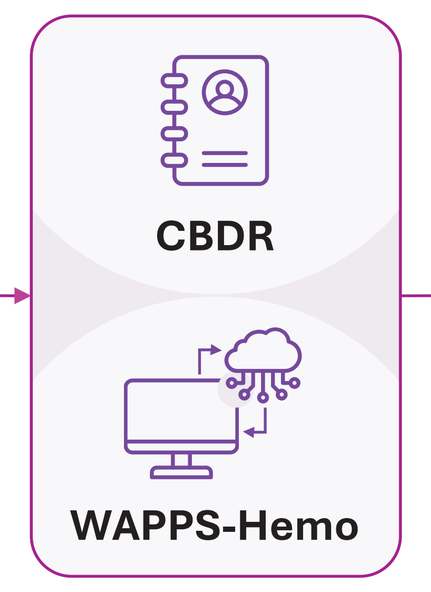
CBDR
WAPPS-Hemo

Statistical Analysis

Modelling Approaches
- Bayesian predictive analysis and population PK models predicted plasma FIX levels at the time of reported bleeds

Bleeder/Non-Bleeder* Classification
- Based on spontaneous bleeds (bleeders)

Shared Frailty Gamma Model
- Adjust for each patient susceptibility to the risk of bleeding that cannot be explained by the observed covariates

Discriminatory Threshold Analysis**
- Sensitivity (Se)
- 1-Specficity (1-Sp)

CBDR, Canadian Bleeding Disorders Registry; FIX, factor IX; N9-GP, nonacog beta pegol; PK, pharmacokinetic; rFIXFc, recombinant FIX fusion protein; WAPPS-Hemo, Web-Accessible Pharmacokinetic Service-Hemophilia Service platform.
*Non-bleeders are individuals who either carry the hemophilia gene without showing symptoms or have hemophilia but experience very few or no bleeding episodes due to mild disease or effective treatment.
**Discriminatory threshold analysis was performed using the highest predicted FIX level at the time of spontaneous bleeds (for bleeders) and the lowest observed trough level during prophylaxis (for non-bleeders).

Results

- Among the 210 PWHB, 72 were treated with rFIXFc and 67 with N9-GP; of these, population PK data were available for 33 and 34 patients, respectively.
- rFIXFc and N9-GP were administered once weekly at median doses of 60 and 41 IU/kg, respectively.
- Predicted FIX plasma levels at the time of spontaneous bleeding episodes for both bleeding and non-bleeding patients were higher for N9-GP than for rFIXFc (**Table 1** and **Table 2**).
- The percentage of spontaneous bleeding event is similar in both groups despite different predicted PK profile.

Table 1. Patient characteristics and bleeding summary among PWHB treated with rFIXFc or N9-GP

Parameter	rFIXFc	N9-GP
Overall characteristics		
No. of PWHB	33	34
Age (years), median (range)	17 (1–88)	42 (0.6–73)
Male, n (%)	33 (100)	34 (100)
BMI (kg/m^2), mean ± SD	24 ± 6	27 ± 5
Hemophilia severity*, n (%)		
Mild	1 (3)	0 (0)
Moderate	5 (15)	13 (38)
Severe	27 (82)	21 (62)
Bleed type summary		
Bleeder, n	24	25
Experienced spontaneous bleed while on treatment	14	18
Experienced traumatic bleed while on treatment	9	5
Spontaneous bleed only while receiving the other product	1	2
Non-bleeder, n	9	9

*Severe (FIX <1 IU/dL), moderate (FIX 1–5 IU/dL).
BMI, body mass index; FIX, factor IX; n, number of patients; N9-GP, nonacog beta pegol; PWHB, people with hemophilia B; rFIXFc, recombinant FIX fusion protein; SD, standard deviation.

Table 2. Predicted FIX at the time of a spontaneous bleed or trough at each infusion when there was no bleed among PWHB

Parameter	Spontaneous bleeders		Non-bleeders	
	rFIXFc	N9-GP	rFIXFc	N9-GP
No. of patients	14	18	9	9
No. of spontaneous bleeds	77	90	0	0
	For bleed treatments		When there was no bleed	
No. of infusions	77	90	709	534
	Predicted FIX (IU/dl) level at the time of spontaneous bleed		Through (IU/dl) at each infusion when there was no bleed	
Mean ± SD	16 ± 14	45 ± 26	16 ± 22	20 ± 17
Median (IQR)	13 (7, 20)	38 (26, 58)	7 (3, 21)	14 (6, 29)
(min, max)	(1, 67)	(1, 132)	(1, 115)	(1, 96)

FIX, factor IX; IQR, interquartile range; N9-GP, nonacog beta pegol; PWHB, people with hemophilia B; rFIXFc, recombinant FIX fusion protein; SD, standard deviation.

- The association between FIX levels (per quartile) and bleeding episodes significantly varied between the two products (p=0.008 for interaction effect) (**Table 3**).
- For rFIXFc, spontaneous bleeding risk was lower in the 2nd and 3rd quartiles and higher in the 4th quartile than in the 1st quartile, indicated by a hazard ratio of <1. Nonetheless, these estimates were imprecise and statistically non-significant (**Table 3**).
- For N9-GP, spontaneous bleeding risk was higher in the 2nd, 3rd, and 4th quartiles than in the 1st quartile, indicated by a hazard ratio of >1. However, these estimates were highly imprecise and statistically non-significant (**Table 3**).

Table 3. Associations of FIX levels with spontaneous bleeding events after fitting an age-adjusted shared frailty model: rFIXFc versus N9-GP

Outcome: recurrent bleeding	rFIXFc						N9-GP						*p*-value for interaction between FIX levels and product type
	No. of bleeds	No. of patients with a bleed	Total no. of patients	Person-days	HR (95% CI)	*p*-value	No. of bleeds	No. of patients with a bleed	Total no. of patients	Person-days	HR (95% CI)	*p*-value	
FIX (IU/dL) categories													0.008
First quartile (FIX level ≤8.6)	22	7	11	4,521	1		2	2	6	3,456	1		
Second quartile (8.6< FIX level ≤20.1)	35	10	12	7,396	0.68 (0.32–1.42)	0.31	10	7	9	3,913	11.73 (0.80–171.38)	0.072	0.045
Third quartile (20.1 <FIX level ≤34.7)	12	7	8	2,873	0.56 (0.21–1.47)	0.24	26	11	13	7,583	9.43 (0.70–127.63)	0.091	0.048
Fourth quartile (FIX level >34.7)	8	3	4	620	3.34 (0.99–11.30)	0.052	52	15	16	13,504	7.33 (0.55–97.35)	0.131	0.59

CI, confidence interval; FIX, factor IX; HR, hazard ratio; N9-GP, nonacog beta pegol; rFIXFc, recombinant FIX fusion protein.

Discriminatory threshold analysis

- For rFIXFc, the trough level of all non-bleeders was ≤25 IU/dL, and approximately 50% of bleeders bled at FIX levels ≤20 IU/dL (**Table 4**).
- For N9-GP, the trough levels for all non-bleeders was ≤15 IU/dL, whereas 50% of bleeders bled at FIX levels ≤60 IU/dL (**Table 4**).

Table 4. Discriminatory ability of predicted FIX level (highest for bleeders and lowest trough for non-bleeders) in predicting spontaneous bleeds (yes or no) among patients who received rFIXFc versus N9-GP

FIX level (IU/dL)	rFIXFc*		N9-GP**	
	Se among bleeders	Sp among non-bleeders	Se among bleeders	Sp among non-bleeders
No. of patients	14	9	18	9
Decile cut points				
≤3.64	0/14 (0.00)	6/9 (66.67)	0/18 (0.00)	6/9 (66.67)
≤6.87				
≤9.94	1/14 (7.14)	6/9 (66.67)	0/18 (0.00)	8/9 (88.89)
≤15.16	4/14 (28.57)	8/9 (88.89)	0/18 (0.00)	9/9 (100.00)
≤20.11	6/14 (42.86)	8/9 (88.89)	1/18 (5.56)	9/9 (100.00)
≤25.27	9/14 (64.29)	9/9 (100.00)	2/18 (11.11)	9/9 (100.00)
≤30.84	10/14 (71.43)	9/9 (100.00)	3/18 (16.67)	9/9 (100.00)
≤40.95	12/14 (85.71)	9/9 (100.00)	6/18 (33.33)	9/9 (100.00)
≤58.42	13/14 (92.86)	9/9 (100.00)	9/18 (50.00)	9/9 (100.00)
Additional cut points				
≤70	-	-	9/18 (50.00)	9/9 (100.00)
≤80	-	-	12/18 (66.67)	9/9 (100.00)
≤95	-	-	15/18 (83.33)	9/9 (100.00)
≤100	-	-	16/18 (88.89)	9/9 (100.00)

N9-GP, nonacog beta pegol; rFIXFc, recombinant FIX fusion protein; Se, sensitivity; Sp, 1-Specificity. *There was no predicted factor level between 6.87 and 9.94 IU/dL.
**There was no predicted factor level between 6.87 and 9.94 IU/dL.

- A similar discriminatory pattern was observed for patients with only severe hemophilia B. Non-bleeders treated with rFIXFc or N9-GP exhibited trough levels of ≤25 IU/dL and ≤15 IU/dL, respectively. Approximately 50% of the bleeders experienced bleeds at FIX level of ≤20 IU/dL for rFIXFc and ≤60 IU/dL for N9-GP.
- **Table 5** presents the median predicted FIX levels for the 1st to subsequent spontaneous bleeds for patients who received rFIXFc or N9-GP, stratified by the severity of hemophilia B.
 - For rFIXFc, FIX levels at repeated bleeding events remained low and comparable across both severe and moderate cases.
 - N9-GP patients had considerably higher predicted FIX levels during the bleeding events, with median levels often in the normal to near-normal range for both severities.

Table 5. Predicted FIX level (IU/dL) at the 1st spontaneous bleed and up to the 9th spontaneous bleed among patients with hemophilia B

	rFIXFc			N9-GP		
	Overall	Severe	Moderate	Overall	Severe	Moderate
Total no. of bleeds	77	40	37	90	60	30
Median predicted FIX level (IU/dL) at						
1st bleed	13 (4, 20), 14	13 (4, 20), 10	8 (1, 21), 4	33 (18, 43), 18	28 (17, 37), 13	40 (40, 43), 5
2nd bleed	17 (9, 24), 13	16 (6, 24), 9	19 (13, 39), 4	38 (23, 66), 16	27 (19, 56), 11	44 (44, 75), 5
3rd bleed	11 (8, 16), 9	10 (8, 16), 6	11 (4, 34), 3	35 (23, 66), 12	32 (22, 81), 8	38 (29, 46), 4
4th bleed	9 (3, 18), 8	9 (5, 17), 5	9 (1, 33), 3	49 (41, 75), 10	65 (39, 78), 7	48 (47, 49), 3
5th bleed	23 (9, 30), 7	16 (10, 26), 4	26 (1, 67), 3	42 (26, 58), 6	53 (37, 59), 4	26 (15, 37), 2
6th bleed	7 (6, 10), 5	7 (6, 7), 2	10 (1, 54), 3	56 (35, 59), 5	59 (35, 71), 3	36 (16, 56), 2
7th bleed	10 (7, 13), 5	10 (7, 13), 2	10 (1, 39), 3	50 (39, 51), 5	39 (25, 51), 3	54 (50, 58), 2
8th bleed	11 (10, 14), 4	14 (11, 17), 2	10 (8, 11), 2	33 (31, 58), 4	32 (29, 34), 2	57 (32, 83), 2
9th bleed	9 (4, 13), 2	-	9 (4, 13), 2	47 (33, 65), 4	33 (30, 36), 2	65 (58, 72), 2

FIX, factor IX; N9-GP, nonacog beta pegol; rFIXFc, recombinant FIX fusion protein.
Data are presented as median (25th, 75th percentiles); n, number of patients.

Strengths and limitations

- A key strength of this study was the availability of plasma FIX activity data, enabling more precise evaluation.
- The study relies on existing medical records, which may not represent the entire population of PWHB.
- The findings may not be generalizable to all PWHB, particularly those outside Canada or on different treatment regimens.

Conclusion

- **In hemophilia B, high plasma levels for different products do not necessarily indicate improved bleed protection, similarly, low plasma FIX levels may not necessarily indicate reduced protection.**
- **For patients experiencing breakthrough bleeds at high plasma FIX levels, switching to an alternative concentrate with a different PK profile may be more effective than increased dosing.**

Funding
The study was funded by Sanofi.

References
1. Iorio A, et al. *Annals of Internal Medicine*. 2019;171 (8): 540–46.
2. Castaman G and Matino D. *Haematologica*. 2019;104 (9):1702.
3. Pasi KJ. *Thrombosis and haemostasis*. 2017;117 (3): 508–18.
4. Carcao M. *Thrombosis and haemostasis.* 2020;120 (5): 737–46.
5. Shapiro A. *Hemophilia*. 2020;26 (6): 975–83.
6. Stafford DW. *Thrombosis Journal.* 2016,14(1):35.
7. Cloesmeijer ME, et al. *CPT Pharmacometrics Syst Pharmacol*. 2024;00:1–11.
8. Mann DM, et al. *Haemophilia*. 2021;00:1–8.

Acknowledgement
Medical editorial assistance was provided by Charu Pundir and Deepshikha Pawar (Sanofi). The authors would like to thank Leah Granby (Sanofi) for her contributions to the poster. Sanofi and Sobi personnel reviewed this poster. The authors had full editorial control of the content and provided their final approval.

COI disclosure
AI: employment with McMaster University; received research funding for the institute from Bayer, Novo Nordisk, Sanofi, Roche, and Pfizer; holds patents and receives royalties from WAPPS-Hemo; and serves as a member of Florio's medical advisory board and WFH advisory board. EI: received consulting fees from Sanofi. QI: employment with McMaster University. AF: an employee of Sanofi at the time of this study and may hold shares and/or stock options in the company. AK: employment with McMaster University; received research funding for the institute from Novo Nordisk and Roche.
AA, KH, DNM, and GN: employees of Sanofi and may hold stocks and/or stock options in the company.

Poster presented at 33rd Congress of the International Society on Thrombosis and Haemostasis (ISTH) 2025, June 21–25, 2025, Washington, D.C., U.S.