SA-P00679

Pharmacokinetic, Pharmacodynamic, and Exposure–Response Analysis of Pegcetacoplan in C3G and Primary IC-MPGN

Keshavarz A,[1] Khankin E,[1] Melnick J,[1] Wachtel D,[1] Voruganti SK[1]

[1]Apellis Inc, a subsidary of Biogen, Cambridge, MA, USA

Scan QR code for a copy of this poster

BACKGROUND

- C3G and primary IC-MPGN comprise dysregulated C3 activation and progressive renal injury.[1,2]
- Pegcetacoplan, a C3/C3b inhibitor,[3–9] showed clinical benefit,[10,11] but PK, PD, and E-R relationships are not well defined.

OBJECTIVE

- To define PK, PD, and E-R relationships of pegcetacoplan for pediatric and adult patients with C3G and primary IC-MPGN.

METHODS

- Data from two Phase 2 and one Phase 3 randomized studies in adolescents and adults with C3G and primary IC-MPGN were analyzed.
- Population PK and pegcetacoplan/C3 target-mediated drug disposition models (data from 14 clinical studies) and E-R models (data from 3 nephrology studies) were developed to assess relationships among exposure, complement biomarkers, UPCR, and safety.

RESULTS

- Pegcetacoplan reached steady-state exposure at weeks 4–8 with consistent concentrations across subgroups.
- Adolescent weight-adjusted regimens achieved exposures comparable to adults and produced similar clinically meaningful UPCR reductions (**Figure 1**).

Figure 1. Simulated Steady-State Pegcetacoplan Exposure by Body Weight

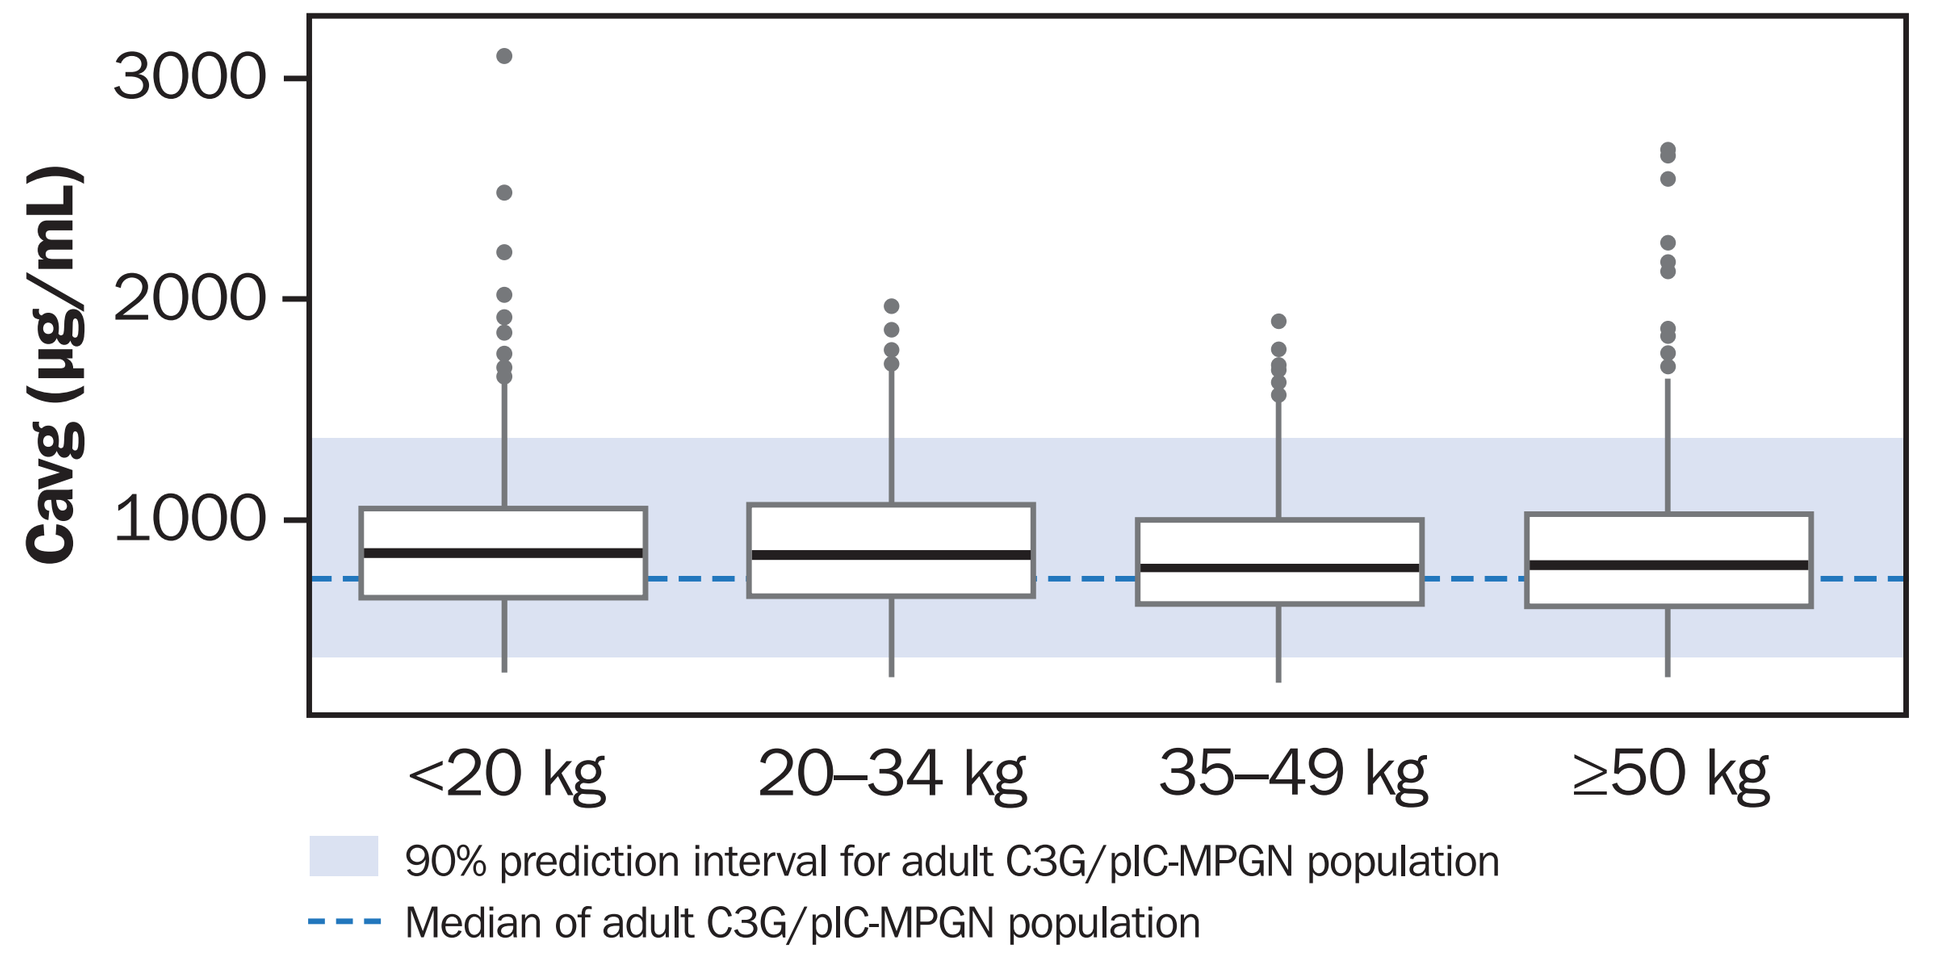

Cavg (µg/mL)
3000
2000
1000
<20 kg
20–34 kg
35–49 kg
≥50 kg
90% prediction interval for adult C3G/pIC-MPGN population
Median of adult C3G/pIC-MPGN population

Weight Group	Pegcetacoplan Dosing
<20 kg	324 mg (6 mL) twice weekly x 4 doses, then 486 mg (9 mL) twice weekly
20 to <35 kg	540 mg (10 mL) twice weekly x 2 doses, then 648 mg (12 mL) twice weekly
35 to <50 kg	648 mg (12 mL) x 1 dose, then 810 mg (15 mL) twice weekly
≥50 kg	1080 mg (20 mL) twice weekly

Simulated steady-state pegcetacoplan exposure in adolescent patients by body weight and dosing regimen compared with adult C3G/pIC-MPGN fixed-dose reference exposure following pegcetacoplan 1080 mg SC twice weekly treatment (blue dashed line: adult median; shaded region: 90% prediction interval for adults receiving fixed-dose pegcetacoplan). Pegcetacoplan is approved for adult and adolescent patients aged 12 years and older with C3G or pIC-MPGN.

- Pegcetacoplan led to rapid and sustained complement inhibition within 4 weeks, with >95% free C3 suppression across the dosing interval, indicating maximal target engagement. Free C3 returned to baseline within 4 weeks of discontinuation (**Figure 2**).

Figure 2. Mean Percent Change in Free C3 and Total C3 With Pegcetcoplan 1080 mg Twice Weekly Treatment in Patients With C3G

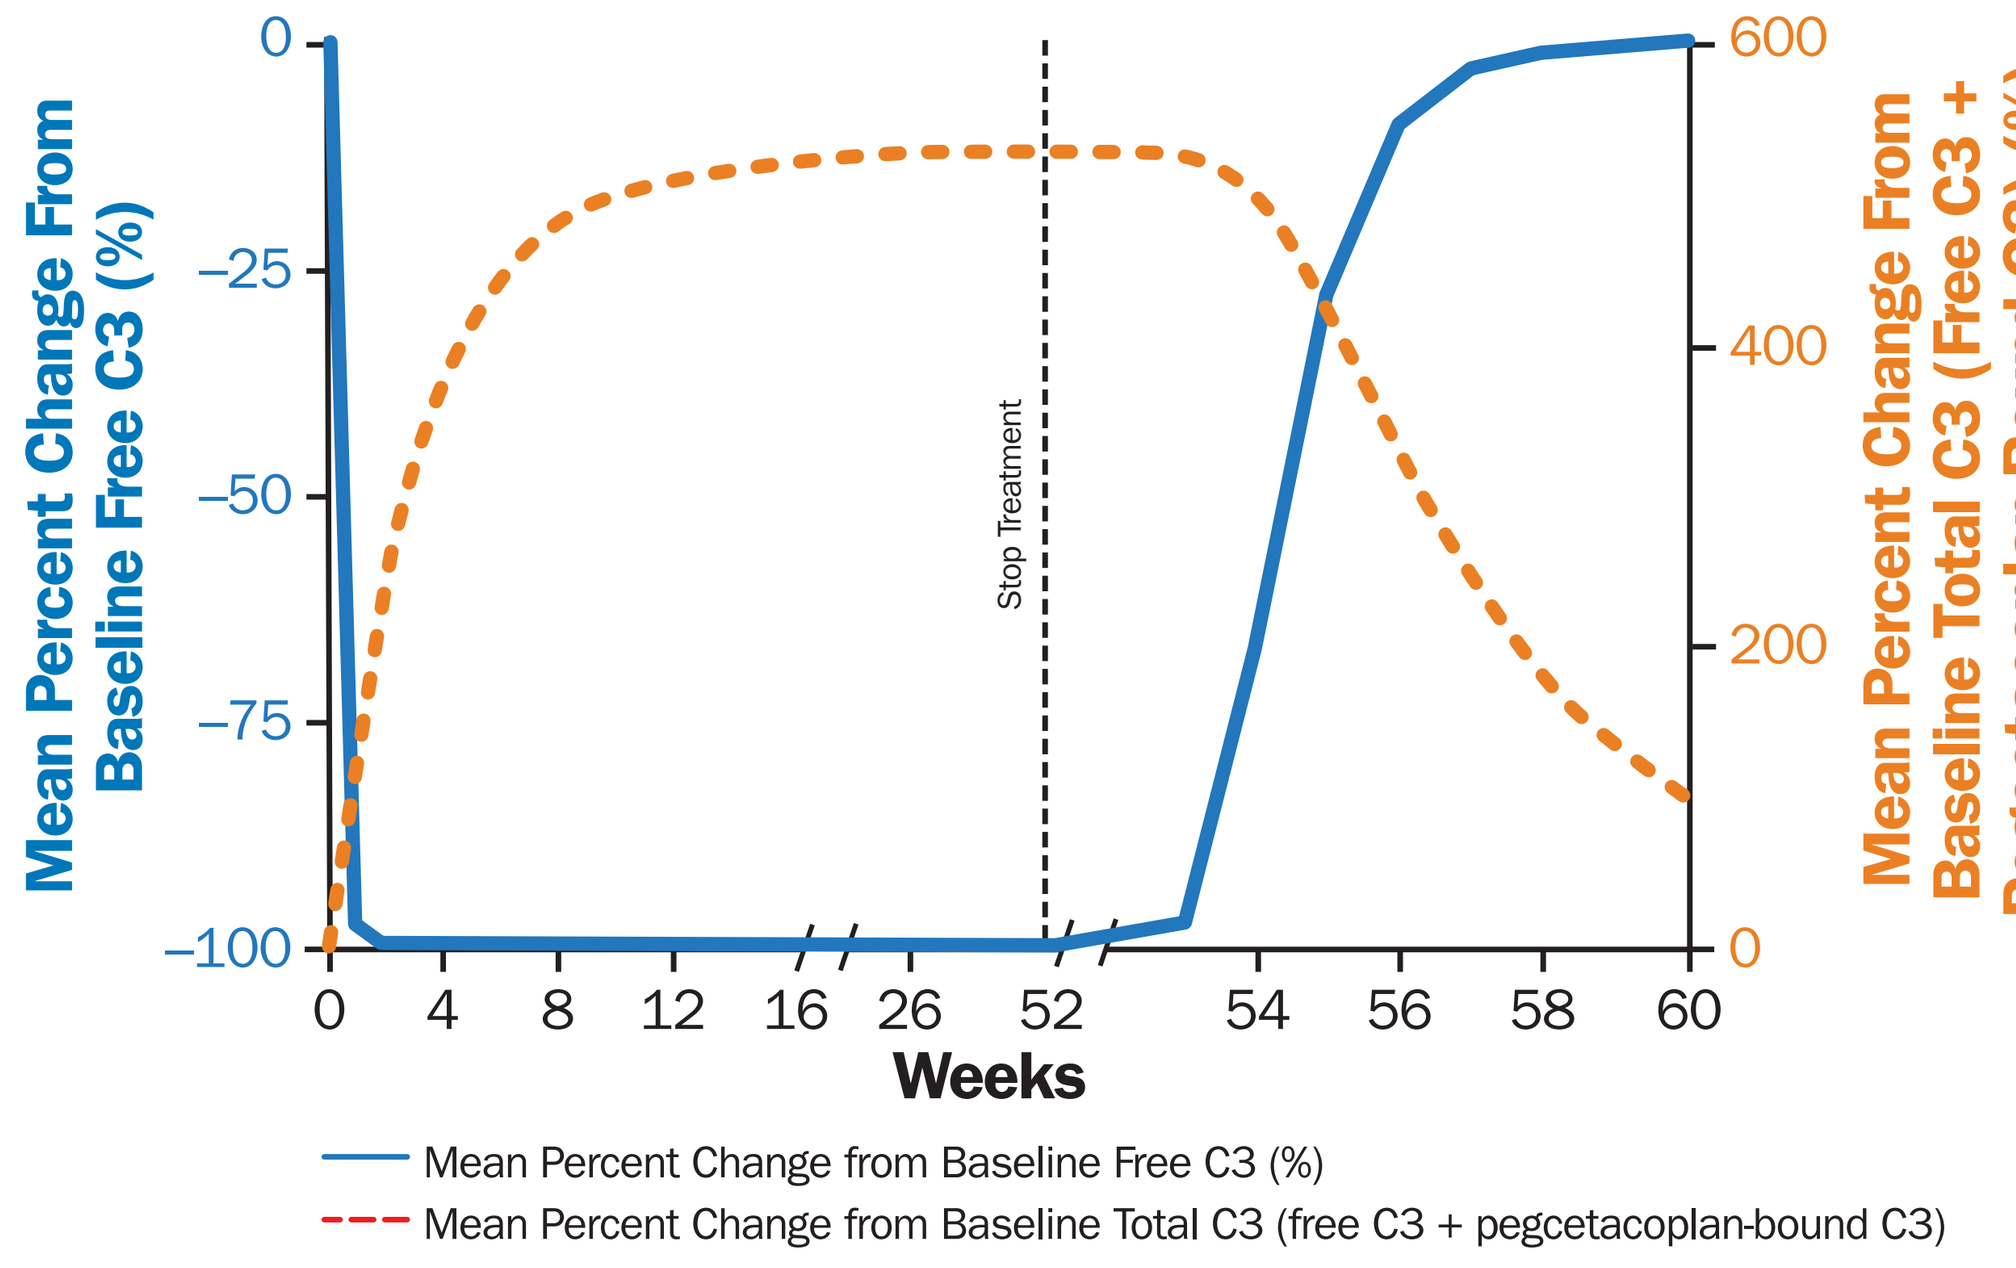

Mean Percent Change From Baseline Free C3 (%)
Mean Percent Change From Baseline Total C3 (Free C3 + Pegcetacoplan-Bound C3) (%)
Stop Treatment
Weeks
0 4 8 12 16 26 52 54 56 58 60
Mean Percent Change from Baseline Free C3 (%)
Mean Percent Change from Baseline Total C3 (free C3 + pegcetacoplan-bound C3)

Mean percent change from baseline in free C3 and total C3 (free C3 plus pegcetacoplan-bound C3) following 52 weeks of pegcetacoplan 1080 mg twice weekly treatment in patients with C3G (last dose on day 364).

- Patient subgroups achieved consistent UPCR reductions. No intrinsic or extrinsic factors, including baseline eGFR or UPCR, had a clinically meaningful impact on exposure or response (**Figures 3** and **4**).
- Immunogenicity had no impact on PK, PD, efficacy, or safety.
- Pegcetacoplan was not associated with adverse safety outcomes within observed exposure and treatment-duration ranges.

Figure 3. Observed Steady-State Pegcetacoplan Concentrations Stratified by (A) Baseline eGFR and (B) Baseline UPCR

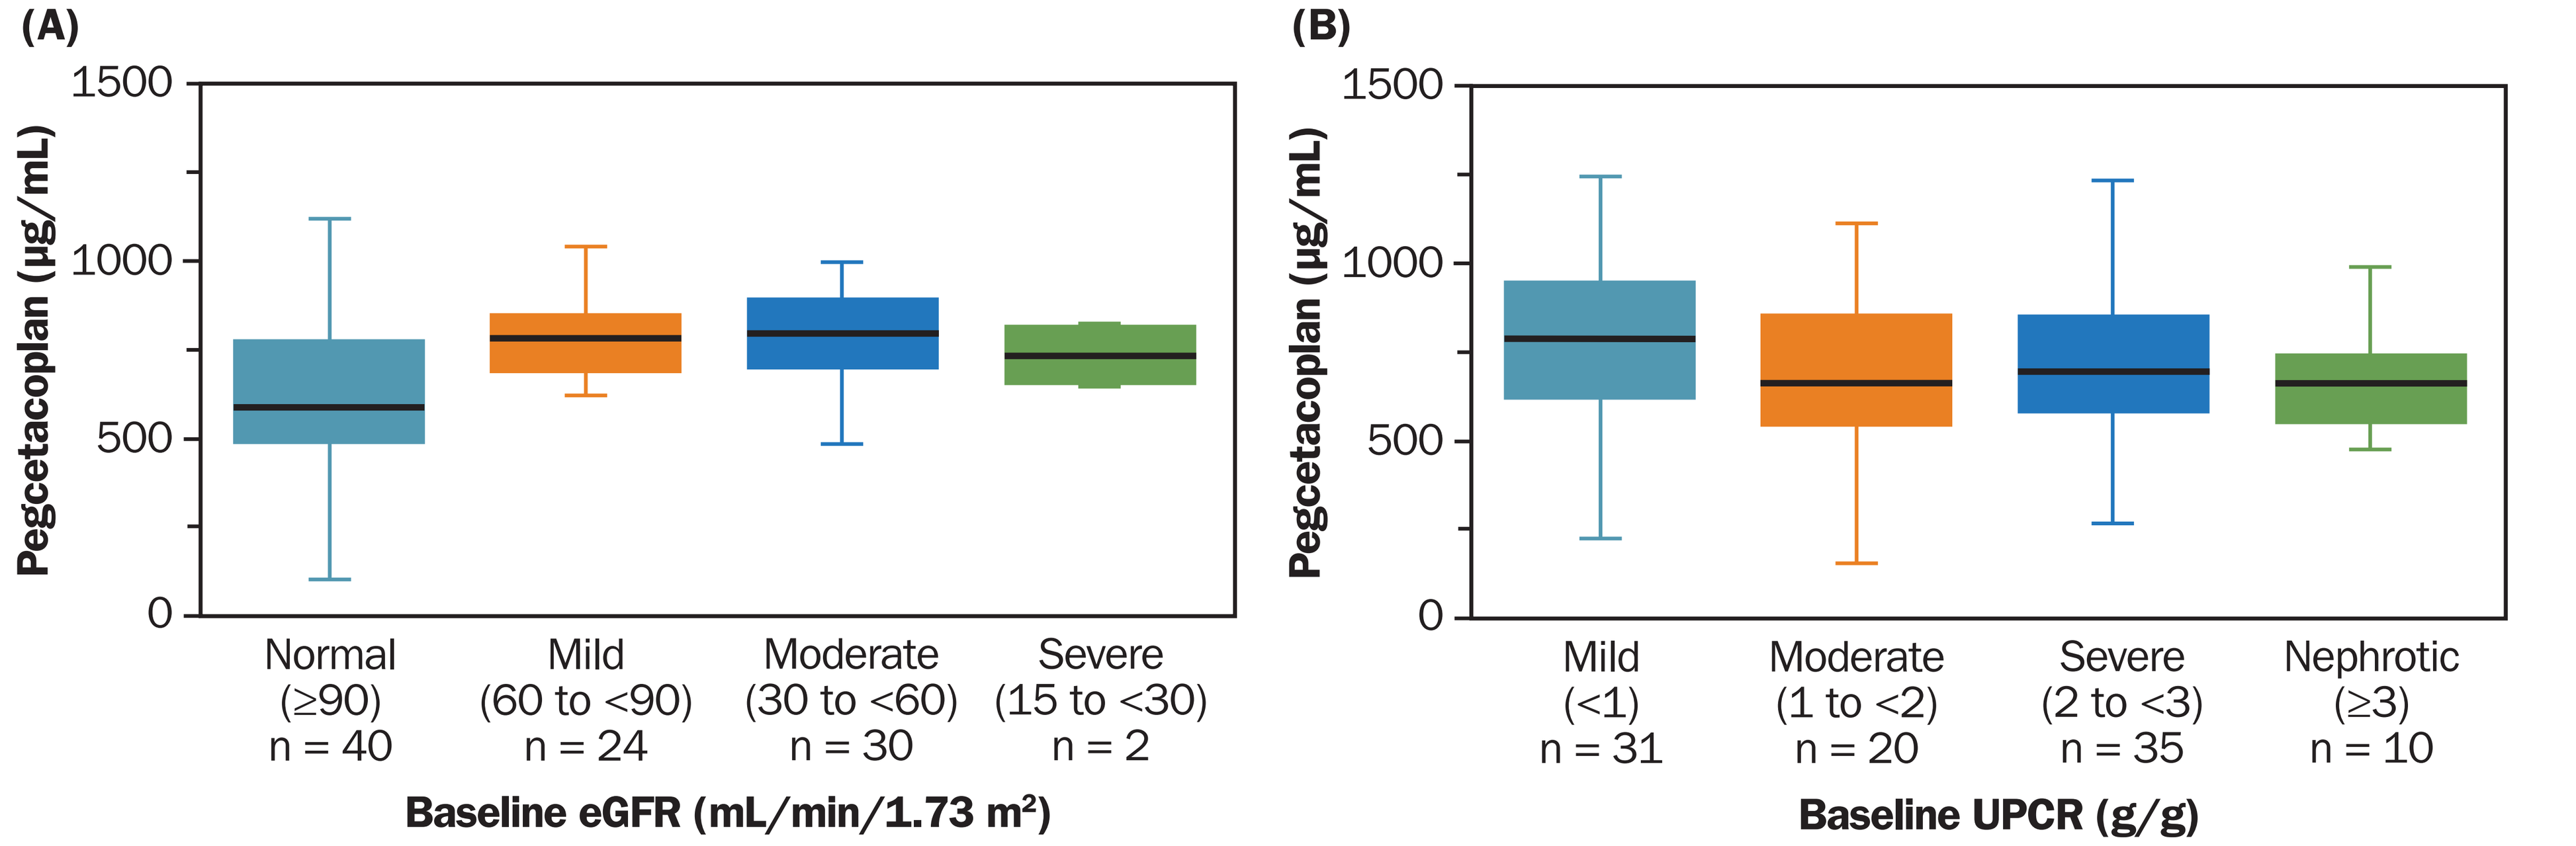

(A)
Pegcetacoplan (µg/mL)
Normal (≥90) n = 40
Mild (60 to <90) n = 24
Moderate (30 to <60) n = 30
Severe (15 to <30) n = 2
Baseline eGFR (mL/min/1.73 m²)
(B)
Pegcetacoplan (µg/mL)
Mild (<1) n = 31
Moderate (1 to <2) n = 20
Severe (2 to <3) n = 35
Nephrotic (≥3) n = 10
Baseline UPCR (g/g)

Observed steady-state pegcetacoplan concentrations (week 26 from the first dose) stratified by baseline eGFR and UPCR in patients with C3G/pIC-MPGN receiving pegcetacoplan 1080 mg SC twice weekly dosing (or weight-adjusted pediatric dosing) in a Phase 3 study.

Note: Lower and upper boundaries of the box represent the 1st quartile (Q1) and 3rd quartile (Q3), respectively. Median is shown as the horizontal line inside the box. Whiskers represent the minimum and maximum values that are within 1.5 × IQR below Q1 or above Q3.

Figure 4. Predicted UPCR Change From Baseline Stratified by (A) Baseline eGFR and (B) Baseline UPCR

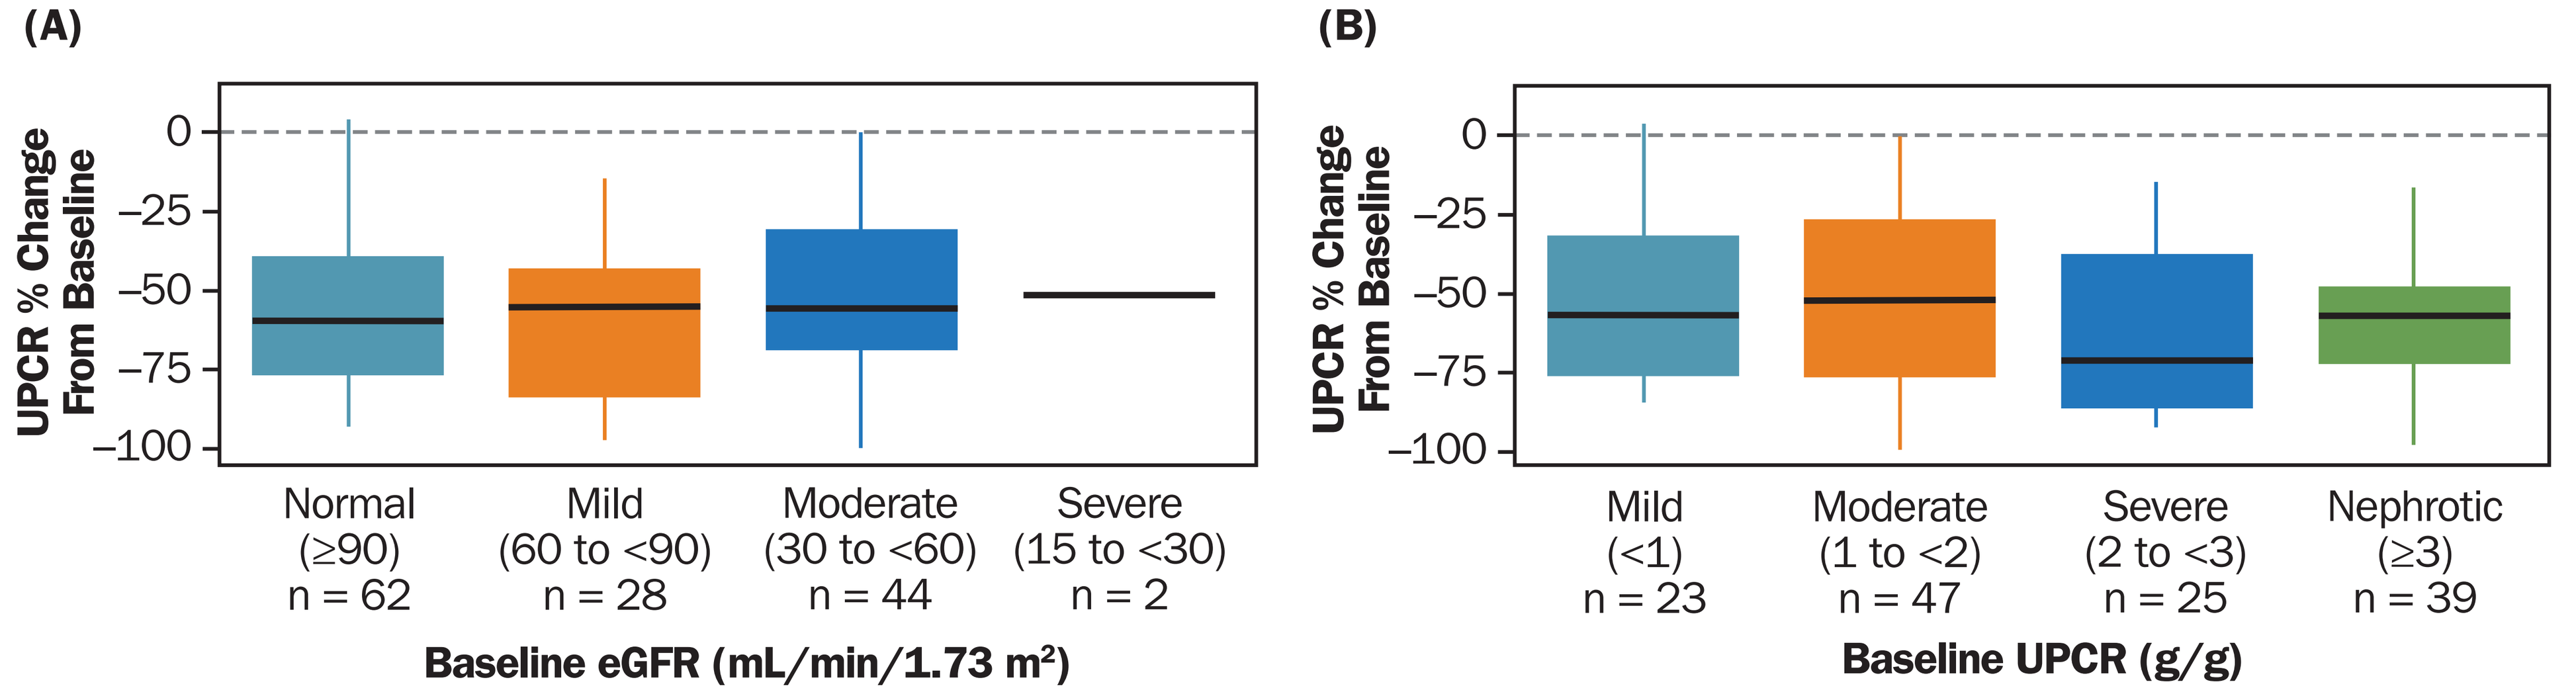

(A)
UPCR % Change From Baseline
Normal (≥90) n = 62
Mild (60 to <90) n = 28
Moderate (30 to <60) n = 44
Severe (15 to <30) n = 2
Baseline eGFR (mL/min/1.73 m²)
(B)
UPCR % Change From Baseline
Mild (<1) n = 23
Moderate (1 to <2) n = 47
Severe (2 to <3) n = 25
Nephrotic (≥3) n = 39
Baseline UPCR (g/g)

Predicted UPCR percent change from baseline stratified by baseline eGFR or UPCR in patients with C3G/pIC-MPGN receiving pegcetacoplan 1080 mg SC twice weekly dosing (or weight-adjusted pediatric dosing).

Note: Lower and upper boundaries of the box represent the 1st quartile (Q1) and 3rd quartile (Q3), respectively. Median is shown as the horizontal line inside the box. Whiskers represent the minimum and maximum values that are within 1.5 × IQR below Q1 or above Q3.

CONCLUSIONS

- At indicated doses for patients ≥12 years, pegcetacoplan provides consistent exposure, rapid, sustained complement inhibition, and clinically meaningful and safe proteinuria reduction.
- This supports a fixed-dose adult regimen of 1080 mg twice weekly and weight-adjusted adolescent regimen without adjustment for renal function or proteinuria.

Acronyms: C3G = C3 glomerulopathy; Cavg = average steady-state concentration; eGFR = estimated glomerular filtration rate; E-R = exposure-response; IQR = interquartile range; pIC-MPGN = primary immune-complex membranoproliferative glomerulonephritis; PD = pharmacodynamic; PK = pharmacokinetic; SC = subcutaneous; UPCR = urine protein-to-creatinine ratio.

References: **1.** Bomback AS, et al. *Kidney Int Rep*. 2024;10:17–28. **2.** Mastrangelo A, et al. *Front Pediatr*. 2020;8:205. **3.** Smith RJH, et al. *Nat Rev Nephrol*. 2019;15:129–143. **4.** Zipfel PF, et al. *Front Immunol*. 2019;10:2166. **5.** Meuleman MS, et al. *Semin Immunol*. 2022;60:101634. **6.** Dixon BP, et al. *Kidney Int Rep*. 2023;8:2284–2293. **7.** Empaveli. Prescribing information. Apellis; 2025. **8.** Aspaveli. Summary of product characteristics. Sobi; 2026. **9.** Lamers C, et al. *Nat Commun*. 2022;13:5519. **10.** Fakhouri F, et al. *N Engl J Med*. 2025;393(22):2210–2220. **11.** Vivarelli M, et al. *Clin J Am Soc Nephrol*. 2026.

Disclosures: **AK**, **EK**, **JM**, **DW**, and **SKV** were employed by Apellis at the time of study conduct and are currently employed by Biogen and hold stock and/or stock options in Biogen.

Acknowledgments: This study was sponsored by Apellis Inc (a subsidiary of Biogen) and Sobi (Swedish Orphan Biovitrum AB).

Writing and editorial support for the preparation of this poster was provided by Jennifer L. Gibson, PharmD, of Kay Square Scientific (Butler, PA, USA): funding was provided by Apellis Inc (a subsidiary of Biogen) and Sobi (Swedish Orphan Biovitrum AB).

The authors thank Priska Kaufmann for her critical support of the study and input on the abstract.

Presented at Annual Meeting of the American Society of Nephrology | October 22–25, 2026 | Denver, CO