Pegcetacoplan population pharmacokinetic and exposure-response in pediatric and adult patients with C3G and primary immune-complex membranoproliferative glomerulonephritis

Ali Keshavarz[1], Priska Kaufmann[2], Brandon Smith[3], Xiaoying Yu[3], Sravan Karumanchi[3], Ryan L. Crass[3]

[1]Apellis Pharmaceuticals, Inc, Waltham, MA, USA; [2]Swedish Orphan Biovitrum AB, Stockholm, Sweden, [3]A2-Ai, Ann Arbor, MI, USA

CONCLUSIONS

- **Pegcetacoplan 1080 mg SC BIW in adults, or corresponding weight-adjusted regimens for adolescents, achieves exposures near the UPCR E–R plateau, with 95% of patients predicted to exceed the concentration for 75% of the maximal response (IC75) without exposure-related safety concerns**

INTRODUCTION

- Pegcetacoplan is a selective C3 & C3b inhibitor that blocks complement dysregulation, the key pathophysiological driver for C3G & primary IC-MPGN, thereby preventing glomerular C3 deposition and activation of downstream effectors (C3 & C5 convertase activity and C5b-9 assembly)[1-7]
- Population PK and E–R analyses for efficacy and safety support pegcetacoplan dosing approved for adolescents and adults with C3G and primary IC-MPGN and identified dosing for pediatric patients based on exposure matching

OBJECTIVES

- **To quantify the serum concentration-time profile of pegcetacoplan in adolescent and adult patients with C3G or primary IC-MPGN using population PK modeling**
- **To assess the relationship between pegcetacoplan exposure and biomarker (UPCR) response using E-R modeling**
- **To evaluate the impact of pegcetacoplan exposure and clinically relevant adverse events**

METHODS

- A PK model was developed using 14 clinical studies (including 3 in C3G or primary IC-MPGN)
- Dose-exposure simulations for pediatric patients (2–<18 years) identified weight-stratified regimens (<50 kg) to match adult exposure of 1080 mg SC BIW
- The PK model was applied to generate empirical Bayesian predictions into E–R models for UPCR and clinically relevant AEs

RESULTS

- Pegcetacoplan PK was described by a one-compartment model with first-order transit absorption (n = 1) and first-order elimination
- Typical PK parameters in a 70-kg patient with a baseline C3 level of 0.7 g/L were: clearance, 0.0123 L/h; volume, 4.49 L; absorption rate constant, 0.0375 1/h; bioavailability, 0.77
- PK parameters were largely consistent across indications, with no eliminating organ impairment
- Pediatric SC dosing regimens match early and steady-state exposure to adults (**Fig A–C**): ≥50 kg, 1080 mg BIW; 35–<50 kg, 648 mg x1 then 810 mg BIW; 20–<35 kg, 540 mg BIW x2 then 648 mg BIW; <20 kg, 324 mg BIW x4 then 486 mg BIW
 - Note that pegcetacoplan is not approved for patients weighing <30 kg

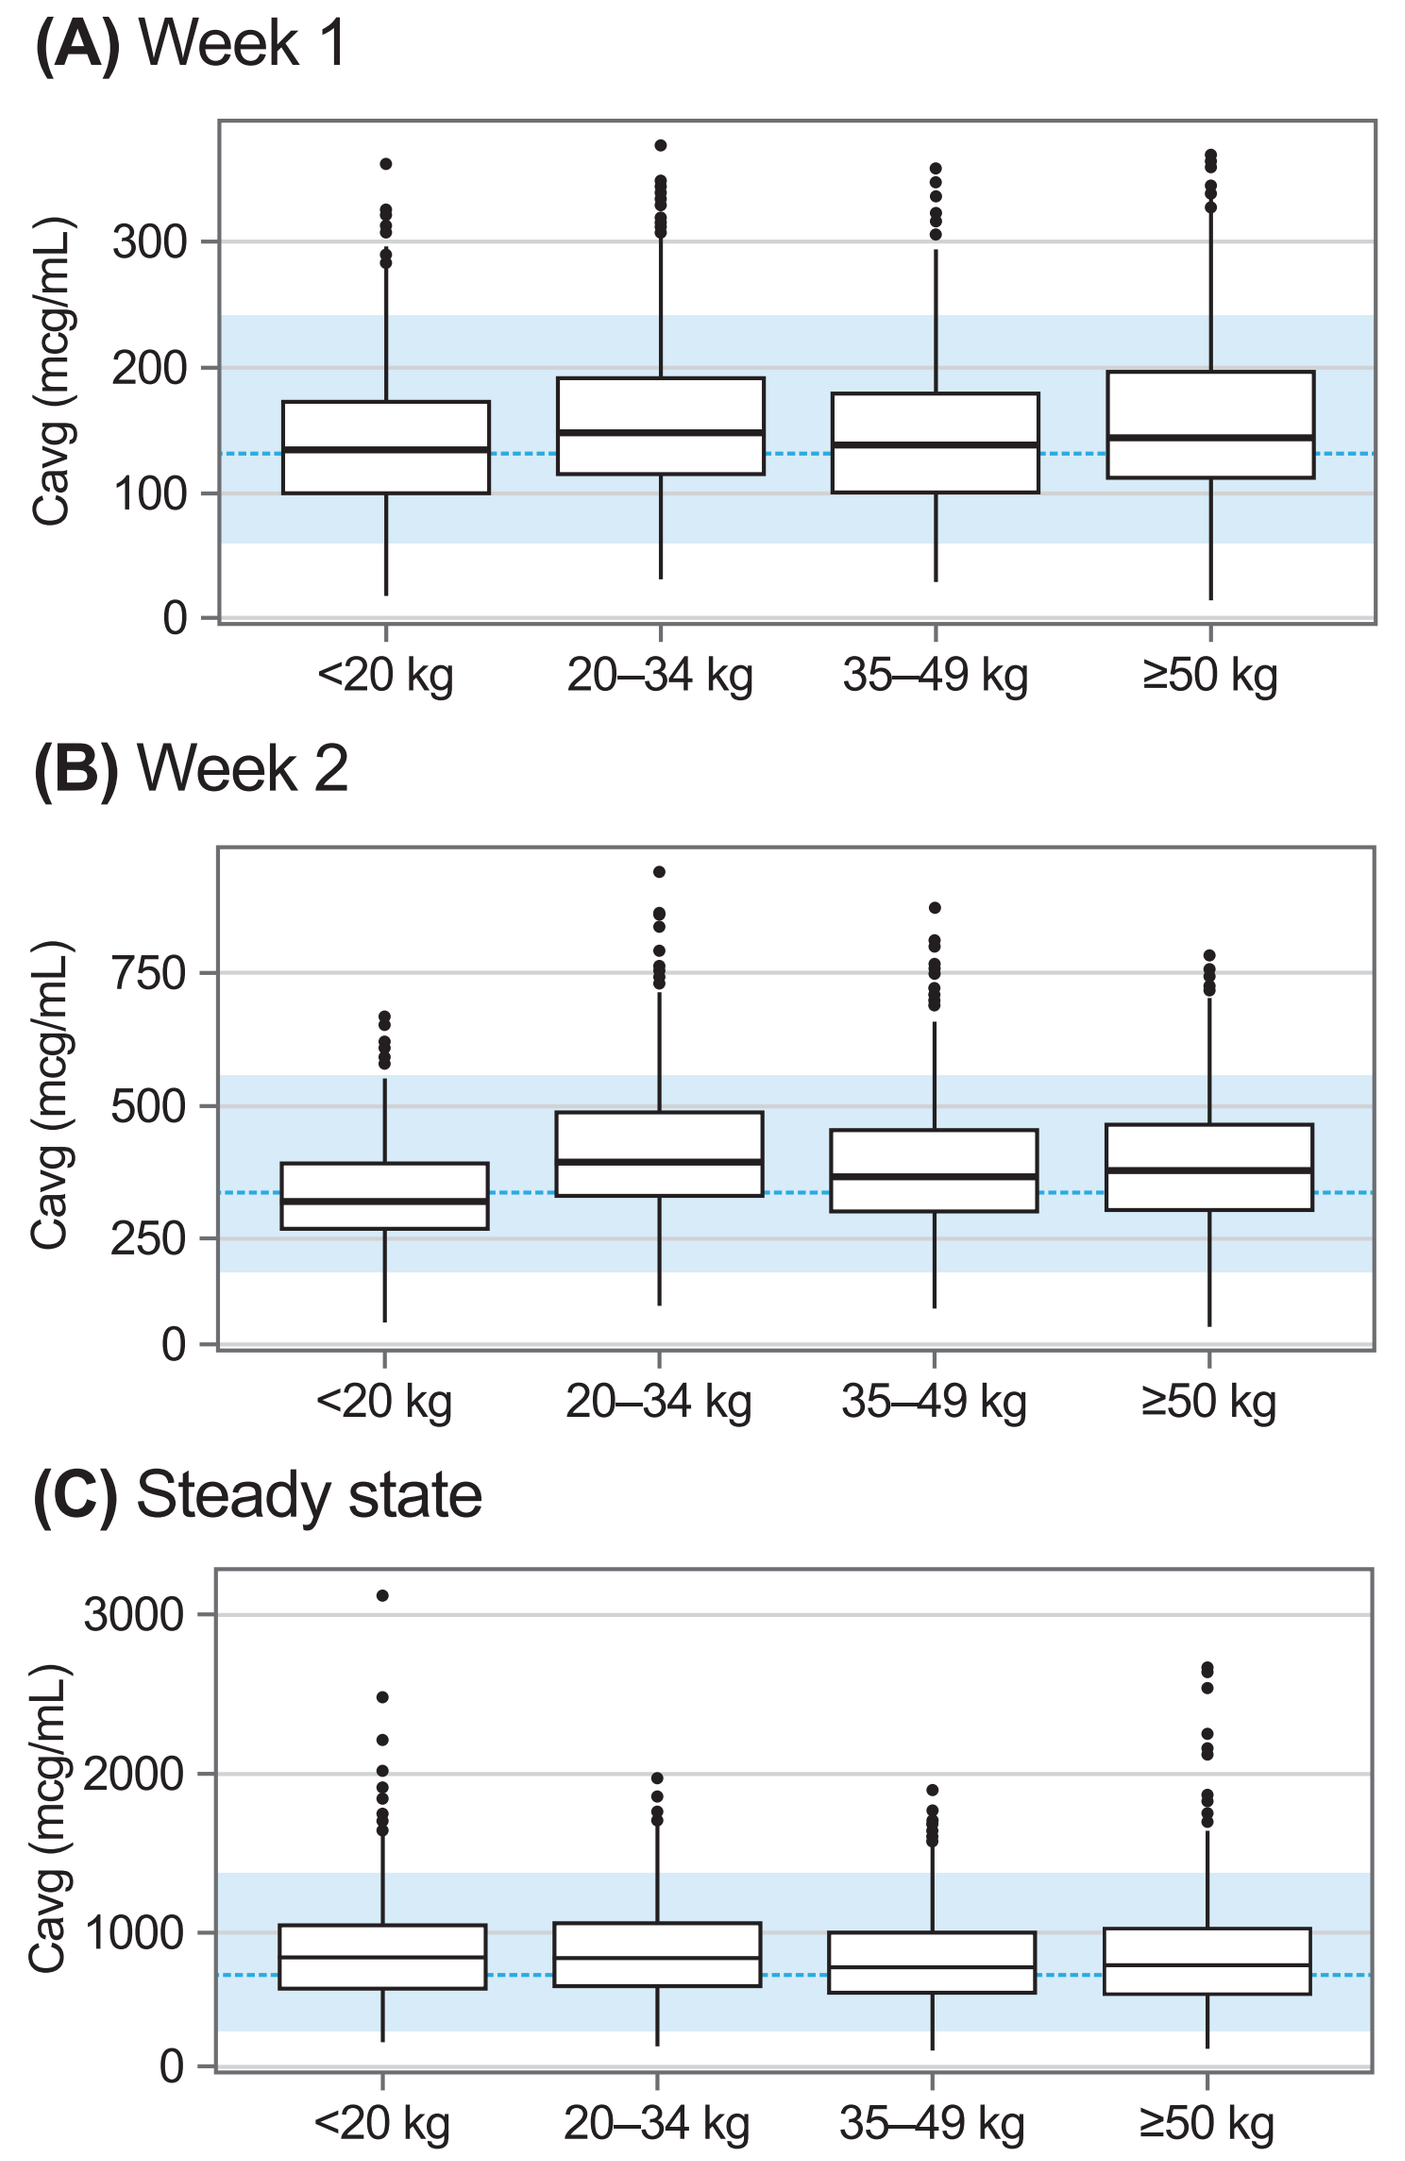
(A) Week 1
(B) Week 2
(C) Steady state
Cavg (mcg/mL)
<20 kg
20–34 kg
35–49 kg
≥50 kg

(A–C) Simulated pegcetacoplan exposure (Cavg) in pediatric patients by weight group at (A) week 1, (B) week 2, and (C) steady-state. Box plots represent the 1st (Q1) and 3rd (Q3) quartiles (box edges), median (horizontal line), and minimum/maximum values within 1.5×IQR below Q1 or above Q3 (whiskers). The blue shaded region represents 90% prediction interval for the adult C3G and primary IC-MPGN population. The blue dashed line represents the adult C3G and primary IC-MPGN population median.

- A non-linear E–R relationship was identified for UPCR, with a maximal reduction of 68% and a pegcetacoplan concentration of 109 µg/mL achieving 50% of this maximum (**Fig D**)

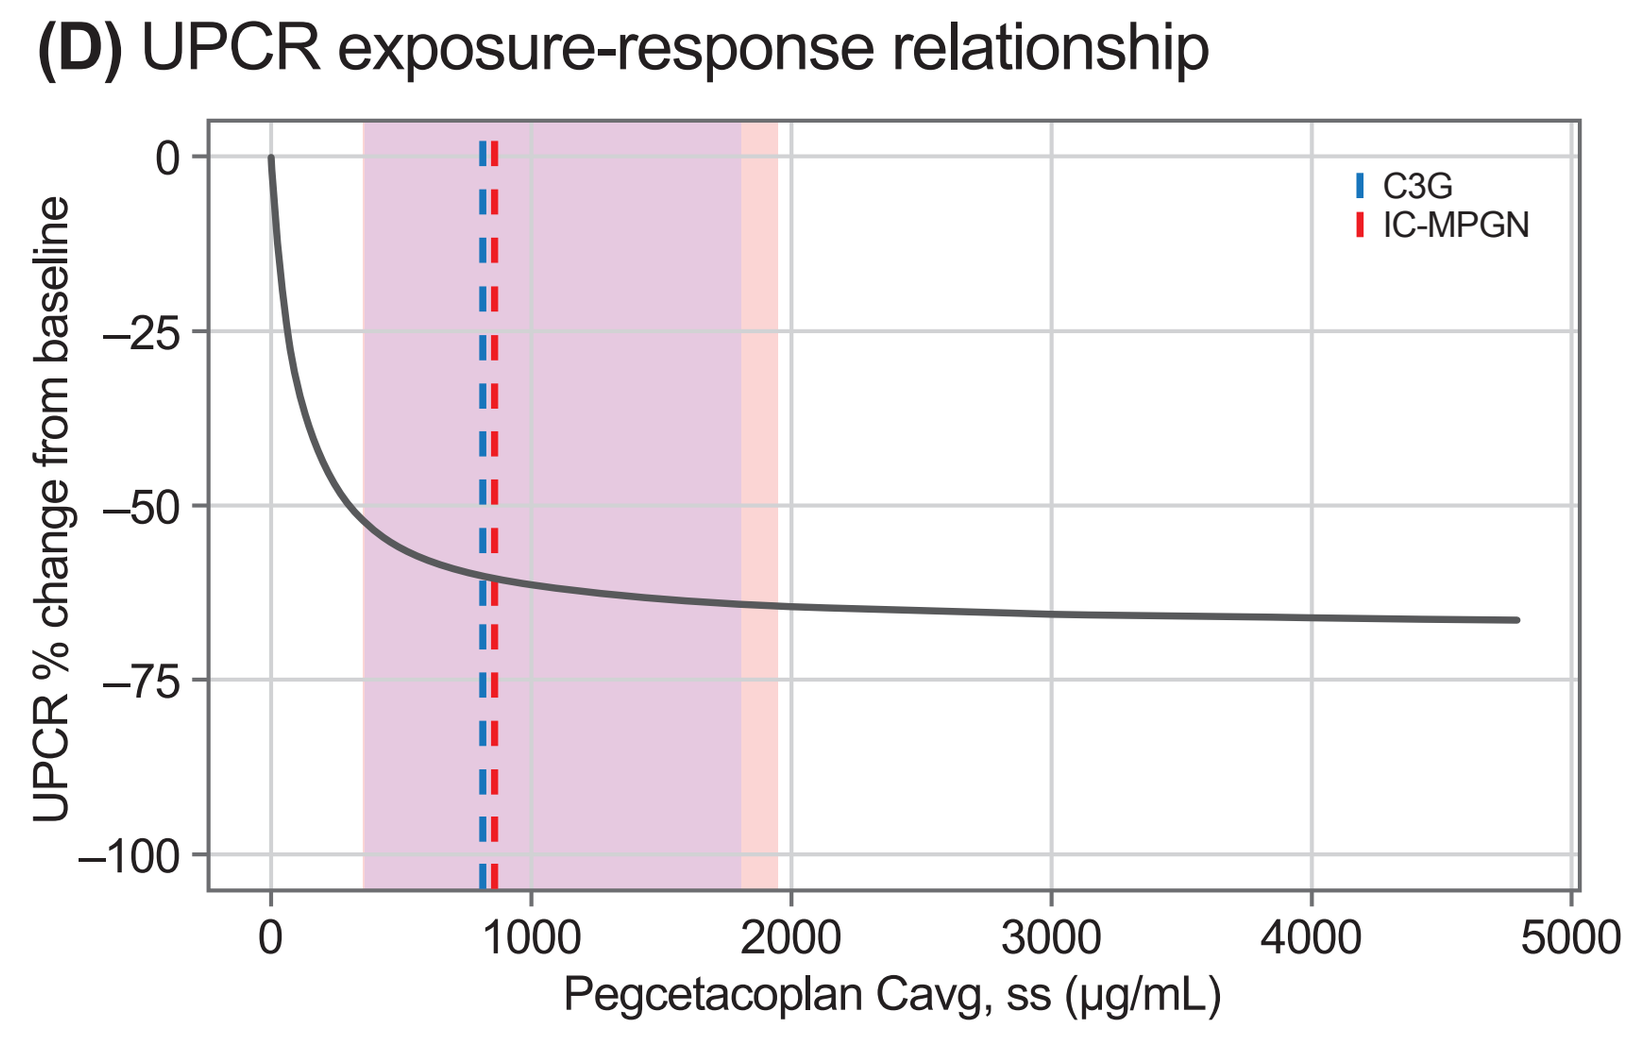
(D) UPCR exposure-response relationship
C3G
IC-MPGN
UPCR % change from baseline
Pegcetacoplan Cavg, ss (µg/mL)

(D) Dose E–R predictions for C3G and primary IC-MPGN patient populations. Vertical dashed lines represent median steady-state Cavg for C3G (blue) and primary IC-MPGN (red); shaded regions represent 5th–95th percentiles for patients receiving pegcetacoplan 1080 mg BIW (or corresponding weight-adjusted adolescent dose). The black line represents the model-predicted E–R curve for UPCR reduction.

RESULTS *(cont.)*

- No intrinsic or extrinsic patient factors (including age [adolescent vs adult], transplant status [transplanted kidney vs native kidney], diagnosis [PNH vs C3G vs primary IC-MPGN]) (**Fig E–G**), baseline UPCR or eGFR (**Fig H–I**), or immunogenicity (anti-pegcetacoplan or anti-peptide antibodies) were predicted to have a clinically relevant impact on exposure or UPCR response, supporting consistent E–R across subgroups
- eGFR and UPCR over time did not have a clinically relevant impact on pegcetacoplan exposure

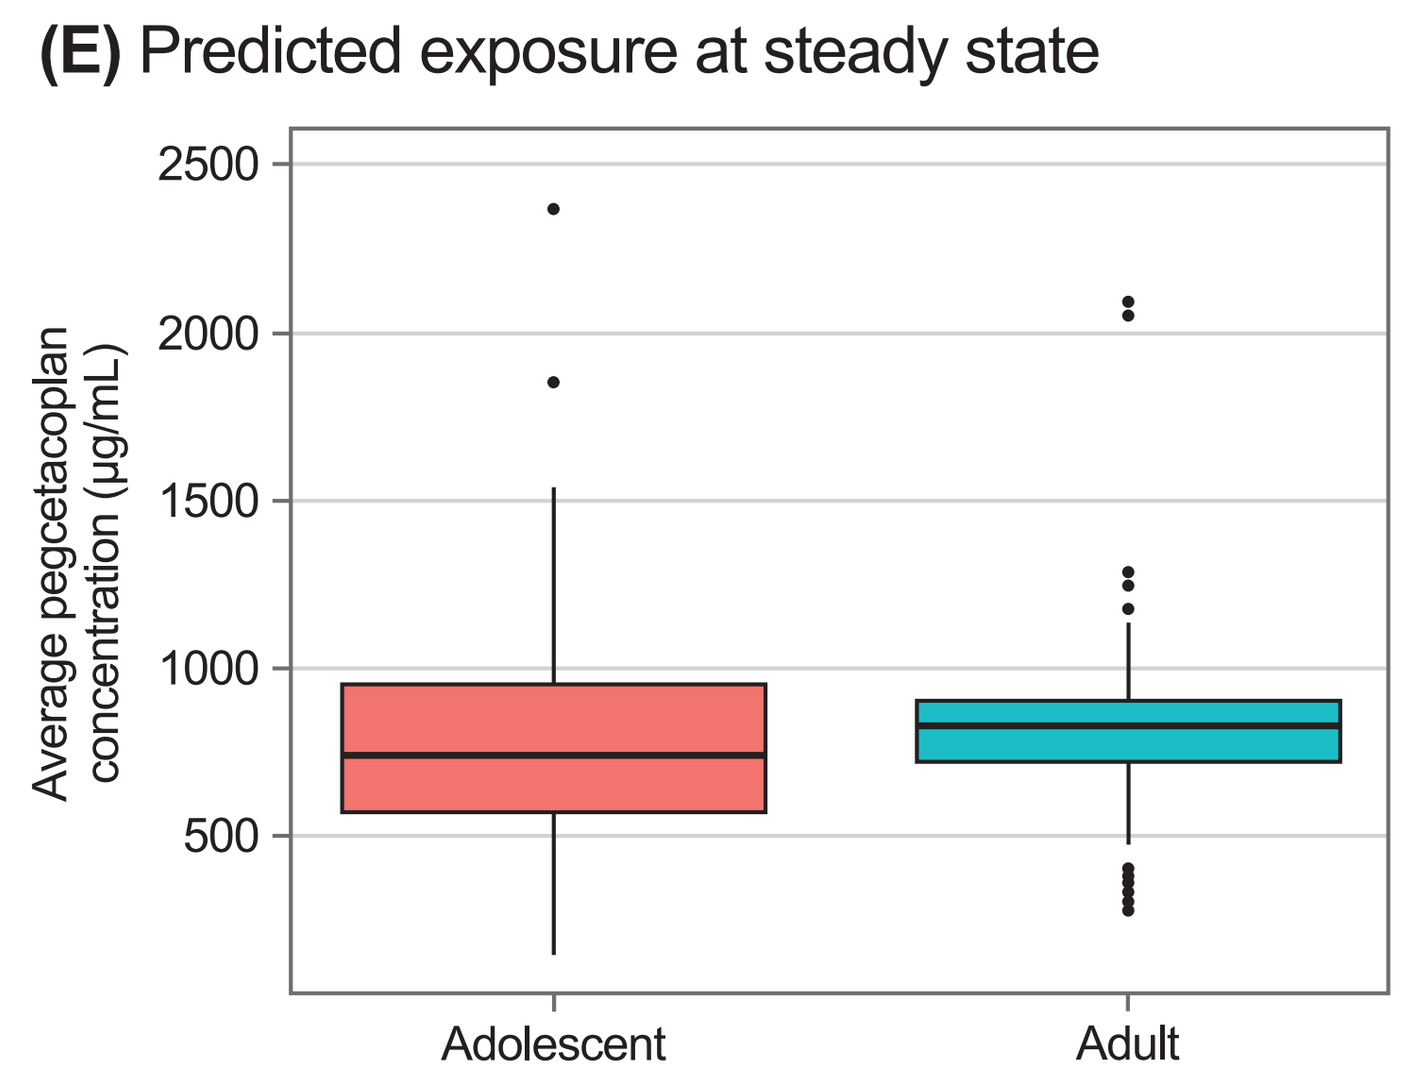
(E) Predicted exposure at steady state
Average pegcetacoplan concentration (µg/mL)
Adolescent
Adult

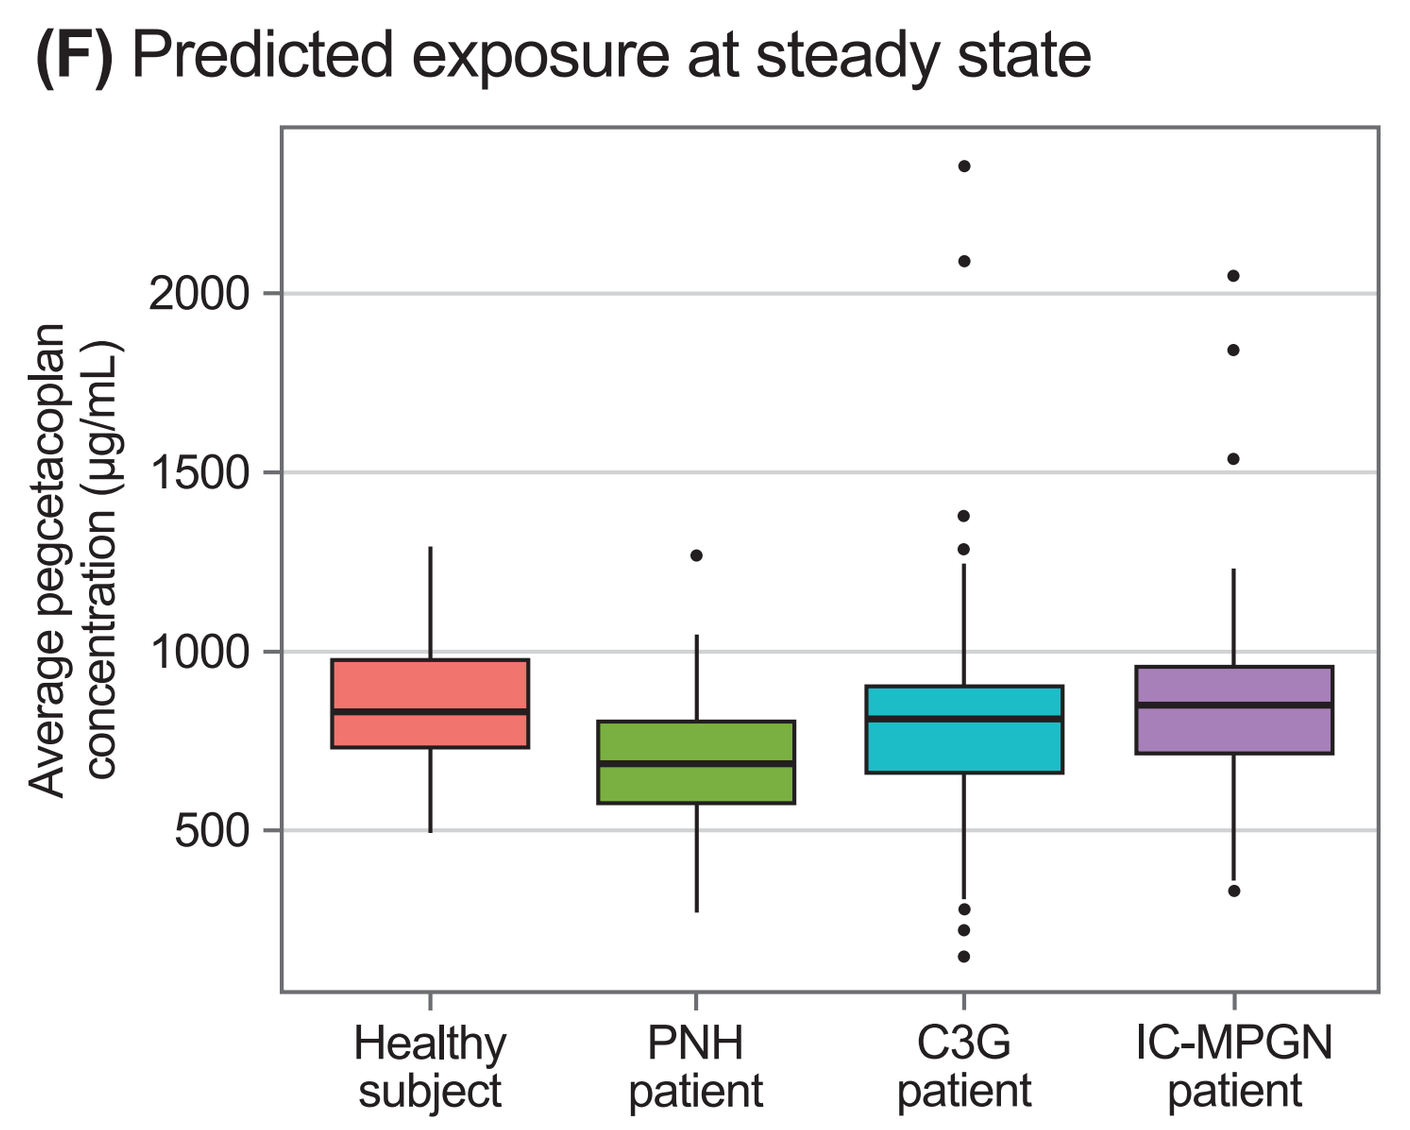
(F) Predicted exposure at steady state
Average pegcetacoplan concentration (µg/mL)
Healthy subject
PNH patient
C3G patient
IC-MPGN patient

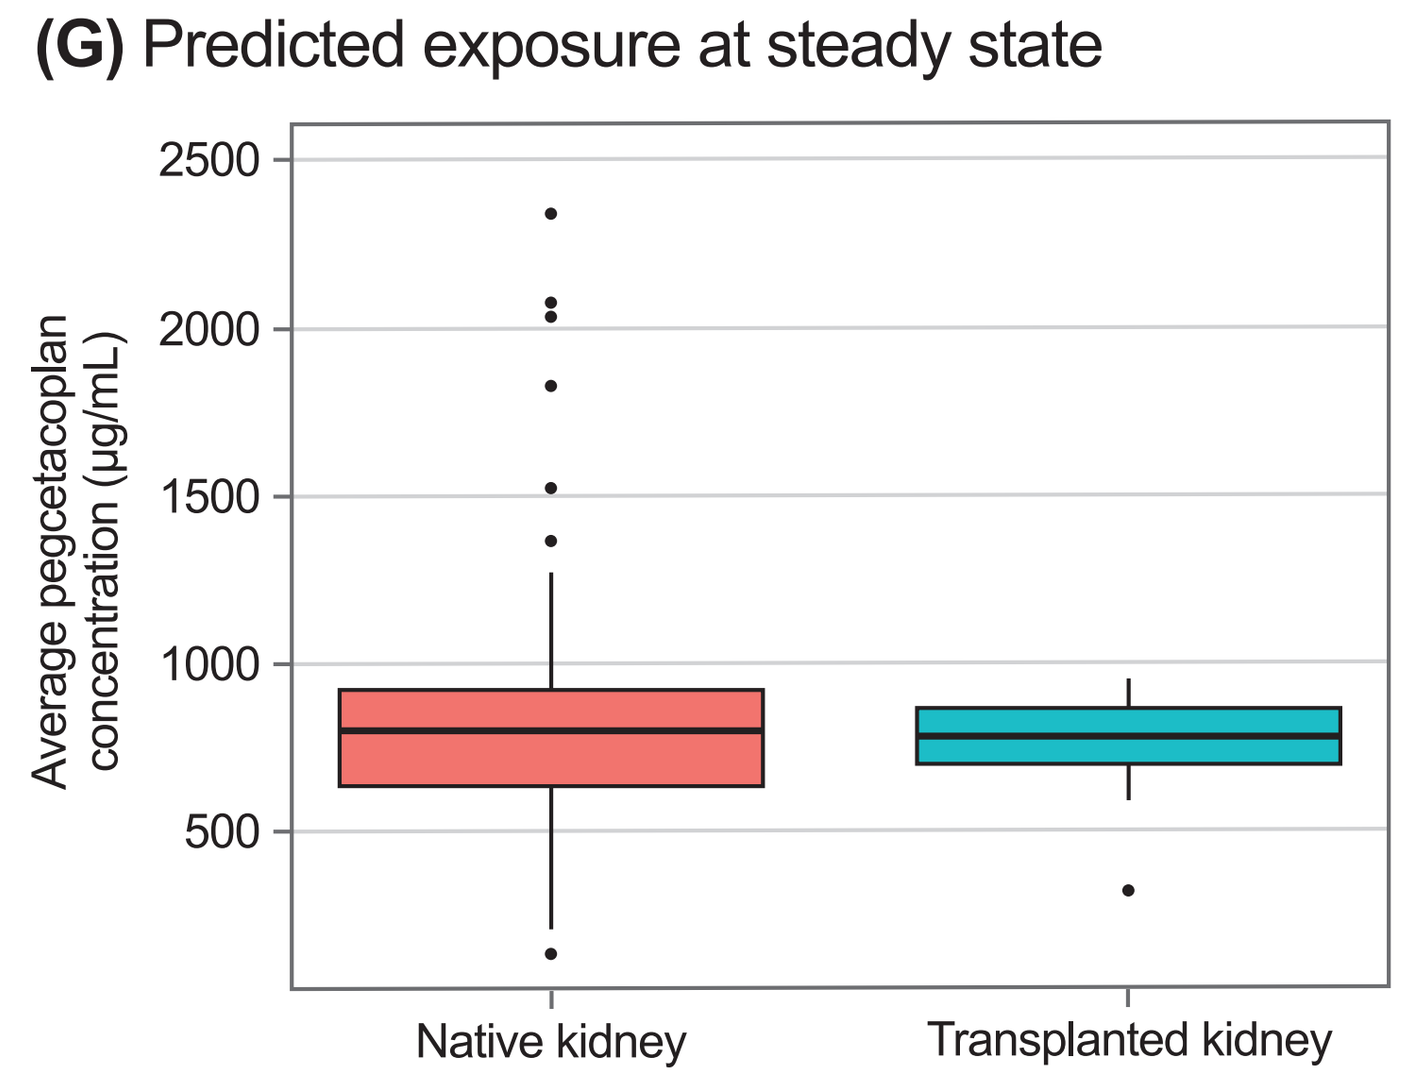
(G) Predicted exposure at steady state
Average pegcetacoplan concentration (µg/mL)
Native kidney
Transplanted kidney

(E–G) Bayesian estimated steady-state pegcetacoplan concentration (µg/mL), stratified by (E) age, (F) diagnosis, and (G) transplant status.

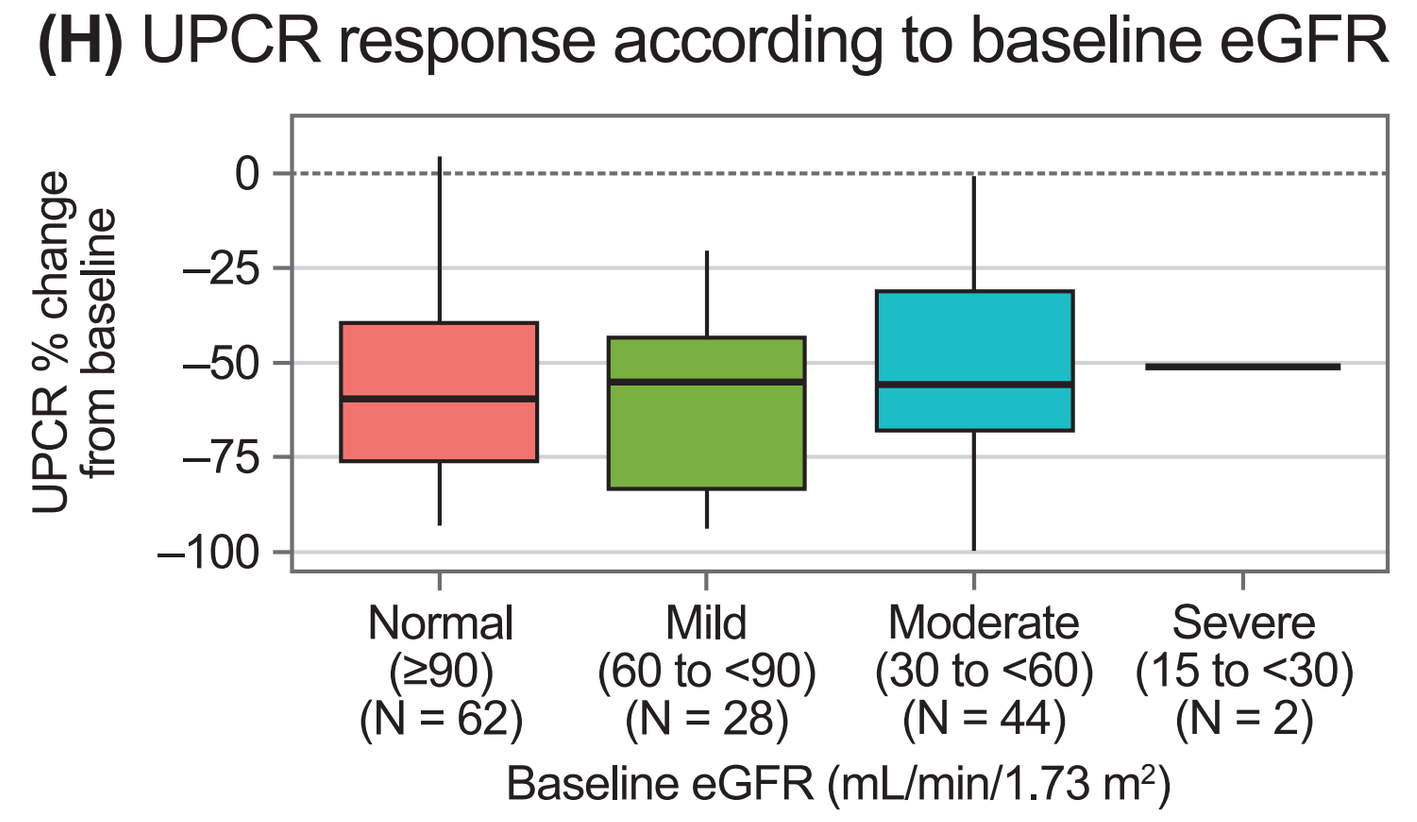
(H) UPCR response according to baseline eGFR
UPCR % change from baseline
Normal (≥90) (N = 62)
Mild (60 to <90) (N = 28)
Moderate (30 to <60) (N = 44)
Severe (15 to <30) (N = 2)
Baseline eGFR (mL/min/1.73 m²)

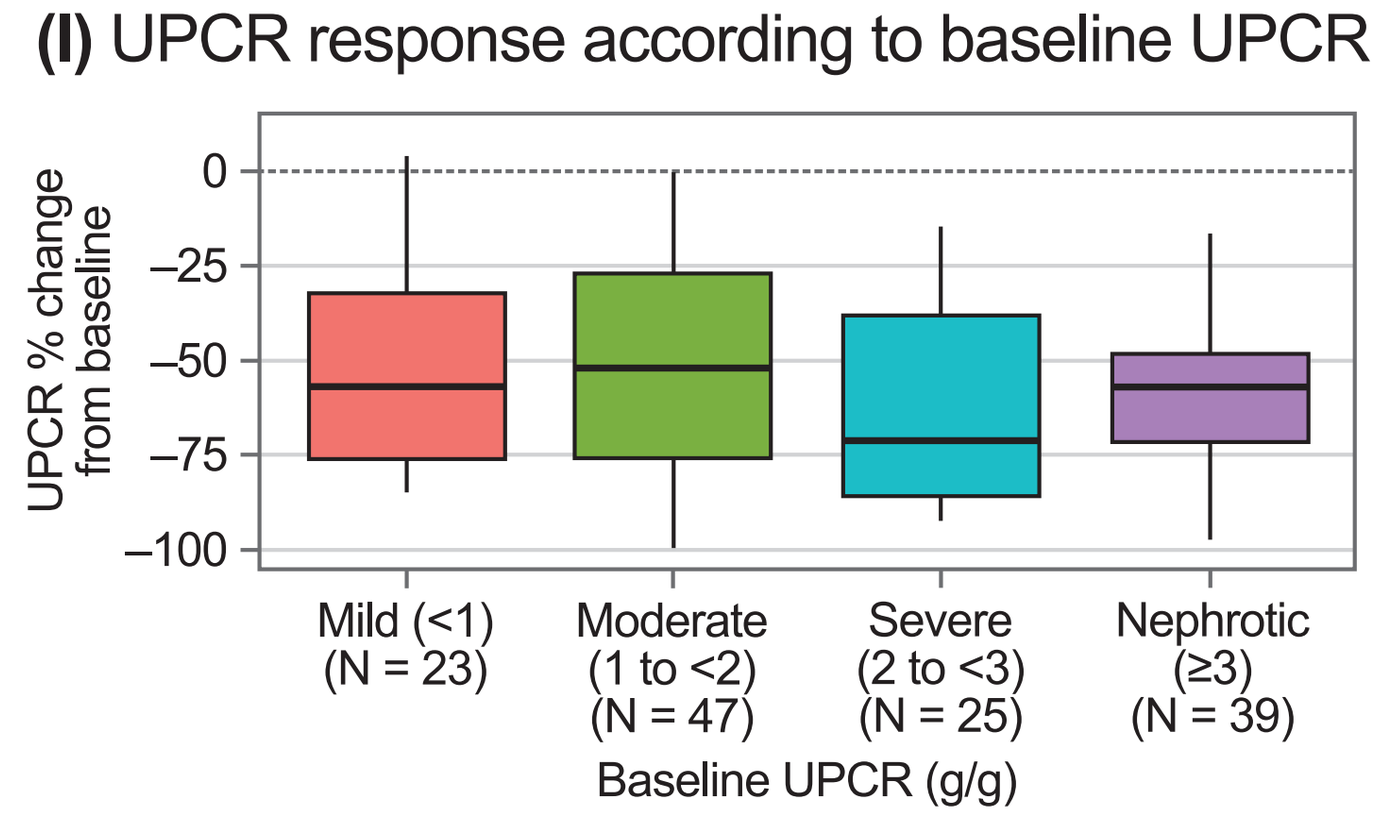
(I) UPCR response according to baseline UPCR
UPCR % change from baseline
Mild (<1) (N = 23)
Moderate (1 to <2) (N = 47)
Severe (2 to <3) (N = 25)
Nephrotic (≥3) (N = 39)
Baseline UPCR (g/g)

(H–I) Bayesian predicted steady-state UPCR percentage change from baseline stratified by (H) baseline eGFR and (I) baseline UPCR. Renal function categories: Normal (≥90 mL/min/1.73 m²), Mild (60–<90 mL/min/1.73 m²), Moderate (30–<60 mL/min/1.73 m²), Severe (15–<30 mL/min/1.73 m²).

RESULTS *(cont.)*

- The safety E–R analyses support the safety of pegcetacoplan in adolescent and adult patients with C3G and primary IC-MPGN
- Exposure to pegcetacoplan does not appear to be associated with AEs (**Fig J–M**)

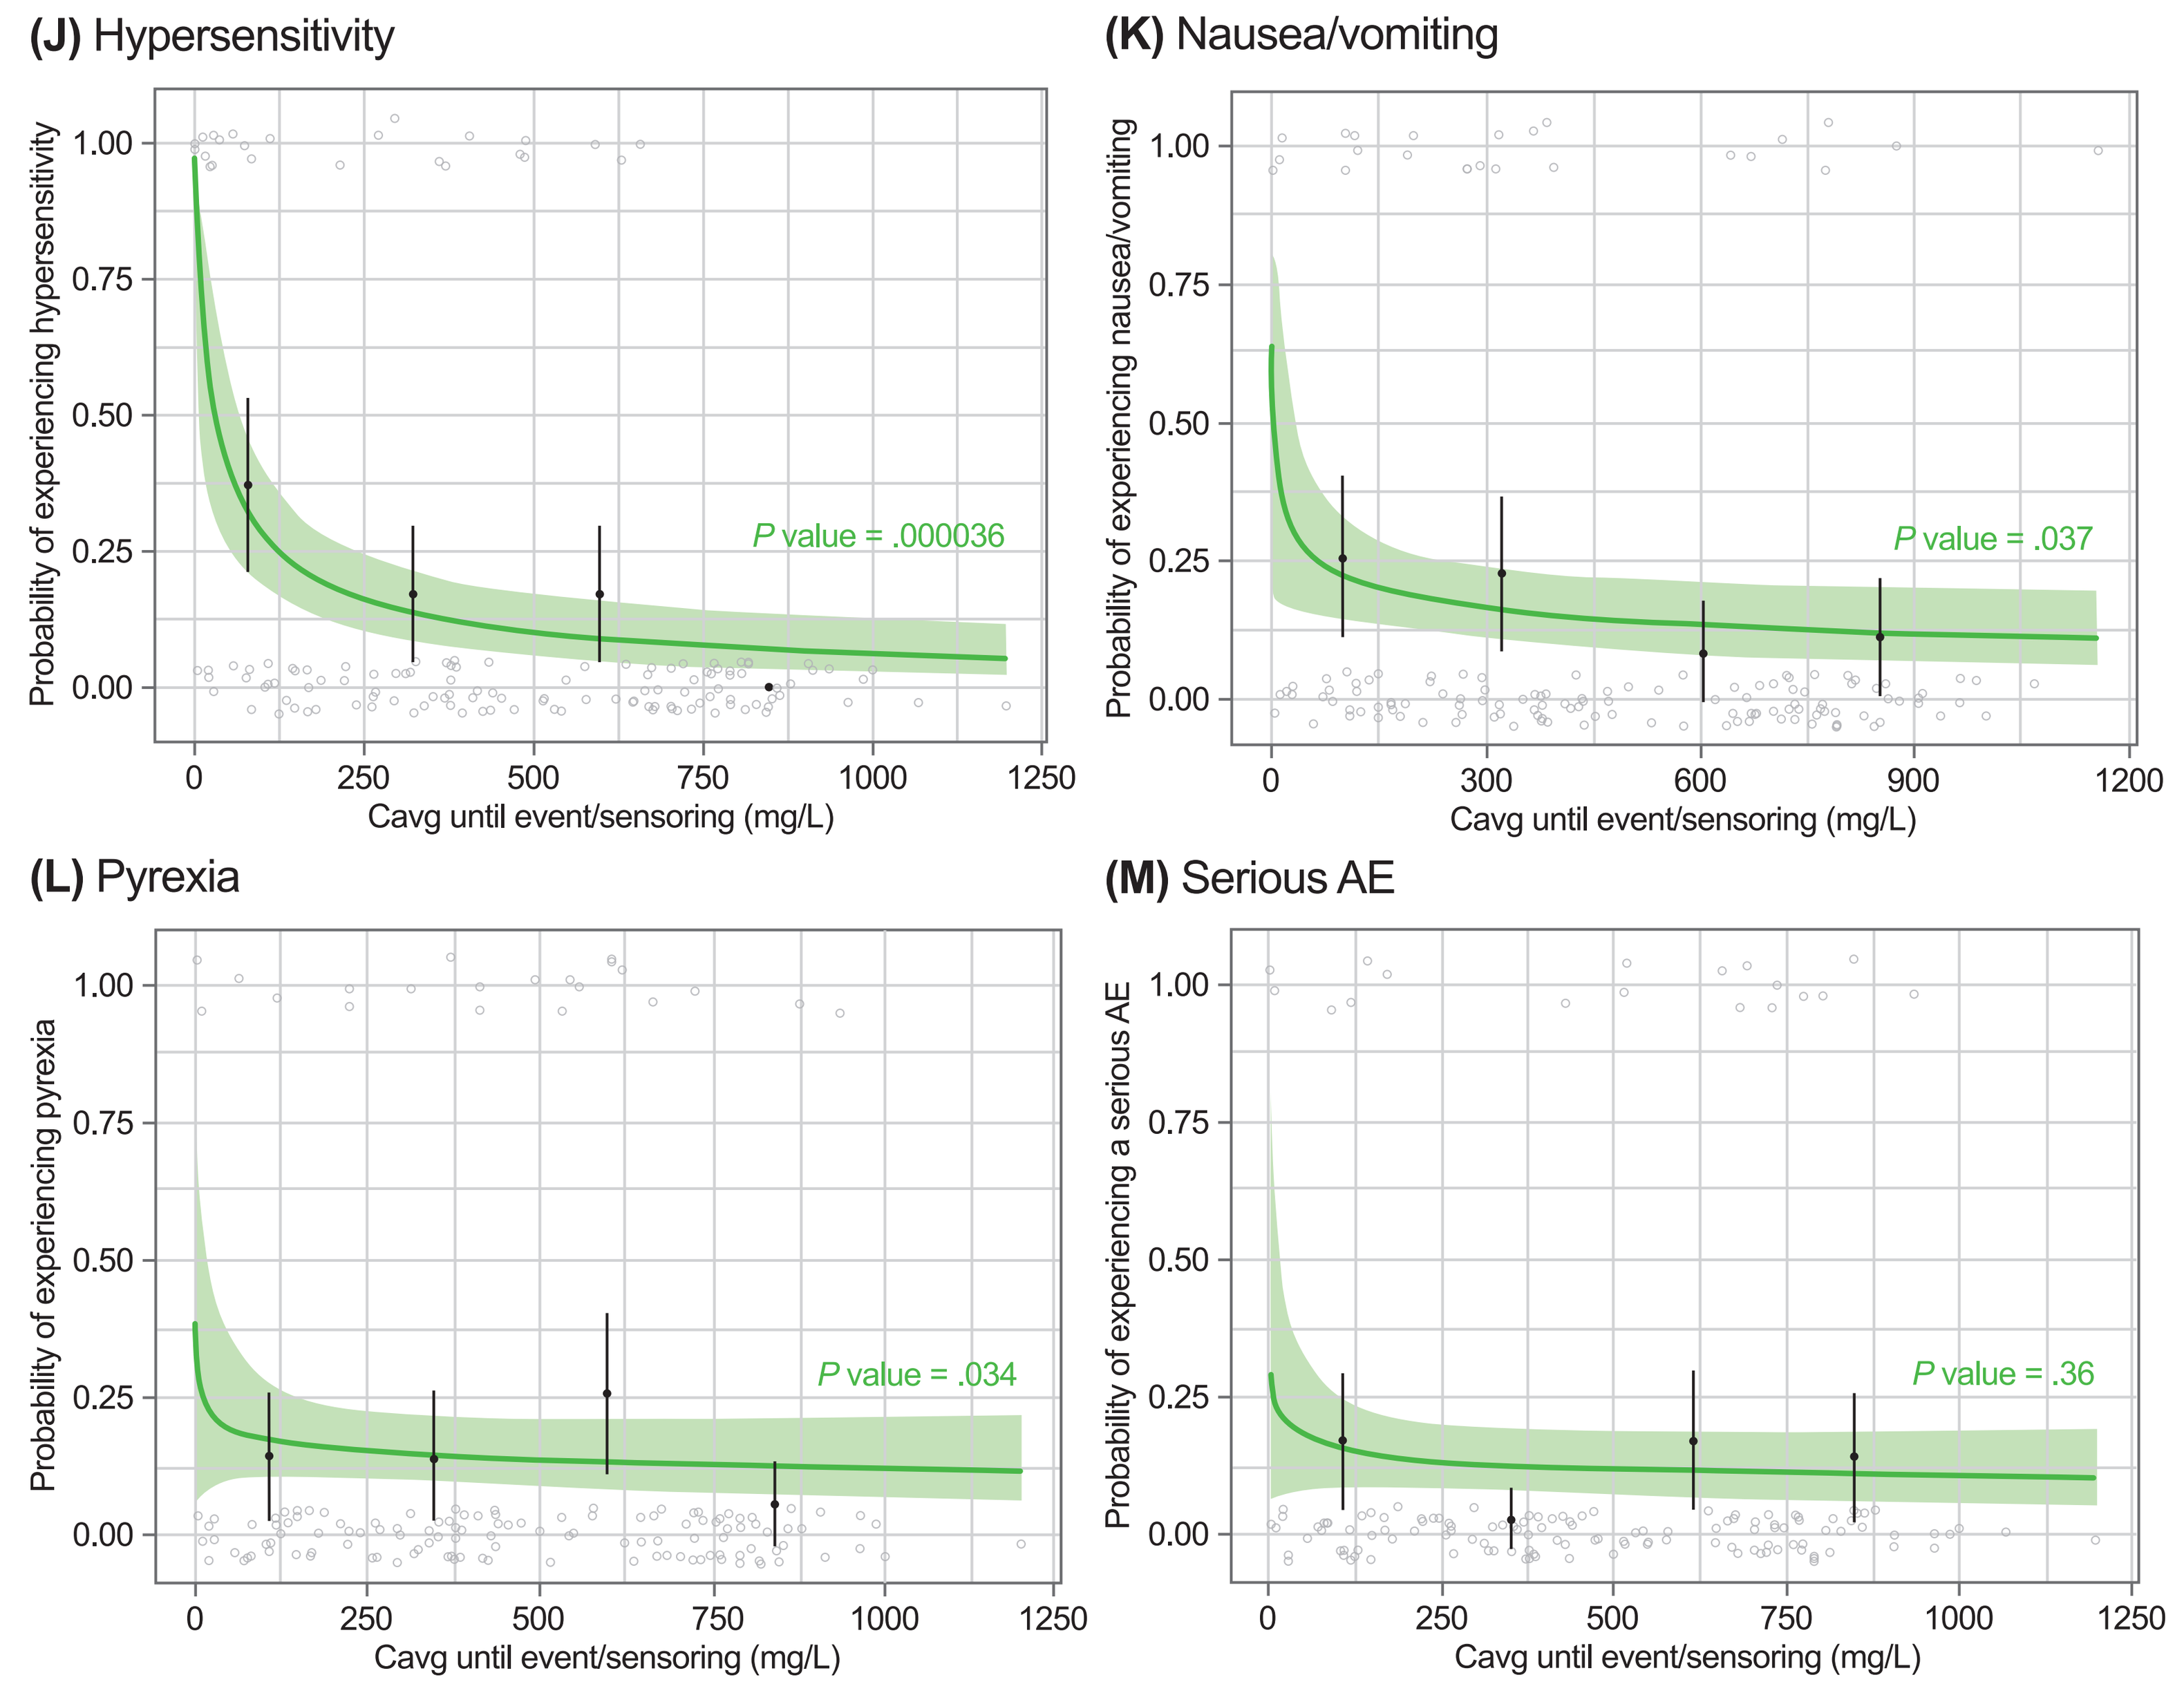
(J) Hypersensitivity
Probability of experiencing hypersensitivity
P value = .000036
(K) Nausea/vomiting
Probability of experiencing nausea/vomiting
P value = .037
(L) Pyrexia
Probability of experiencing pyrexia
P value = .034
(M) Serious AE
Probability of experiencing a serious AE
P value = .36
Cavg until event/sensoring (mg/L)

(J–M) Exposure–AE relationships: (J) hypersensitivity, (K) nausea/vomiting, (L) pyrexia, (M) serious AEs. Gray circles represent observed binary outcomes (1 = AE occurred, 0 = no AE). Green line represents model-predicted AE probability; green shading represents 95% CI. Black dots represent observed proportion of patients who experienced the AE of interest in the 4 exposure quartiles; the x-axis locations are the median values of the predicted exposure quartiles. The vertical black bars represent the normal approximation of the 95% CI for the observed proportion of patients experiencing the AE of interest. The *P* value corresponds to the hypothesis test concerning the significance of the exposure metric.

References: 1. Smith RJH, et al. *Nat Rev Nephrol*. 2019;15:129–43. **2.** Zipfel PF, et al. *Front Immunol*. 2019;10:2166. **3.** Meuleman MS, et al. *Semin Immunol*. 2022;60:101634. **4.** Dixon BP, et al. *Kidney Int Rep*. 2023;8:2284–93. **5.** EMPAVELI (pegcetacoplan). Apellis Pharmaceuticals, Inc; 2024. **6.** ASPAVELI (pegcetacoplan). Swedish Orphan Biovitrum AB; 2024. **7.** Lamers C, et al. *Nat Commun*. 2022;13:5519.

ABBREVIATIONS: AE, adverse event; BIW, twice weekly; C3G, C3 glomerulopathy; Cavg, average concentration; eGFR, estimated glomerular filtration rate; E–R, exposure-response; IQR, interquartile range; IC-MPGN, immune-complex membranoproliferative glomerulonephritis; PNH, paroxysmal nocturnal hemoglobinuria; PK, pharmacokinetics; SC, subcutaneous; UPCR, urine protein-to-creatinine ratio.

Disclosures: **AK** is an employee of Apellis and may hold stock or stock options; **PK** is an employee of Sobi (Swedish Orphan Biovitrum AB) and may hold stock or stock options; **BS**, **XY**, **SK**, and **RLC** are employees of A2-Ai.

ACKNOWLEDGMENTS Research funding source: This study was funded by Apellis Pharmaceuticals, Inc and Sobi (Swedish Orphan Biovitrum AB). Writing assistance was provided by Jennifer L. Gibson, PharmD, of Kay Square Scientific (Butler, PA, USA), and was funded by Apellis Pharmaceuticals, Inc and Sobi (Swedish Orphan Biovitrum AB).

127th Annual Meeting of the American Society for Clinical Pharmacology and Therapeutics (ASCPT)
March 4–6, 2026 | Denver, CO