Human factors validation of a pegcetacoplan on-body injector demonstrates ease of use by adolescents with C3 glomerulopathy or primary immune-complex membranoproliferative glomerulonephritis

Eliyahu Khankin[1]; Chirayu Mehta[1]; Chauncey Clemons[1]

[1]Apellis Pharmaceuticals, Inc, Waltham, MA, USA

FR-PO0890

CONCLUSIONS

- **The on-body injector was safe and effective for independent self-administration of pegcetacoplan by adolescents with C3G or primary IC-MPGN**

INTRODUCTION

- Pegcetacoplan, a C3/C3b inhibitor, was approved by the US Food and Drug Administration (FDA) in July 2025 for adults and adolescents aged ≥12 years with C3 glomerulopathy (C3G) or primary immune-complex membranoproliferative glomerulonephritis (IC-MPGN)[1–4]
- Previously approved for adults with paroxysmal nocturnal hemoglobinuria (PNH),[4] US adults with PNH can self-administer pegcetacoplan subcutaneously using an approved on-body, autoinjector, after transferring the prescribed pegcetacoplan dose from a vial to the injector with a syringe (**Figure 1**)[5,6]
- Pegcetacoplan dosage in adolescents is based on body weight,[4] requiring different filling steps compared with adults

Figure 1. On-body, injector for pegcetacoplan (A) and critical injection steps (B)

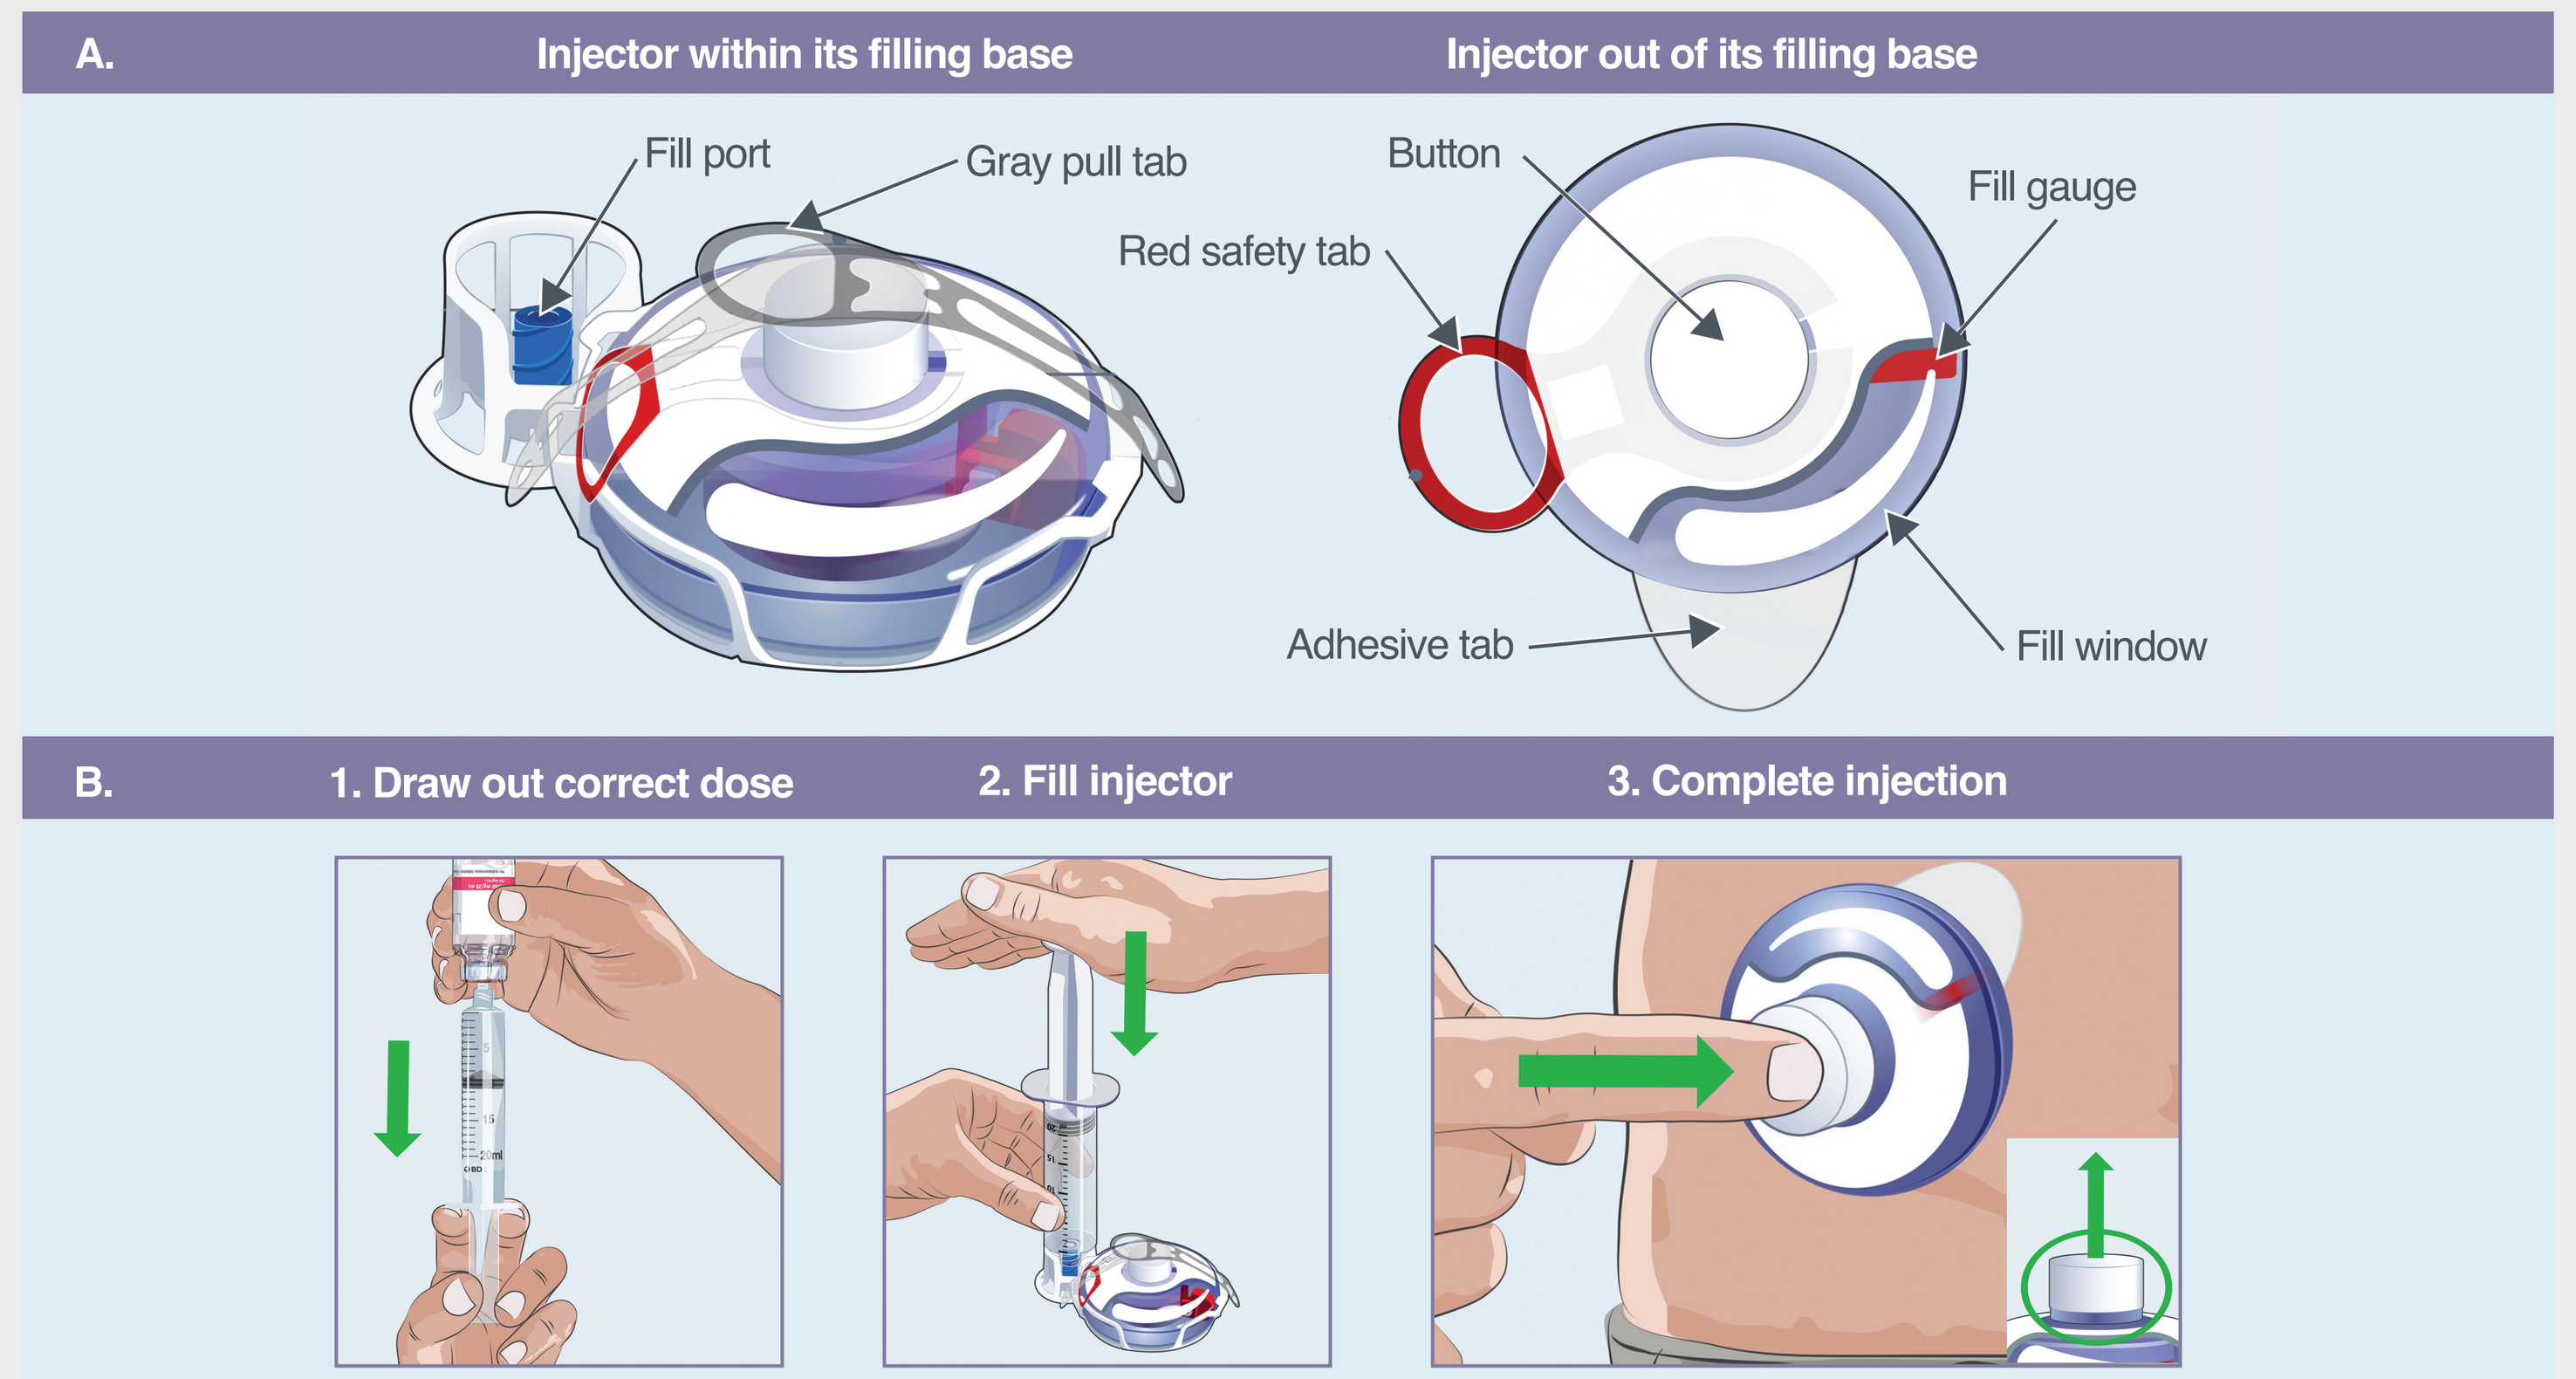
A.
Injector within its filling base
Injector out of its filling base
Fill port
Gray pull tab
Red safety tab
Button
Fill gauge
Adhesive tab
Fill window
B.
1. Draw out correct dose
2. Fill injector
3. Complete injection

Reproduced from Shahin H, Laurence L, Korkuch D. Human-factors validation study for a wearable, single-use injector for patients with paroxysmal nocturnal hemoglobinuria. *TheResearchPost*. Published online October 8, 2024. https://doi.org/10.53705/241008

OBJECTIVE

This human factors study aimed to validate the use of the pegcetacoplan injector by adolescents aged 12–17 years with chronic conditions like C3G or primary IC-MPGN

METHODS

- The design of this human factor validation study followed the FDA guidance to evaluate a drug injection device[7] (**Figure 2**)

Figure 2. Design of a human factors validation study to evaluate a drug injection device[7]

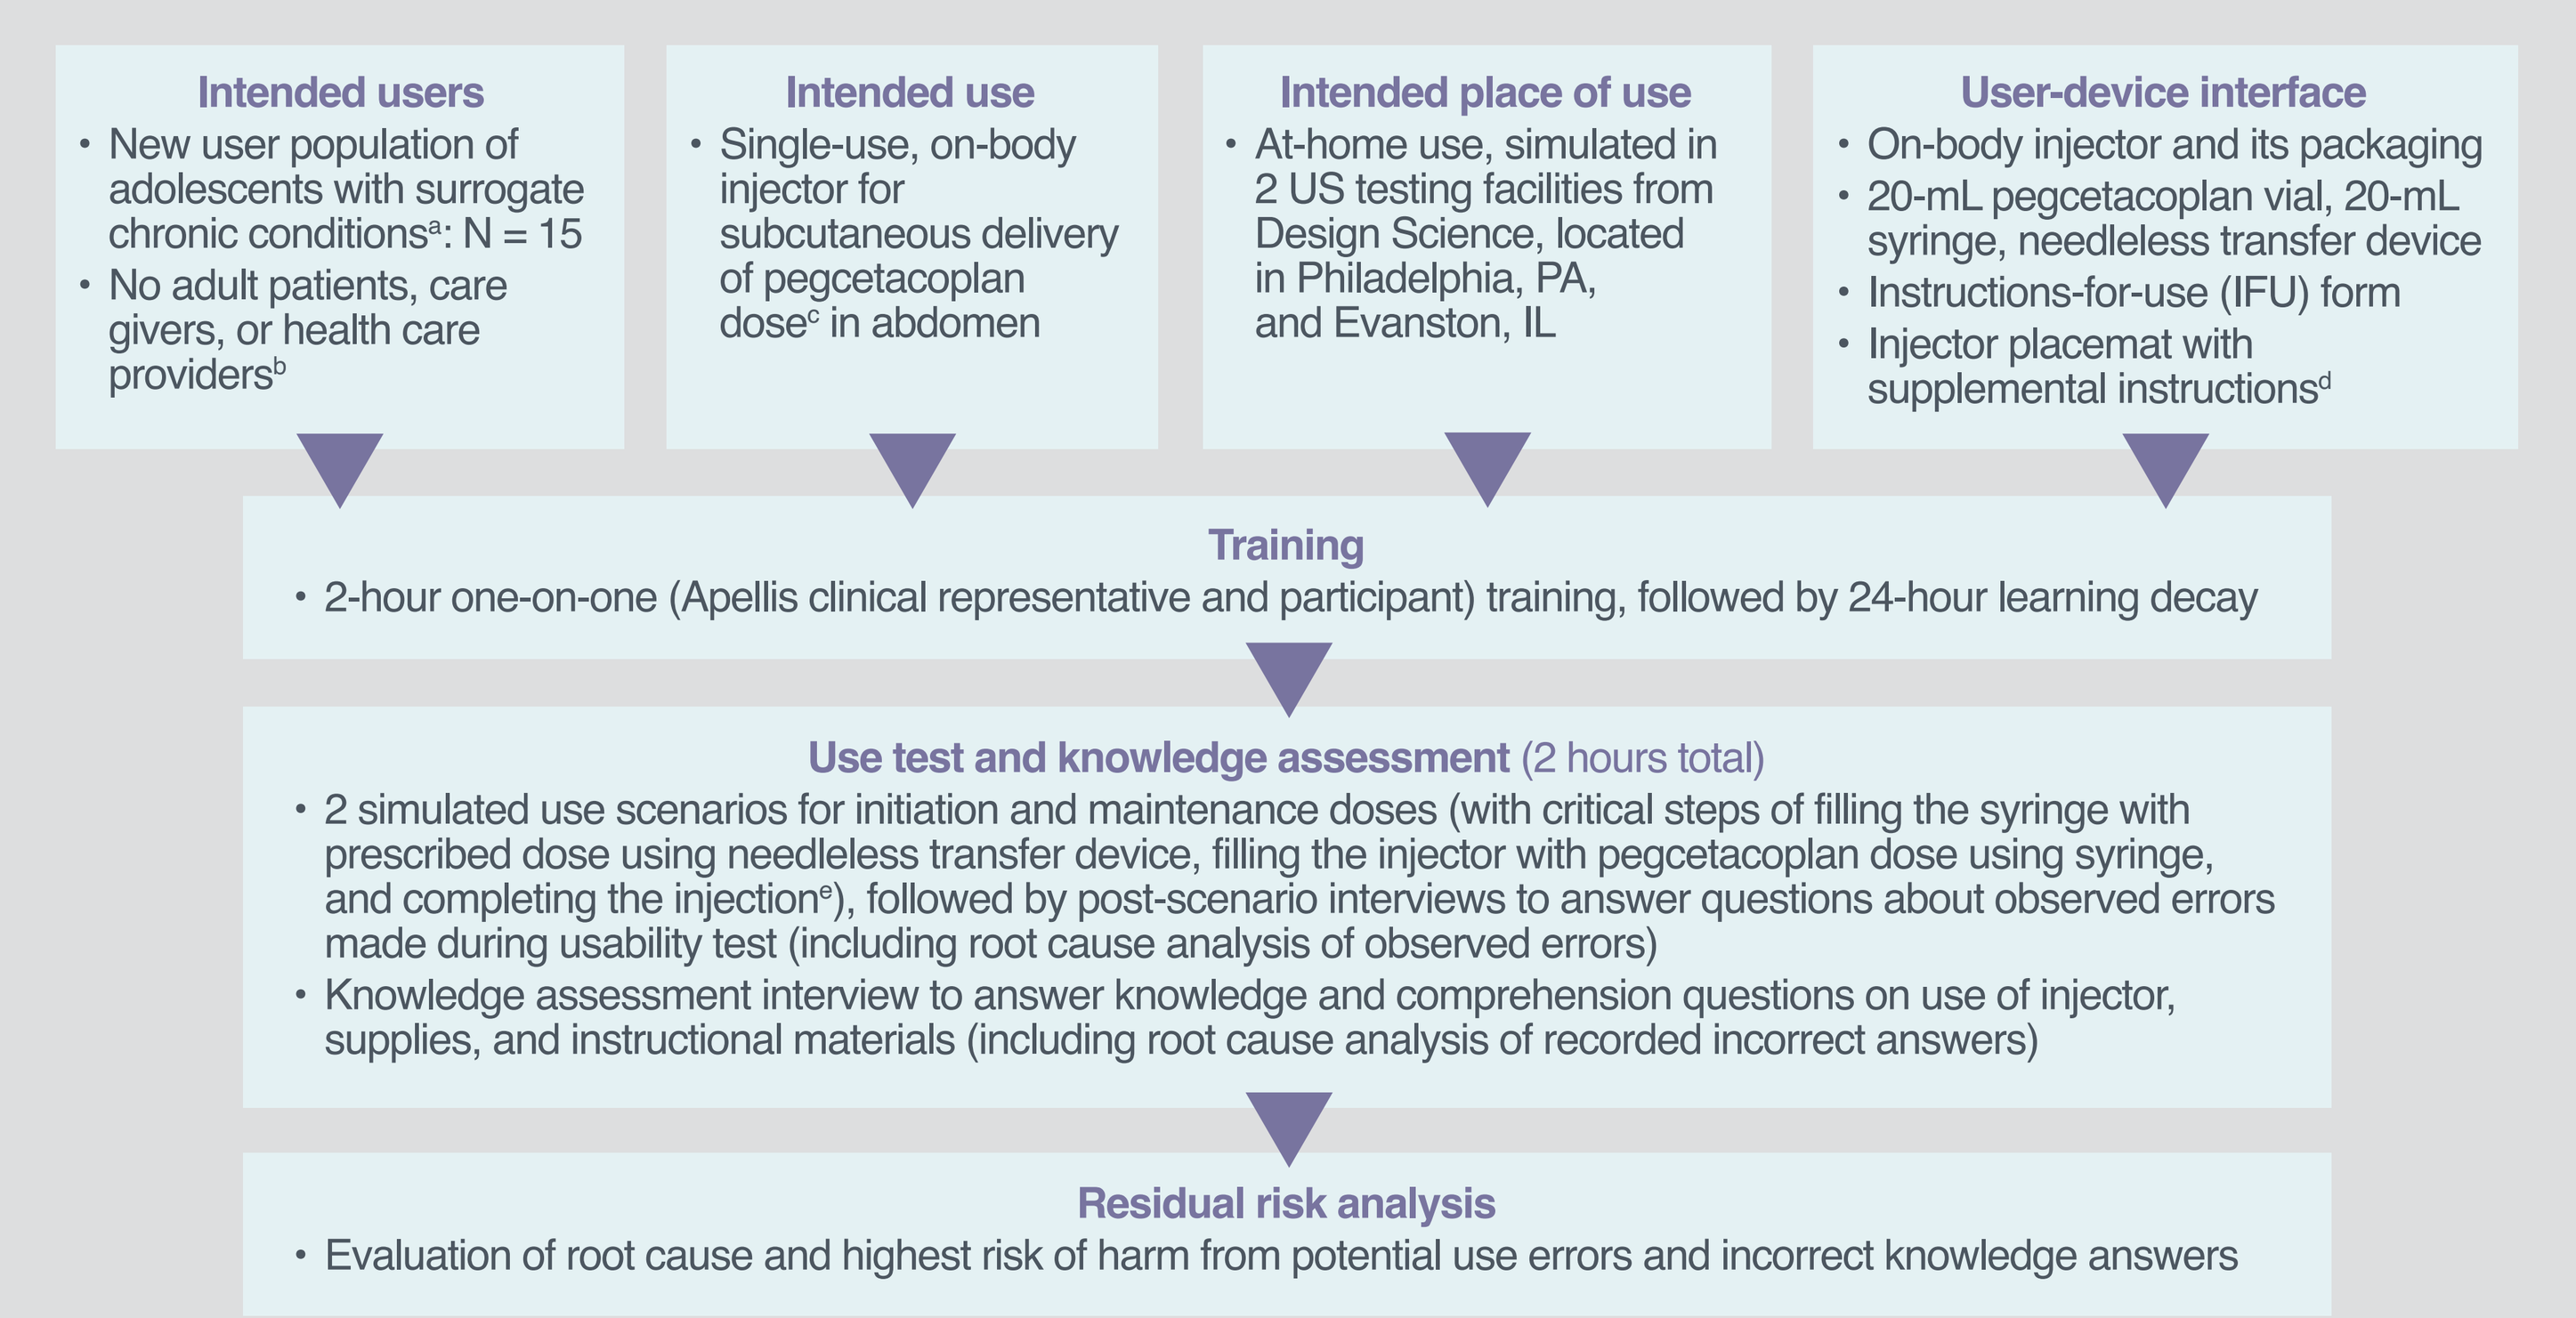
Intended users
• New user population of adolescents with surrogate chronic conditions[a]: N = 15
• No adult patients, care givers, or health care providers[b]
Intended use
• Single-use, on-body injector for subcutaneous delivery of pegcetacoplan dose[c] in abdomen
Intended place of use
• At-home use, simulated in 2 US testing facilities from Design Science, located in Philadelphia, PA, and Evanston, IL
User-device interface
• On-body injector and its packaging
• 20-mL pegcetacoplan vial, 20-mL syringe, needleless transfer device
• Instructions-for-use (IFU) form
• Injector placemat with supplemental instructions[d]
Training
• 2-hour one-on-one (Apellis clinical representative and participant) training, followed by 24-hour learning decay
Use test and knowledge assessment (2 hours total)
• 2 simulated use scenarios for initiation and maintenance doses (with critical steps of filling the syringe with prescribed dose using needleless transfer device, filling the injector with pegcetacoplan dose using syringe, and completing the injection[e]), followed by post-scenario interviews to answer questions about observed errors made during usability test (including root cause analysis of observed errors)
• Knowledge assessment interview to answer knowledge and comprehension questions on use of injector, supplies, and instructional materials (including root cause analysis of recorded incorrect answers)
Residual risk analysis
• Evaluation of root cause and highest risk of harm from potential use errors and incorrect knowledge answers

[a]Surrogate chronic diseases (chronic kidney disease, diabetes, anemia, juvenile arthritis) representing similar physical and mental burden of disease to C3G or primary IC-MPGN, which are rare diseases. [b]Included in initial injector validation study. [c]As in Table 1. [d]New to this study compared with primary human factor study. [e]Participants used injection pads over their clothing rather than their skin for the test injections. IFU, instructions for use.

- The study was approved by a local Institutional Review Board and all participants provided written informed consent

RESULTS

Participants: 15 adolescents with chronic diseases representative of C3G or primary IC-MPGN and aged 12–17 years were enrolled in 2 US testing facilities in October 2024 (**Table 1**)

Table 1. Participant characteristics

Characteristics, n	All adolescents, N = 15
Age range, years	12–13, n = 4; 14–15, n = 3; 16–17, n = 8
Sex	Female, n = 8; male, n = 7
Handedness	Right-handed, n = 13; left-handed, n = 1; ambidextrous, n = 1
Dexterity	Sprained wrist, n = 1
Injection experience[a]	Experienced, n = 6; naive, n = 9

[a]Injection-experienced was defined as having performed an injection with any drug or any needle-injection device within the last 12 months and injection-naive was defined as never having injected themselves or another person with prescribed medication or non–health-related substance from a needle-based injection device.

Simulated use results: (**Table 2**)

- In scenario 1 (first 12-mL dose), 14 of 15 participants drew out the correct pegcetacoplan dose, 11 filled the injector correctly, and 14 completed the injection

RESULTS *(cont.)*

Simulated use results (cont.):

- In scenario 2 (maintenance 15-mL dose), 15 drew out the correct pegcetacoplan dose, 10 filled the injector correctly, and 15 completed the injection
- On second tries, all participants completed the tasks correctly and independently without serious harm

Table 2. Analysis of critical use errors

Critical tasks[a] with use errors[b]	Participants with use errors[b]	Root cause[c]	Highest risk of harm
Withdraw prescribed dose of pegcetacoplan into syringe	▪ 1 in scenario 1: drew 14 mL instead of 12 mL ▪ 0 in scenario 2	**Incorrect mental model** and **slip** (intentionally drew same dose as their current medication)	**Overdose** of medicinal product, with potential harm of **systemic reactions**; however, this is not considered an overdose for pegcetacoplan and is expected to be safe and generally well tolerated based on PK/PD modeling
Fill injector with pegcetacoplan from syringe (push syringe plunger down to fill injector with all medicinal product and do not remove syringe from filling base)	▪ 4 in scenario 1: – 3 removed syringe from filling base after filling injector, – 1 removed injector from filling base before filling injector[d] ▪ 5 in scenario 2: – 3 removed syringe from filling base after filling injector, – 2 struggled pushing plunger down	**Insufficient salience of instructions** or training (removal of syringe from filling base), or **incorrect mental model** (muscle memory from current insulin pump use)	**Exposure of skin or eyes** to medicinal product with a potential harm of **skin irritation or inflammation**; however, no skin irritation is expected with minor pegcetacoplan spillage on participants' skin
		Lapse (forgot to fill injector before removing it from its filling base)	**Insignificant delay** in medicinal product delivery with a potential harm of **minimal to no impact on therapy**; however, participant was able to fill injector correctly on a subsequent attempt
		Device design (insufficient force to push plunger down)	**Exertion of excessive ergonomic force** with a potential harm of **minor strain or bruising and inconvenience**; however, participants were able to complete this task correctly on subsequent attempts
Push injector button and wait for it to pop to complete injection	▪ 1 in scenario 1[d]	**Previous use error** (button did not press because injector was not filled properly after being prematurely removed from filling base)	**Insignificant delay** in medicinal product delivery with a potential harm of **minimal to no impact on therapy** and **exposure to pinch points, needle, spike, or other sharp edges** with a potential harm of **minor bruising, contusion, minor puncture injury, or laceration**; however, participant realized their error and completed a subsequent injection correctly

[a]Tasks that if performed incorrectly or not at all could cause harm, including compromising medical care. [b]Action or lack of action that differed from manufacturer's expectations or instructions. [c]Verbiage taken from FDA guidance.[7] [d]Same participant

Knowledge assessment: (**Table 3**)

- 4 of 21 questions received the most (>4) incorrect answers
- Most incorrect answers were due to incorrect mental models (incorrect answer caused by participant's inaccurate perceptions of how the device should be used), lapses (oversight), and user state (mental fatigue) and resulted in low or no residual risk

RESULTS *(cont.)*

Knowledge assessment (cont.):

Table 3. Analysis of critical use errors

Questions with errors	Participants with errors	Root cause(s)[c]	Highest risk of harm
Q1: How should the wearable device be stored? [away from heat and sunlight and stored in the original box]	6	Incorrect mental model (3) Lapse (2) User state (1)[a]	Underdose of medicinal product due to damage to injector Exposure to pathogens, with potential harm of systemic infection
Q7: Where on your body should you inject the wearable device? [abdomen/stomach/belly at least 1 inch from the belly button, not along the belt line or folds of skin]	7	Lapse (2) Test artifact (2)[b] Incorrect mental model (1) Insufficient salience of instructions (1)[c] User state (1)[a]	Exposure to blood-borne pathogens with a potential harm of systemic infection
Q15: What should you not do while wearing the wearable device? [do not swim, exercise, bathe, shower, use hot tubs, whirlpools, saunas, or sleep]	7	Incorrect mental model (4) Insufficient salience of instructions (1)[c] Test artifact (1)[b] User state (1)[a]	Exposure to blood-borne pathogens with a potential harm of systemic infection
Q21: Who should not use the wearable device? [people with an acrylic allergy]	11	Test artifact (4)[b] Incorrect mental model (3) Insufficient salience of instructions (1)[c] Information location (1) User state (1)[a]	Acrylic allergy with potential harm of toxicity or local or systemic allergic reaction

[a]Same participant who experienced mental fatigue during the session. [b]Ambiguous study question. [c]Information did not stand out to participants in instructions for use.

Risk mitigation measures:

- Results from the residual risk analysis indicate that design changes were not expected to further mitigate risk associated with the observed use problems or incorrect knowledge answers
- Minor adjustments to the injector user interface, specifically the instructions for use, were implemented, to provide additional information on acceptable medication storage and preparation, directions for syringe preparation steps and filling the injector, as well as instructions on proper syringe disposal, among others

References: 1. Dixon BP, et al. *Kidney Int Rep*. 2023;8:2284. **2.** Bomback AS, et al. *Kidney Int Rep*. 2024;10:87. **3.** Fakhouri F, et al. *New Eng J Med*. 2025. Accepted for publication. **4.** EMPAVELI® (pegcetacoplan) US PI 2025. https://pi.apellis.com/files/PI_Empaveli.pdf. **5.** Shahin H, et al. *TheResearchPost*. 2024. https://doi.org/10.53705/241008. **6.** Empaveli® (pegcetacoplan) Injector Instructions for Use.2025. https://pi.apellis.com/files/IFU_EMPAVELIinjector.pdf. **7.** US Food and Drug Administration. Applying human factors and usability engineering to medical devices. February 2016. https://www.fda.gov/media/80481/download.

ABBREVIATIONS: C3G, C3 glomerulopathy; FDA, US Food and Drug Administration; IC-MPGN, immune-complex membranoproliferative glomerulonephritis; IFU, instructions for use; PNH, paroxysmal nocturnal hemoglobinuria.

Disclosures: **EK**, **CM**, and **CC** are employees of Apellis and may hold stock or stock options.

ACKNOWLEDGMENTS This study was funded by Apellis Pharmaceuticals, Inc. Writing assistance was provided by Catherine Champagne, PhD, of Kay Square Scientific (Butler, PA, USA), and was funded by Apellis Pharmaceuticals, Inc and Sobi (Swedish Orphan Biovitrum AB).

58th Annual Meeting of the American Society of Nephrology (ASN 2025) | November 5–9, 2025 | Houston, TX