Pegcetacoplan for adolescents and adults with C3 glomerulopathy or primary immune-complex membranoproliferative glomerulonephritis: Enrollment status of the VALE extension

INFO15-TH

Eliyahu Khankin[1]; Carla Nester[2]; Gema Ariceta[3]; Anwesha Mukherjee[1]; Johan Szamosi[4]; Luis Lopez Lazaro[4]

[1]Apellis Pharmaceuticals, Inc, Waltham, MA, USA; [2]University of Iowa, Stead Family Children's Hospital, Iowa City, IA, USA; [3]Autonomous University of Barcelona, University Hospital Vall d'Hebron, Barcelona, Spain; [4]Swedish Orphan Biovitrum AB, Stockholm, Sweden

CONCLUSIONS

- **The ongoing VALE extension study assesses long-term efficacy and safety of pegcetacoplan in a broad population of patients ≥12 years old with C3G or primary IC-MPGN**

OBJECTIVE

To describe the study design and enrollment status of VALE (NCT05809531), a phase 3 extension study to evaluate long-term efficacy and safety of pegcetacoplan in C3G and primary IC-MPGN

INTRODUCTION

- C3G and primary (idiopathic) IC-MPGN are rare diseases caused by C3 dysregulation that results in abnormal deposition of C3 breakdown products in the kidney, causing inflammation, kidney damage, and kidney failure[1–5]
- Pegcetacoplan blocks overactivation of alternative, classical, and lectin complement pathways by binding to C3 and C3b to regulate C3 cleavage and downstream activation[5,6]
- In the phase 3 VALIANT (NCT05067127) trial in patients with C3G or primary IC-MPGN, pegcetacoplan reduced proteinuria by 68.1% (vs placebo), reduced C3 staining to 0 in 71.4% of patients, and stabilized eGFR at week 26[7]
 - Improvements were maintained at week 52
- Pegcetacoplan has recently been approved in the United States for treatment of adult and pediatric patients aged 12 years and older with C3G or primary IC-MPGN, to reduce protienuria[8]
- VALE (NCT05809531), a phase 3 extension of the VALIANT trial, was initiated to assess long-term efficacy and safety of pegcetacoplan in patients with C3G or primary IC-MPGN[9]

METHODS

- In the open-label, nonrandomized VALE extension study, participants ≥12 years old who completed VALIANT through week 52 and had clinical benefit will continue twice-weekly doses of pegcetacoplan for a minimum of approximately 2.5 years (**Figure 1**)

Figure 1. Design of the VALIANT phase 3 trial and subsequent VALE extension trial of pegcetacoplan treatment for participants with C3G or primary IC-MPGN

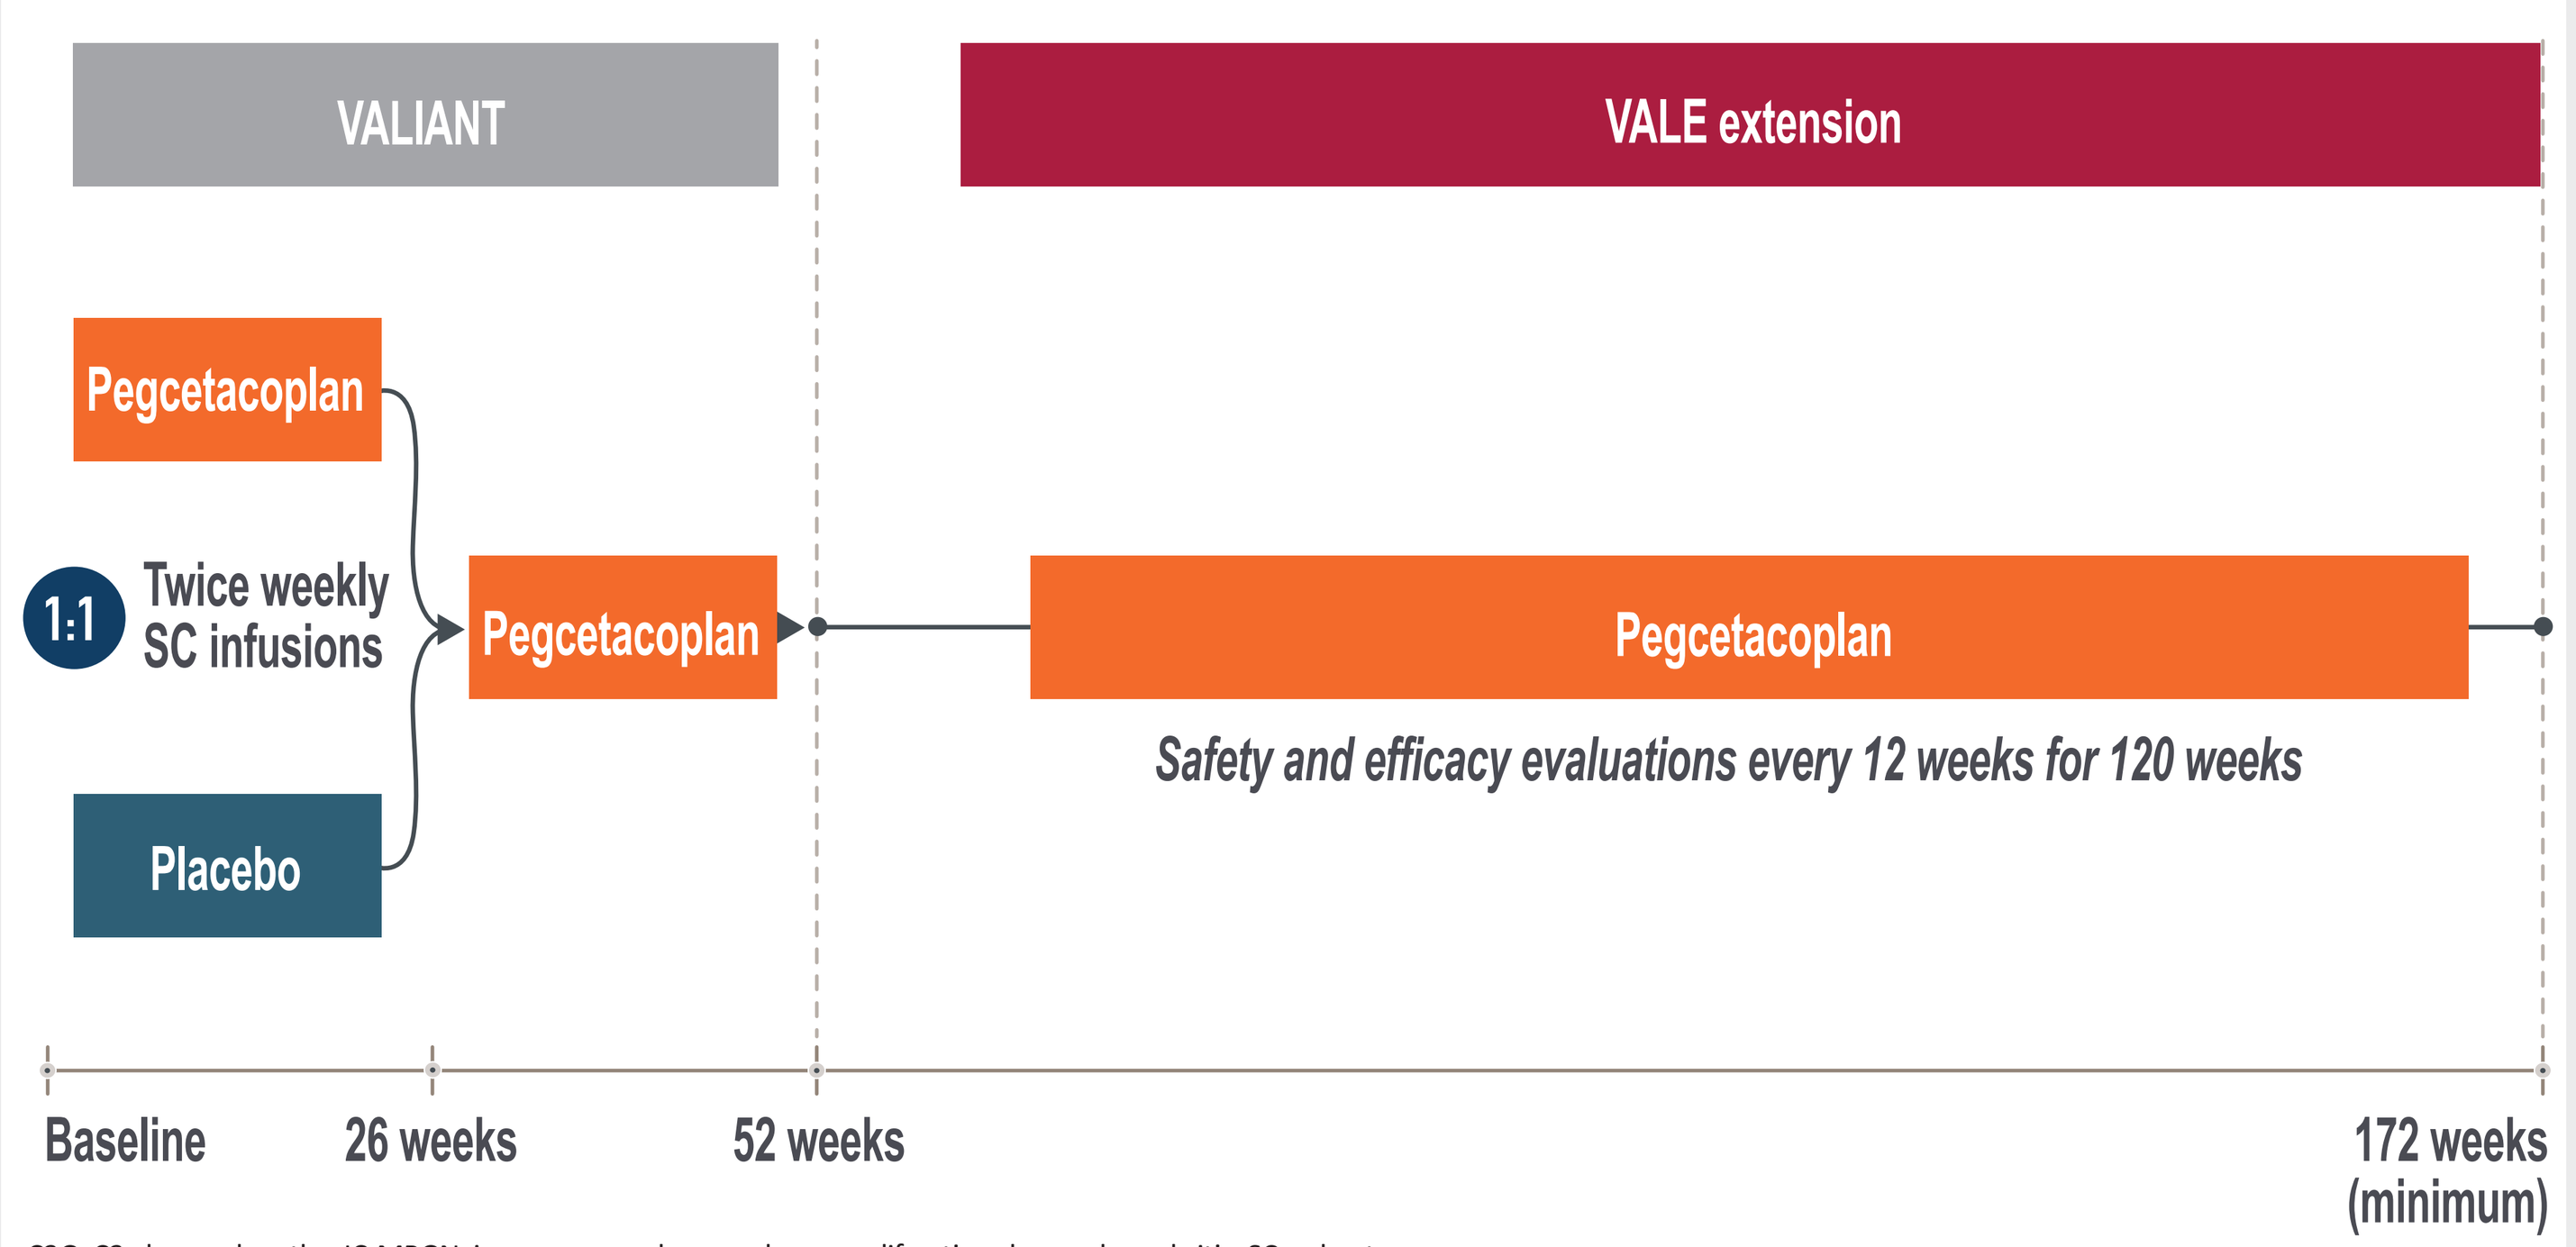
VALIANT
VALE extension
Pegcetacoplan
1:1
Twice weekly SC infusions
Pegcetacoplan
Placebo
Pegcetacoplan
Safety and efficacy evaluations every 12 weeks for 120 weeks
Baseline
26 weeks
52 weeks
172 weeks (minimum)

C3G, C3 glomerulopathy; IC-MPGN, immune-complex membranoproliferative glomerulonephritis; SC, subcutaneous.

- Key VALIANT inclusion and exclusion criteria and additional inclusion criteria for VALE participants are shown in **Table 1**

METHODS *(cont.)*

Table 1. Key inclusion and exclusion criteria for VALIANT and key inclusion criteria for VALE

VALIANT Key inclusion criteria	VALIANT Key exclusion criteria
▪ Adolescents (12-17 y) weighing ≥30 kg or adults (≥18 y) ▪ Diagnosis of primary C3G or IC-MPGN (with or without previous renal transplant) ▪ Evidence of active renal disease ▪ Willingness and ability to self-administer pegcetacoplan or have a specified caregiver available to administer pegcetacoplan ▪ Vaccination against *Streptococcus pneumoniae*, *Neisseria meningitidis* (types A, C, W, Y, and B), and *Haemophilus influenzae* (type B) were required to be initiated ≥14 days prior to randomization; it was recommended for all series to be completed with coadministration of prophylactic antibiotics while receiving pegcetacoplan per the ACIP immunization schedule for complement deficiencies	▪ Previous exposure to pegcetacoplan ▪ Evidence of improving kidney disease in the 8 weeks prior to screening or during the screening period ▪ C3G or IC-MPGN secondary to another condition in the opinion of the investigator ▪ Inability or unwillingness to follow the study protocol ▪ Evidence of ongoing drug or alcohol abuse or dependence, per investigator opinion ▪ Presence of a condition that creates an undue risk for the participant or will confound interpretation of the study, per investigator opinion

VALE Key inclusion criteria
▪ Completed the VALIANT trial through week 52 ▪ Experienced clinical benefit from pegcetacoplan in the VALIANT trial, per investigator opinion ▪ Must remain on a stable regimen for treatment according to the requirements of the VALIANT trial ▪ Vaccinated per VALIANT requirements and agreed to receive additional vaccinations recommended by ACIP or other similar local applicable guidelines for adults or children with complement deficiencies and/or immunocompromising conditions

ACIP, Advisory Committee on Immunization Practices; C3G, C3 glomerulopathy; IC-MPGN, immune-complex membranoproliferative glomerulonephritis.

- Pegcetacoplan dosages were determined based on participant age, with adjustments for body weight in adolescents (ie, pediatric patients 12–17 years of age) (**Table 2**)

Table 2. Pegcetacoplan SC dosage regimen for participants in the VALE extension trial

Age, body weight	Pegcetacoplan dosage
Adults (18 years of age and older)	
▪ All weights	▪ 1080 mg twice weekly
Adolescent patients 12–17 years of age[a]	
▪ ≥50 kg	▪ 1080 mg twice weekly
▪ 35 to <50 kg	▪ 810 mg twice weekly
▪ 30 to <35 kg	▪ 648 mg twice weekly

SC, subcutaneous.
[a]Dosing will be adjusted as appropriate for adolescents/pediatric participants who enter adulthood or who have weight changes during the study.

METHODS *(cont.)*

- Key end points are shown in **Table 3**

Table 3. Key outcomes to be assessed in the VALE extension trial

Key outcomes
Primary efficacy end point
▪ Log-transformed ratio of UPCR over time versus pretreatment baseline
Secondary efficacy end points
▪ Change from pretreatment in eGFR values over time ▪ Proportion of patients with a UPCR reduction of ≥50% from the pretreatment value over time ▪ Proportion reaching proteinuria of <1 g/day over time ▪ Change from pretreatment in the FACIT-Fatigue Scale score over time ▪ Time to progression to a composite clinical outcome (≥1 of the following: sustained doubling of serum creatinine, progression to chronic kidney disease stage 5 or ESRD, kidney transplant, death from kidney or cardiovascular disease)
Key safety end points
▪ Incidence and severity of TEAEs and changes from baseline in vital signs, clinical laboratory tests, and ECG results

ECG, electrocardiogram; eGFR, estimated glomerular filtration rate; ESRD, end-stage renal disease; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy – Fatigue; TEAEs, treatment-emergent adverse events; UPCR, urine protein-to-creatinine ratio.

RESULTS

- Of 114 patients who completed VALIANT, 113 enrolled in VALE
- As of June 2025, 101 of the 113 (89%) patients who entered VALE remain in VALE
- Results of this ongoing study are not yet available

References: 1. Kidney Disease: Improving Global Outcomes (KDIGO) Glomerular Diseases Work Group. *Kidney Int*. 2021;100(4S):S1–276. **2.** Medjeral-Thomas NR, et al. *Clin J Am Soc Nephrol*. 2014;9:46–53. **3.** Kovala M, et al. *Cells*. 2023;12(5):712. **4.** Noris M, et al. *Nephrol Dial Transplant*. 2023;38:283–90. **5.** Dixon B, et al. *Kidney Int Rep*. 2023;8(11):2284–93. **6.** Smith RJH, et al. *Nat Rev Nephrol*. 2019;15:129–43. **7.** Fakhouri F, et al. In progress. **8.** Empaveli® Prescribing information. Apellis; 2025. **9.** ClinicalTrials.gov. VALE. Accessed August 21, 2025. clinicaltrials.gov/study/NCT05809531.

ABBREVIATIONS: C3G, C3 glomerulopathy; ECG, electrocardiogram; eGFR, estimated glomerular filtration rate; ESRD, end-stage renal disease; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy – Fatigue; IC-MPGN, immune-complex membranoproliferative glomerulonephritis; SC, subcutaneous; TEAEs, treatment-emergent adverse events; UPCR, urine protein-to-creatinine ratio.

Disclosures: **CN** is the Associate Director for Molecular Otolaryngology and Renal Research Laboratory; receives NIH grant support (2R01DK110023-07); serves on advisory boards for Novartis, Apellis, Biocryst, and Alexion; participates as a site investigator for Novartis, Apellis, Biocryst, and Retrophin; is a member of the data safety monitoring board for Kira; serves as Chair of a data safety monitoring board for FIT4KID; and receives author royalties for UpToDate. **GA** serves on advisory boards for Alexion, AstraZeneca Rare Diseases, Dicerna, Alnylam, Chiesi, and Advicenne; and participates as a site investigator for Apellis. **AM** and **EK** are employees of Apellis and hold stock options. **JS** and **LLL** are employees of Swedish Orphan Biovitrum AB and hold stock options.

ACKNOWLEDGMENTS Research funding source: This study was funded by Apellis Pharmaceuticals, Inc and Sobi (Swedish Orphan Biovitrum AB). Writing assistance was provided by Kathryn Fogarty, PhD, of Kay Square Scientific (Butler, PA, USA), and was funded by Apellis Pharmaceuticals, Inc and Sobi (Swedish Orphan Biovitrum AB).

58th Annual Meeting of the American Society of Nephrology (ASN 2025) | November 5–9, 2025 | Houston, TX