Joint Health Outcomes With Efanesoctocog Alfa Prophylaxis for Patients With Severe Hemophilia A: Third Interim Analysis of the Phase 3 XTEND-ed Study

Christoph Königs,[1] Annette von Drygalski,[2] Cedric Hermans,[3] Barbara A. Konkle,[4] María Teresa Álvarez Román,[5] Liane Khoo,[6] Nobuaki Suzuki,[7] Graham Neill,[8] Linda Bystrická,[9] Meredith Foster,[10] Elena Santagostino,[9] Sriya Gunawardena,[11] Robert Klamroth[12]

[1]Goethe University, Department of Pediatrics and Adolescent Medicine, University Hospital, Frankfurt, Germany; [2]Division of Hematology/Oncology, Department of Medicine, University of California, San Diego, San Diego, CA, USA; [3]Division of Haematology, Haemostasis and Thrombosis Unit, Saint-Luc University Hospital, Université Catholique de Louvain, Brussels, Belgium; [4]Washington Center for Bleeding Disorders and the University of Washington, Seattle, WA, USA; [5]Thrombosis and Haemostasis Unit, Hospital Universitario La Paz, Universidad Autónoma de Madrid, Madrid, Spain; [6]Institute of Haematology, Royal Prince Alfred Hospital, Sydney, NSW, Australia; [7]Department of Transfusion Medicine, Nagoya University Hospital, Aichi, Japan; [8]Sanofi, Cambridge, UK; [9]Sobi, Basel, Switzerland; [10]Sanofi, Cambridge, MA, USA; [11]Sanofi, Morristown, NJ, USA; [12]Vivantes Klinikum, Friedrichshain, Berlin, Germany

HJHS total score
improved or remained constant in
85.6%
of evaluable participants (n=90) from XTEND-1 baseline to XTEND-ed Month 36

Plain language summary

1 **This research studied joint health in adults and teenagers with severe hemophilia A who took efanesoctocog alfa once weekly for up to 4 years in the XTEND clinical trial program**

2 **Approximately 8 out of 10 people taking efanesoctocog alfa experienced stable or improved joint health**

Introduction

- Efanesoctocog alfa is a first-in-class, high-sustained factor VIII (FVIII) replacement therapy (also referred to as ultra-long half-life FVIII) that overcomes the von Willebrand factor–imposed half-life ceiling[1-3]
- The Phase 3 XTEND-1 study[4] (NCT04161495) showed once-weekly efanesoctocog alfa (50 IU/kg) prophylaxis was well tolerated in adults/adolescents with severe hemophilia A, providing superior bleed prevention over prior factor VIII prophylaxis
- Participants completing XTEND-1 could continue weekly efanesoctocog alfa (50 IU/kg) in the long-term extension study, XTEND-ed (NCT04644575) Arm A
- The second interim analysis of XTEND-ed showed that overall improvements in total Hemophilia Joint Health Score (HJHS) observed after 12 months of efanesoctocog alfa prophylaxis in XTEND-1 were maintained at Month 24[5]

Objective

- To report long-term joint health with efanesoctocog alfa in adults/adolescents aged ≥12 years from XTEND-1 baseline through XTEND-ed third interim analysis (data cut: February 21, 2025)

Methods

Study design and population

- Participants provided informed consent; the study was approved by applicable review boards
- The XTEND clinical trial program is shown in **Figure 1**

Figure 1: XTEND clinical trial program

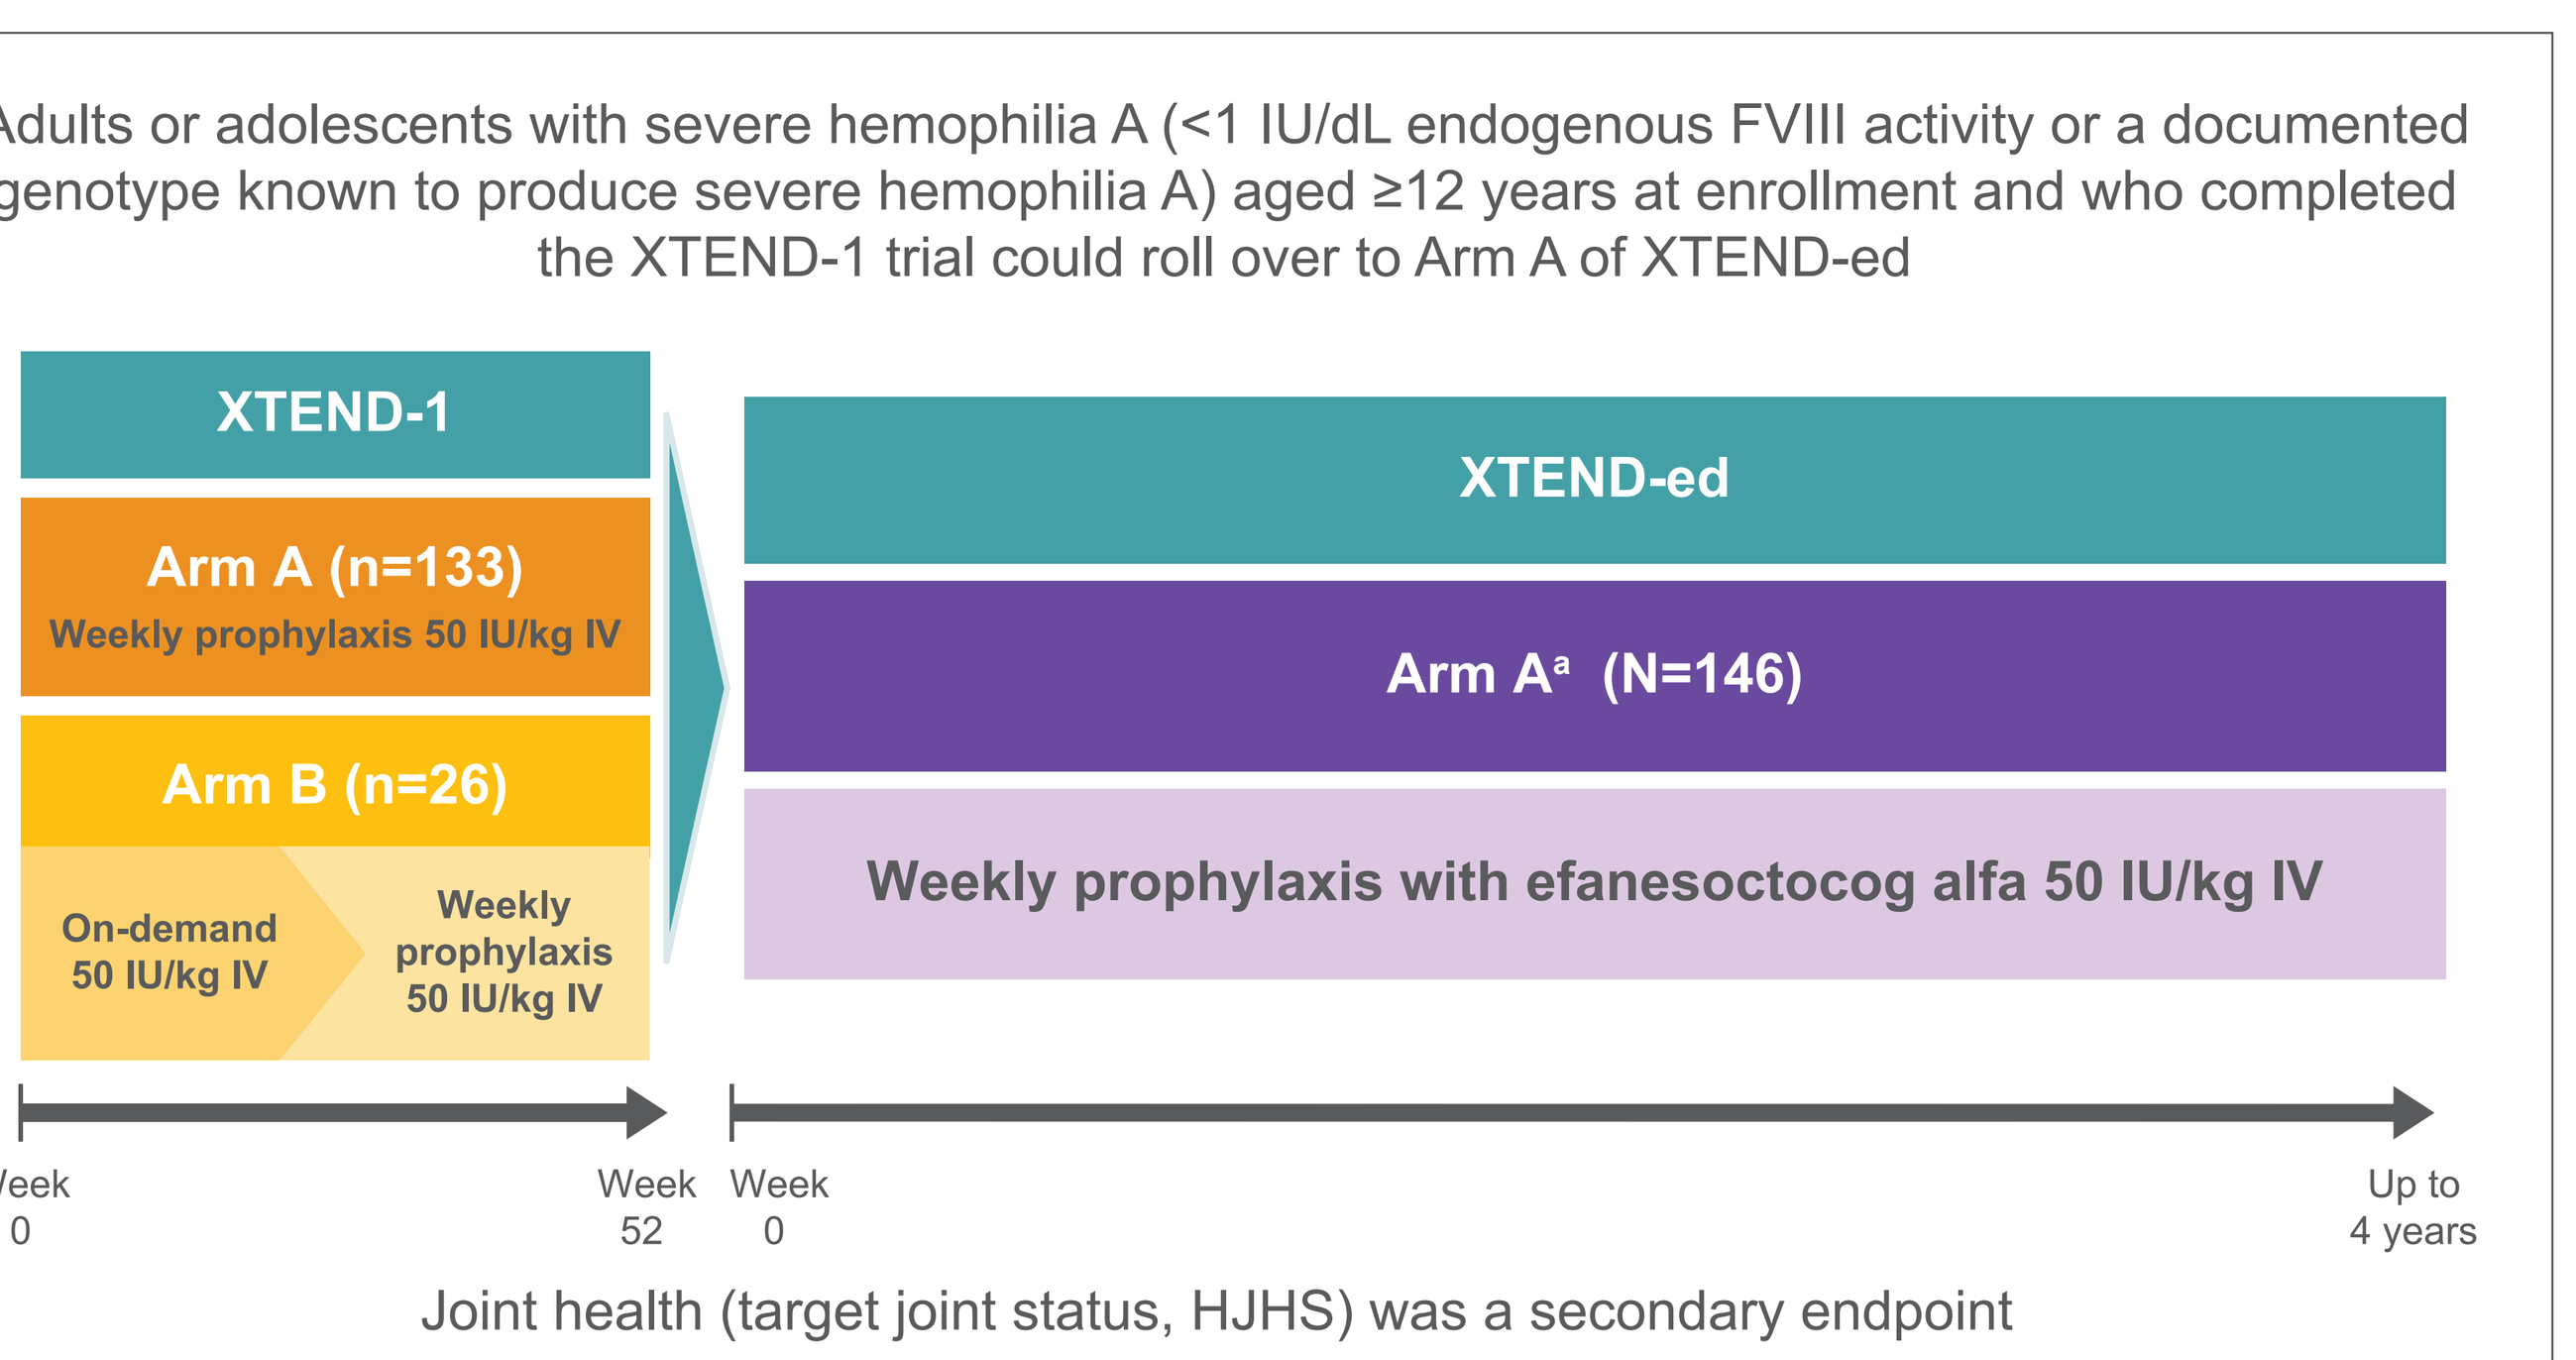
Adults or adolescents with severe hemophilia A (<1 IU/dL endogenous FVIII activity or a documented genotype known to produce severe hemophilia A) aged ≥12 years at enrollment and who completed the XTEND-1 trial could roll over to Arm A of XTEND-ed
XTEND-1
Arm A (n=133)
Weekly prophylaxis 50 IU/kg IV
Arm B (n=26)
On-demand 50 IU/kg IV
Weekly prophylaxis 50 IU/kg IV
XTEND-ed
Arm A[a] (N=146)
Weekly prophylaxis with efanesoctocog alfa 50 IU/kg IV
Week 0
Week 52
Week 0
Up to 4 years
Joint health (target joint status, HJHS) was a secondary endpoint

[a]Patients in XTEND-ed Arm A will continue to receive efanesoctocog alfa prophylaxis for up to 4 years, unless efanesoctocog alfa is commercially available in their participating country.
FVIII, factor VIII; HJHS, Hemophilia Joint Health Score; IU, international unit; IV, intravenous.

Outcome measures

- Changes in HJHS (range 0–124) from parent study (XTEND-1) baseline up to XTEND-ed Month 36 are presented descriptively
- Higher HJHS denotes worse joint health
- Individual joints (left/right elbows, knees, ankles) were scored according to 8 HJHS domains to obtain total joint score; gait score was determined based on walking and climbing stairs and ranged from 0 to 4
 - HJHS assessments within 2 weeks after a joint or muscle bleed were excluded
 - Joint scores following joint surgeries were replaced using the last observation carried forward method
- Improved/worsened HJHS was defined as decrease/increase of ≥4 points, and unchanged (or constant) HJHS as decrease/increase of -3 to +3 points[6]

Results

- Overall, 146 adults/adolescents rolled over from XTEND-1 to Arm A of XTEND-ed (**Table 1**)
 - Median (range) treatment duration and exposure days from XTEND-1 baseline were 212.2 (46.3–244.8) weeks and 216.5 (47–254) days, respectively

Table 1: Participant demographics and baseline characteristics

	Overall cohort (N=146)
Mean (SD) age (at XTEND-ed enrollment), years	37.0 (15.1)
Age category (at XTEND-ed enrollment), n (%)	
12–17 years	21 (14.4)
18–64 years	120 (82.2)
≥65 years	5 (3.4)
Sex, n (%)	
Male	145 (99.3)
Female	1 (0.7)
Race, n (%)	
White	100 (68.5)
Black or African American	4 (2.7)
Asian	27 (18.5)
Not reported	13 (8.9)
Other	2 (1.4)
Mean (SD) weight, kg	78.0 (19.4)
Mean (SD) body mass index, kg/m²	25.6 (5.2) *(n=145)*

HJHS total and domain scores

- In total, 132/146 participants had evaluable HJHS scores at XTEND-1 baseline
 - Median (range) HJHS total score was 19 (0–98)
 - Mean (standard deviation [SD]) HJHS total score was 20.5 (18.4)
 - Gradual improvements in HJHS total score at 12-month intervals were observed with up to 4 years of efanesoctocog alfa prophylaxis (**Figure 2**)

Figure 2: Change in HJHS total score from XTEND-1 baseline to XTEND-ed Month 36 according to target joint status

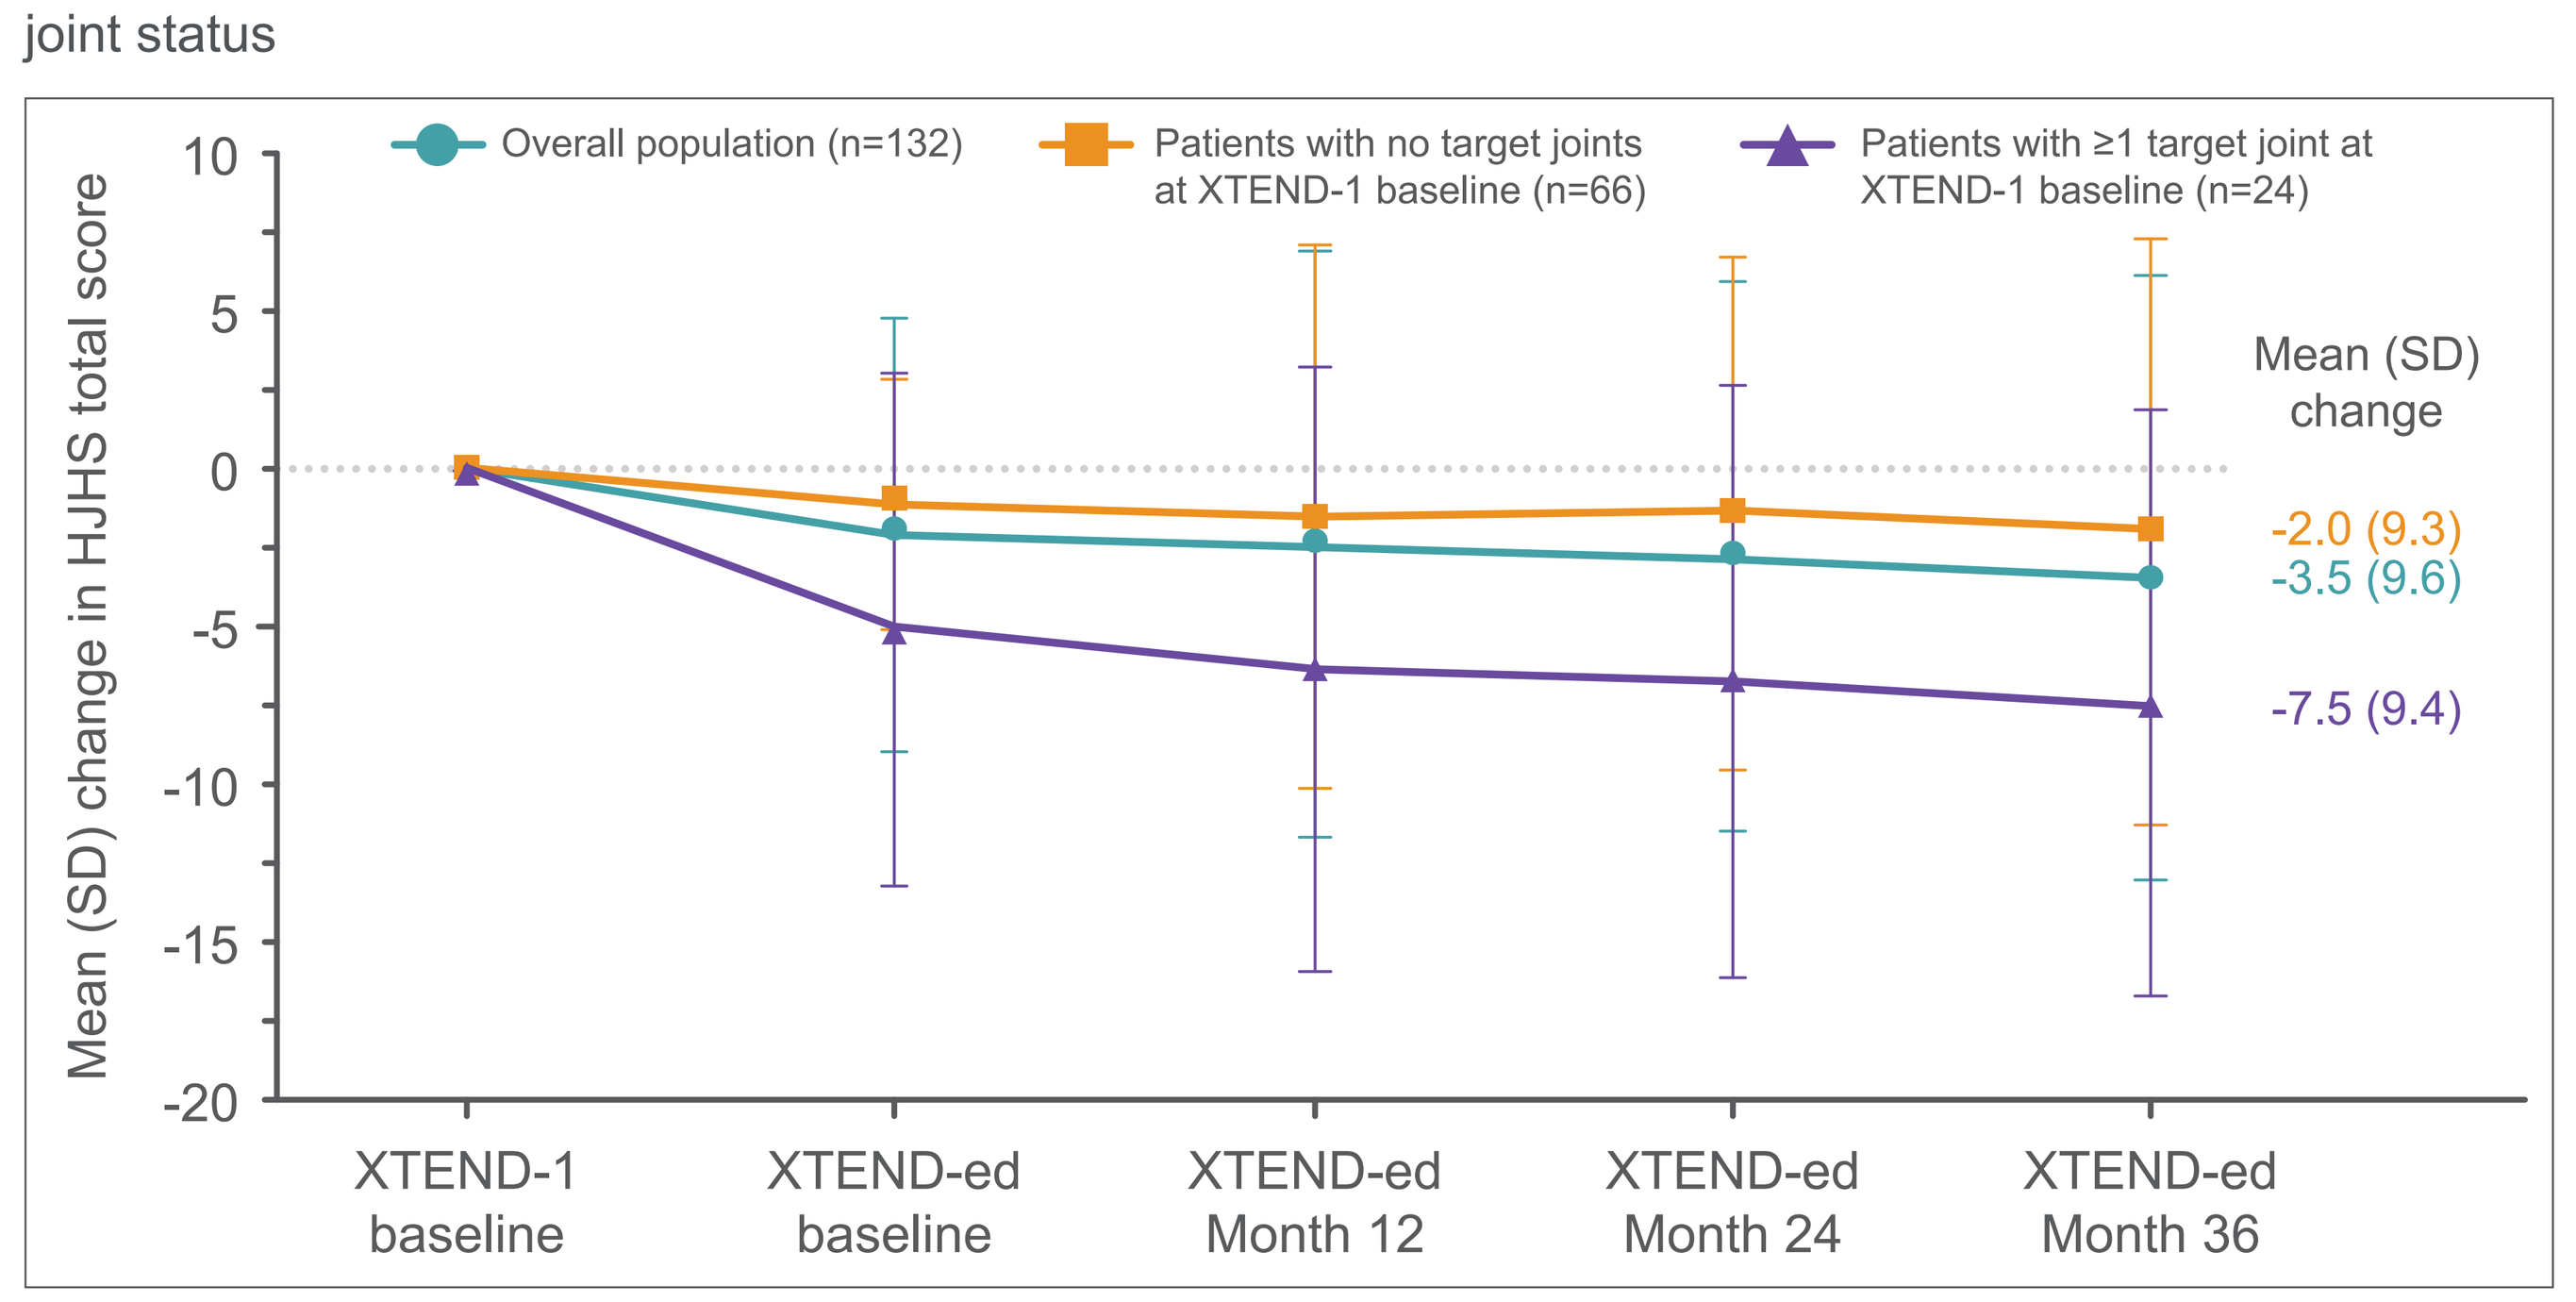
Overall population (n=132)
Patients with no target joints at XTEND-1 baseline (n=66)
Patients with ≥1 target joint at XTEND-1 baseline (n=24)
Mean (SD) change in HJHS total score
Mean (SD) change
-2.0 (9.3)
-3.5 (9.6)
-7.5 (9.4)
XTEND-1 baseline
XTEND-ed baseline
XTEND-ed Month 12
XTEND-ed Month 24
XTEND-ed Month 36

Mean (SD) HJHS Total Score (0–124) Overall population	XTEND-1 Baseline		XTEND-ed Baseline		XTEND-ed Month 12		XTEND-ed Month 24		XTEND-ed Month 36	
	N	Mean (SD)	N	Mean (SD)	N	Mean (SD)	N	Mean (SD)	N	Mean (SD)
Total score	132	20.5 (18.4)	133	17.6 (17.1)	114	16.5 (15.9)	122	17.0 (16.7)	96	14.8 (14.9)
Change from XTEND-1 baseline	–	–	128	-2.1 (6.9)	107	-2.4 (9.3)	115	-2.8 (8.7)	90	-3.5 (9.6)

An HJHS total score could be calculated if all 48 individual item scores (8 domains × 6 joints, total joint score) and the gait score were present.
HJHS, Hemophilia Joint Health Score; SD, standard deviation.

Conclusions

1 **Long-term once-weekly prophylaxis with efanesoctocog alfa for up to 4 years continues to improve or maintain joint health in adults and adolescents with severe hemophilia A**

2 **The greatest improvements in HJHS total score were observed in people with the worst joint health at XTEND-1 baseline (HJHS >29; quartile 4) and those aged ≥50 years**

- At Month 36, relative to XTEND-1 baseline:
 - Greatest mean (SD) changes in HJHS domain scores were observed for strength -0.6 (2.6) and swelling -0.5 (1.7) (**Figure 3**)
 - Mean (SD) HJHS gait score remained constant at 1.5 (1.6) (XTEND-1 baseline, 1.7 [1.6])
 - HJHS total score improved or remained constant[6] in 85.6% of participants (77/90)

Figure 3: Change in HJHS domain scores from XTEND-1 baseline to XTEND-ed Month 36

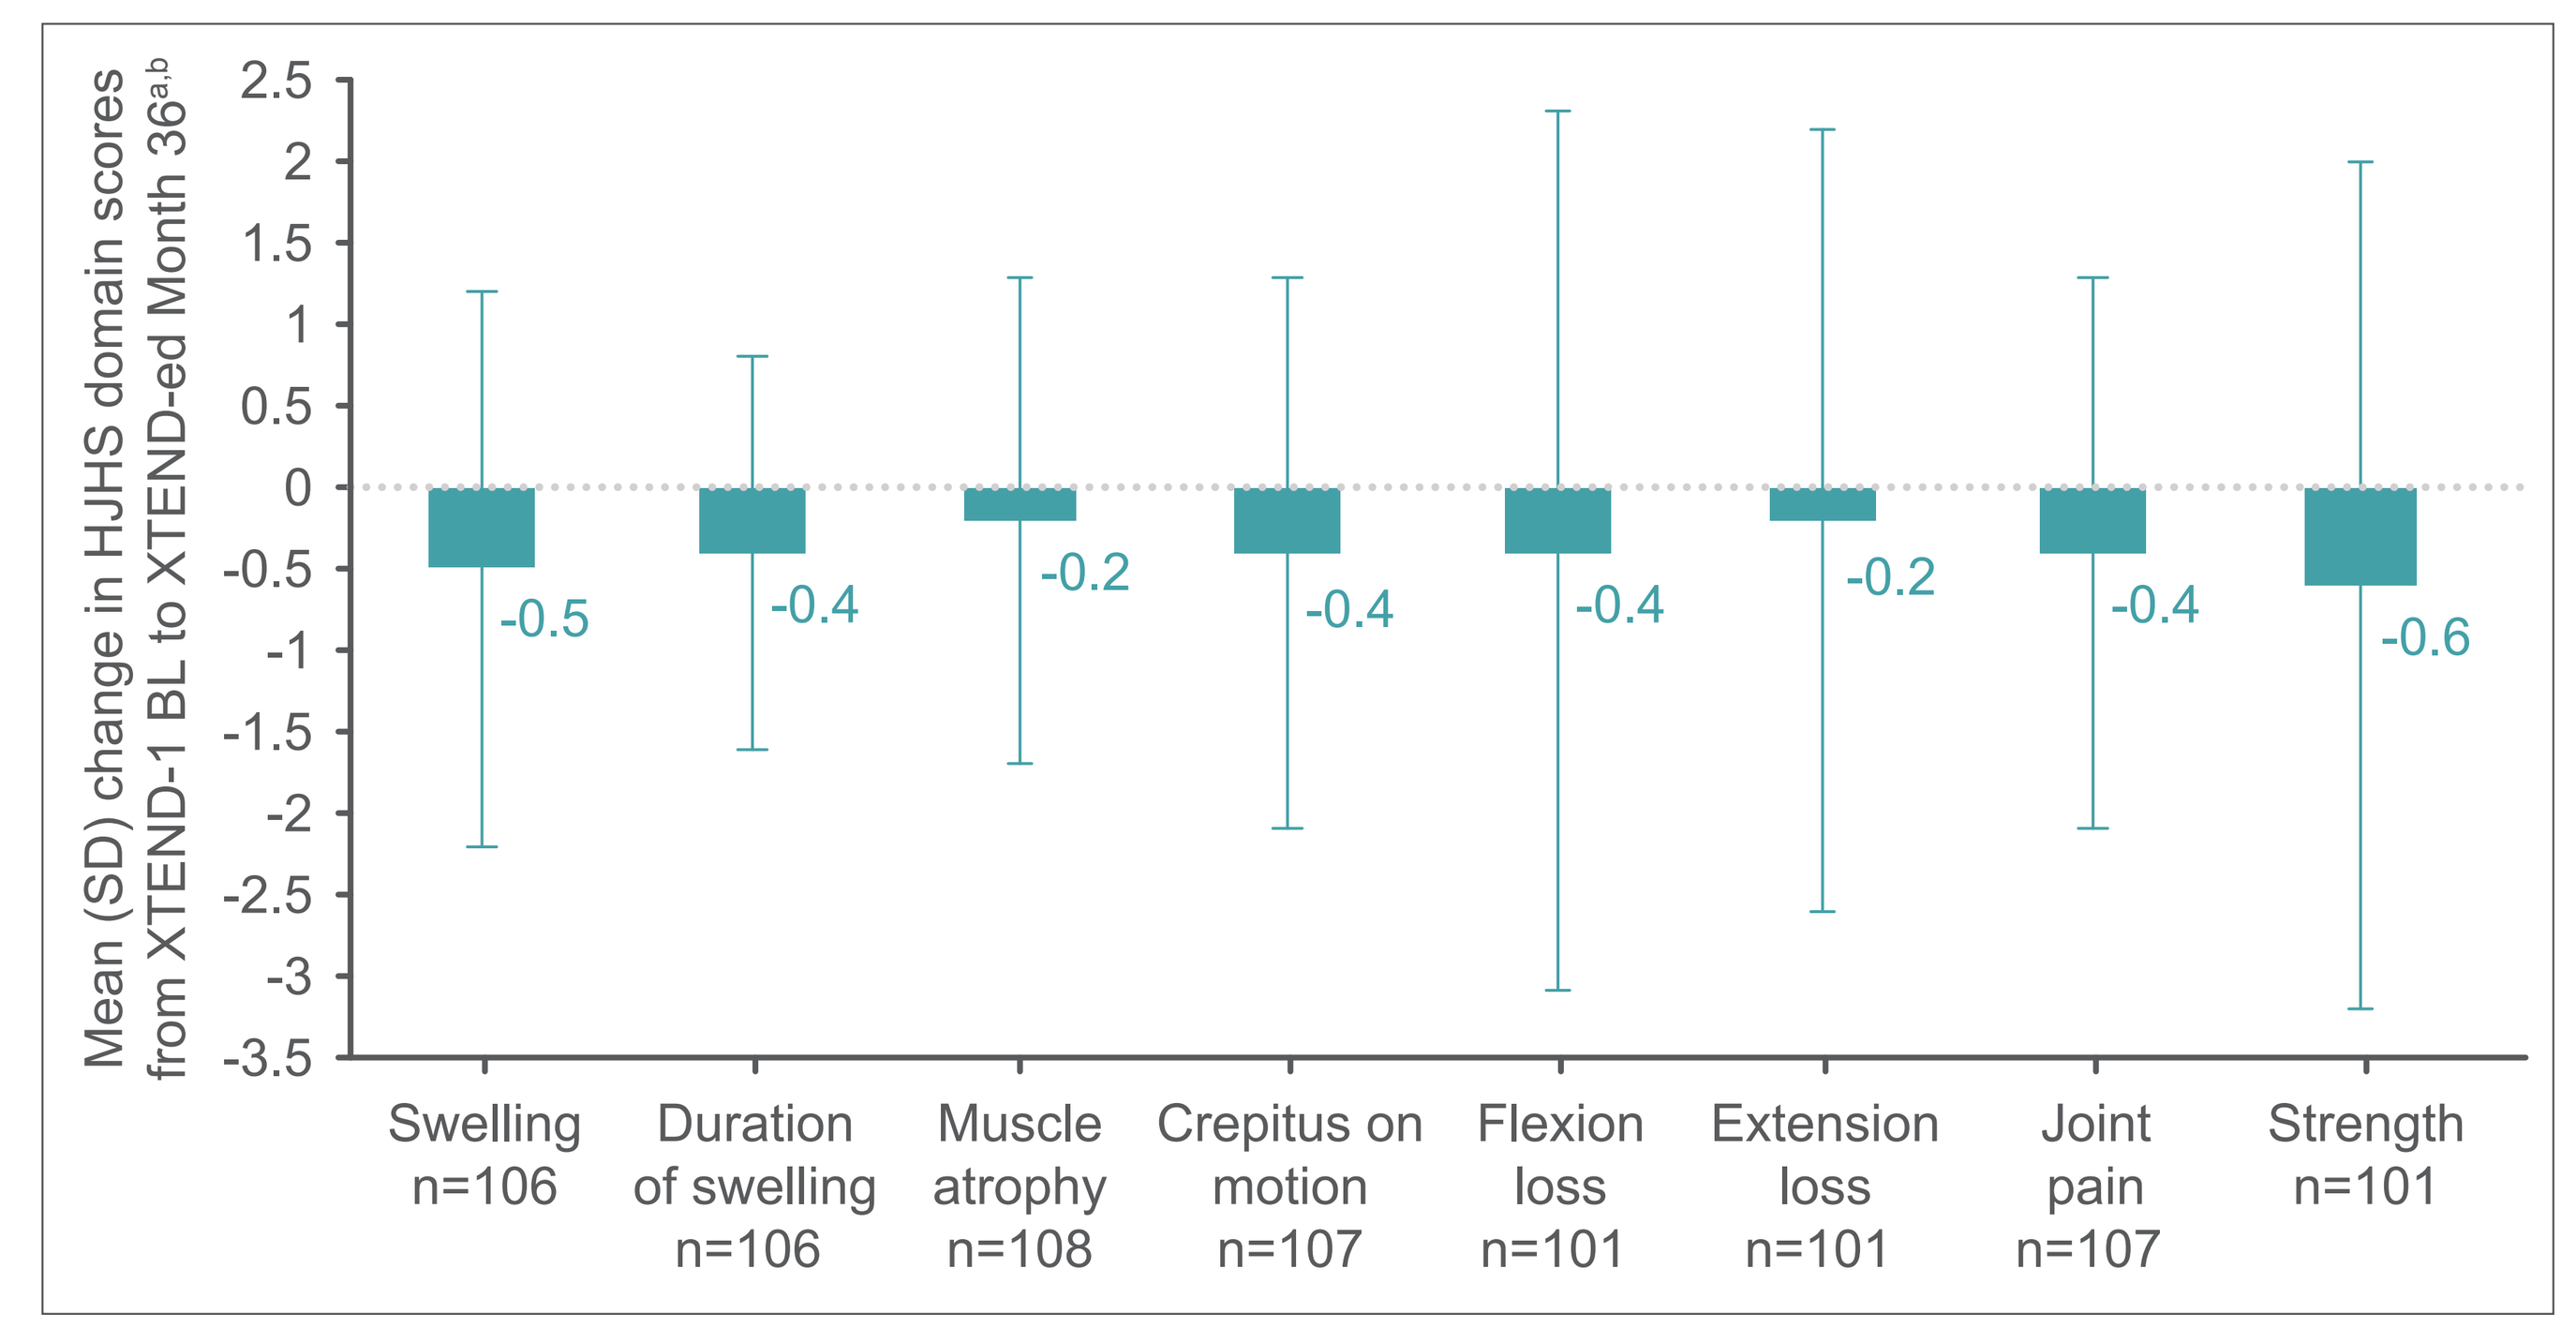
Mean (SD) change in HJHS domain scores from XTEND-1 BL to XTEND-ed Month 36[a,b]
-0.5
-0.4
-0.2
-0.4
-0.4
-0.2
-0.4
-0.6
Swelling n=106
Duration of swelling n=106
Muscle atrophy n=108
Crepitus on motion n=107
Flexion loss n=101
Extension loss n=101
Joint pain n=107
Strength n=101

[a]Mean change only determined for participants for whom data were available at XTEND-1 baseline and at each respective XTEND-ed follow-up time; [b]HJHS domains are assessed on the following ranges of scores: swelling (0–3); duration of swelling (0 or 1); muscle atrophy (0–2); crepitus on motion (0–2); flexion loss (0–3); extension loss (0–3); joint pain (0–2); strength (0–4).
BL, baseline; HJHS, Hemophilia Joint Health Score; SD, standard deviation.

Change in HJHS total score according to baseline HJHS quartile

- The greatest improvements in HJHS total score were observed in participants with the highest HJHS (>29; quartile 4) at XTEND-1 baseline (**Figure 4**)

Figure 4: Mean change in HJHS total score per XTEND-1 baseline HJHS quartile

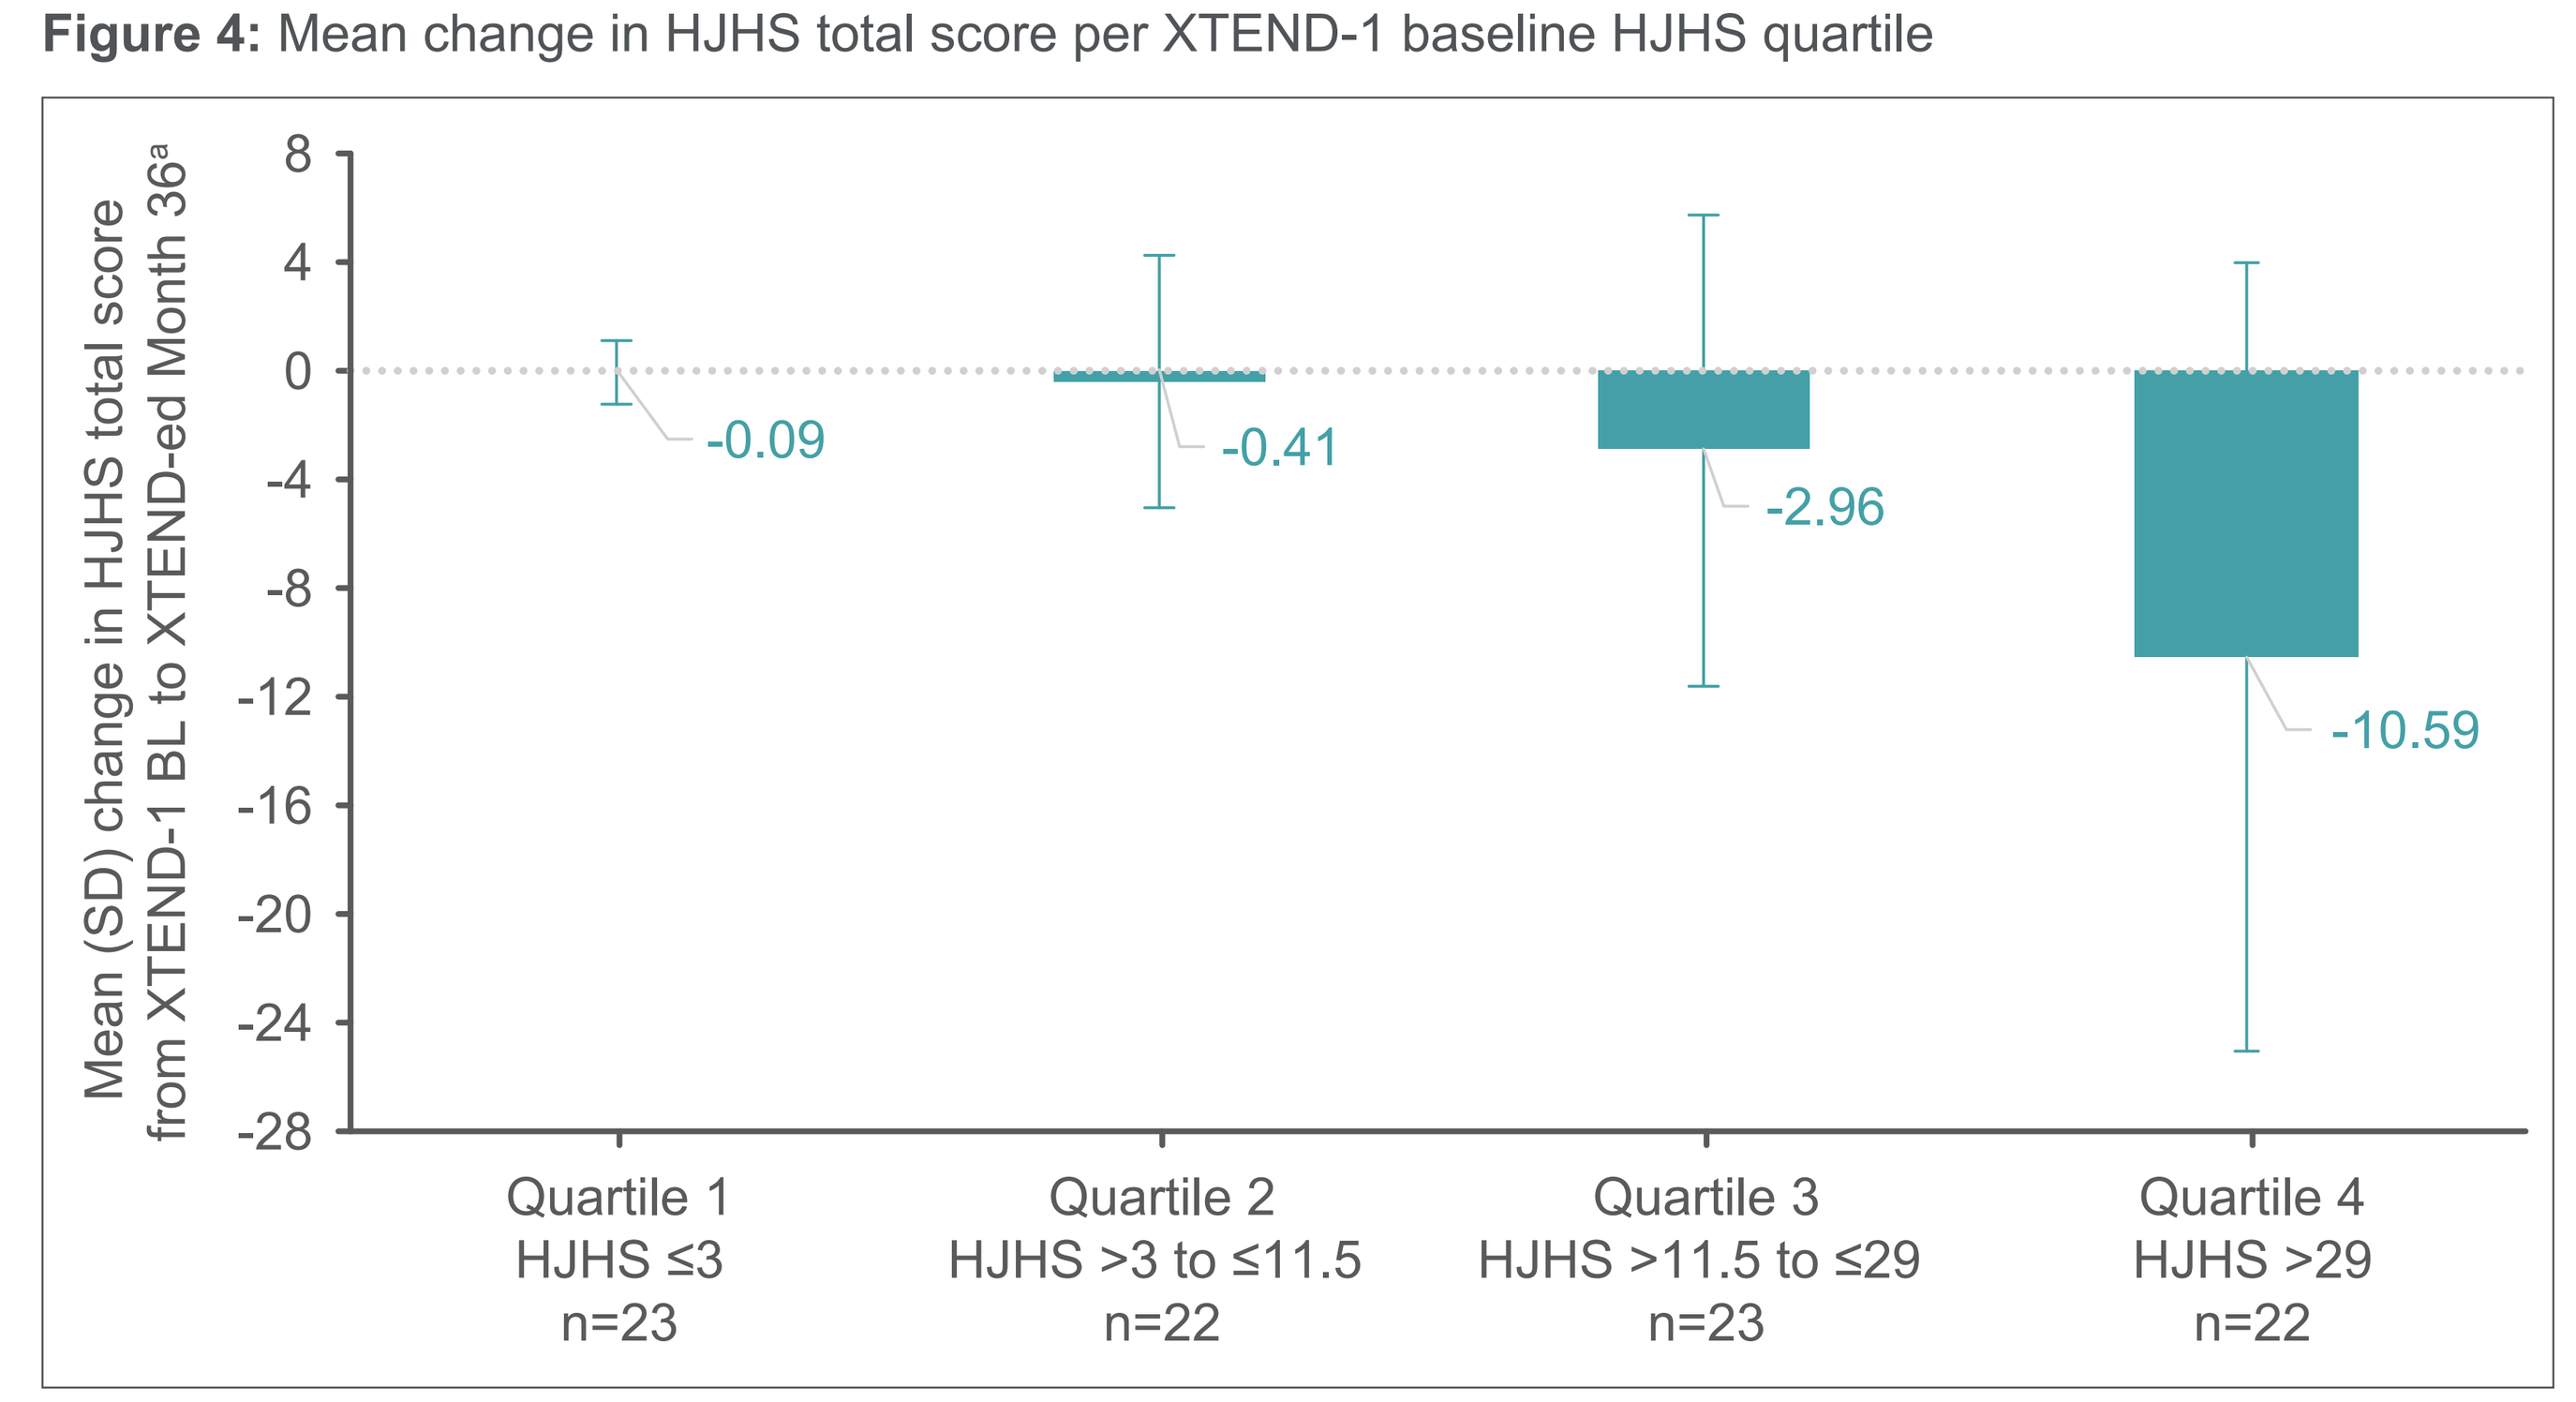
Mean (SD) change in HJHS total score from XTEND-1 BL to XTEND-ed Month 36[a]
-0.09
-0.41
-2.96
-10.59
Quartile 1 HJHS ≤3 n=23
Quartile 2 HJHS >3 to ≤11.5 n=22
Quartile 3 HJHS >11.5 to ≤29 n=23
Quartile 4 HJHS >29 n=22

Mean (SD) HJHS Total Score (0–124)	XTEND-1 Baseline HJHS Quartile			
	Quartile 1	Quartile 2	Quartile 3	Quartile 4
XTEND-1 Baseline	0.61 (0.94)	7.09 (2.54)	22.04 (5.51)	41.45 (14.82)
XTEND-ed Month 36	0.52 (0.95)	6.68 (5.40)	19.09 (9.20)	30.86 (11.78)

Higher HJHS score denotes worse joint health. aIncludes patients who had HJHS data at both XTEND-1 baseline and XTEND-ed Month 36.
BL, baseline; HJHS, Hemophilia Joint Health Score; SD, standard deviation.

Change in HJHS total score according to baseline age

- The greatest improvements in HJHS total score were reported among participants aged ≥50 years, followed by those aged 30 to 39 years (**Figure 5**)
- Mean (SD) change in HJHS total score from XTEND-1 baseline to XTEND-ed Month 36 was -2.90 (6.7; median, 0) in participants aged <40 years (n=63) and -4.78 (14.3; median, 3.0) in participants aged ≥40 years (n=27)

Figure 5: Mean change in HJHS total score per XTEND-1 baseline age

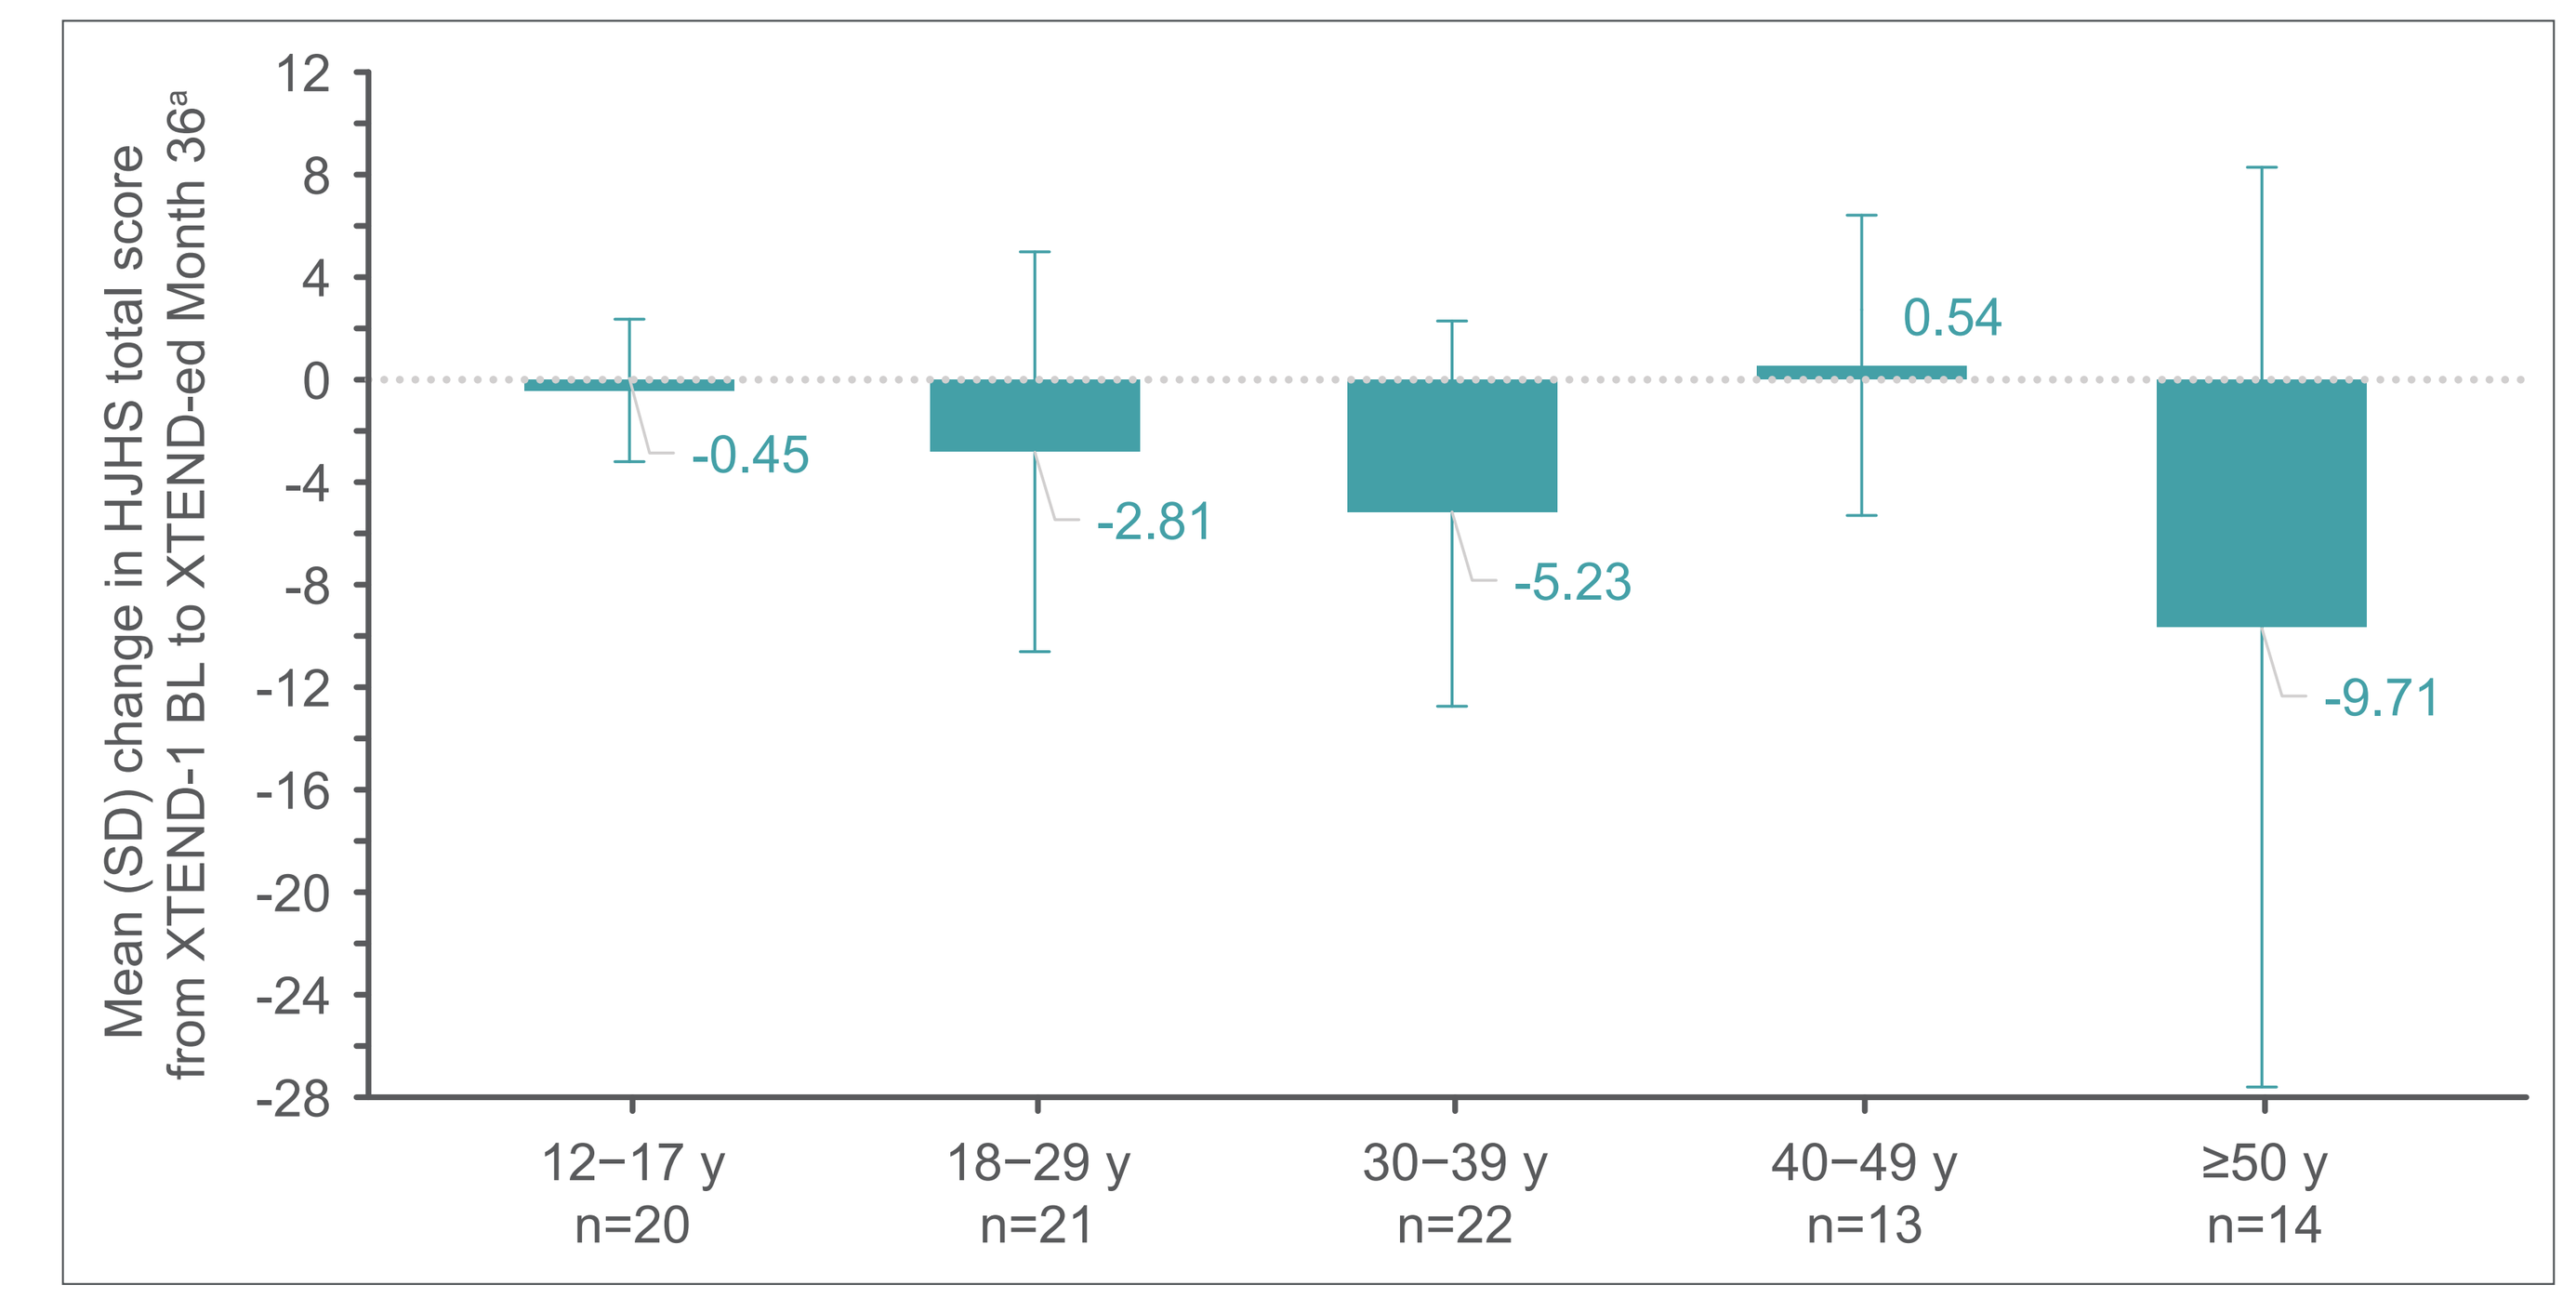
Mean (SD) change in HJHS total score from XTEND-1 BL to XTEND-ed Month 36[a]
-0.45
-2.81
-5.23
0.54
-9.71
12–17 y n=20
18–29 y n=21
30–39 y n=22
40–49 y n=13
≥50 y n=14

Age group at XTEND-1 Baseline	Mean (SD) HJHS Total Score (0–124)	
	XTEND-1 Baseline	XTEND-ed Month 36
12–17 y	3.25 (5.76)	2.80 (6.73)
18–29 y	9.24 (11.07)	6.43 (6.71)
30–39 y	20.68 (12.59)	15.45 (11.13)
40–49 y	22.23 (12.88)	22.77 (13.91)
≥50 y	41.86 (19.15)	32.14 (10.97)

Higher HJHS score denotes worse joint health. [a]Includes patients who had HJHS data at both XTEND-1 baseline and XTEND-ed Month 36.
BL, Baseline; HJHS, Hemophilia Joint Health Score; SD, standard deviation; y, year.

References

1. Chhabra ES, et al. *Blood*. 2020;135(17):1484-1496.
2. Konkle BA, et al. *N Engl J Med*. 2020;383(11):1018-1027.
3. Lissitchkov T, et al. *Blood Adv*. 2022;6(4):1089-1094.
4. von Drygalski A, et al. *N Engl J Med*. 2023;388(4):310-318.
5. Königs C, et al. *Res Pract Thromb Haemost*. 2025;9(Suppl 2):e102923.
6. Kuijlaars IAR, et al. *Haemophilia*. 2017;23(6):934-940.

Funding

This study and medical writing support were funded by Sanofi and Sobi.

Acknowledgments

Medical writing support for the development of this poster was provided Kimberly Fung, PhD, CMPP™, of IMPRINT Science, and was funded by Sanofi and Sobi. The authors thank Monique Bidell, PharmD, CMPP™ (Sanofi), and Nick Fulcher, PhD, CMPP™ (Sobi), for publication coordination, and Alejandro Fernández (Sanofi) for early guidance on the concept. Sanofi and Sobi personnel reviewed this poster. The authors had full editorial control of the poster and provided their final approval of all content.

Disclosures

CK has received personal fees (speaker/advisory boards) from Bayer, CSL Behring, Florio, Novo Nordisk, Pfizer, Roche/Chugai, Sanofi/Sobi, and Takeda; research and/or clinical trial support from Bayer, Biotest, CSL Behring, EU H2020 ITN, Haemophilia Foundation, Intersero, Novo Nordisk, Pfizer, Roche/Chugai, Sanofi/Sobi, and Takeda. **AvD** has received fees from BioMarin, Bioverativ/Sanofi-Genzyme, Novo Nordisk, Takeda, and uniQure for participation in industry-sponsored educational events and advisory boards. She has received research funding from Bioverativ/Sanofi-Genzyme and Pfizer. She is co-founder and a member of the board of directors of Hematherix Inc. She holds a patent for a super FVa, and she is the inventor and physician lead for the Joint Activity and Damage Examination (JADE) ultrasound measurement tool. JADE is copyrighted through the Office of Innovation and Commercialization at the University of California, San Diego. **CH** has received research funding from Bayer, CSL Behring, Novo Nordisk, Pfizer, Shire/Takeda, and Sobi, and honoraria/speakers bureau fees from LFB, Bayer, CAF-DCF, CSL Behring, Hoffmann-La Roche, Novo Nordisk, Octapharma, Pfizer, Shire/Takeda, Sobi, uniQure, Regeneron, and Sanofi. **BAK** has acted as a consultant for Be Biopharma, BioMarin, HC Bioscience, Novo Nordisk, Regeneron, Pfizer, Sanofi, and Takeda, and has received research grants from CSL Behring, Pfizer, Sanofi, Takeda, and uniQure. **MTÁR** has received speakers bureau fees from Amgen, CSL Behring, Novartis, Novo Nordisk, Octapharma, Pfizer, Roche, Sobi, and Takeda, and honoraria from Amgen, CSL Behring, Novartis, Novo Nordisk, Octapharma, Pfizer, Roche, Sobi, and Takeda. **LK** has received honoraria and/or been a member of advisory committees for BioMarin, CSL Behring, Novo Nordisk, Roche/Chugai, and Sanofi. **NS** has received honoraria from Bayer, CSL Behring, Chugai, KM Biologics, Japan Blood Products Organization, Novo Nordisk, Pfizer, Sanofi, and Takeda Yakuhin. **GN, MF,** and **SG** are employees of Sanofi and may hold shares and/or stock options in the company. **LB** and **ES** are employees of Sobi and may hold shares and/or stock options in the company. **RK** has received honoraria and/or been a member of advisory committees for Bayer, BioMarin, Biotest, CSL Behring, Grifols, Novo Nordisk, Octapharma, Pfizer, Roche/Chugai, Sanofi, Sobi, and Shire/Takeda.

Presented at the International Society on Thrombosis and Haemostasis (ISTH) 2026 Congress; July 11–15, 2026; Paris, France

PB3282