QSP Modeling of Loncastuximab Tesirine-lpyl Combined with T Cell-Dependent Bispecific Antibodies Bridges Knowledge and Dose Regimen Strategy

Yuezhe Li[1], A. Katharina Wilkins[1], Jimena Davis[1], Tim Knab[1], Marie Toukam[2], Daniel C. Kirouac[1], Joseph P. Boni[2]
[1]*Metrum Research Group, Boston, MA, USA*, [2]*ADC Therapeutics America, New Providence, NJ*

METRUM RESEARCH GROUP

Abstract

Background
Antibody drug conjugates (ADCs) and T cell-dependent bispecific antibodies (TDBs) have proven single-agent efficacy in relapsed/refractory (R/R) lymphomas. Coadministering therapeutics with orthogonal mechanisms of action is a potential means of enhancing efficacy while minimizing dose-limiting toxicities. However, it is not feasible to empirically test every potential drug combination and coadministration regimen in clinical trials. Predictive, quantitative systems pharmacology (QSP) models could thus prove valuable as a means of simulating alternate treatment regimens to guide optimal clinical trial design.

Methods
An integrated systems pharmacology model of a CD19-targeted ADC (loncastuximab tesirine-lpyl, or Lonca) and CD3/CD20-targeted TDBs (mosunetuzumab, glofitamab, and epcoritamab) was developed to predict combination regimen efficacy in R/R diffuse large B cell lymphoma (DLBCL). Clinically validated models of loncastuximab tesirine and mosunetuzumab were taken from the literature, simplified, integrated while maintaining core functionality, and extended to additional TDBs. A virtual population was constructed by randomizing five key model parameters and prevalence weighting against reported monotherapy response data for the individual drugs. Combination effects on tumor volume dynamics were then simulated under multiple dosing regimens and patient population scenarios.

Results
Simulations predicted combination antitumor effects between loncastuximab tesirine and TDBs appearing by the fourth treatment cycle outperforming either TDB or loncastuximab tesirine monotherapy. Antitumor effects increased with subsequent treatment cycles in an additive manner, were insensitive to reductions in loncastuximab tesirine dose, and were maintained in lymphopenic patients.

Conclusions
The virtual population-based lymphoma QSP model enabled systematic exploration of alternate drug combinations, dosing schemes, clinical covariates and resultant effect on anti-tumor activity in silico. The results of the LOTIS-7 study (NCT04970901), a platform study evaluating loncastuximab tesirine in combination with glofitamab, will soon be available to assess the accuracy of the model predictions.

Methods

Model Construction
The loncastuximab tesirine QSP model was based on the previously validated and published loncastuximab tesirine PBPK-QSP model [1]. This model was reduced in physiological complexity to be compatible with a published QSP model of mosunetuzumab [2], while maintaining the core functionality of predicting tumor dynamics during Lonca monotherapy (**Figure 1**).

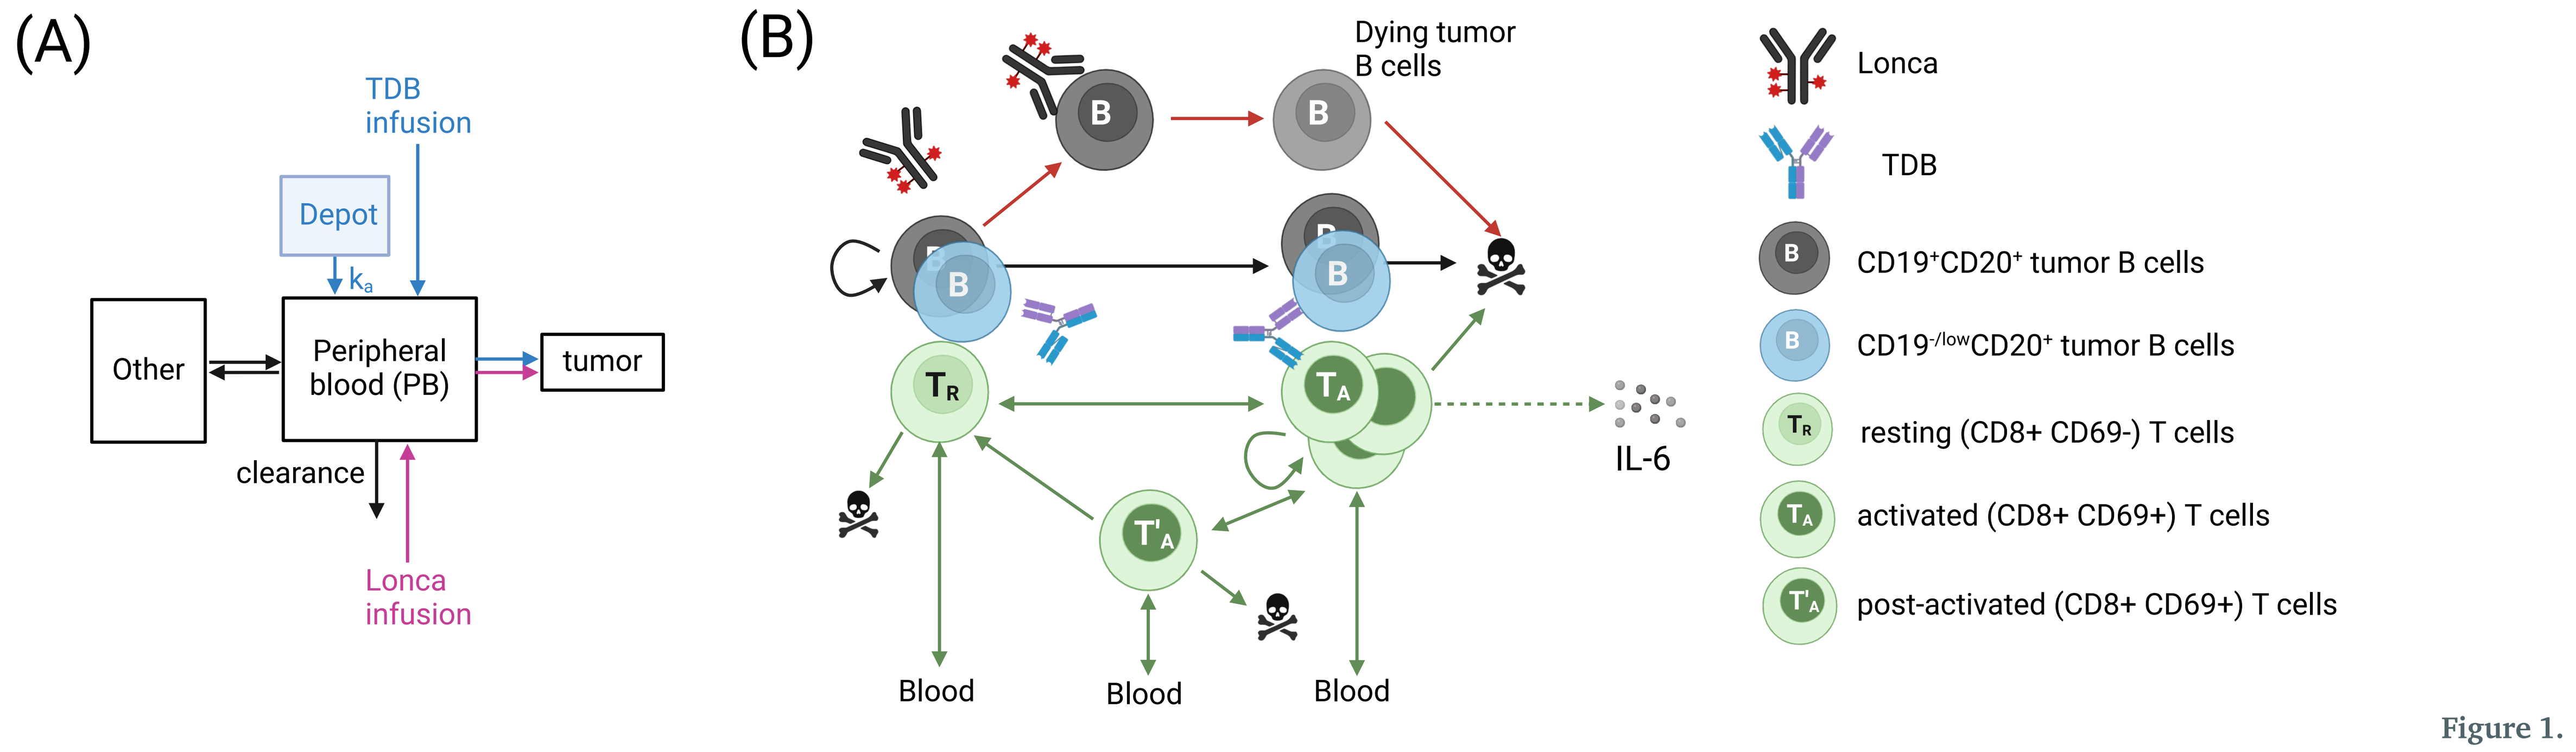
(A)
TDB infusion
Depot
k_a
Other
Peripheral blood (PB)
tumor
clearance
Lonca infusion
(B)
Dying tumor B cells
B
T_R
T_A
T'_A
IL-6
Blood
Blood
Blood
Lonca
TDB
CD19+CD20+ tumor B cells
CD19-/lowCD20+ tumor B cells
resting (CD8+ CD69-) T cells
activated (CD8+ CD69+) T cells
post-activated (CD8+ CD69+) T cells

Figure 1.

Schematic diagrams for the TDB + loncastuximab tesirine combination therapy model.
A: Compartmental PK diagram. The PK model includes 3 compartments: a central compartment (i.e., peripheral blood), a peripheral tissue compartment to account for drug distribution, and a deposit compartment to account for subcutaneous injection. **B: tumor B cell and T cell dynamics in the presence of TDB and loncastuximab tesirine.** T cells in the tumor cycle through resting state, activated state, and postactivated (exhausted) state, and may die at any state. Only activated T cells proliferate and induce both healthy and cancerous B cell death. All T cells could enter or leave the tumor from/into the bloodstream. Tumor cells killed by loncastuximab tesirine transition to a dying stage before being removed from the tumor.

Virtual DLBCL Patient Population
Virtual populations were created by resampling (prevalence weighting) from a larger virtual cohort, such that monotherapy tumor responses matched reported clinical observations from three respective clinical trials, as described in [3]. A virtual cohort of 20,000 subjects was generated by Latin Hypercube sampling five key parameters specifying:

1. Initial tumor volumes: 0.1 to 100 mL
2. Malignant B cell proliferation rates: 0.01 to 0.05 day^{-1}
3. CD19-/Low fraction: 0 to 10%
4. Lonca-induced maximal cell killing rate on CD19-/Low cells: 0.01 to 0.07 day^{-1}
5. Apoptotic transition rate: 0.01 to 0.05 day^{-1}

Prevalence weights were computed by matching simulated tumor responses to mosunetuzumab, epcoritamab, and loncastuximab tesirine monotherapy treatments from the approved regimen to their corresponding clinical observations [4,5,6]. Prevalence weights were then assigned via quadratic programming, and a Virtual Population of 500 patients created by resampling with proportional frequencies (Table 1) .

Dosing Schemes
Loncastuximab tesirine was given every 3 weeks (Q3W) at the following doses:
1. 150 μg/kg for 2 doses followed by 75 μg/kg
2. 120 μg/kg for 2 doses followed by 75 μg/kg
3. 90 μg/kg

Mosunetuzumab was dosed subcutaneously at 5 mg on cycle 1 day 1 (C1D1), 45 mg on C1D8, 45 mg on C1D15, and 45 mg Q3W for C2 onward.
Glofitamab was dosed intravenously at 2.5 mg on C1D8 following obinutuzumab pretreatment on C1D1, Glofitamab 10 mg on C1D15, and Glofitamab 30 mg Q3W for C2 onward.
Epcoritamab was dosed intravenously at 0.16 mg, 0.8 mg, 48 mg on C1D1, C1D8, C1D15, respectively; then 48 mg on D1, D8, D15 between cycles 2 and 3; and then 48 mg Q3W onward.

Combination index
Combination indices (CIs) were calculated to assess combination synergy/antagonism relative to Bliss independence [7] based on response classification [8] at the end of each cycle. CIs were calculated by dividing the simulated overall response rates (ORR=CR+PR) to combination treatments by the ORR predicted by independent activities: $ORR_{AB} = ORR_A + ORR_B \times (1\text{-}ORR_A)$.

Results Tables and Figures

Table 1. Observed versus simulated patient response rates to epcoritamab, mosunetuzumab, loncastuximab terisine, and glofitamab monotherapy treatments in the virtual population.

	Epcoritamab		Mosunetuzumab		Loncastuximab terisine		Glofitamab (predicted)	
Response	Sim	Obs	Sim	Obs	Sim	Obs	Sim	Obs
CR %	43.48	43	18.63	20	28.05	29	33.8	33
PR %	25.05	27	19.06	17	32.33	29	13.28	15
SD %	3.21	4	7.92	7	16.7	17	38.97	
PD %	28.05	27	54.39	56	22.91	25	13.92	

Resp: Response. CR: complete response. PR: partial response. SD: stable disease. PD: progressive disease. Sim: simulated result. Obs: observed result. Response classifications are based on [8]. Epcoritamab clinical data from n=157 r/r NHL patients [6]. Mosunetuzumab clinical data from n=129 aggressive-NHL patients [5]. Loncastuximab tesirine clinical data from n=145 r/r DLBCL patients [4]. Glofitamab clinical data from n=127 aggressive-NHL patients [9].

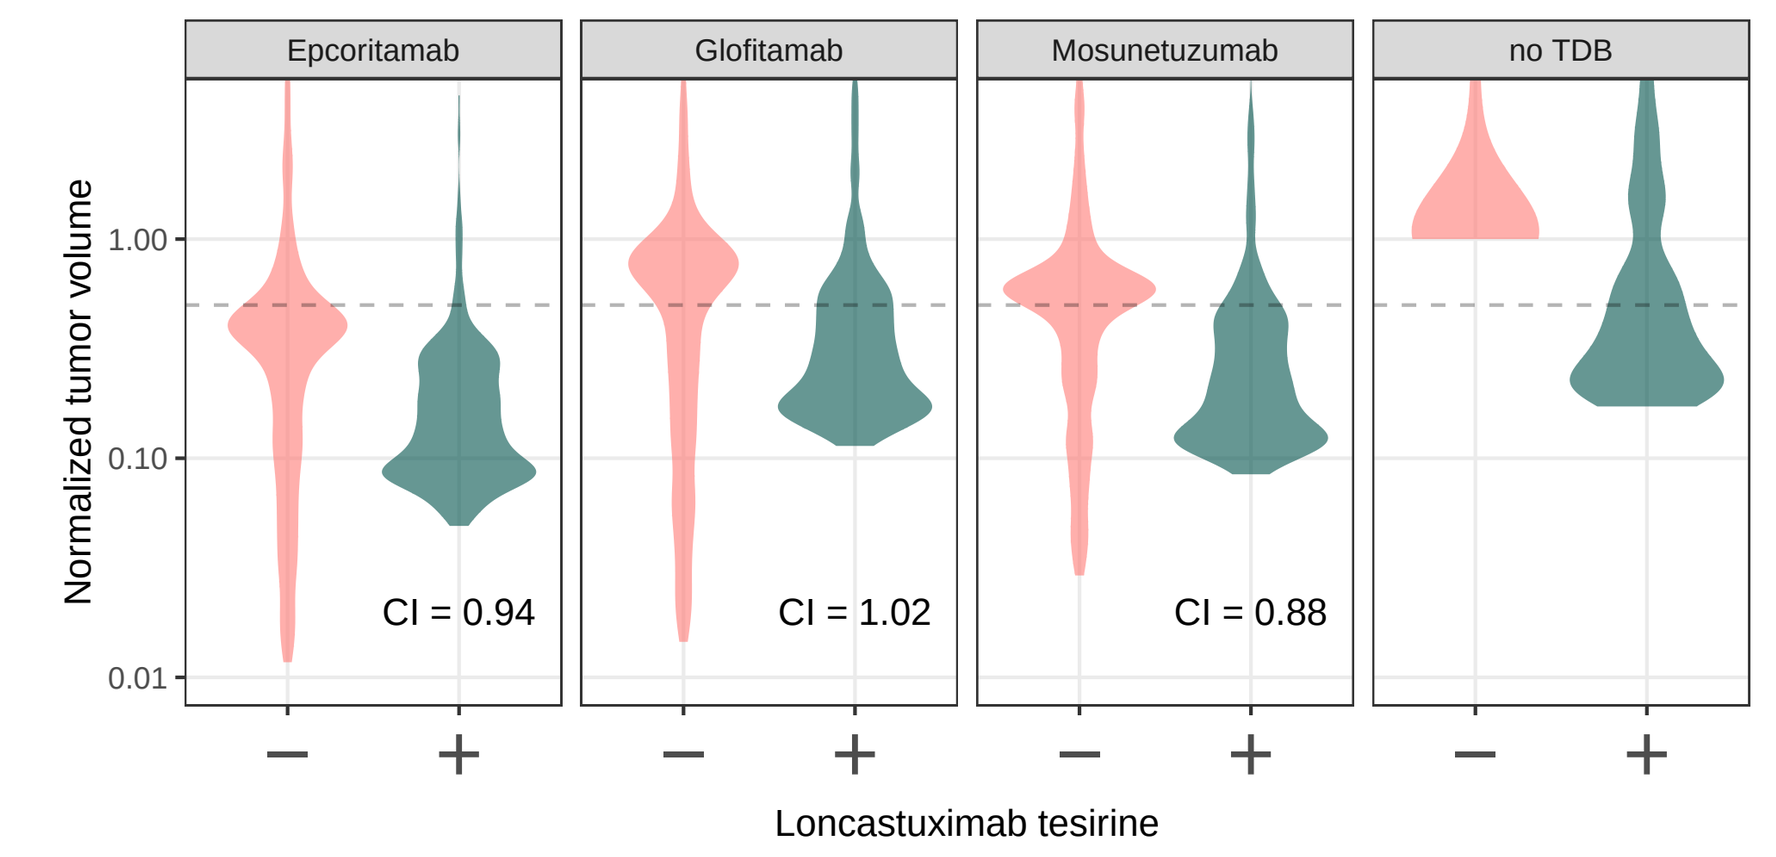
Epcoritamab
Glofitamab
Mosunetuzumab
no TDB
Normalized tumor volume
1.00
0.10
0.01
CI = 0.94
CI = 1.02
CI = 0.88
Loncastuximab tesirine

Figure 2. Virtual population responses to TDB and loncastuximab tesirine combinations following 2 cycles of therapy (Day 42). CIs were computed to quantify the degree of non-additive effects resulting from the combinations (CI > 1 indicates synergistic). Dashed line: separation between non-responders and responders. Tumor response classification based on [8].

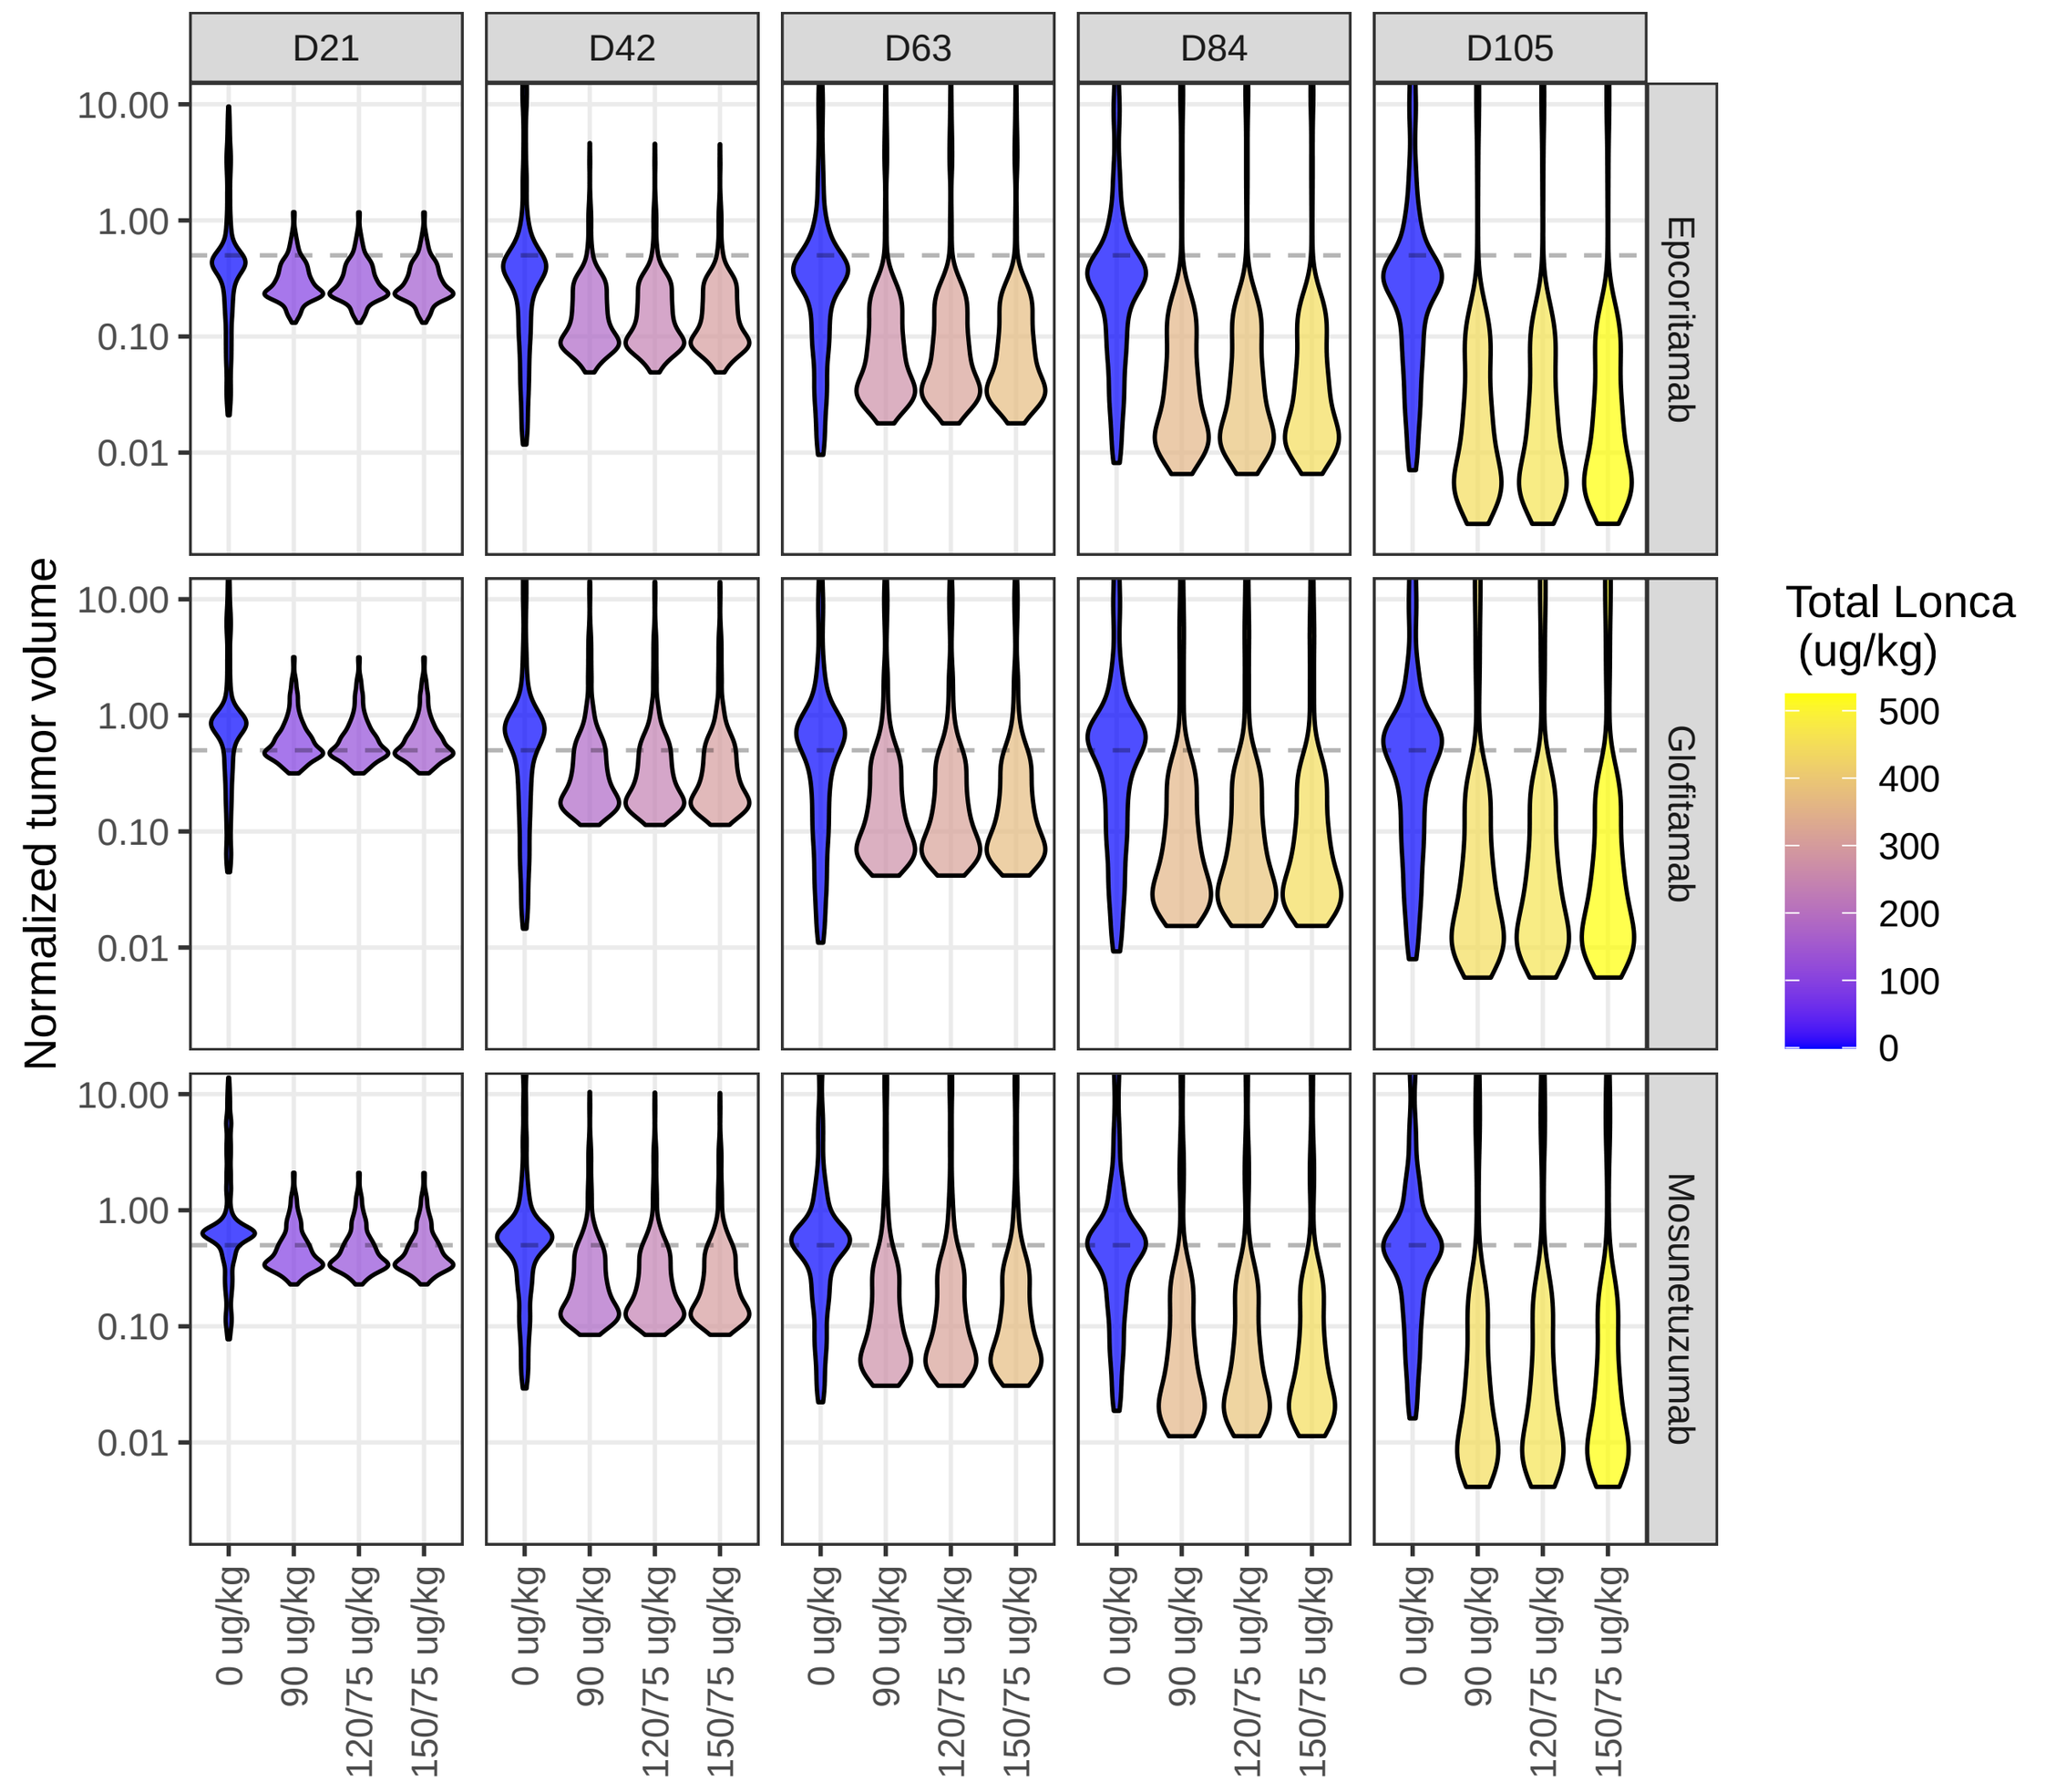
D21
D42
D63
D84
D105
Epcoritamab
Glofitamab
Mosunetuzumab
Normalized tumor volume
10.00
1.00
0.10
0.01
Total Lonca (ug/kg)
500
400
300
200
100
0
0 ug/kg
90 ug/kg
120/75 ug/kg
150/75 ug/kg

Figure 3. Virtual population tumor volume reduction to T cell-dependent bispecific antibody and loncastuximab tesirine combination therapy by cycle and loncastuximab tesirine (Lonca) dosing regimen. Dashed line: separation between non-responders and responders. Tumor response classification based on [8].

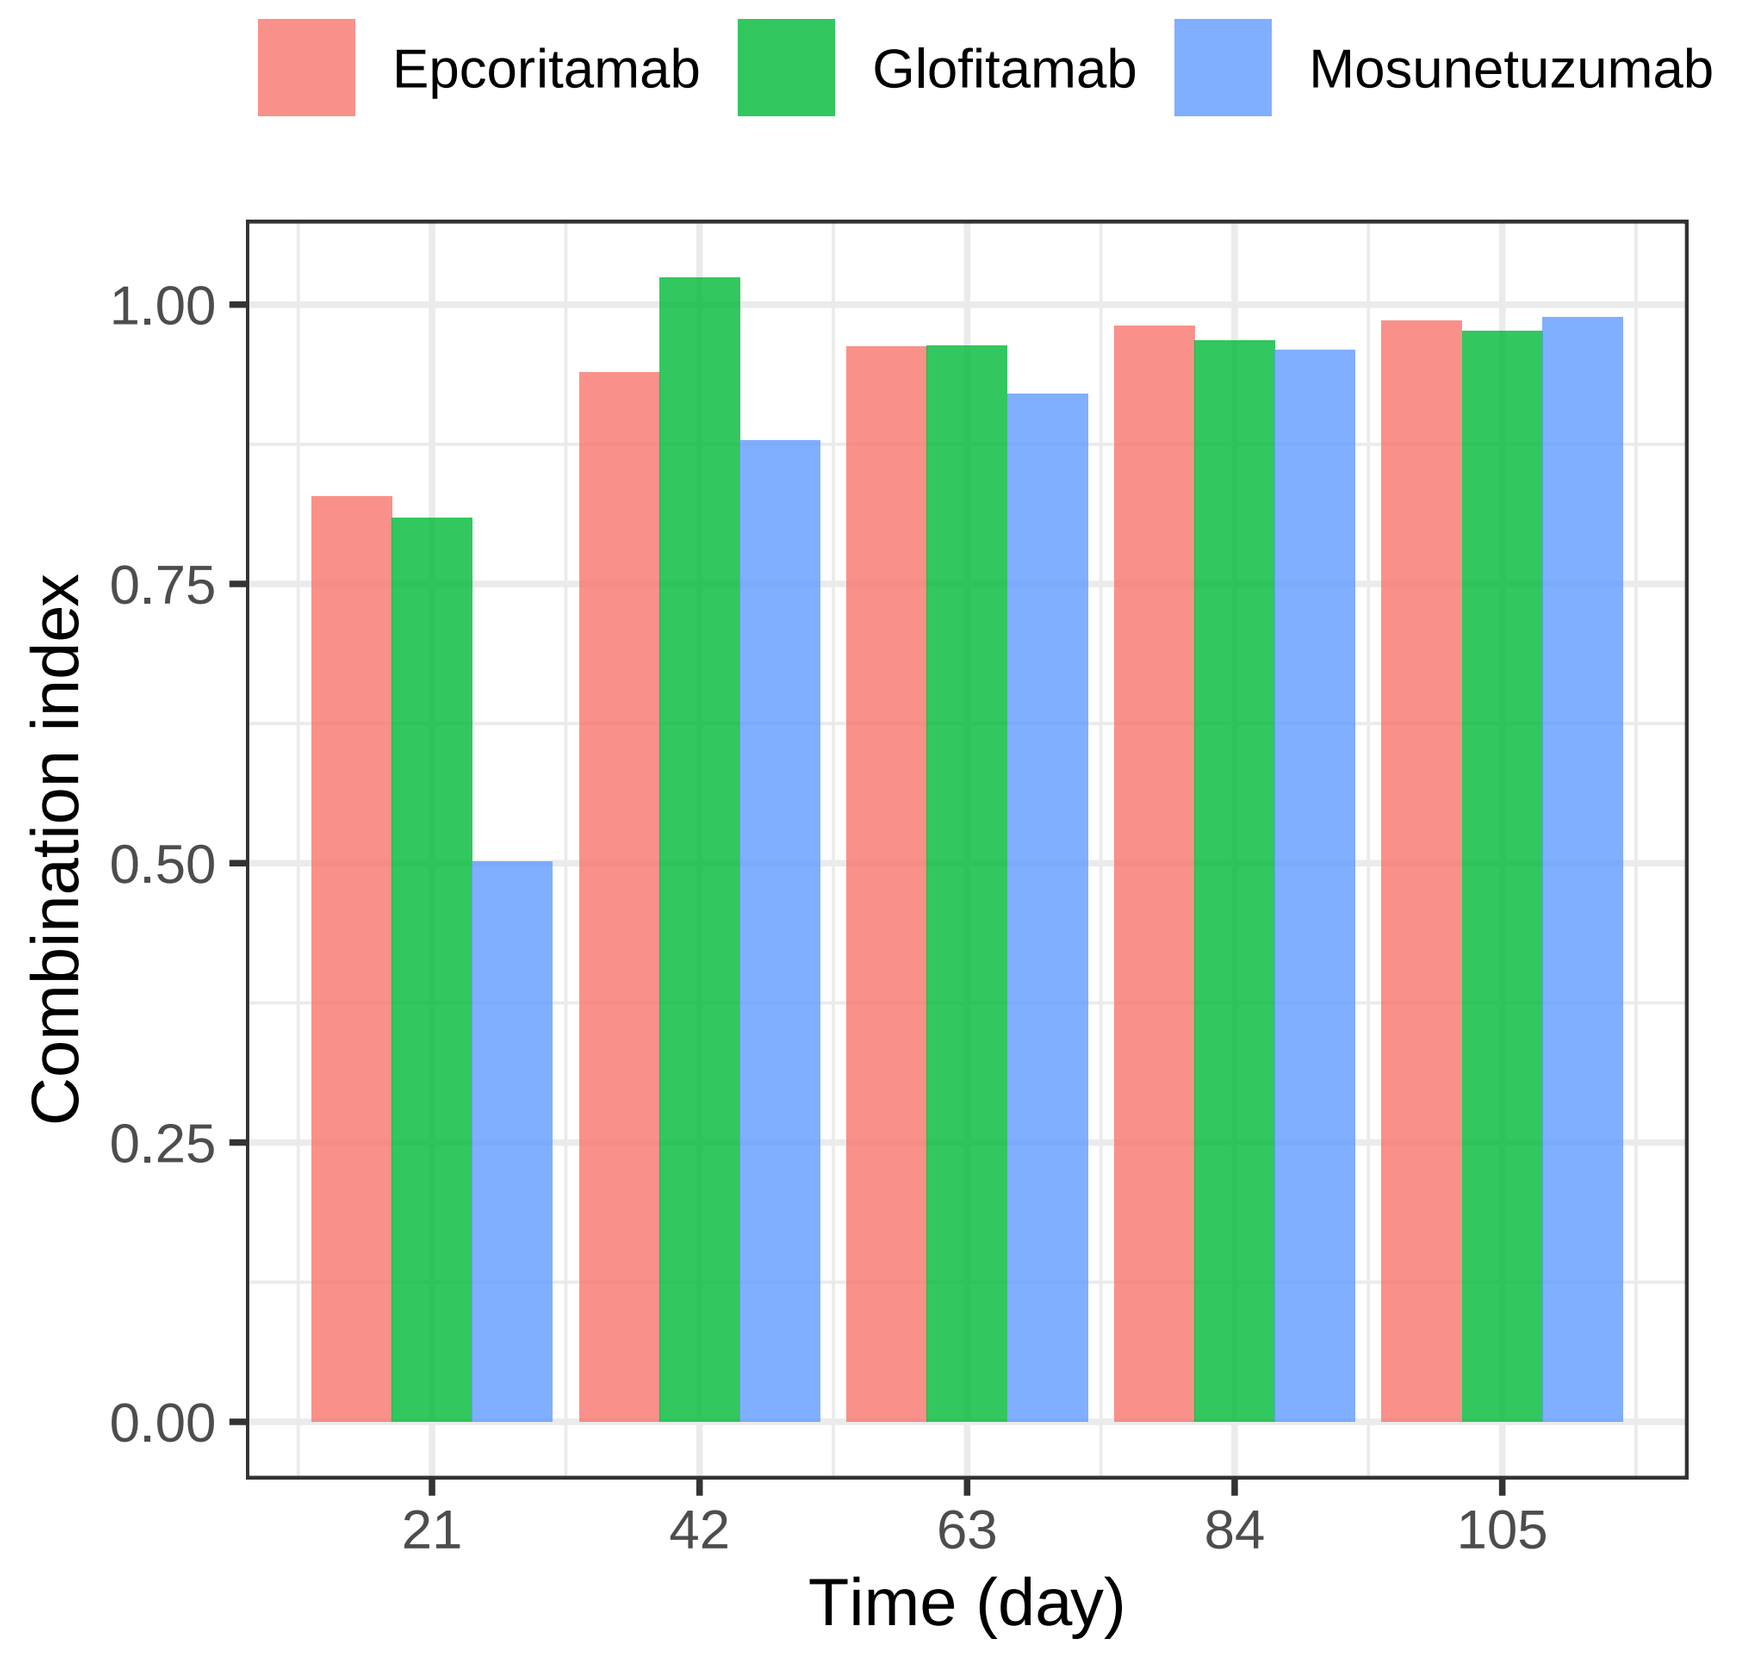
Epcoritamab
Glofitamab
Mosunetuzumab
Combination index
1.00
0.75
0.50
0.25
0.00
21
42
63
84
105
Time (day)

Figure 4. Combination indices (CI) computed based on virtual population responses to TDB and loncastuximab tesirine combinations over 5 cycles (D21, D42, D64, D84, D105). Combination indices (CI) were computed [7] to quantify the degree of non-additive effects resulting from the combinations (CI > 1 indicates synergistic). In all three combinations, the loncastuximab tesirine dose was 150 ug/kg Q3W for the first 2 cycles, followed by 75 ug/kg Q3W.

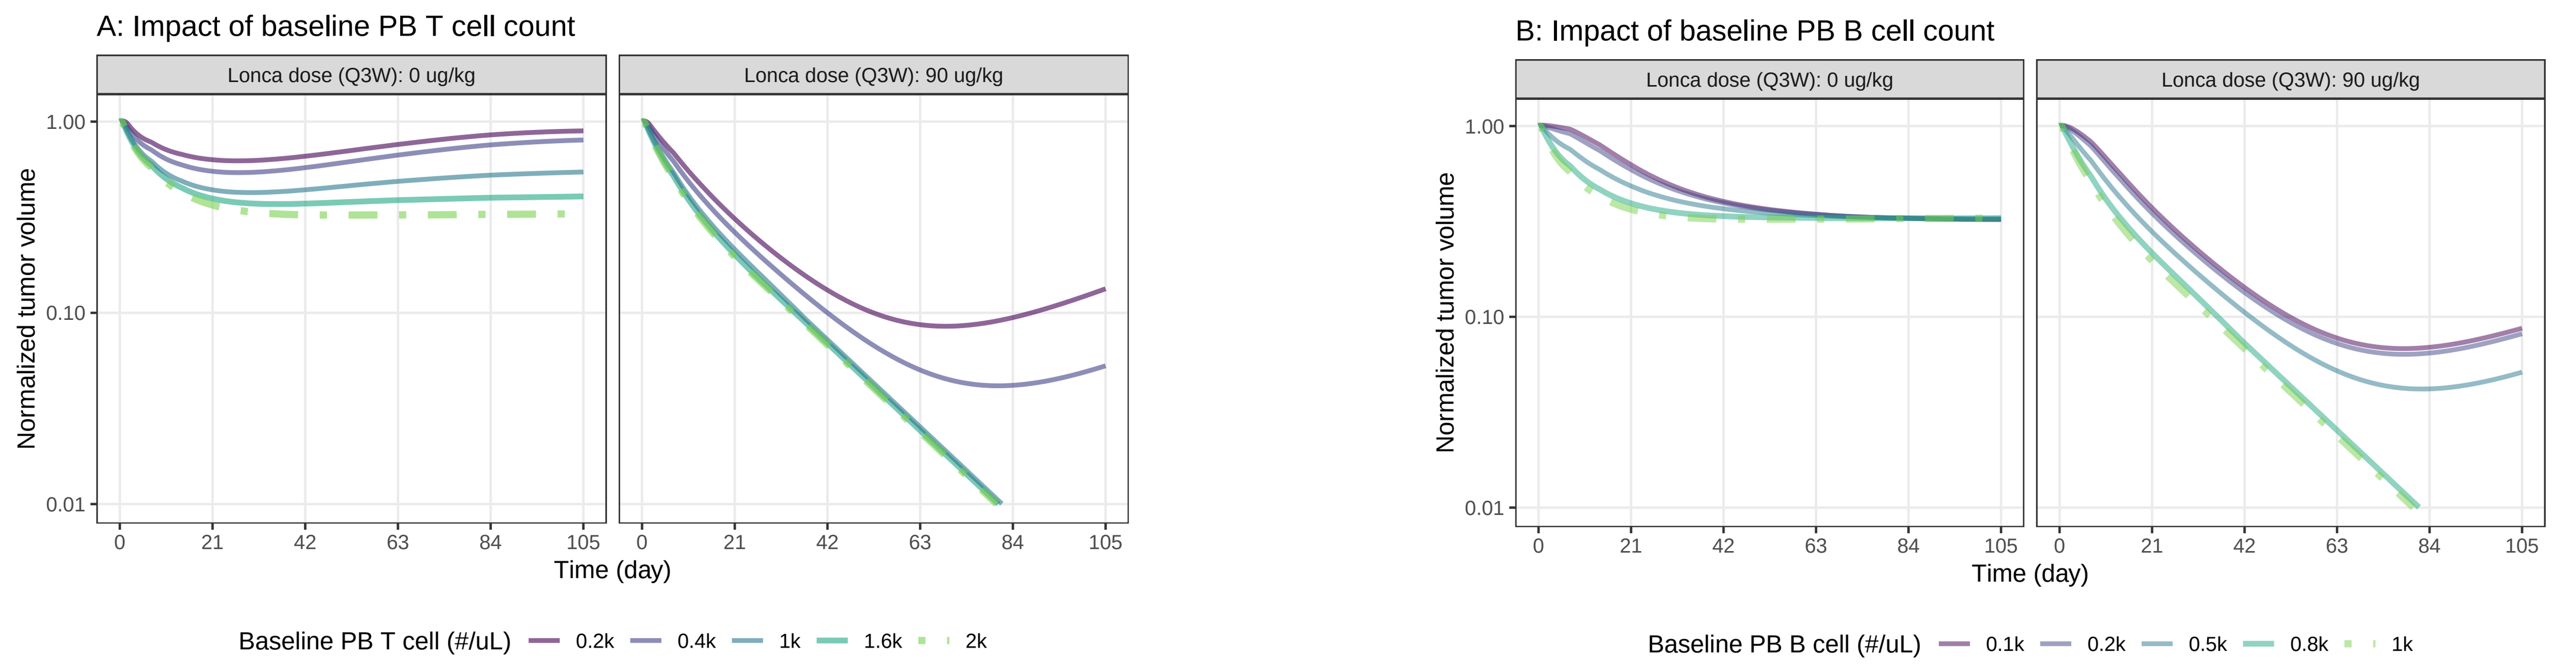
A: Impact of baseline PB T cell count
B: Impact of baseline PB B cell count
Lonca dose (Q3W): 0 ug/kg
Lonca dose (Q3W): 90 ug/kg
Normalized tumor volume
1.00
0.10
0.01
0
21
42
63
84
105
Time (day)
Baseline PB T cell (#/uL)
0.2k
0.4k
1k
1.6k
2k
Baseline PB B cell (#/uL)
0.1k
0.2k
0.5k
0.8k
1k

Figure 5. Sensitivity of epcoritamab and epcoritamab + loncastuximab tesirine (Lonca) tumor responses to differences in initial circulating T cell (A) and B cell (B) counts. Default healthy baseline T cell counts (A) were set at 2000 cells/uL, and default baseline B cell counts (B) at 1000 cells/uL. The loncastuximab tesirine dose was set to be either 0 μg/kg or 90 μg/kg Q3W, with the epcoritamab dose following as described in the Methods. The tumor was assumed to have an initial volume of 7.5 mL with 80% CD19+CD20+ tumor B cells and 20% CD19-CD20+ cells.

Key Results

- The virtual DLBCL population accurately recapitulated clinical tumor response data (**Table 1**).
- Greater efficacy was predicted for combination therapies with loncastuximab tesirine (**Figure 2**).
- Greater efficacy was predicted for combination therapies with more cycles, instead of higher accumulative dose of loncastuximab tesirine (**Figure 3**).
- Combination index indicated loncastuximab tesirine-TDB combination effects were largely additive, but depend on cycle (**Figure 4**).
- Efficacy of combination treatment was maintained in lymphopenic virtual patients (**Figure 5**). A decrease in circulating T cells (by 50% of baseline) or circulating B cell counts (by 20% of baseline) in peripheral blood (PB) was predicted to minimally alter the epcoritamab + loncastuximab tesirine combination efficacy.

Conclusions

The virtual population-based lymphoma QSP model enabled systematic exploration of alternate drug combinations, dosing schemes, clinical covariates and resultant effect on anti-tumor activity *in silico*, infeasible to test in a clinical trial. The results of the LOTIS-7 study (NCT04970901), a platform study evaluating loncastuximab tesirine in combination with glofitamab, will soon be available to assess the accuracy of the model predictions.

References

[1] Caimi *et al.* E J Haem, 5(1):76-83, 2024. [2] Susilo *et al.* Clin. Transl. Sci., 16(7):1134-1148, 2023. [3] Gadkar *et al.* CPT Pharmacometrics Syst. Pharmacol., 5(5):235-249, 2016. [4] Caimi *et al.* Haematologica, 109(4):1184-1193, 2023. [5] Budde *et al.* J. Clin. Oncol., 40(5):481-491, 2022. [6] Thieblemont *et al.* J. Clin. Oncol., 41(12):2238-2247, 2023. [7] Foucquier and Mickael Guedj. Pharmacol. Res. Perspect., 3(3):e00149, 2015. [8] Cheson *et al.* J. Clin. Oncol., 25(5):579-586, 2007. [9] Hutchings *et al.* J. Clin. Oncol., 39(18):1959-1970, 2021.

Acknowledgements

The analysis was funded by ADC Therapeutics SA and partially funded by Sobi.

Metrum Research Group Publications and Posters

Presented at the American Society for Clinical Pharmacology & Therapeutics 2025 Annual Meeting; 28 May - 30 May 2025
Copies available at: www.metrumrg.com/all-publications