Pegcetacoplan for 52 weeks maintained proteinuria reduction regardless of immunosuppressant use or nephrotic range proteinuria at baseline: VALIANT subgroup analysis

FR-PO0836

Carla M Nester[1], Andrew S Bomback[2], Gema Ariceta[3], Yahsou Delmas[4], Bradley P Dixon[5], Daniel P Gale[6], Larry A Greenbaum[7], Seung Hyeok Han[8], Nicole Isbel[9,10], Christoph Licht[11], Antonio Mastrangelo[12], Masashi Mizuno[13], Maria Izabel Neves de Holanda[14], Matthew C Pickering[15], Giuseppe Remuzzi[16], Nicole Van De Kar[17], Marina Vivarelli[18], Patrick D Walker[19], Dean Wallace[20], Daniel Zecher[21], Moglie Le Quintrec[22], Yael Borovitz[23], Nabil Melhem[24], Fadi Fakhouri[25]

[1]University of Iowa, Stead Family Children's Hospital, Iowa City, IA, USA; [2]Columbia University Irving Medical Center, New York, NY, USA; [3]Hospital Vall d'Hebron, Barcelona, Spain; [4]Bordeaux University Hospital, Bordeaux, France; [5]University of Colorado School of Medicine, Aurora, CO, USA; [6]University College London, London, UK; [7]Emory School of Medicine, Atlanta, GA, USA; [8]Yonsei University College of Medicine, Seoul, Republic of Korea; [9]Princess Alexandra Hospital, Brisbane, Australia; [10]The University of Queensland, Brisbane, Australia; [11]The Hospital for Sick Children and University of Toronto, Toronto, ON, Canada; [12]Unit Fondazione IRCCS Ca' Granda Ospedale Maggiore Policlinico, Milan, Italy; [13]Department of Renal Replacement Therapy, Nagoya University Graduate School of Medicine, Nagoya, Japan; [14]Hospital Federal de Bonsucesso, Ruschel Medicina, Rio de Janeiro, Brazil; [15]Imperial College, London, UK; [16]Istituto di Ricerche Farmacologiche Mario Negri IRCCS, Bergamo, Italy; [17]Radboudumc Amalia Children's Hospital, Nijmegen, The Netherlands; [18]Bambino Gesù Children's Hospital, IRCCS, Rome, Italy; [19]Arkana Laboratories, Little Rock, AR, USA; [20]Royal Manchester Children's Hospital, Manchester, UK; [21]Regensburg University Hospital, Regensburg, Germany; [22]CHU Lapeyronier, Montpellier, France; [23]Schneider Children's Medical Center of Israel, Petah-Tikva, Israel; [24]Evelina London Children's Hospital, Guy's and St Thomas' NHS Foundation Trust, London, UK; [25]Lausanne University Hospital and University of Lausanne, Lausanne, Switzerland

CONCLUSIONS

- This post hoc analysis of VALIANT shows that patients with C3G or primary (idiopathic) IC-MPGN who received pegcetacoplan for up to 52 weeks had strong, prompt UPCR reductions and slowed eGFR decline, comparable to the overall population, regardless of nephrotic range proteinuria at baseline or concomitant IS treatment
- Patients with nephrotic range proteinuria whose treatment was delayed by only 26 weeks had marked UPCR reductions and slowed eGFR decline after switching to pegcetacoplan; the magnitude of proteinuria reduction may have been impacted by this delayed treatment, indicating the need for urgent intervention, especially in patients with nephrotic range proteinuria who are known to have worse outcomes
- No new safety signals were identified in these key patient groups

INTRODUCTION

- C3G and primary IC-MPGN are driven by complement dysregulation, resulting in the accumulation of C3 deposits in the glomeruli (in addition to immunoglobulins in IC-MPGN), leading to inflammation and ultimately kidney failure[1,2]
- In the phase 3 VALIANT trial (NCT05067127) RCP, patients with C3G or primary IC-MPGN who received pegcetacoplan for 26 weeks improved in all components of the C3G outcome triad established by an expert panel convened by the Kidney Health Initiative, as shown below[3,4]

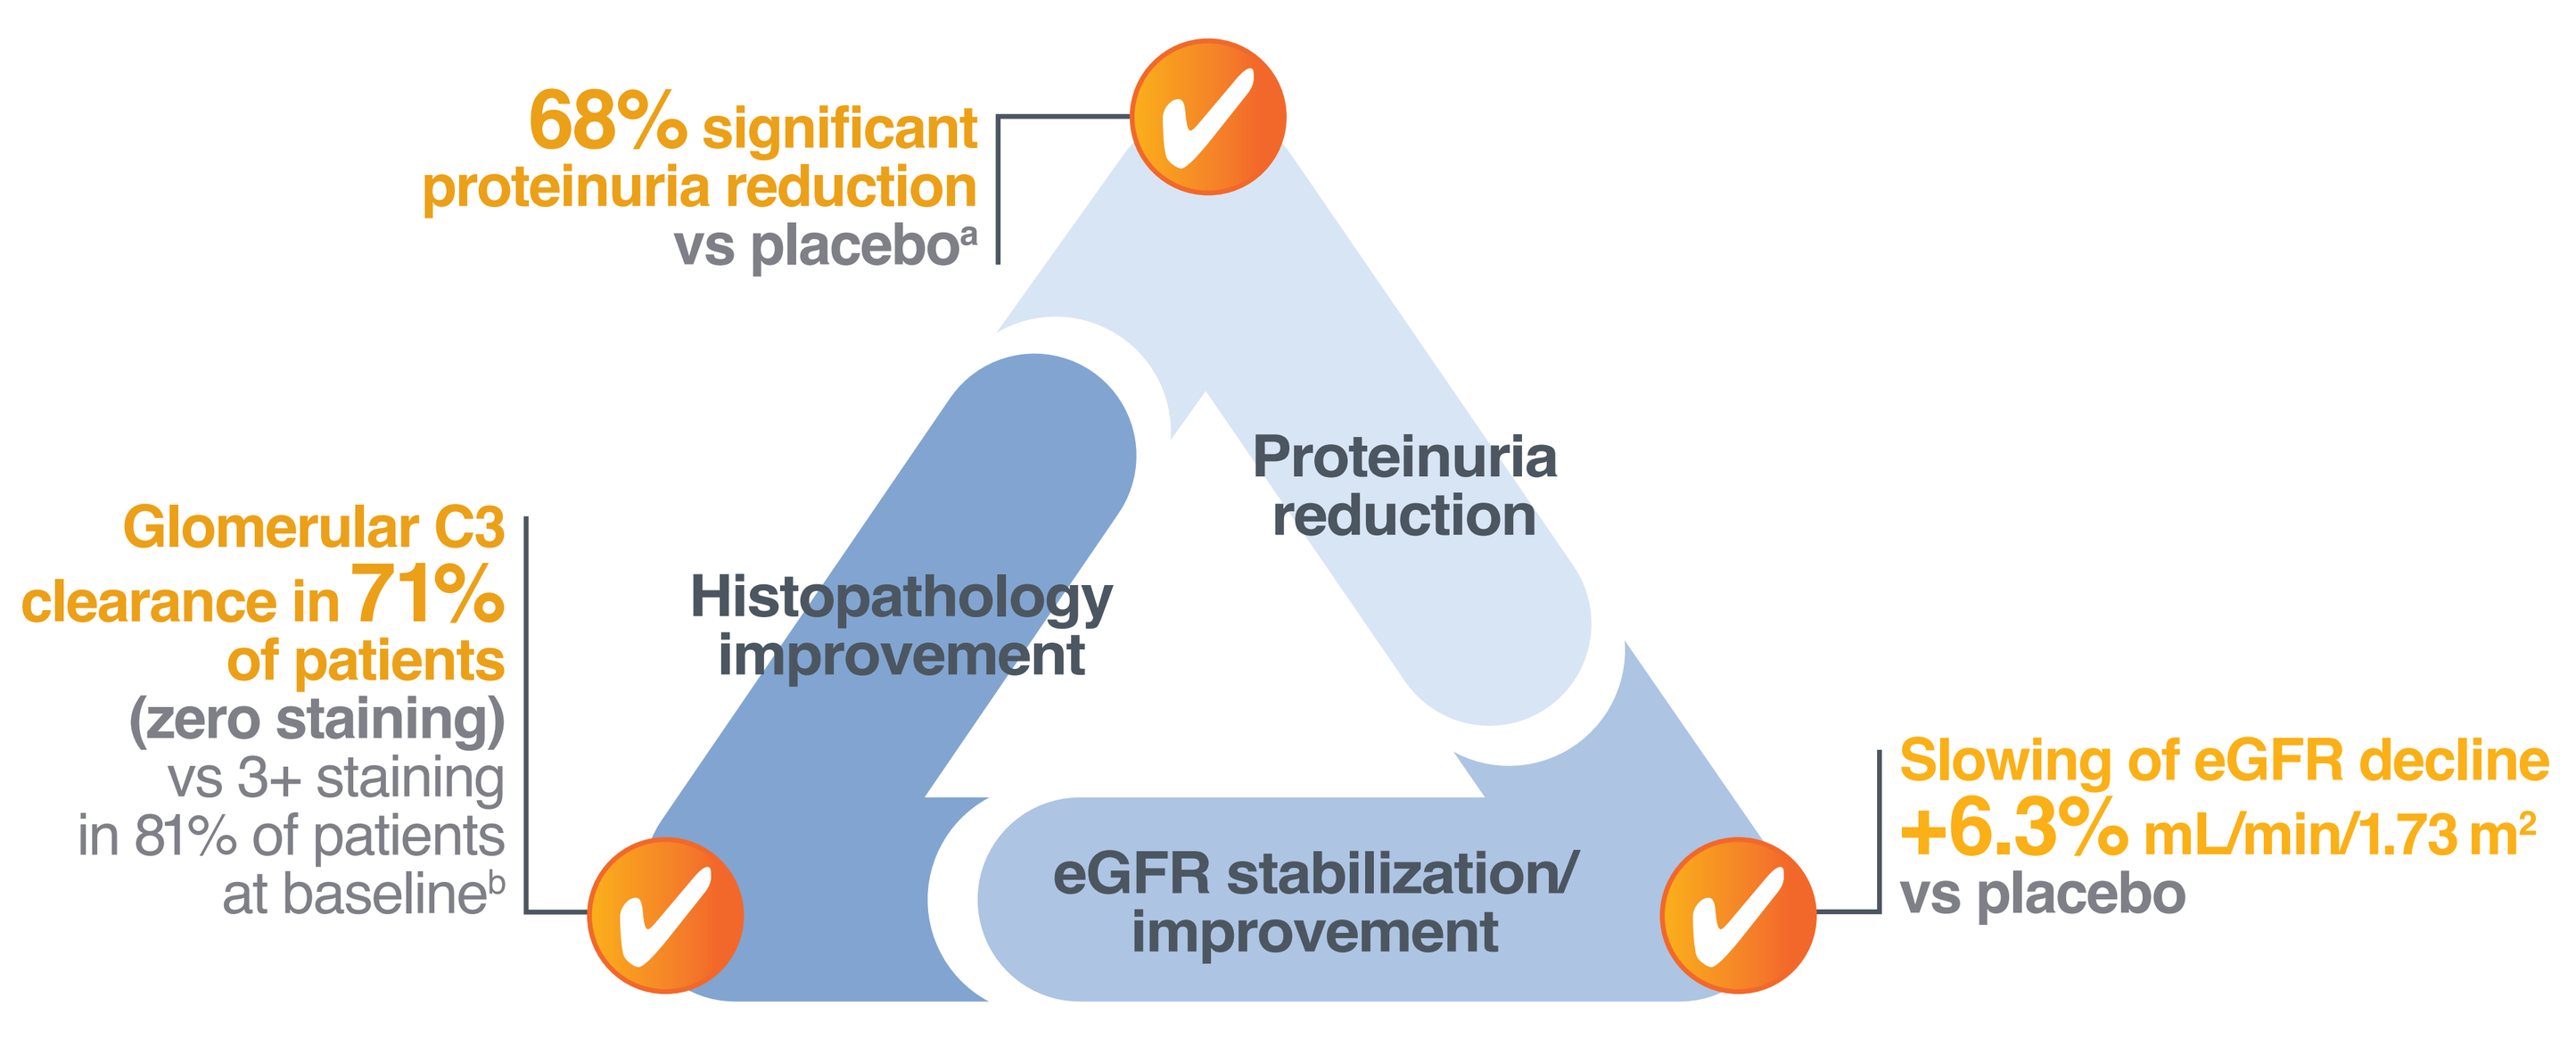
68% significant proteinuria reduction vs placebo[a]
Proteinuria reduction
Histopathology improvement
eGFR stabilization/ improvement
Glomerular C3 clearance in 71% of patients (zero staining) vs 3+ staining in 81% of patients at baseline[b]
Slowing of eGFR decline +6.3% mL/min/1.73 m² vs placebo

[a]Consistent across subgroups (age, disease type, transplant status). [b]Adult patients only. Biopsies at week 26 were not required for adolescents.
eGFR, estimated glomerular filtration rate.

- As IS treatment is often recommended for patients with C3G and primary IC-MPGN who have a higher risk for progression, such as those with persistent nephrotic-range proteinuria or declining kidney function[5], it is of great interest to investigate pegcetacoplan's efficacy in patients with concomitant IS use
- Patients with nephrotic-range proteinuria are known to have particularly poor outcomes[6]; treatment options for this difficult-to-treat group are urgently needed
- In these post hoc analyses, we assessed efficacy in these key patient subgroups (concomitant IS treatment, including patients with recurrence after transplantation [yes/no] or nephrotic range proteinuria at baseline [UPCR <3 g/g vs ≥3 g/g]) after up to 52 weeks of pegcetacoplan

OBJECTIVE

Determine if the proteinuria reduction and the slowing of eGFR decline observed with 52 weeks of pegcetacoplan in VALIANT was consistent in patients with or without concomitant IS use and patients with or without nephrotic range proteinuria at baseline

METHODS

- VALIANT tested pegcetacoplan in patients ≥12 years of age with biopsy-confirmed C3G or primary IC-MPGN[3] (**Figure 1**)

Figure 1. VALIANT study design

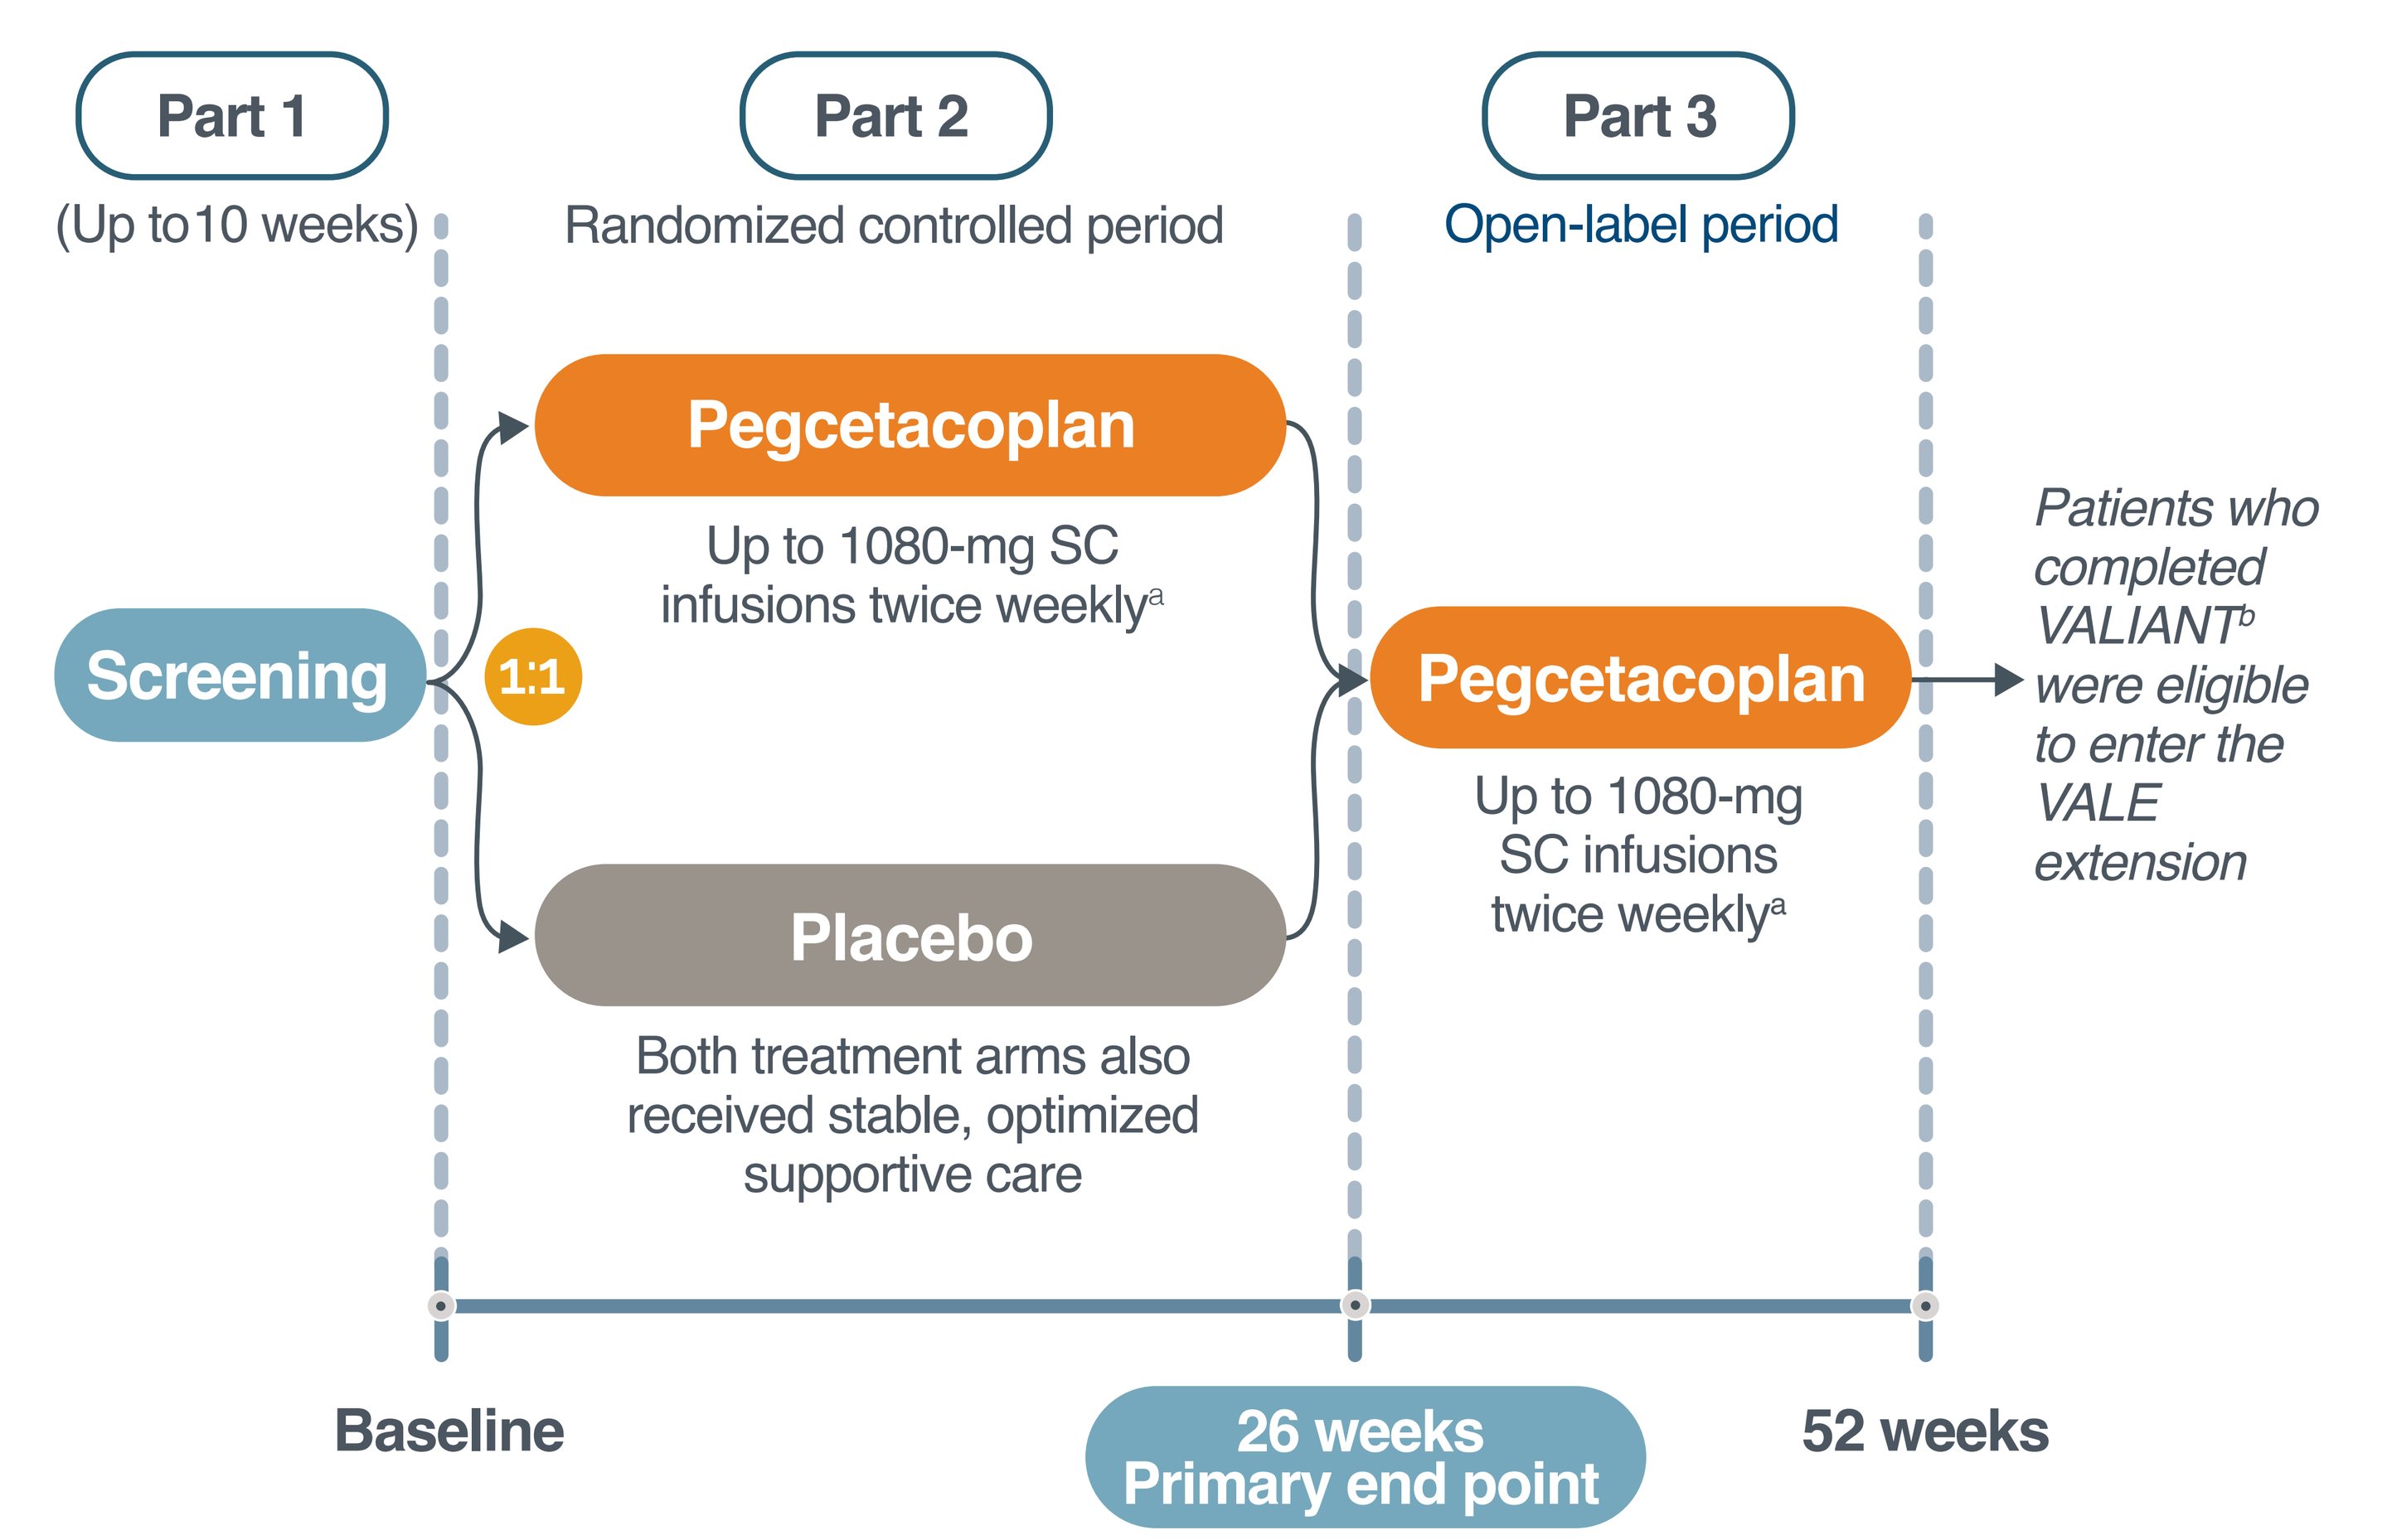
Part 1
(Up to10 weeks)
Part 2
Randomized controlled period
Part 3
Open-label period
Screening
1:1
Pegcetacoplan
Up to 1080-mg SC infusions twice weekly[a]
Placebo
Both treatment arms also received stable, optimized supportive care
Pegcetacoplan
Up to 1080-mg SC infusions twice weekly[a]
Patients who completed VALIANT[b] were eligible to enter the VALE extension
Baseline
26 weeks Primary end point
52 weeks

[a]All adults and adolescents weighing ≥50 kg self administered 1080 mg/20 mL twice weekly. Adolescent patients weighing 30 to <35 kg received 540 mg/10 mL for the first 2 doses, then 648 mg/12 mL twice weekly. Adolescent patients weighing 35 to <50 kg received 648 mg/12 mL for the first dose, 810 mg/15 mL for the second dose, then 810 mg/15 mL twice weekly. Biopsies were not required at week 52. [b]61 of 63 patients randomized to pegcetacoplan entered the OLP and 59 completed the study. 57 of 61 patients randomized to placebo entered the OLP and 55 completed the study. Patients who completed the OLP could continue to the VALE extension.[7] OLP, open-label period; SC, subcutaneous.

RESULTS

- Patients receiving pegcetacoplan for 52 weeks had sustained proteinuria decreases (mean [95% CI] change from baseline: week 26, –67.2% [–74.9 to –57.2]; week 52, –67.2% [–75.8 to –55.4])
- Patients in the placebo-to-pegcetacoplan group had a similar result after switching to pegcetacoplan: week 26, 2.9% (–8.6 to 15.9); week 52, –51.3% (–62.1 to –37.5)
- Proteinuria reductions among pegcetacoplan-treated patients by concomitant IS use and nephrotic range proteinuria at baseline were consistent with the overall population to week 52 (**Figure 2**). Placebo-to-pegcetacoplan patients in these subgroups also achieved UPCR reduction and eGFR stabilization after 26 weeks of pegcetacoplan treatment. The magnitude of proteinuria reduction in the nephrotic range subgroup, after a 26-week delay in treatment, indicates the importance of early treatment

RESULTS *(cont.)*

Figure 2. Change in proteinuria during VALIANT by concomitant IS use (A) and nephrotic range proteinuria (B)

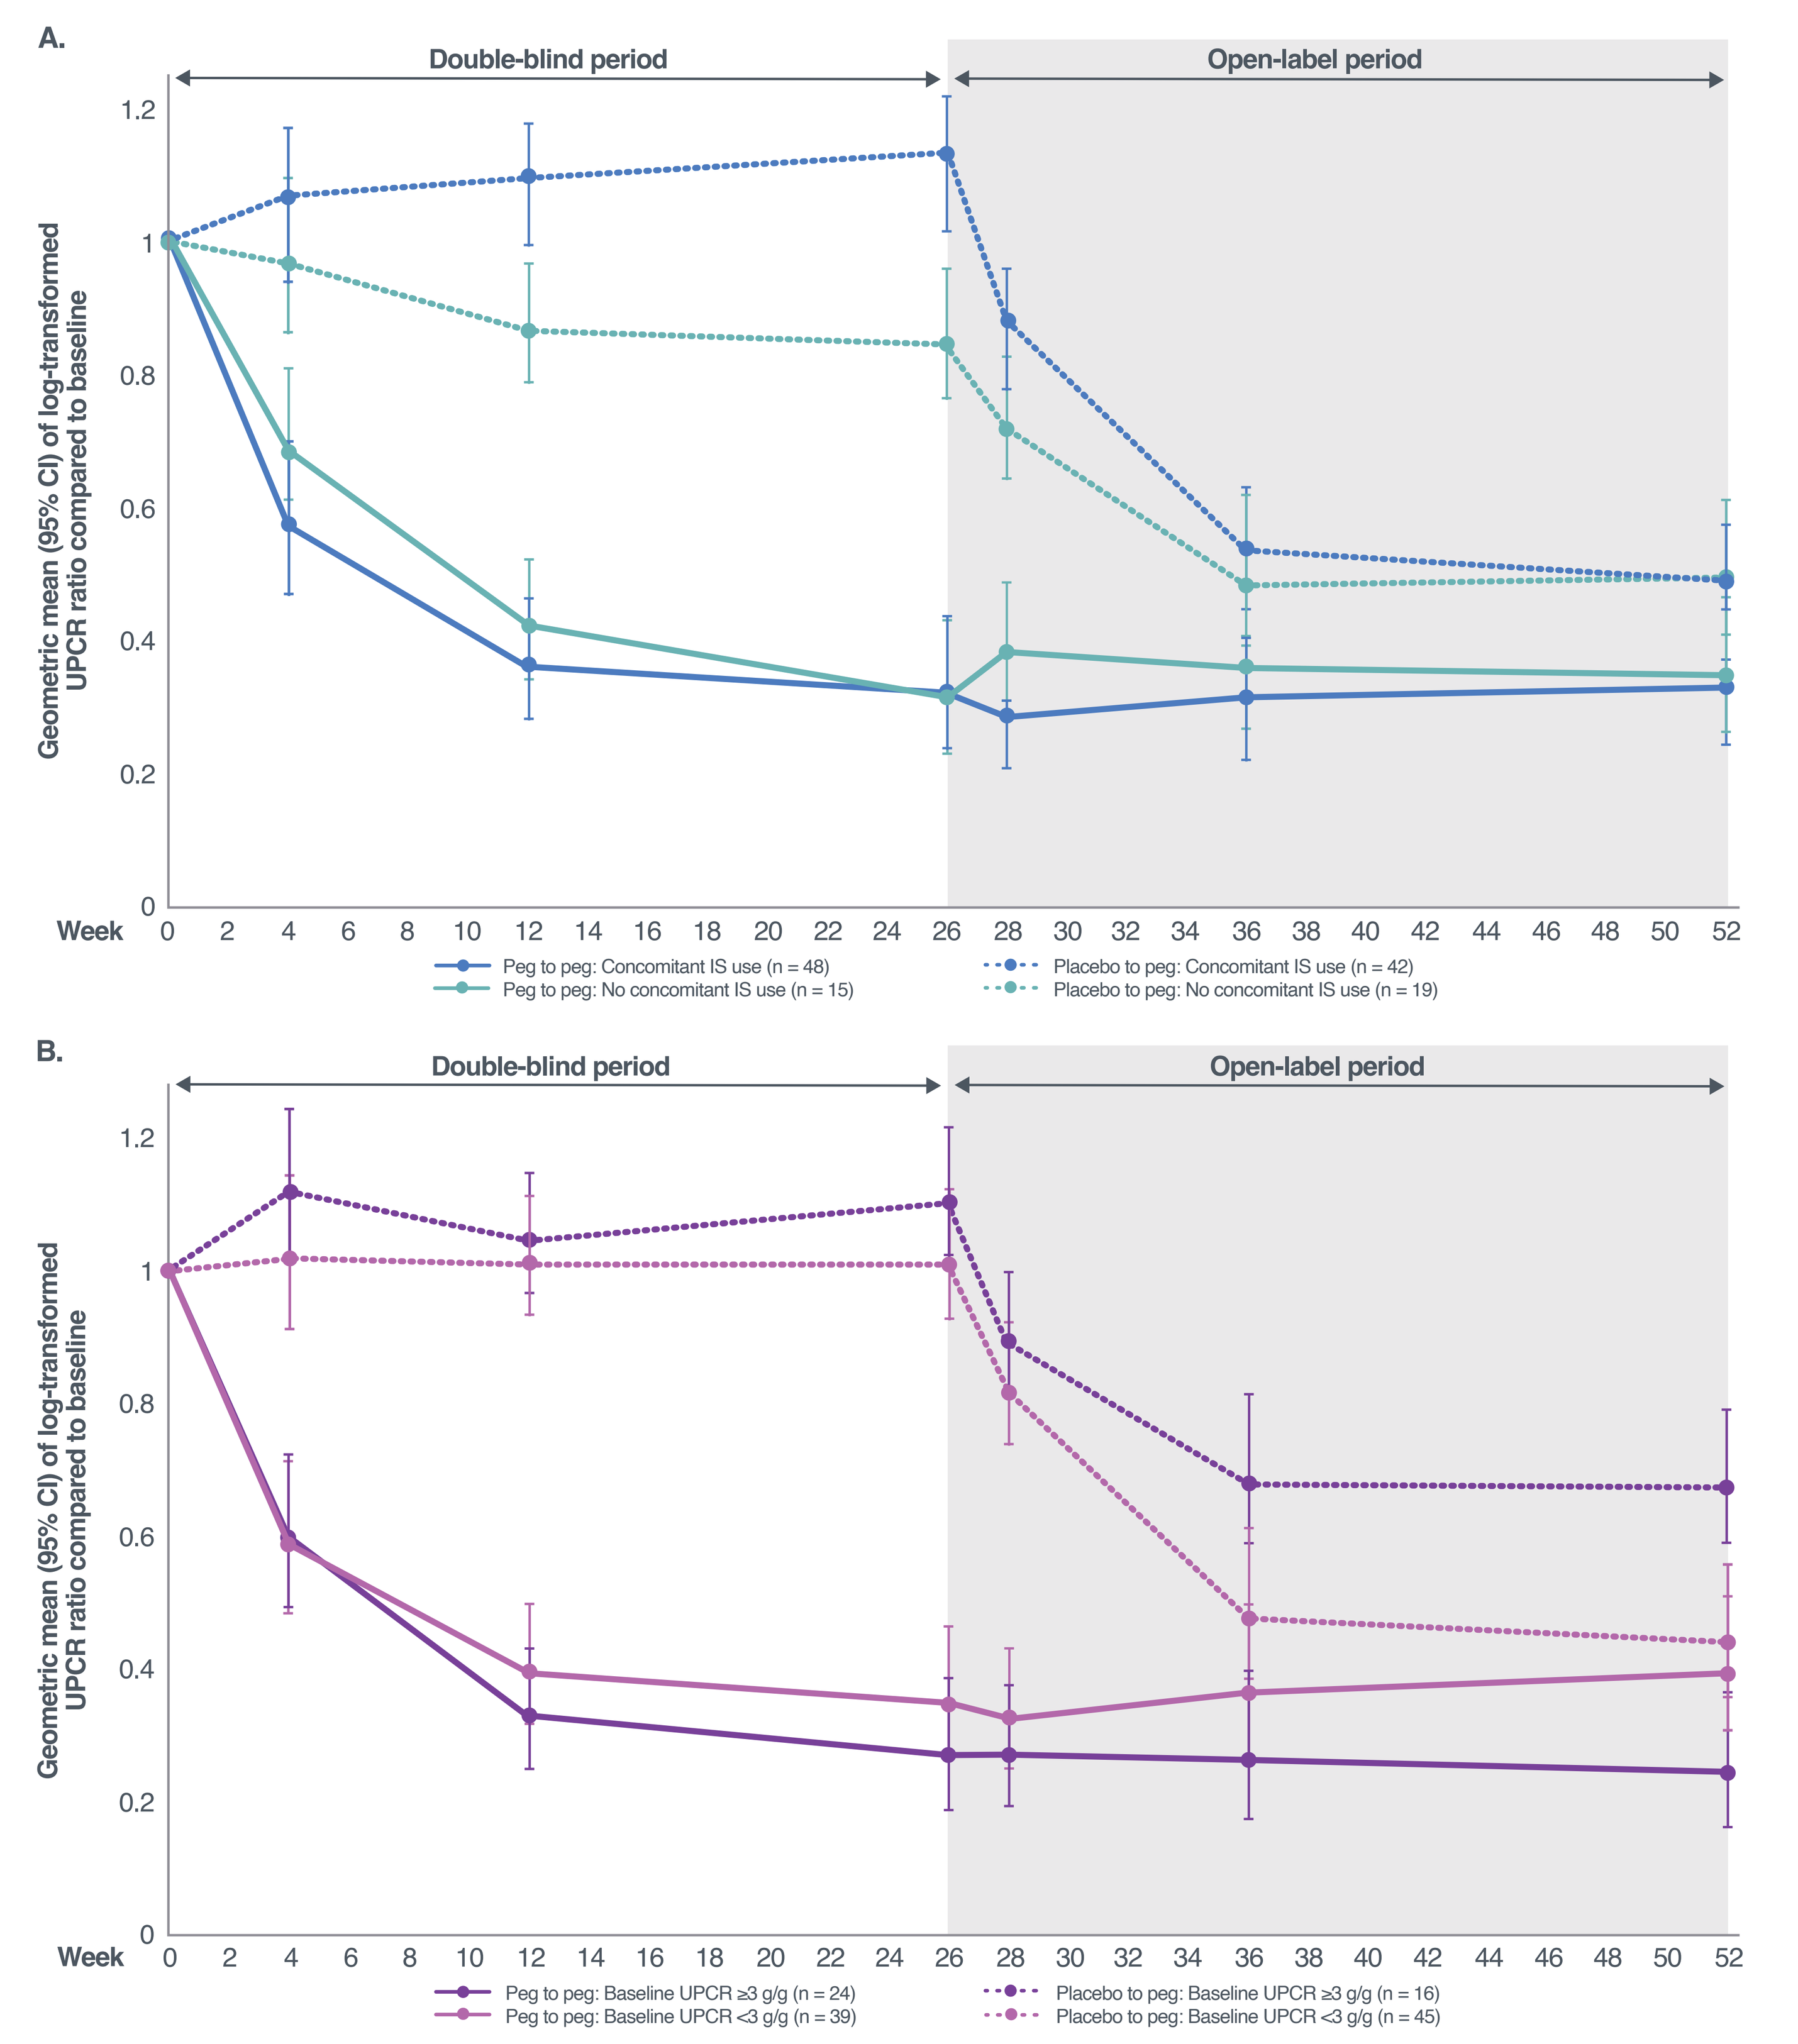
A.
Double-blind period
Open-label period
Geometric mean (95% CI) of log-transformed UPCR ratio compared to baseline
Week
Peg to peg: Concomitant IS use (n = 48)
Peg to peg: No concomitant IS use (n = 15)
Placebo to peg: Concomitant IS use (n = 42)
Placebo to peg: No concomitant IS use (n = 19)
B.
Double-blind period
Open-label period
Geometric mean (95% CI) of log-transformed UPCR ratio compared to baseline
Week
Peg to peg: Baseline UPCR ≥3 g/g (n = 24)
Peg to peg: Baseline UPCR <3 g/g (n = 39)
Placebo to peg: Baseline UPCR ≥3 g/g (n = 16)
Placebo to peg: Baseline UPCR <3 g/g (n = 45)

IS use was defined as receiving concomitant medication within the ATC Level 2 categories of Immunosuppressants or Corticosteroids for Systemic Use using the WHO Drug Dictionary version WHODrug-Global-B3-202209.
IS, immunosuppressant; peg, pegcetacoplan; UPCR, urine protein-to-creatinine ratio.

- eGFR was stable for the pegcetacoplan-to-pegcetacoplan group (LS mean [SE] change from baseline, mL/min/1.73 m^2: week 26, –1.5 [2.2]; week 52, –3.7 [2.7]); placebo-to-pegcetacoplan patients had a slowing of eGFR decline after switching to pegcetacoplan (week 26, –7.8 [1.9]; week 52, –4.7 [2.2]) (**Figure 3**)
- LS mean (SE) eGFR changes from baseline for patients in the pegcetacoplan-to-pegcetacoplan treatment group with concomitant IS treatment or nephrotic range proteinuria were consistent with the total population

RESULTS *(cont.)*

Figure 3. Change in eGFR during VALIANT in the total population

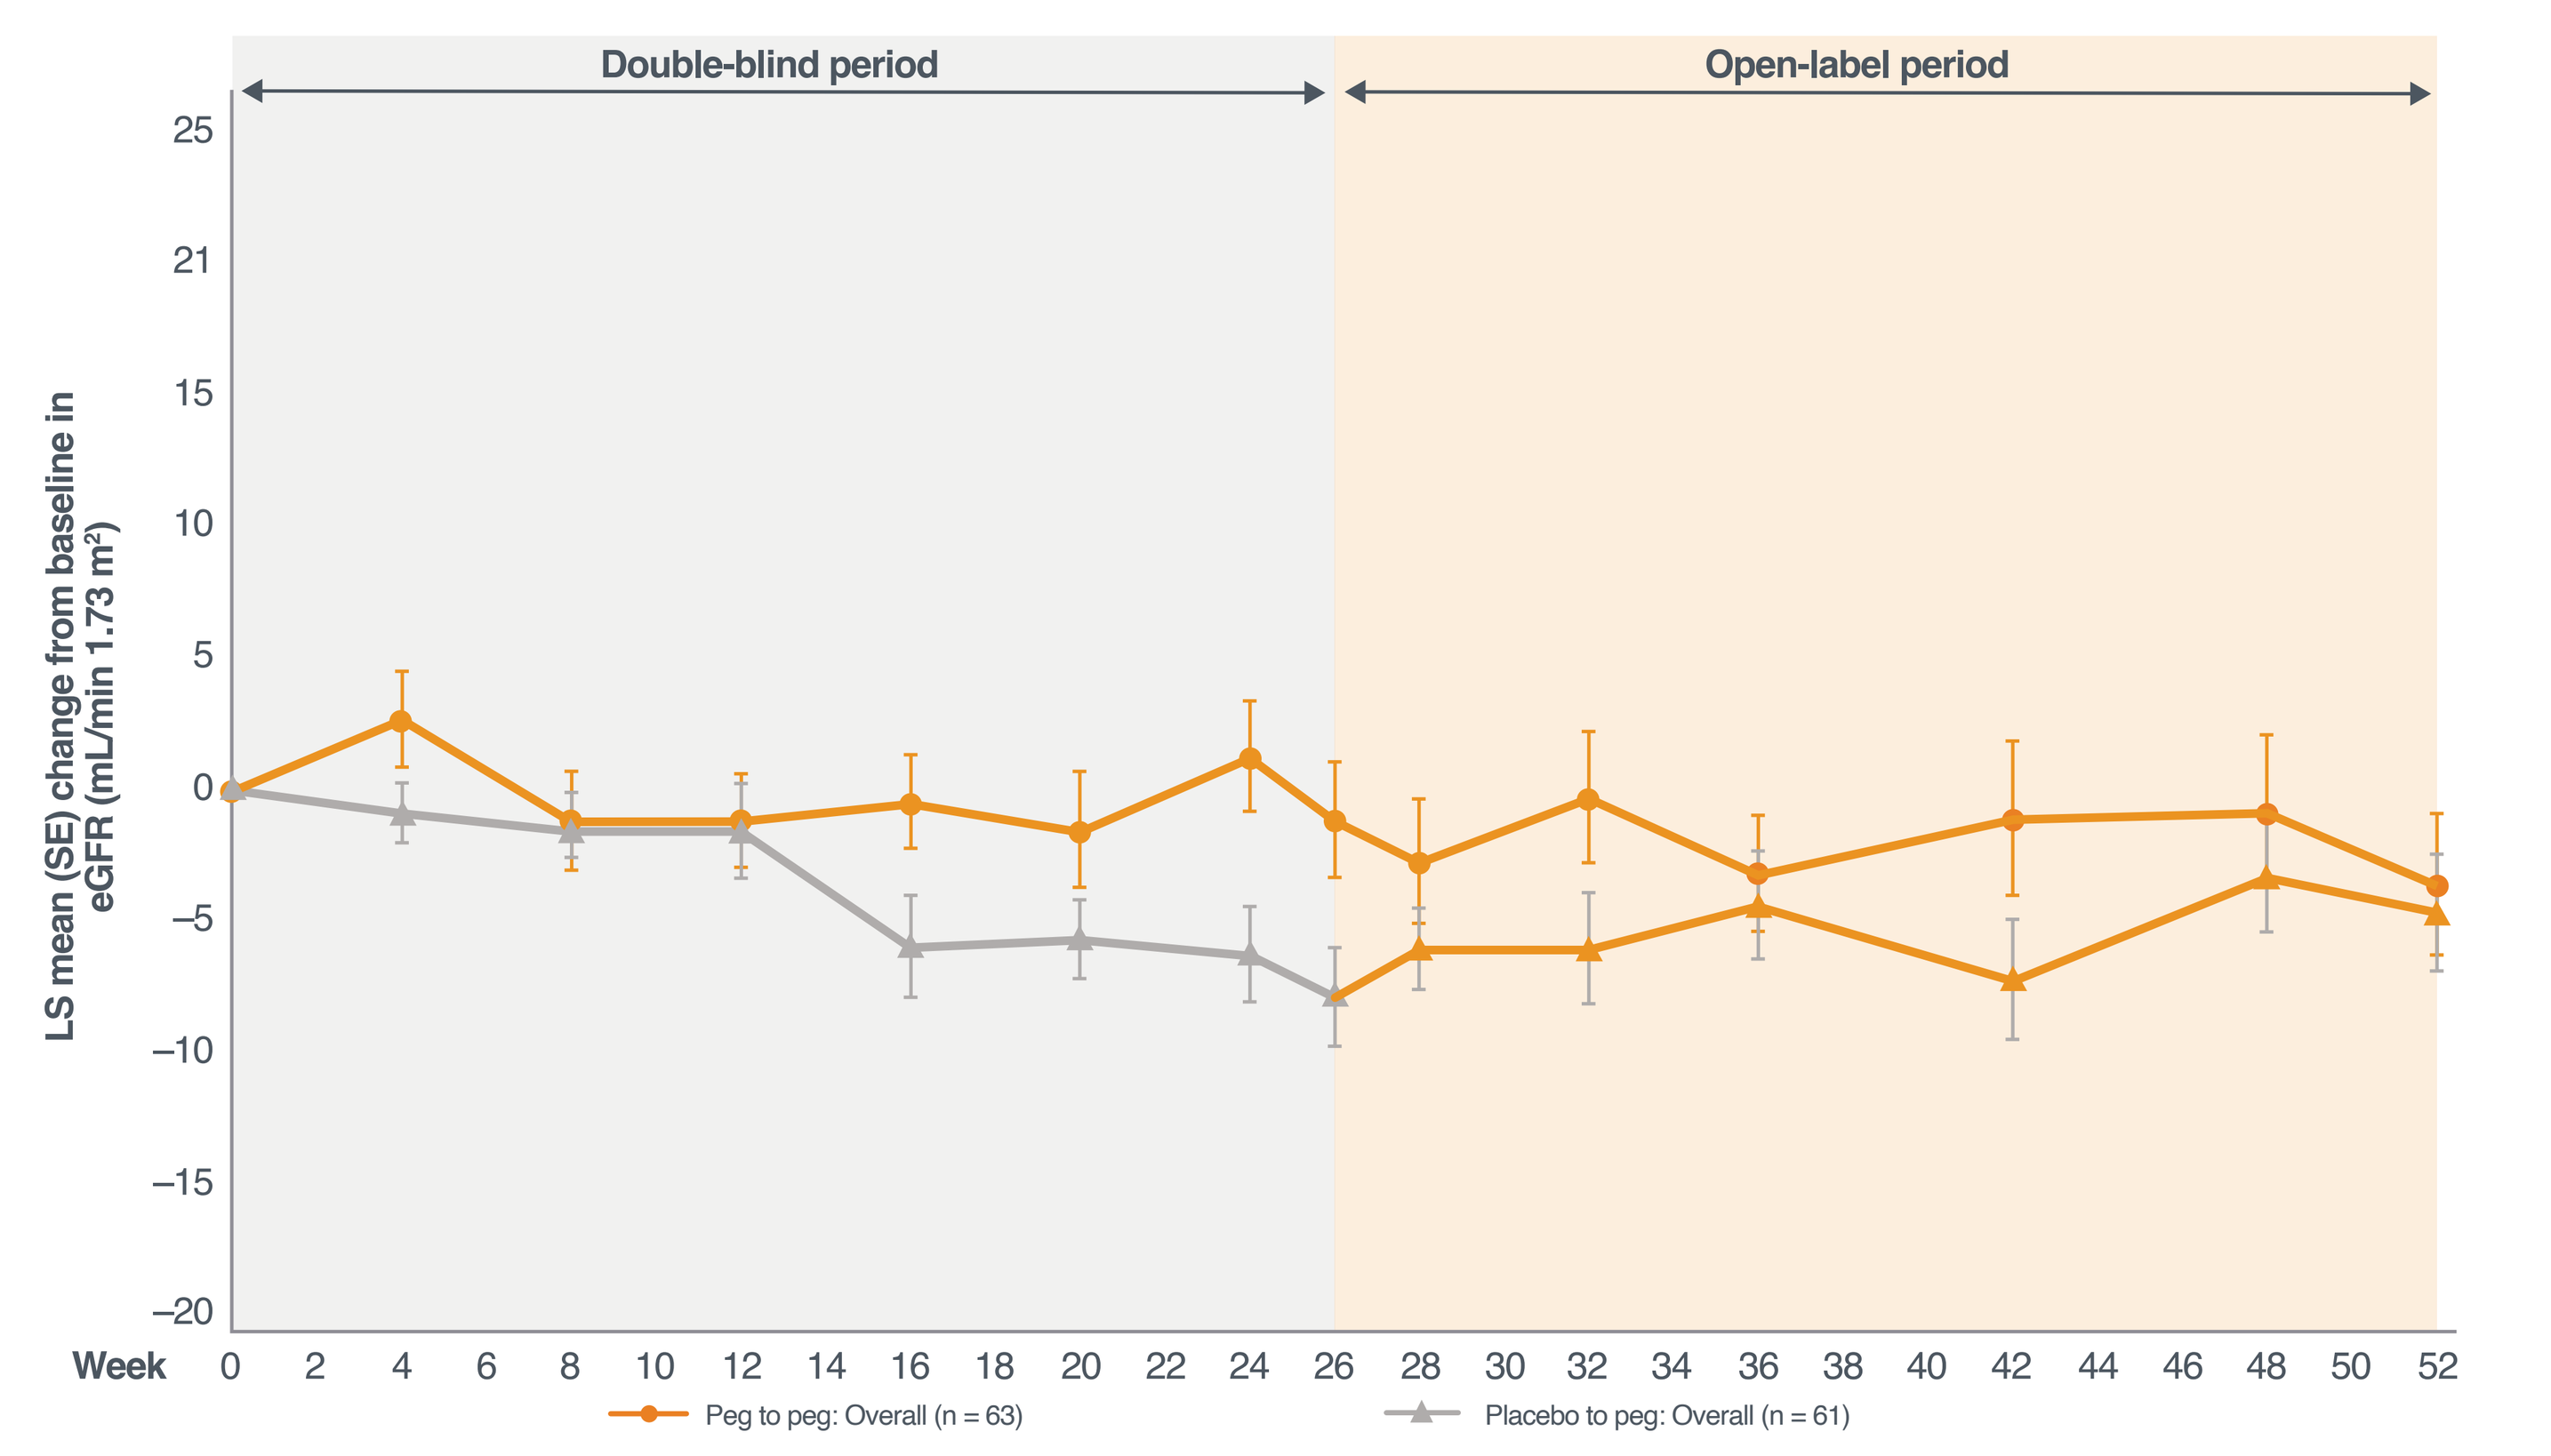
Double-blind period
Open-label period
LS mean (SE) change from baseline in eGFR (mL/min 1.73 m²)
Week
Peg to peg: Overall (n = 63)
Placebo to peg: Overall (n = 61)

eGFR, estimated glomerular filtration rate; LS, least-squares; SE, standard error.

- Overall, patients had 99% adherence to pegcetacoplan dosing
- No new safety signals were identified through 52 weeks
- No graft losses occurred during the trial
- No encapsulated meningococcal infections were reported

LIMITATIONS

- Small numbers in the post hoc analyses limit interpretation
- The heterogeneity of IS regimens and treatment durations were not accounted for in this analysis

References: 1. Bomback AS, et al. *Kidney Int Rep*. 2024;10:17–28. **2.** Mastrangelo A, et al. *Front Pediatr*. 2020;8:205. **3.** Fakhouri F, et al. 62nd ERA Congress. June 4–7, 2025. **4.** Nester C, et al. *Clin J Am Soc Nephrol*. 2024;19(9):1201. **5.** Rovin BH, et al. *Kidney Int*. 2021;100(4):S1. **6.** Iimori S, et al. *PLoS One*. 2018;13(1):e0190493. 7. ClinicalTrials.gov. VALIANT. Accessed August 21, 2025. clinicaltrials.gov/study/NCT05809531.

ABBREVIATIONS: ACEis, angiotensin-converting enzyme inhibitors; ARBs, angiotensin receptor blockers; C3G, C3 glomerulopathy; eGFR, estimated glomerular filtration rate; IC-MPGN, immune-complex membranoproliferative glomerulonephritis; IS, immunosuppressant; LS, least-squares; MMF, mycophenolate mofetil; OLP, open-label period; RCP, randomized controlled period; SGLT2is, sodium-glucose cotransporter-2 inhibitors; SC, subcutaneous; UPCR, urine protein-to-creatinine ratio.

Disclosures: **CMN** is the Associate Director for Molecular Otolaryngology and Renal Research Laboratory; receives NIH grant support (2R01DK110023-07); serves on advisory boards for Novartis, Apellis, Biocryst, and Alexion; participates as a site investigator for Novartis, Apellis, Biocryst, and Retrophin; is a member of the data safety monitoring board for Kira; serves as Chair of a data safety monitoring board for FIT4KID; and receives author royalties for UpToDate. **ASB** has received consulting fees from Amgen, Apellis, Catalyst, Genentech, Kezar, Novartis, Q32, Silence Therapeutics, and Visterra. **GA** received honoraria for lectures, educational events, or advisory boards for AstraZeneca (Alexion), Recordati Rare Disease, Advicenne, Chiesi, Kyowa Kirin, Alnylam, and Dicerna; and served as site investigator for Apellis. **YD** has received consulting fees from Alexion, Novartis, Sanofi, and Takeda. **BpD** received consulting fees and honoraria from Alexion (AstraZeneca Rare Disease), Apellis, Novartis, and Arrowhead. **LAG** receives research support from Alexion and Apellis. He has served as a consultant for Novartis, Alexion, and Roche. **NI** served on an advisory board for Alexion. **DPG**, **SHH**, **CL**, **MINdH**, **PDW**, **DW**, **MLQ**, and **NM** have nothing to disclose. **AM** received support from Sobi. **MM** has received consultant and/or advisory bord fees from Apellis, Sobi, and Novartis; speakers' bureau fees from Novartis; and received Novartis grant support. **MCP** has received consulting fees from Alexion, Achillion, Annexon, Apellis, Biocryst, ChemoCentryx, Complement Therapeutics, Gemini, Gyroscope, MIRNA Therapeutics, Ormeros and Q32 Bio. He is also supported by a Wellcome Trust Senior Fellow in Clinical Science grant 212252/Z/18/Z. This research was supported by the National Institute for Health and Care Research (NIHR) Imperial Biomedical Research Centre. The views expressed are those of the authors and not necessarily those of the NIHR or the Department of Health and Social Care. **GR** has received consulting fees from BioCryst and Silence Therapeutics and speakers' bureau fees from Novartis. **NvdeK** has received consultancy fees from Sobi, Roche, Novartis, Alexion, and Samsung. **MV** received consultancy fees from Novartis Travere, Roche, Apellis, Alexion, BioCryst, PureSpring, Bayer, and WebMD; received research funding from Alexion, Chemocentrix, Bayer, Novartis, Roche, Chinook, Apellis, and Travere for participation in clinical studies; and served on speakers' bureaus for Novartis, Roche, Vifor, Travere, and GSK. **DZ** has received consulting fees from Apellis. **FF** has received consulting fees from Apellis, Sobi Novartis, Roche, Alexion, and AstraZeneca.

ACKNOWLEDGMENTS Research funding source: This study was funded by Apellis Pharmaceuticals, Inc and Sobi (Swedish Orphan Biovitrum AB). Writing assistance was provided Jennifer Gibson, PharmD, and Kathryn Fogarty, PhD, of Kay Square Scientific (Butler, PA, USA), and was funded by Apellis Pharmaceuticals, Inc and Sobi (Swedish Orphan Biovitrum AB).

58th Annual Meeting of the American Society of Nephrology (ASN 2025) | November 5–9, 2025 | Houston, TX