Healthcare Resource Utilization in Patients with Primary Immune Thrombocytopenia Treated with Avatrombopag in the United States: REAL-AVA 2.0 Study Results

#359

Abiola Oladapo,[1] Srikanth Nagalla,[2] M Y Levy,[3] Shruti Chaturvedi,[4] Scott Kolodny,[1] Chelsea Bernheisel,[1] Elyse Swallow,[5] Debbie Goldschmidt,[5] Alexandra Greatsinger,[5] Loren Ormenaj,[5] Sinia Sareen,[5] Michael Vredenburg[1]

[1]Sobi Inc., Durham, NC, USA; [2]Miami Cancer Institute, Miami, FL, USA; [3]Texas Oncology, Baylor Charles A. Sammons Cancer Center, Dallas, TX, USA; [4]Division of Medicine-Hematology, Johns Hopkins School of Medicine, Baltimore, MD, USA; [5]Analysis Group, Boston, MA, USA

CONCLUSIONS

- This real-world observational study of patients with primary immune thrombocytopenia (ITP) demonstrated consistent reductions in ITP-related hospitalizations, emergency room (ER) visits, and outpatient (OP) visits following avatrombopag (AVA) initiation, including in patients with varying ITP disease durations and prior thrombopoietin receptor agonist (TPO-RA) exposure.
- The findings support AVA's broad applicability and effectiveness in real-world ITP management. The decline in hospitalizations and ER visits suggests a lower incidence of serious ITP-related events requiring urgent or intensive medical care, and the decrease in OP visits reflects a reduction in ongoing care needs.
- Overall, the study provides real-world evidence that AVA is an effective treatment option for patients with primary ITP that may be associated with a reduction in healthcare resource utilization (HRU) across a clinically diverse patient population.

INTRODUCTION

- Primary ITP is an autoimmune disorder marked by low platelet counts, leading to an elevated risk of bleeding that ranges from minor episodes to a life-threatening hemorrhage.[1]
- Management of ITP and its associated bleeding complications is often resource-demanding, involving frequent medical visits and contributing to significant clinical and economic burden.[2]
- For adults with ITP, TPO-RAs such as AVA are widely used as treatments to enhance platelet production in patients with inadequate response to prior ITP therapy.
- AVA's safety and efficacy have been well established in clinical studies;[3-5] however, limited real-world data are available on the HRU of patients receiving AVA in routine clinical practice.

Study objective

- To assess ITP-related hospitalizations, ER visits, and OP visits among adult patients who were treated with AVA for primary ITP.

METHODS

Data Source

- REAL-AVA 2.0 was a multicenter, retrospective, non-interventional chart review study which evaluated real-world outcomes in patients treated with AVA for primary ITP in the US.
- Participating healthcare centers included academic medical centers and community-based practices that were geographically dispersed across US. Each healthcare center submitted de-identified, patient-level data from up to 30 patient charts into a secure online electronic chart review form (eCRF) from June 2023 to December 2024.

Study Design and Population

- The analysis included adults with primary ITP treated at U.S. healthcare centers who initiated AVA between July 2019 and June 2024.
- Eligible patient charts included those with ≥3 months of medical records prior to AVA initiation and ≥6 months of follow-up after, except in cases of shorter follow-up due to death. Patients were excluded if they had secondary ITP or prior participation in an AVA clinical trial.
- The **index date** was defined as AVA initiation. The **baseline period** was the 3 months prior, and the **follow-up period** spanned from index through the earliest of AVA discontinuation, death, last chart entry, or data cutoff (December 31, 2024). Analyses were conducted for the full cohort and within two stratified subgroups:
 - ITP duration at AVA initiation: acute (<3 months), persistent (3–12 months), or chronic (≥12 months).
 - TPO-RA exposure prior to AVA initiation: yes/no.

Study Outcomes and Analyses

- ITP-related hospitalizations, ER visits, and OP visits were summarized descriptively before and after AVA initiation. A visit was considered "ITP-related" if the purpose was to discuss or address the patient's ITP condition, treatment, or related complications, as determined by the abstractor.
- Visit rates were reported on a per patient per month (PPPM) basis to account for variable follow-up. Results were reported for patients with ≥1 visit of each type.
- Pre- vs. post-AVA visit rates were compared using Wilcoxon signed-rank tests. No alpha adjustment was made.
- ITP-related HRU was also assessed longitudinally by 3-month intervals post-AVA (months 1–3, 4–6, 7–9, 10–12), including only patients with sufficient follow-up for each interval.

RESULTS

Patient characteristics

- 177 patients from 11 U.S. healthcare centers met inclusion criteria and were included in the analysis.
- The mean age was 56.4 (SD 18.9) years; 54% female; 75% White. Patients were geographically distributed across the country (38% South, 34% West). Half (50%) had commercial insurance.
- The median [IQR] disease duration of ITP was 2.2 [0.3, 6.8] years.
- Based on time from initial ITP diagnosis to AVA initiation, 22% (n = 39) were categorized as having acute ITP, 16% (n = 29) had persistent ITP, and 58% (n = 103) had chronic ITP.
 - 6 patients (3%) had missing ITP duration and were excluded from this stratification analysis.
- 117 patients (66%) had received at least one other TPO-RA before initiating AVA, while 60 patients (34%) had no prior TPO-RA exposure.

Table 1. Patient demographic and clinical characteristics

	All Patients N = 177	ITP Type at AVA Initiation: Acute ITP N = 39	Persistent ITP N = 29	Chronic ITP N = 103	Prior TPO-RA Treatment: Prior TPO-RA N = 117	No Prior TPO-RA N = 60
Demographic Characteristics						
Age at index date, mean ± SD, years	56.4 ± 18.9	58.4 ± 17.5	61.3 ± 18.6	54.6 ± 19.5	58.4 ± 19.0	52.4 ± 18.3
Female, n (%)	96 (54.2%)	18 (46.2%)	16 (55.2%)	60 (58.3%)	67 (57.3%)	29 (48.3%)
Race/ethnicity[1], n (%)						
White	132 (74.6%)	27 (69.2%)	22 (75.9%)	78 (75.7%)	93 (79.5%)	39 (65.0%)
Hispanic, Latino or Spanish origin	20 (11.3%)	5 (12.8%)	7 (24.1%)	8 (7.8%)	10 (8.5%)	10 (16.7%)
Black or African American	17 (9.6%)	2 (5.1%)	2 (6.9%)	12 (11.7%)	10 (8.5%)	7 (11.7%)
Other/unknown	14 (7.9%)	7 (17.9%)	1 (3.4%)	6 (5.8%)	6 (5.1%)	8 (13.3%)
Geographic region, n (%)						
South	67 (37.9%)	12 (30.8%)	11 (37.9%)	41 (39.8%)	44 (37.6%)	23 (38.3%)
West	60 (33.9%)	14 (35.9%)	13 (44.8%)	31 (30.1%)	39 (33.3%)	21 (35.0%)
Northeast	33 (18.6%)	10 (25.6%)	3 (10.3%)	20 (19.4%)	22 (18.8%)	11 (18.3%)
Midwest	17 (9.6%)	3 (7.7%)	2 (6.9%)	11 (10.7%)	12 (10.3%)	5 (8.3%)
Insurance type[1], n (%)						
Commercial/private insurance	89 (50.3%)	15 (38.5%)	16 (55.2%)	56 (54.4%)	60 (51.3%)	29 (48.3%)
Medicare	65 (36.7%)	17 (43.6%)	11 (37.9%)	35 (34.0%)	46 (39.3%)	19 (31.7%)
Medicaid	42 (23.7%)	11 (28.2%)	7 (24.1%)	21 (20.4%)	27 (23.1%)	15 (25.0%)
None	5 (2.8%)	3 (7.7%)	0 (0.0%)	2 (1.9%)	2 (1.7%)	3 (5.0%)
Other/unknown	14 (7.9%)	2 (5.1%)	1 (3.4%)	10 (9.7%)	10 (8.5%)	4 (6.7%)
Clinical Characteristics						
ITP disease duration, median [IQR] years	2.2 [0.3, 6.8]	0.1 [0.1, 0.2]	0.5 [0.4, 0.6]	5.3 [2.7, 10.6]	2.8 [0.8, 8.5]	0.4 [0.2, 2.2]
ITP treatments during baseline, n (%)						
Any TPO-RA	81 (45.8%)	12 (30.8%)	14 (48.3%)	53 (51.5%)	81 (69.2%)	0 (0.0%)
Steroids[2]	86 (48.6%)	26 (66.7%)	16 (55.2%)	41 (39.8%)	44 (37.6%)	42 (70.0%)
Intravenous immunoglobulin (IVIG)	47 (26.6%)	21 (53.8%)	10 (34.5%)	15 (14.6%)	27 (23.1%)	20 (33.3%)
None	29 (16.4%)	6 (15.4%)	2 (6.9%)	19 (18.4%)	15 (12.8%)	14 (23.3%)

Notes:
[1] Categories are not mutually exclusive.
[2] Reported steroids included dexamethasone, prednisone, prednisolone, and methylprednisolone.

HRU

Overall Population

- The median [IQR] duration of follow-up for the HRU analysis in the overall population was 12.8 [5.9, 23.2] months.
- 25% (n=45) of patients had ≥1 ITP-related hospitalization during baseline, compared to 18% (n=31) during follow-up. The PPPM rate of ITP-related hospitalizations among patients with any hospitalizations decreased significantly from the baseline period to the follow-up period (median [mean]: 0.3 [0.5] vs. 0.1 [0.3] visits per month; p < 0.01) (**Figure 1**).
- While the proportion of patients with at least one ITP-related ER visit was 19% in both the baseline and follow-up periods, the PPPM rate among these patients declined significantly from baseline to follow-up (median [mean]: 0.3 [0.4] vs. 0.1 [0.1] visits per month; p < 0.01).
- During the 3-month baseline period, 64% of patients had ≥1 OP visit, compared to 76% during follow-up (p<0.05). However, the PPPM rate of ITP-related OP visits among patients with ≥1 visit decreased significantly from baseline to follow-up with a median [mean] PPPM rate of 1.0 [1.6] during baseline and 0.4 [0.9] during follow-up (p < 0.001).

Figure 1. PPPM rates of ITP-related HRU among patients with ≥1 visit

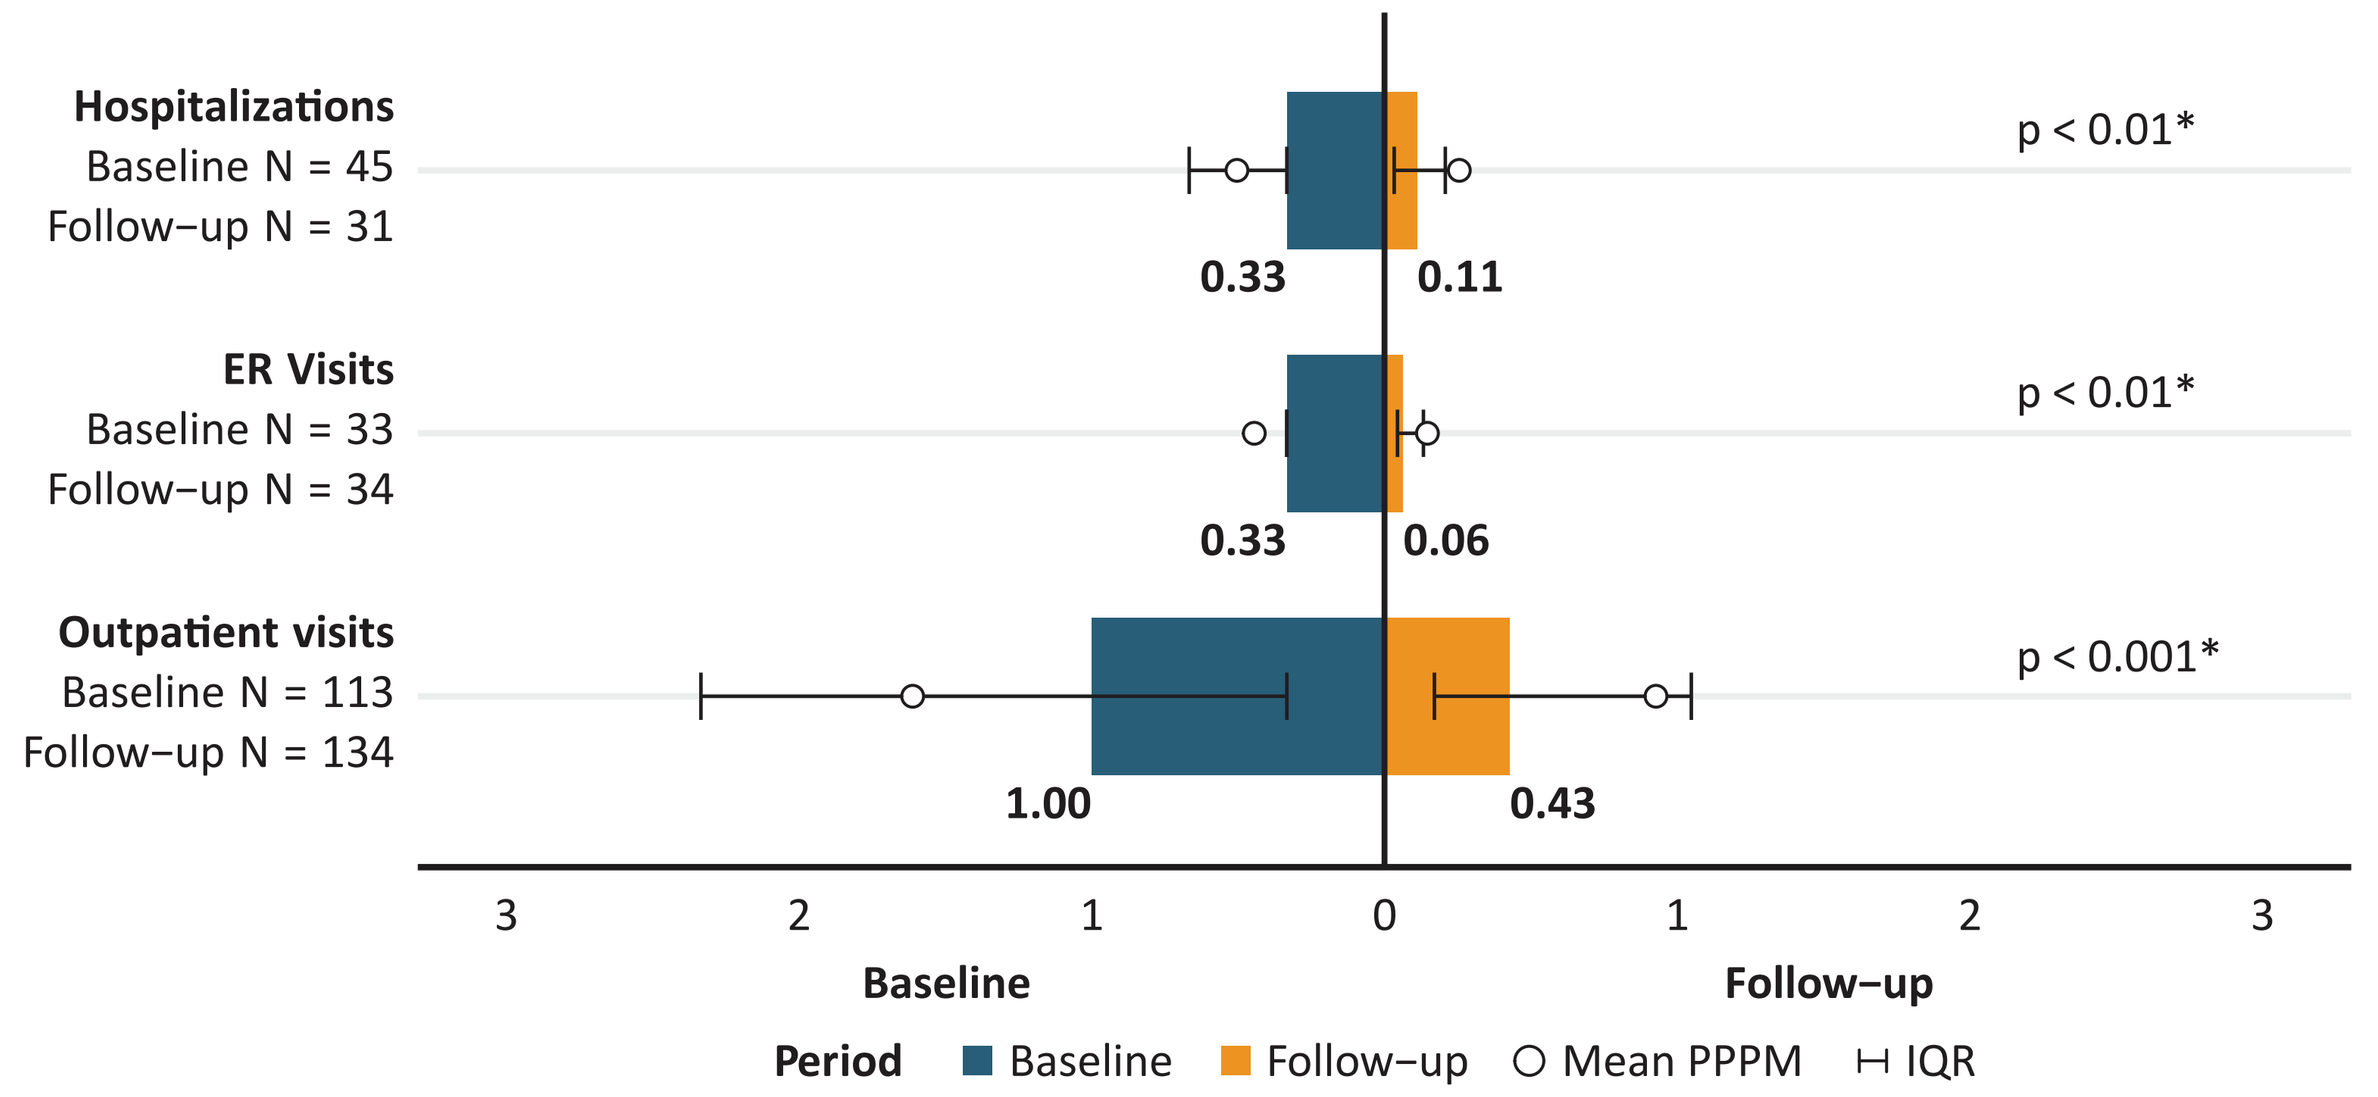
Hospitalizations
Baseline N = 45
Follow−up N = 31
0.33
0.11
p < 0.01*
ER Visits
Baseline N = 33
Follow−up N = 34
0.33
0.06
p < 0.01*
Outpatient visits
Baseline N = 113
Follow−up N = 134
1.00
0.43
p < 0.001*
3 2 1 0 1 2 3
Baseline
Follow−up
Period Baseline Follow−up Mean PPPM IQR

Notes: Median PPPM rates are bolded. * denotes statistical significance at α=0.05.

- The quarterly analysis of HRU during the baseline period through 12 months after AVA initiation showed a marked reduction in HRU after AVA initiation (**Figure 2**).
- The incidence of ITP-related hospitalizations and ER visits dropped sharply post-AVA initiation. During the baseline period, 25% of patients had ≥1 hospitalization and 19% had ≥1 ER visit; both fell to <5% by months 4–6 and remained low. Among these patients, mean hospitalization rates declined 1.5 to 1.0 and ER visits from 1.3 to 1.0 from the baseline period to months 7-9.
- OP visits declined gradually: 64% of patients had ≥1 OP visit at baseline, decreasing to ~50% by months 7-9. Mean OP visit rates fell from 4.8 (baseline) to 2.0 (months 10–12).

Figure 2. Quarterly ITP-related HRU

A. Proportion of patients with ≥1ITP-related visit by quarter

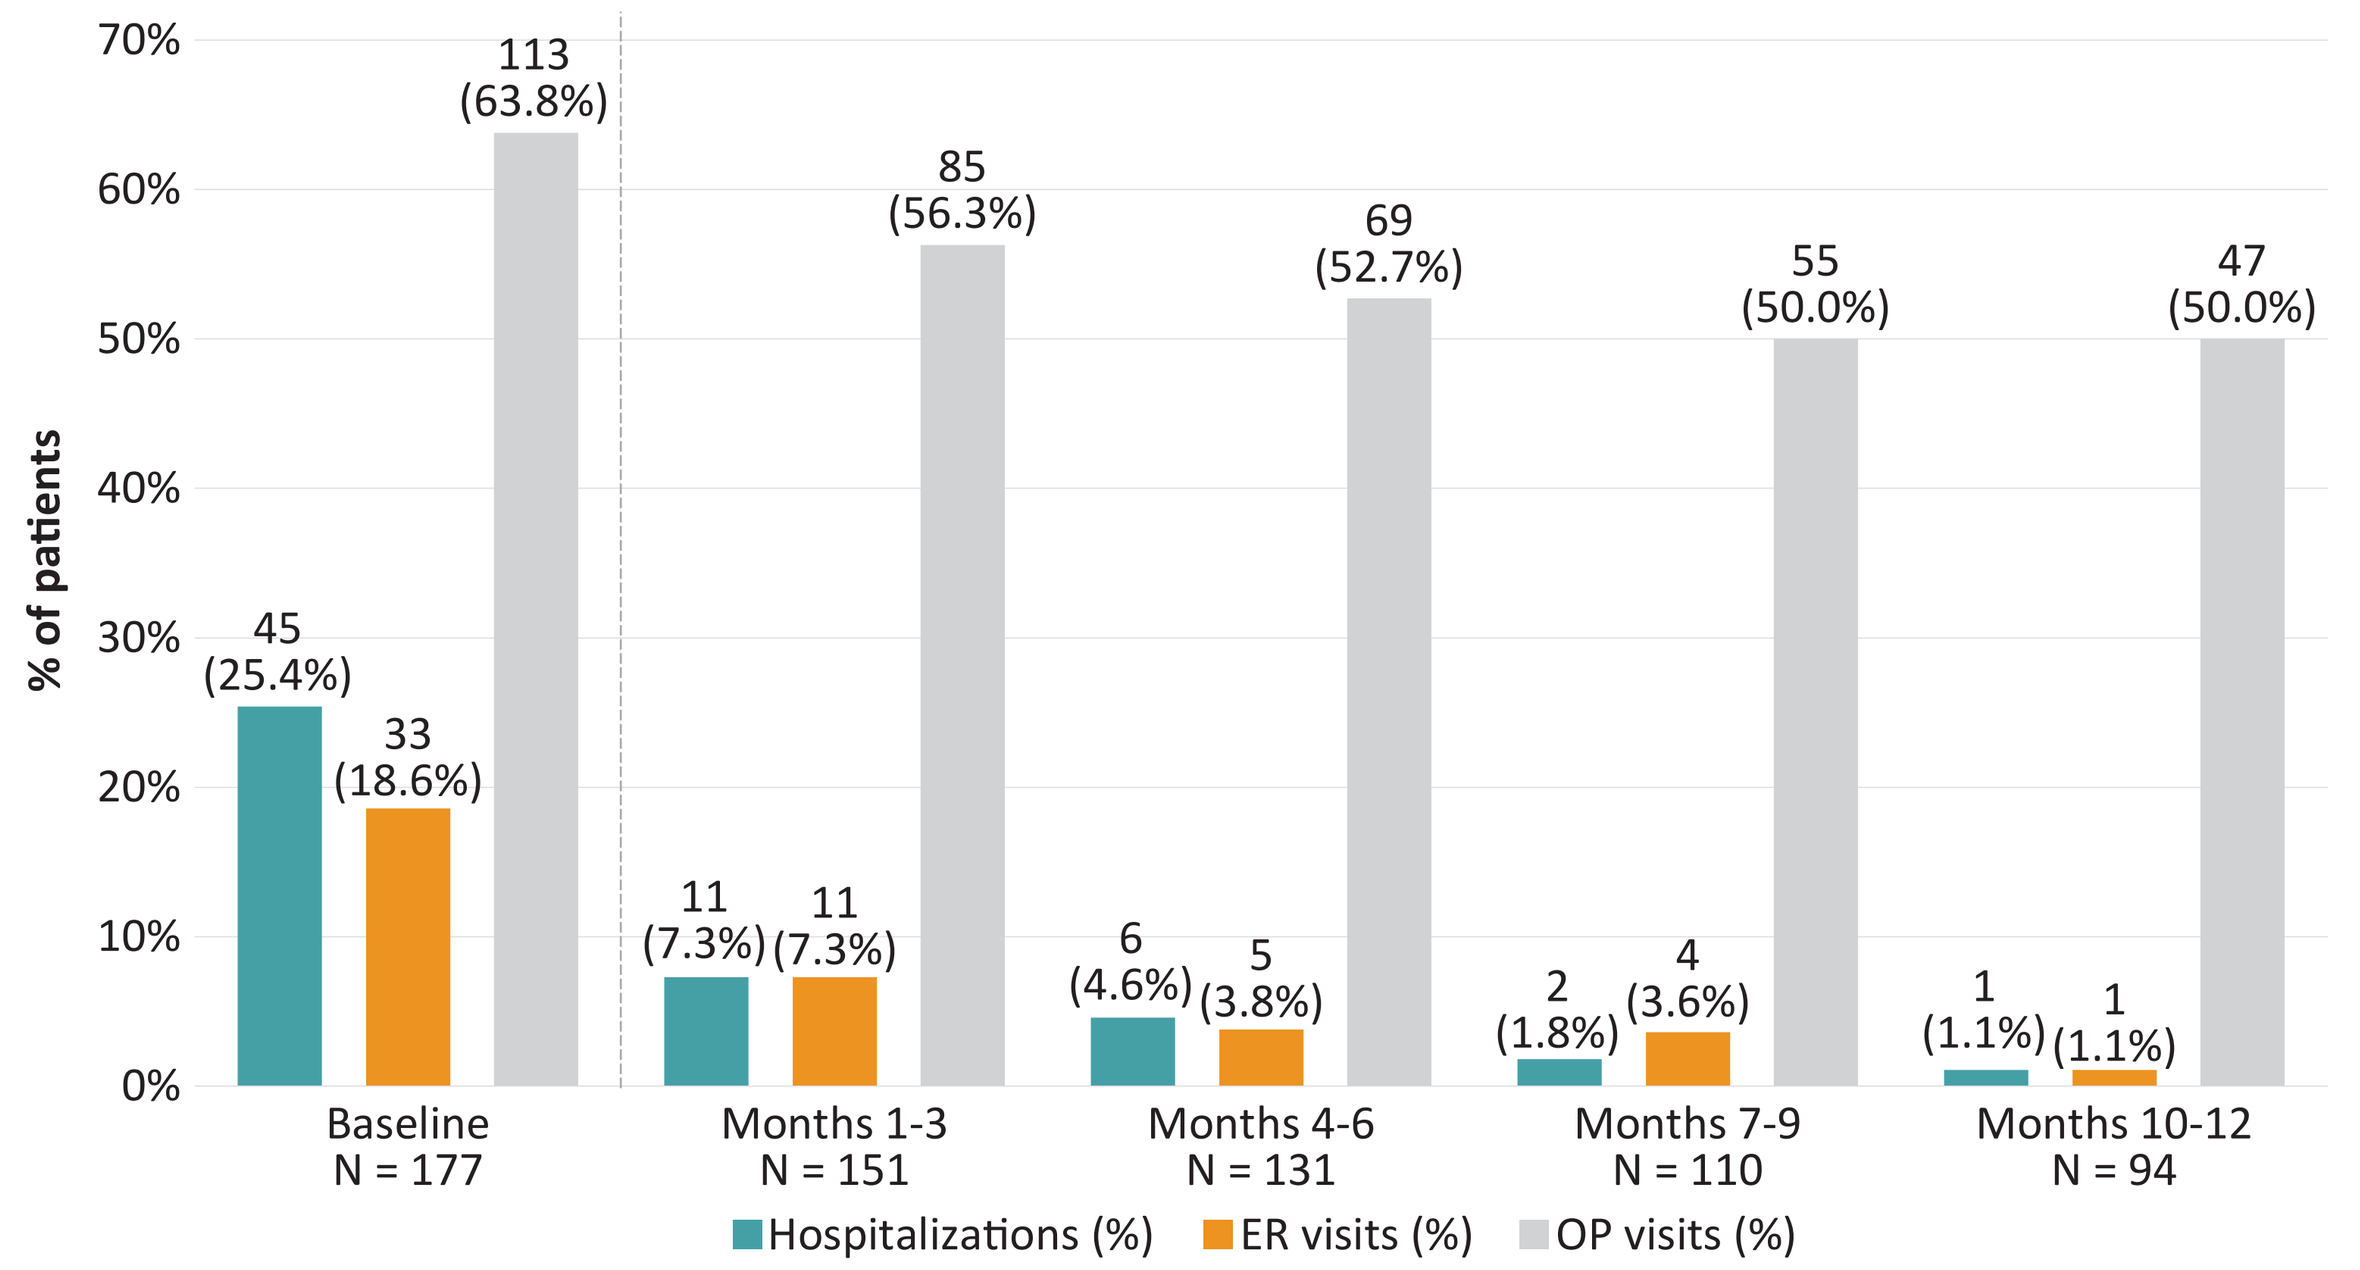
% of patients
70%
60%
50%
40%
30%
20%
10%
0%
45 (25.4%)
33 (18.6%)
113 (63.8%)
11 (7.3%)
11 (7.3%)
85 (56.3%)
6 (4.6%)
5 (3.8%)
69 (52.7%)
2 (1.8%)
4 (3.6%)
55 (50.0%)
1 (1.1%)
1 (1.1%)
47 (50.0%)
Baseline N = 177
Months 1-3 N = 151
Months 4-6 N = 131
Months 7-9 N = 110
Months 10-12 N = 94
Hospitalizations (%)
ER visits (%)
OP visits (%)

B. Number of ITP-related visits by quarter, among patients with ≥1 visit

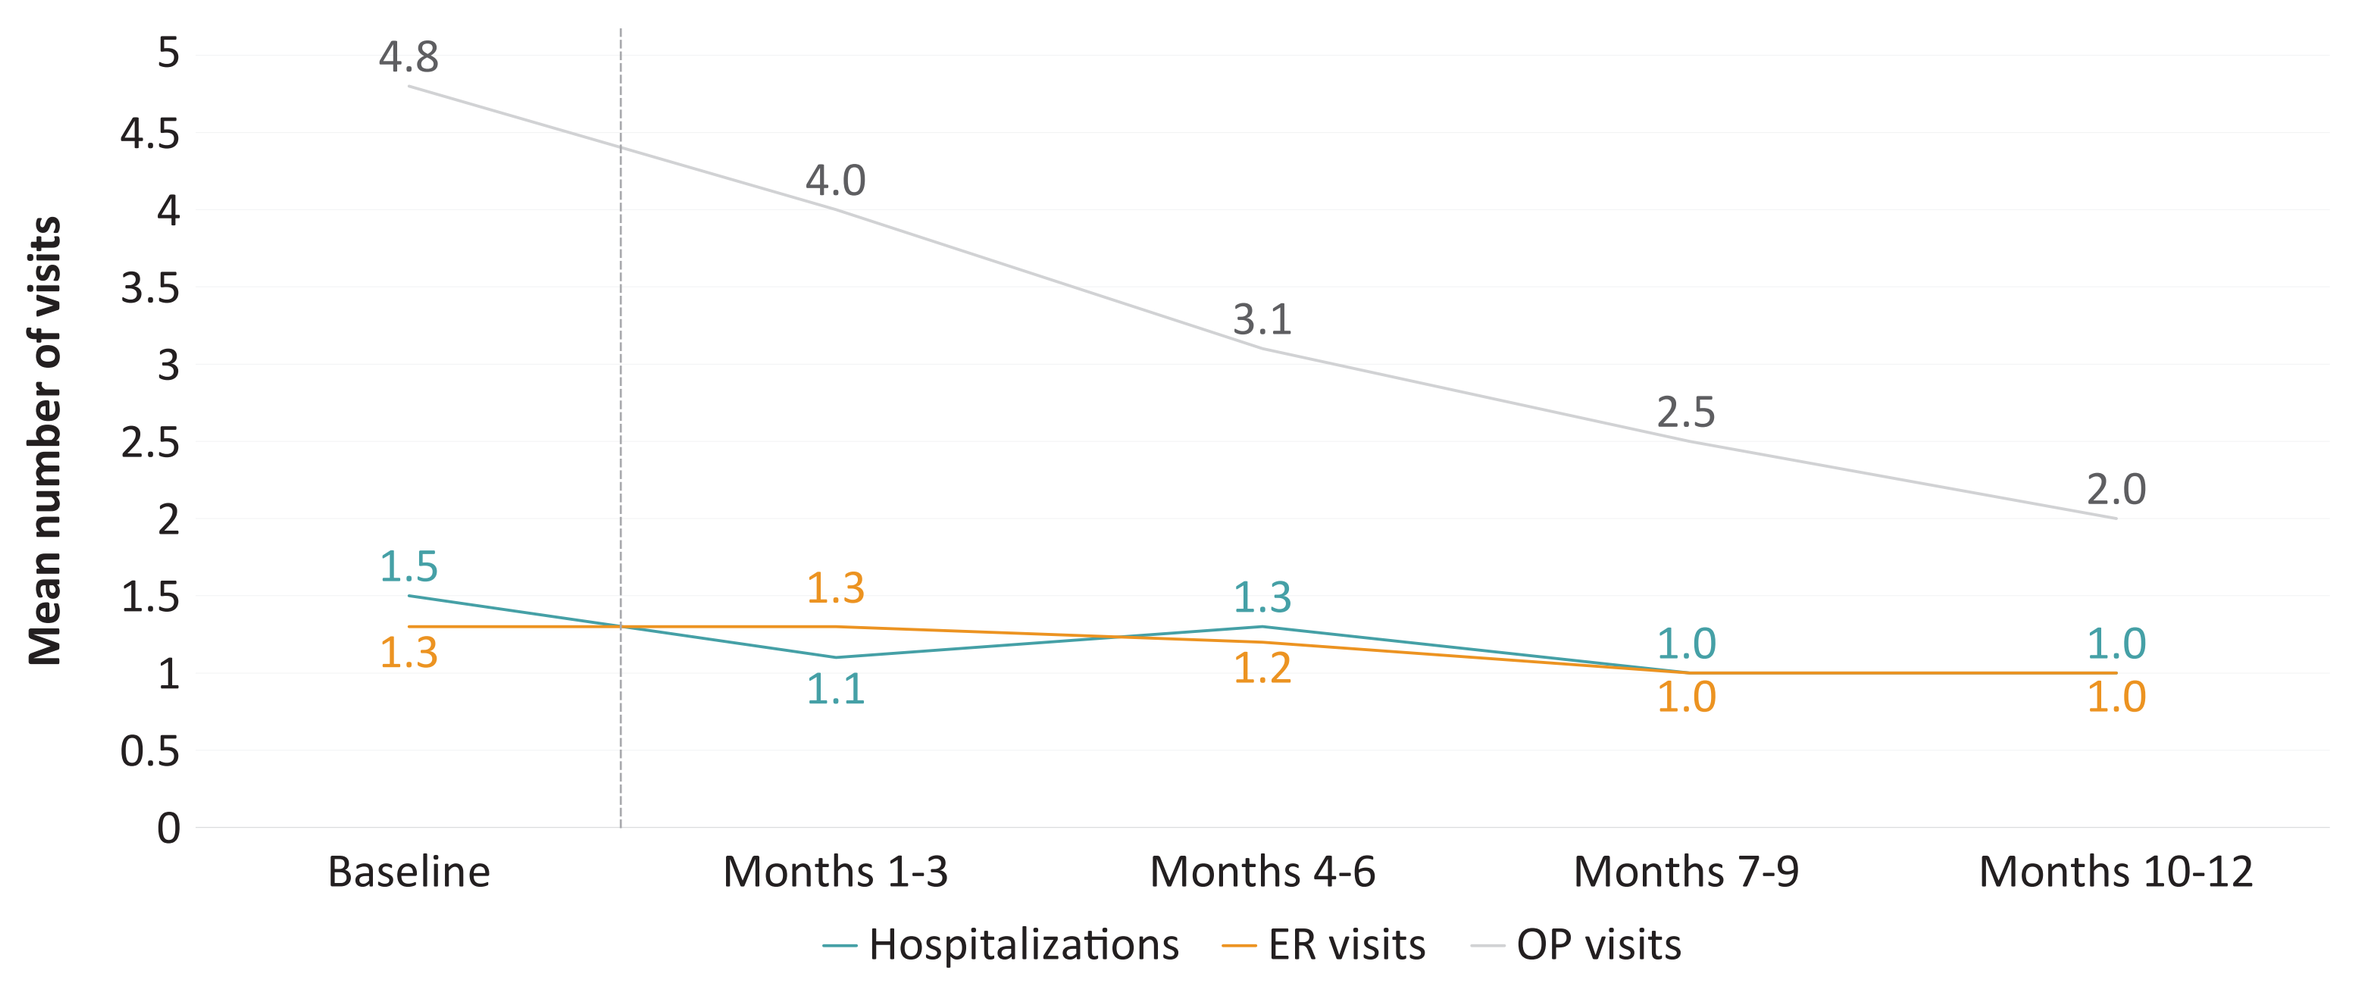
Mean number of visits
5
4.5
4
3.5
3
2.5
2
1.5
1
0.5
0
4.8
4.0
3.1
2.5
2.0
1.5
1.3
1.1
1.3
1.3
1.2
1.0
1.0
1.0
1.0
Baseline
Months 1-3
Months 4-6
Months 7-9
Months 10-12
Hospitalizations
ER visits
OP visits

Stratification by ITP disease duration

- Patients with acute, persistent, and chronic ITP experienced numerical declines in the median PPPM rates of ITP-related hospitalizations and ITP-related ER visits among patients with ≥1 visit from the baseline to the follow-up period (**Figure 3**).
- Patients with persistent and with chronic ITP experienced a significant decline in the median PPPM rate of ITP-related OP visits, decreasing from 1.0 in the baseline period to 0.6 (persistent ITP) and 0.3 (chronic ITP) in the follow-up period (both p < 0.01). Acute ITP patients had a consistent rate of ITP-related OP visits across the same time periods.

Figure 3. PPPM rates of ITP-related HRU among patients with ≥1 visit, by ITP disease duration

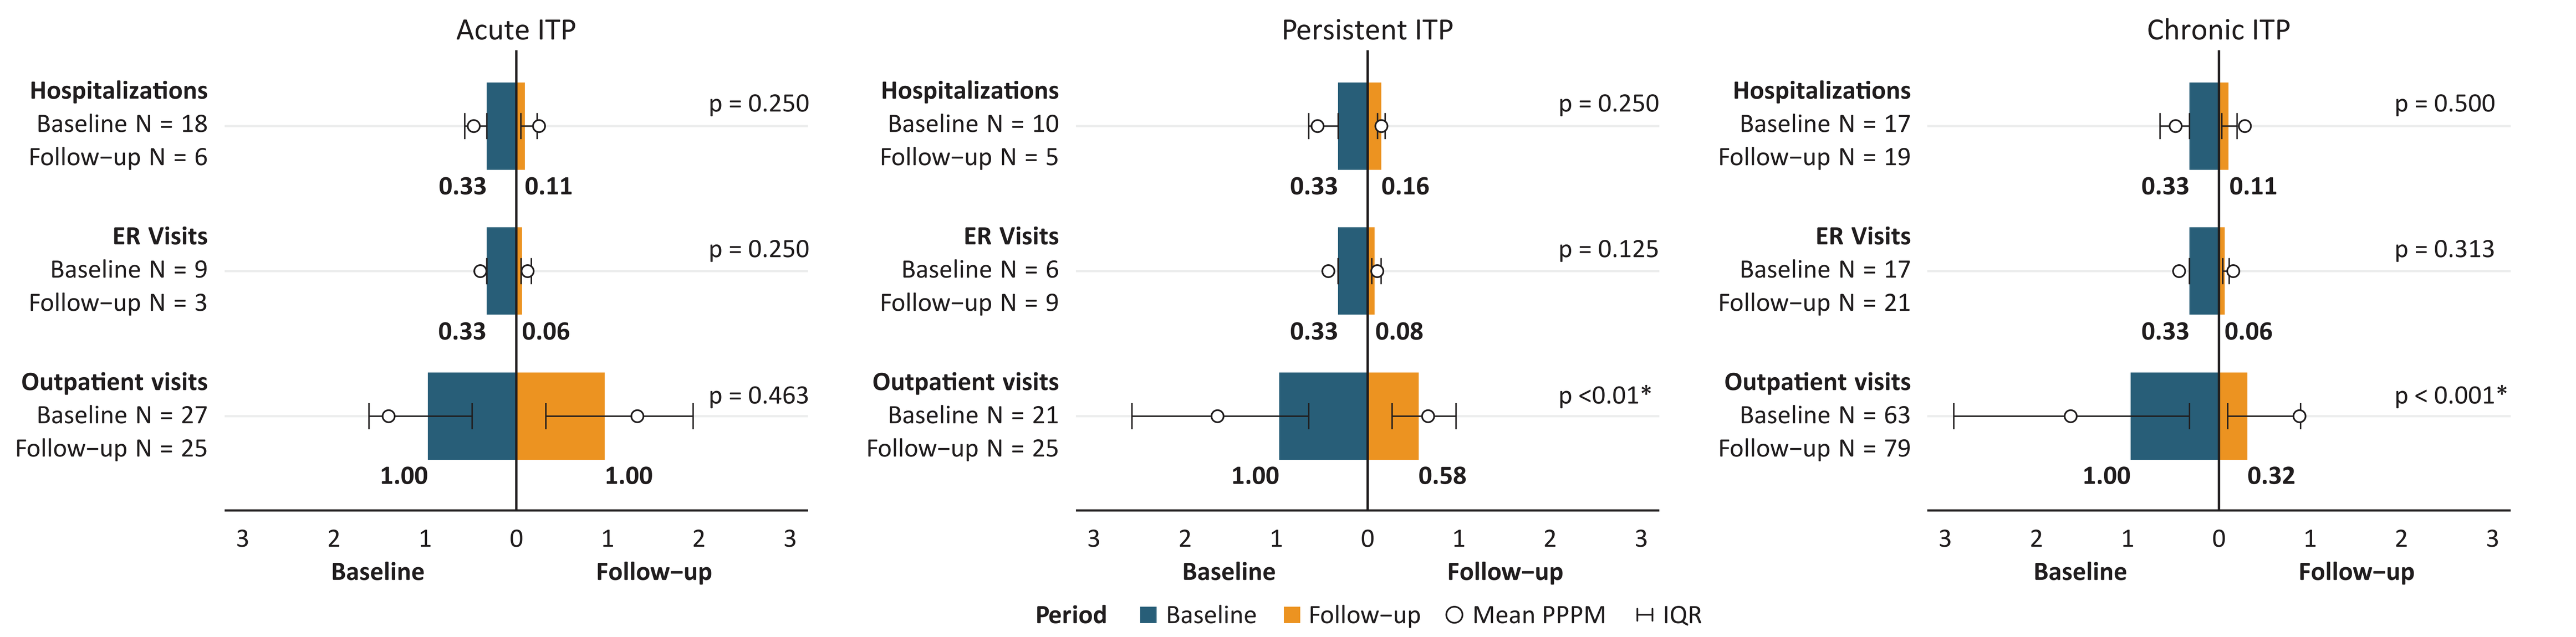
Acute ITP
Hospitalizations
Baseline N = 18
Follow−up N = 6
0.33
0.11
p = 0.250
ER Visits
Baseline N = 9
Follow−up N = 3
0.33
0.06
p = 0.250
Outpatient visits
Baseline N = 27
Follow−up N = 25
1.00
1.00
p = 0.463
Persistent ITP
Hospitalizations
Baseline N = 10
Follow−up N = 5
0.33
0.16
p = 0.250
ER Visits
Baseline N = 6
Follow−up N = 9
0.33
0.08
p = 0.125
Outpatient visits
Baseline N = 21
Follow−up N = 25
1.00
0.58
p <0.01*
Chronic ITP
Hospitalizations
Baseline N = 17
Follow−up N = 19
0.33
0.11
p = 0.500
ER Visits
Baseline N = 17
Follow−up N = 21
0.33
0.06
p = 0.313
Outpatient visits
Baseline N = 63
Follow−up N = 79
1.00
0.32
p < 0.001*
3 2 1 0 1 2 3
Baseline
Follow−up
Period Baseline Follow−up Mean PPPM IQR

Notes: Median PPPM rates are bolded. * denotes statistical significance at α=0.05.

Stratification by TPO-RA exposure status

- Both TPO-RA–experienced and TPO-RA–naïve patients experienced numerical declines in hospitalization rates (**Figure 4**), with median PPPM rates decreasing from 0.3 to 0.1 in both cohorts (both p=0.13).
- TPO-RA–experienced patients had a significant decrease in the rate of ITP-related ER visits from baseline to follow-up, with a decrease in the median PPPM rate from 0.3 to 0.1 (p<0.05). TPO-RA–naïve patients had a similar, though non-significant, drop in the ER visit rate (p=0.13).
- Both subgroups experienced significant reductions in PPPM OP visit rates among patients with ≥1 OP visit. Median PPPM rates fell from 1.2 to 0.4 visits/month in TPO-RA–experienced patients (p<0.01) and from 0.7 to 0.4 in naïve patients (p<0.05).

Figure 4. PPPM rates of ITP-related HRU among patients with ≥1 visit, by prior TPO-RA exposure

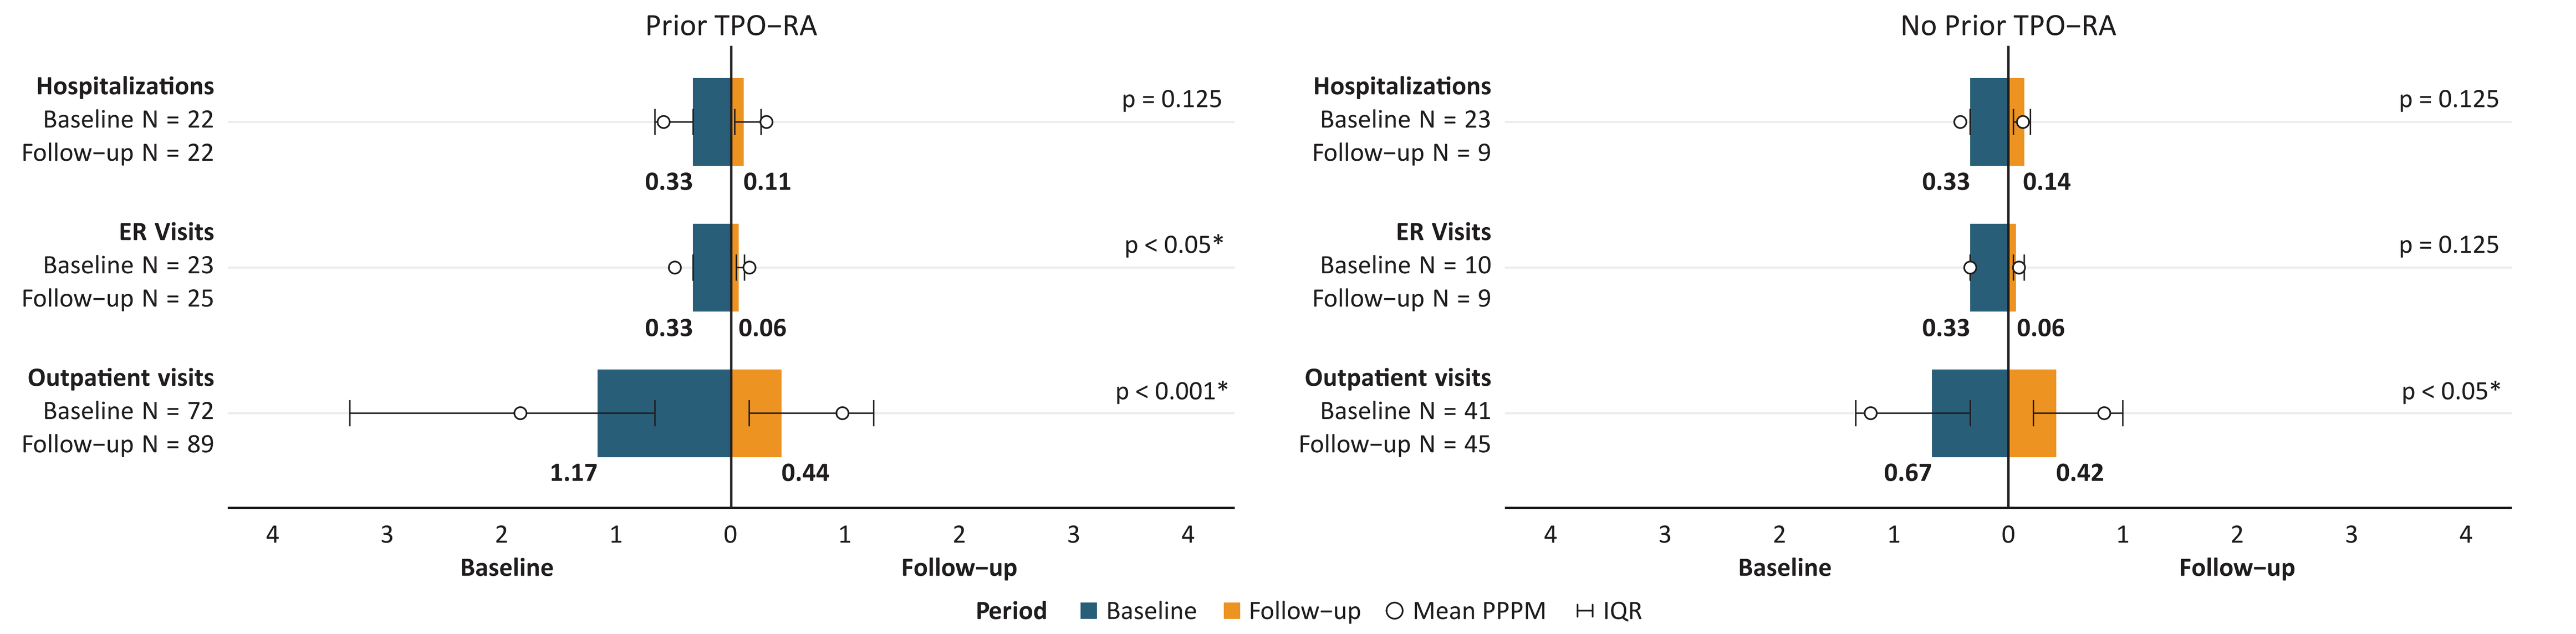
Prior TPO−RA
Hospitalizations
Baseline N = 22
Follow−up N = 22
0.33
0.11
p = 0.125
ER Visits
Baseline N = 23
Follow−up N = 25
0.33
0.06
p < 0.05*
Outpatient visits
Baseline N = 72
Follow−up N = 89
1.17
0.44
p < 0.001*
No Prior TPO−RA
Hospitalizations
Baseline N = 23
Follow−up N = 9
0.33
0.14
p = 0.125
ER Visits
Baseline N = 10
Follow−up N = 9
0.33
0.06
p = 0.125
Outpatient visits
Baseline N = 41
Follow−up N = 45
0.67
0.42
p < 0.05*
4 3 2 1 0 1 2 3 4
Baseline
Follow−up
Period Baseline Follow−up Mean PPPM IQR

Notes: Median PPPM rates are bolded. * denotes statistical significance at α=0.05.

LIMITATIONS

- Care received outside participating centers may not have been visible, leading to potential underreporting of HRU if such visits were not reported to the treating clinician.
- Healthcare cost data were unavailable; additional research is needed to determine whether reductions in HRU translate into cost savings.
- As with any chart review study, abstractor error or misinterpretation (e.g., defining "ITP-related HRU") was possible. To reduce this risk, all centers received standardized CRF training and rigorous data quality checks were performed.

REFERENCES

1. Singh A, et al. Primary immune thrombocytopenia: Novel insights into pathophysiology and disease management. *J Clin Med.* Feb 16 2021;10(4) doi:10.3390/jcm10040789
2. Danese MD, et al. Cost and mortality associated with hospitalizations in patients with immune thrombocytopenic purpura. *Am J Hematol.* Oct 2009;84(10):631-5. doi:10.1002/ajh.21500
3. Al-Samkari H, Nagalla S. Efficacy and safety evaluation of avatrombopag in immune thrombocytopenia: analyses of a phase III study and long-term extension. *Platelets.* Feb 17 2022;33(2):257-264. doi:10.1080/09537104.2021.1881952
4. Jain S, Gernsheimer T, et al. Additional efficacy analysis of avatrombopag phase III data for the treatment of adults with immune thrombocytopenia. *Platelets.* Dec 2023;34(1):2195016. doi:10.1080/09537104.2023.2195016
5. Jurczak W, Chojnowski K, Mayer J, et al. Phase 3 randomised study of avatrombopag, a novel thrombopoietin receptor agonist for the treatment of chronic immune thrombocytopenia. *Br J Haematol.* Nov 2018;183(3):479-490. doi:10.1111/bjh.15573

FUNDING

The REAL AVA 2.0 study was supported by Sobi, Inc.

ABBREVIATIONS

AVA, avatrombopag; CI, confidence interval; ER, emergency room; HRU, healthcare resource utilization; IQR, interquartile range; ITP, immune thrombocytopenia; IVIG, Intravenous immunoglobulin; OP, outpatient; PPPM, per patient per month; SD, standard deviation; TPO-RA, thrombopoietin receptor agonists.

ACKNOWLEDGEMENTS

Participating centers included the City of Hope, Clearview Cancer Institute, Dr. Ilya Blokh, Illinois Bleeding and Clotting Disorders Institute, Johns Hopkins Medicine, Rush University, SLUCare Physician Group, Tennessee Oncology, University of Pennsylvania, University of Utah, and University of Washington.

DISCLOSURES

Authors SN, ML, SC, ES, DG, AG, LO, and SS are consultants of Sobi, Inc.
Authors AO, CB, and MV are employees of Sobi, Inc.; author SK was an employee of Sobi, Inc. at the time of the study.

Copies of the presentation obtained through this QR Code are for personal use only.

Presented at the Academy of Managed Care Pharmacy Nexus 2025 (AMCP Nexus) • October 27-30, 2025 • National Harbor, MD, USA